INTRAPUTAMINAL *AADC* GENE THERAPY (VY-AADC01) FOR ADVANCED PARKINSON'S DISEASE: INTERIM RESULTS OF A PHASE 1B TRIAL

Chadwick Christine MD[1], Krystof Bankiewicz MD PhD[1], Amber Van Laar MD[2], R Mark Richardson MD PhD[2], Bernard Ravina MD[3], Adrian Kells PhD[3], Brendon Boot MBBS[3], Alastair Martin PhD[1], Marin Thompson MS[1], John Nutt MD[4], and Paul Larson MD[1]

[1]Department of Neurology (CC) and Interventional Neuro Center (KB, AM, MT, PL), University of California – San Francisco, CA; [2]Departments of Neurology (AL) and Neurosurgery (RR), University of Pittsburg Medical Center, Pittsburgh, PA; [3]Voyager Therapeutics, Cambridge, MA; [4]Department of Neurology, Oregon Health & Science University, Portland OR.

INTRODUCTION

Levodopa therapy for Parkinson's Disease (PD) becomes less effective over time, possibly due to the loss of the enzyme that converts levodopa into dopamine: aromatic L-amino acid decarboxylase (AADC; Figure 1A). Nigrostriatal axons are the major source of AADC in the human brain, so as PD progresses and these cells die, AADC is lost. This prevents consistent, reliable synthesis and release of dopamine in response to exogenous levodopa, resulting in motor fluctuations.

VY-AADC01 is an AAV gene therapy that replaces AADC. Proof-of-concept for this approach in PD was shown in MPTP-lesioned Non-Human Primates (NHP) that demonstrated impaired conversion of levodopa to dopamine (Figure 1B). Adeno-associated-virus serotype 2 (AAV2) encoding AADC injected into the putamen of these animals with 30% or more coverage, reverses this impairment and durably restores the behavioral response to levodopa[1].

In this ongoing, open label, phase 1b dose escalation trial, we injected AAV2 encoding AADC (VY-AADC01) into the putamen to create an alternative source of AADC in medium spiny neurons that do not degenerate as PD progresses.

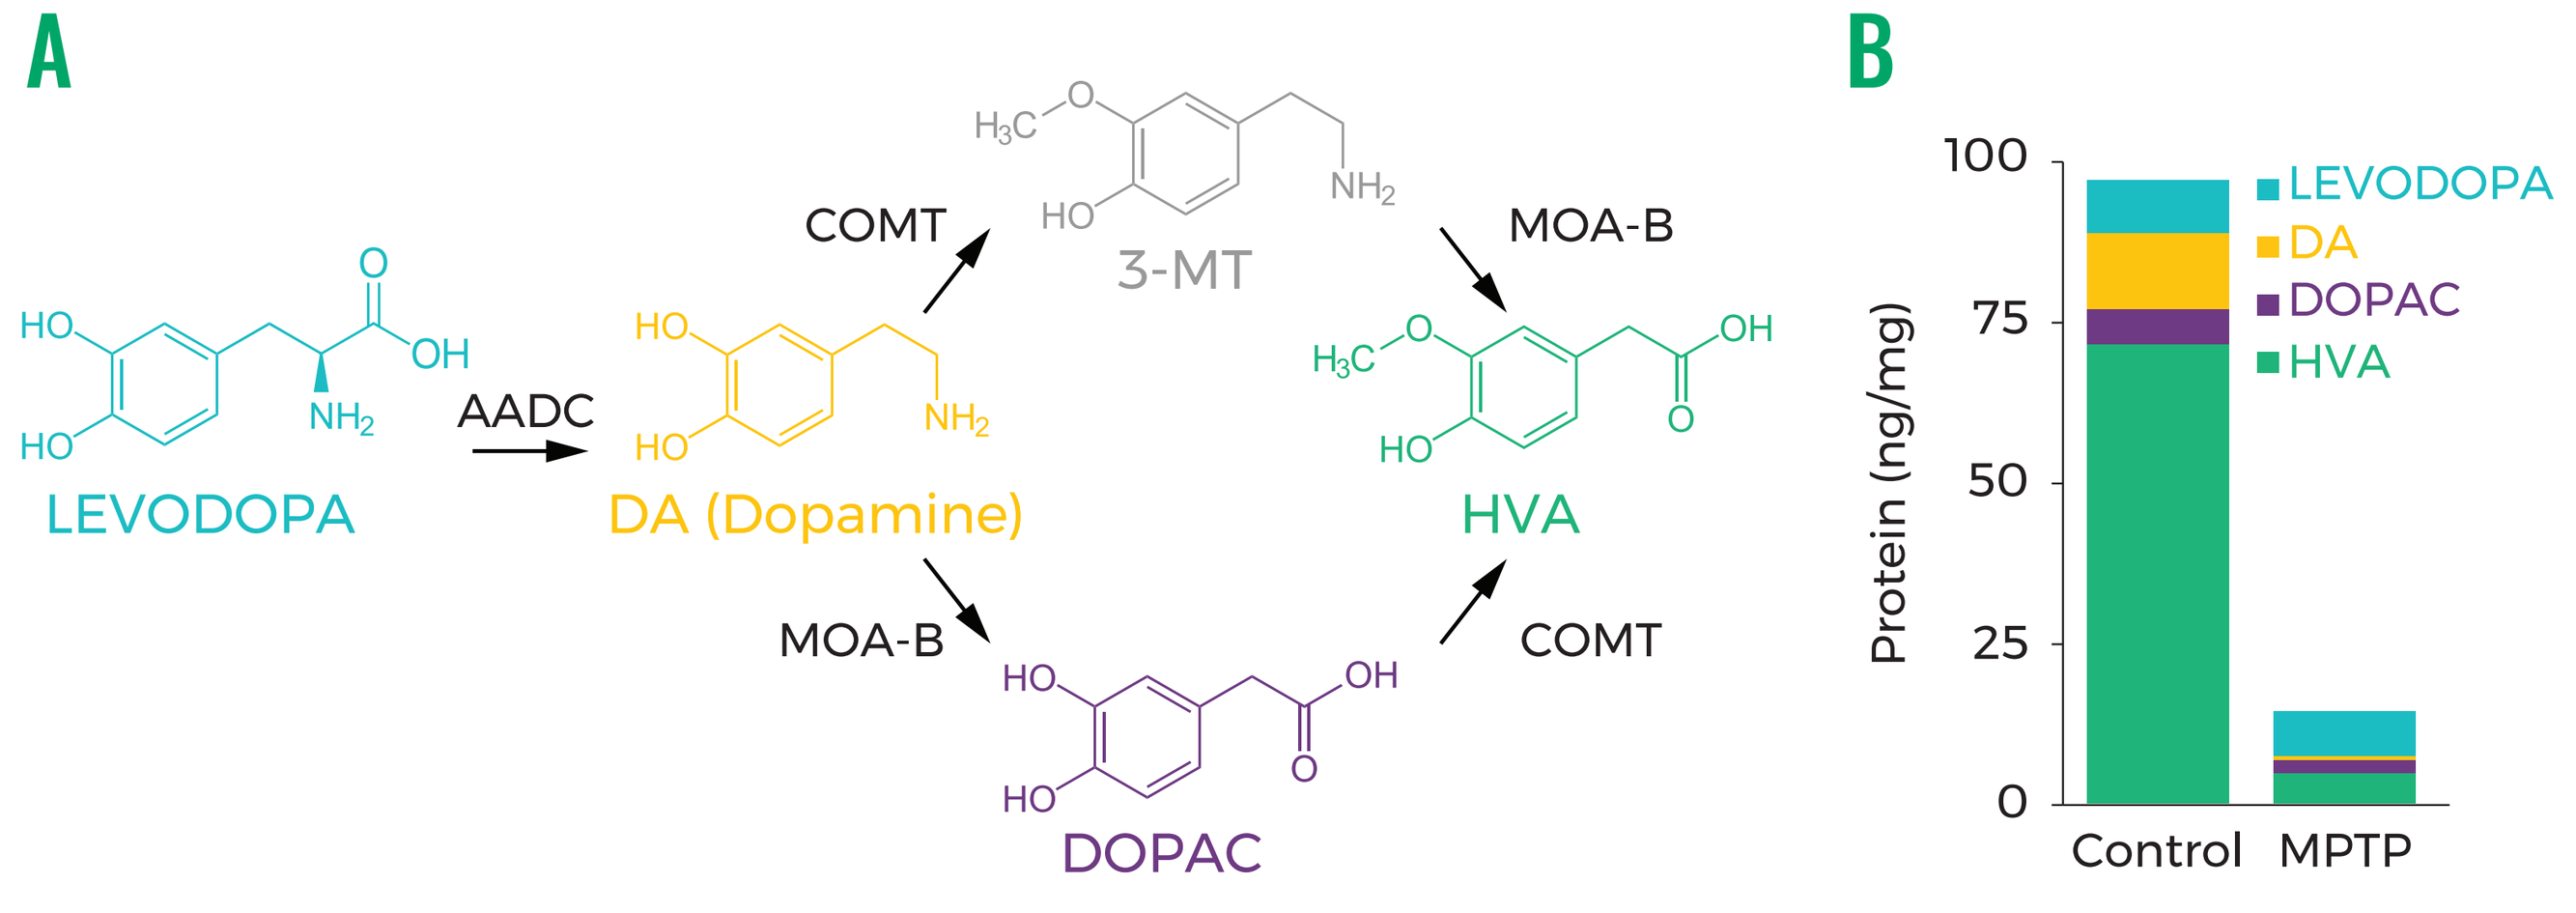

A
B
COMT
MOA-B
3-MT
AADC
LEVODOPA
DA (Dopamine)
HVA
MOA-B
COMT
DOPAC
Protein (ng/mg)
LEVODOPA
DA
DOPAC
HVA
Control
MPTP

Figure 1: Metabolism of levodopa. (**A**) Metabolic pathway. (**B**) Biochemical analyses of the putamen tissue 45 minutes after oral levodopa in control vs. mildly MPTP-lesioned NHPs. Lesioning induces a reduction in the conversion of levodopa to dopamine and metabolites in the putamen (PUT)[2].

AIMS

- Determine the safety of MRI-guided infusion of up to 900µL/ putamen of VY-AADC01.
- Determine the volume of the putamen surgically covered with VY-AADC01, using real time MRI-guided convection enhanced delivery.
- Assess changes in expression of AADC by ^{18}F-DOPA PET, response to intravenous (IV) levodopa and requirements for oral dopaminergic therapy, and motor function as assessed by standard rating scales and motor diaries.

METHODS

Fifteen subjects with disabling motor fluctuations despite optimal medical management were recruited at the University of California- San Francisco and the University of Pittsburgh. VY-AADC01 vector was co-infused with gadoteridol to visualize distribution using bilateral skull-mounted MRI compatible devices. The surgeries were conducted in a sterile field in a 1.5T diagnostic MRI scanner.

Dose is outlined in Table 1. There were two to three trajectories per putamen. Since gadoteridol co-distributes with VY-AADC01 when co-injected, we used real-time imaging to maximize coverage. T1 weighted images obtained during of the infusions were used to calculate volume of coverage of the putamen.

Table 1: VY-AADC01 dosing

Cohort	N	Concentration (vg/mL)	Total Volume (µL per putamen)
1	5	0.83×10^{12}	450
2	5	0.83×10^{12}	up to 900
3	5	2.6×10^{12}	up to 900

IV Levodopa Infusion Test

Before, and 6 months after VY-AADC01 treatment, an independent center (Oregon Health and Science University) assessed the response to IV levodopa[3]. After overnight withdrawal from their PD medications, subjects received low (0.6mg/kg/h) and high (1.2mg/kg/h) dose levodopa infusions for two hours on separate days. The order of the infusions was blinded and randomized. An independent rater performed UPDRS assessments every 30 mins.

Follow-Up

Safety and clinical outcomes occur at 3, 6, 12, 18, 24, 30 and 36 months. All 5 subjects in Cohort 1, and 3 of the 5 subjects in Cohort 2, have 12-month follow-up data, reported below. Two of the 5 subjects in Cohort 2 have not yet reached the 12-month follow-up period at the time of this presentation. Cohort 3 subjects completed treatment, clinical data are not yet available.

Table 2: Baseline characteristics

	Cohort 1	Cohort2
Age	57.4 (7.2)	58.4 (8.6)
Sex	1 Female, 4 Male	5 Male
PD Duration (years)	9.9 (4.6)	10.1 (1.6)
UPDRS II off	13.6 (2.1)	16.0 (1.7)
UPDRS II on	3.0 (2.9)	3.6 (1.7)
UPDRS III off	37.2 (5.9)	35.8 (7.6)
UPDRS III on	7.6 (5.1)	17.0 (3.8)
UPDRS IV	7.8 (2.1)	7.6 (1.8)
PDQ39	18.2 (10.2)	16.6 (12.7)
Avg. Diary off-time (hrs)[a]	4.9 (1.7)	4.2 (1.4)
Avg. Diary on-time (hrs)[a]	10.5 (2.1)	10.7 (1.8)
Hoehn and Yahr Stage	3.0	3.0
LED[b] mg	1467.5 (615.0)	1635.5 (687.3)

Data are mean (SD). (a) Diary times normalized to 16-hour waking day. (b) Levodopa equivalent dose.

RESULTS

Safety

The infusions were well-tolerated with no vector-related SAEs. Fourteen of 15 subjects were discharged to home within 2 days. One subject experienced a thromboembolic event and subsequent atrial fibrillation due to immobility during the surgical procedure. All symptoms resolved in this subject. Subsequent cases had MRI compatible DVT prophylaxis.

Increasing Coverage of the Putamen

Surgical coverage of the putamen was volume dependent and increased with each cohort.

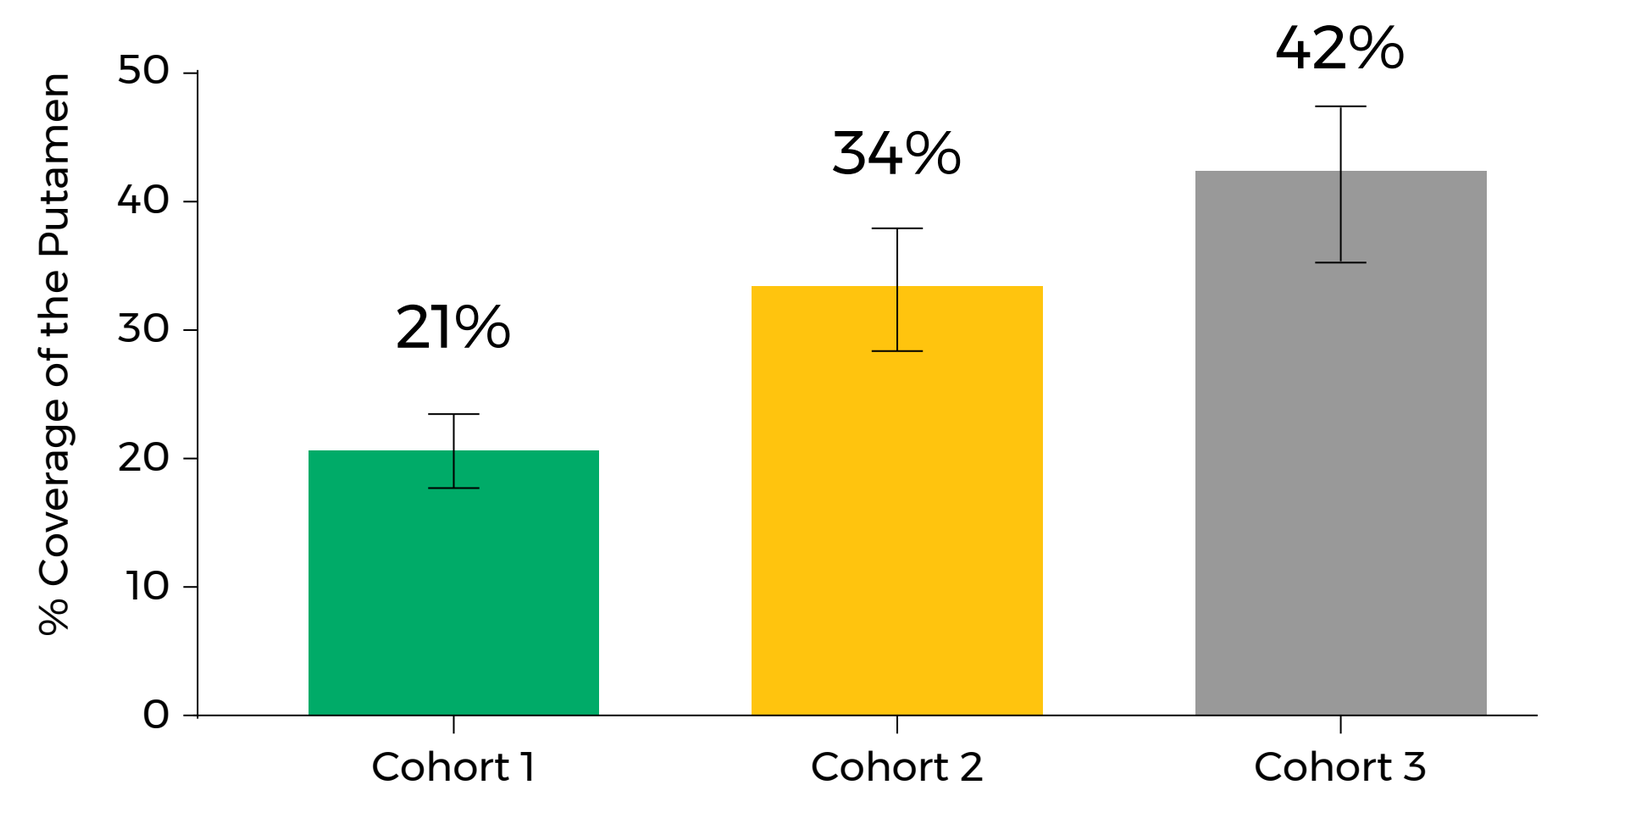

% Coverage of the Putamen
21%
34%
42%
Cohort 1
Cohort 2
Cohort 3

Figure 2: Percent coverage of the putamen (mean +/- SD).

Cohort-Related Increase in ^{18}F-DOPA PET Imaging

Cohort-related increases in ^{18}F-DOPA PET in the putamen were highly correlated with surgical coverage (r=0.81, p<0.05).

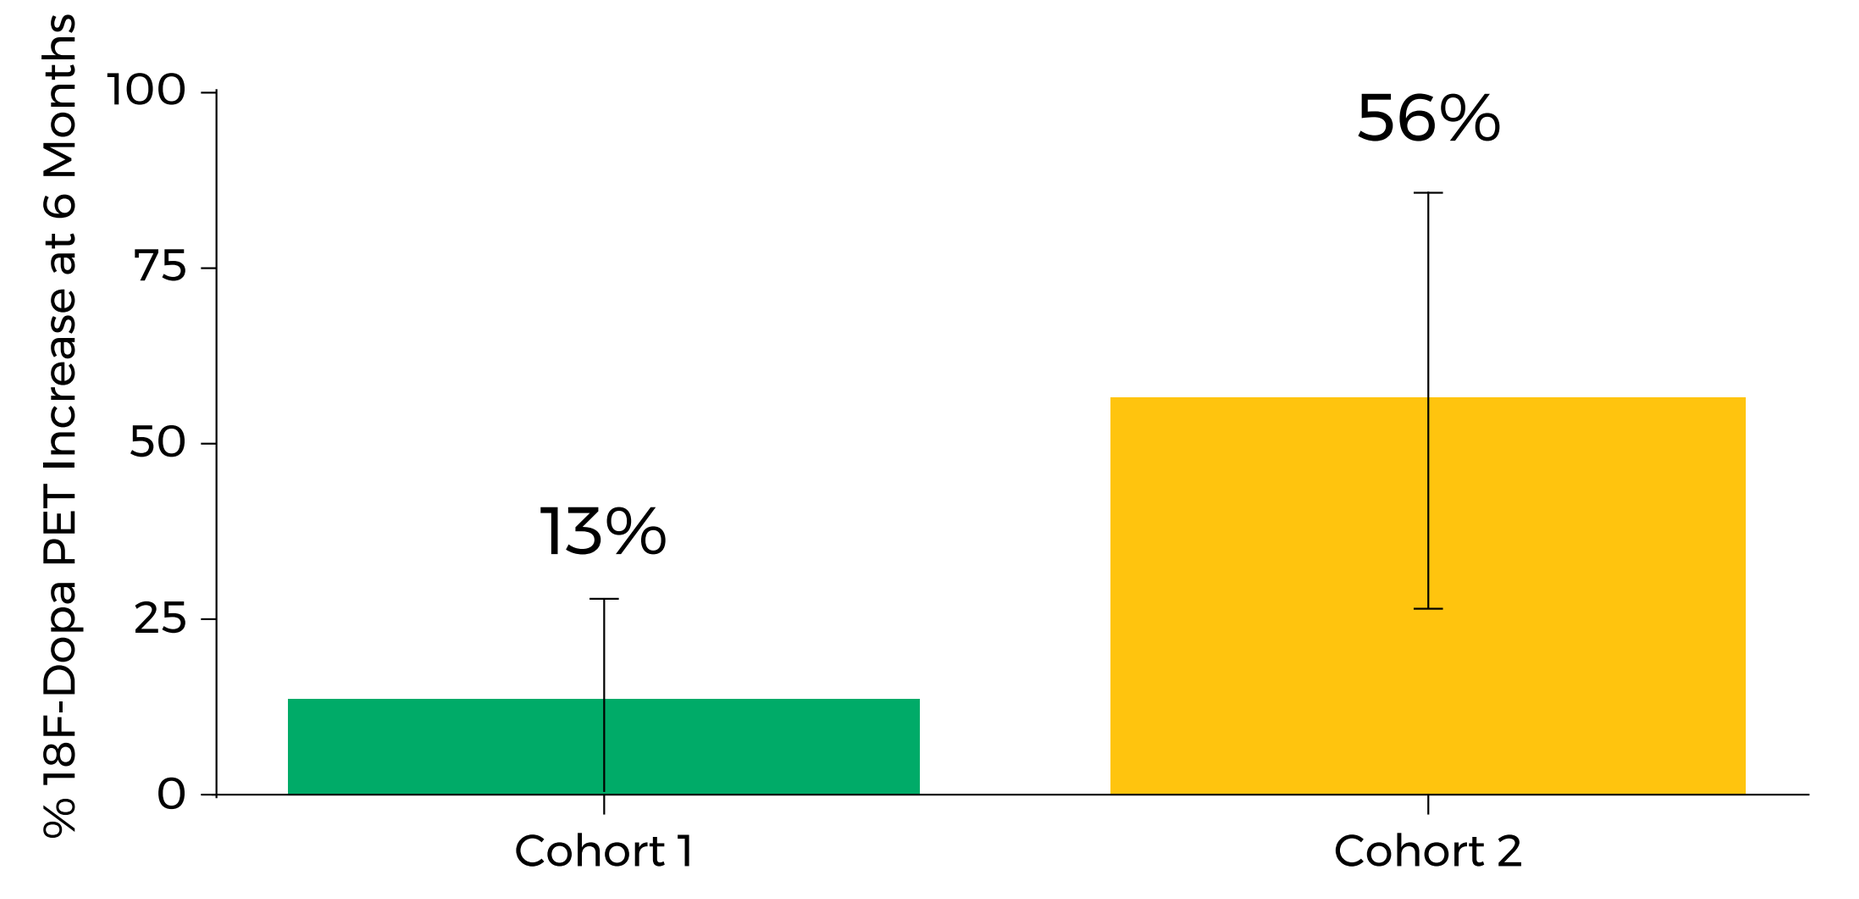

% 18F-Dopa PET Increase at 6 Months
13%
56%
Cohort 1
Cohort 2

Figure 3: Percent change from baseline to six months in ^{18}F-Dopa PET signal within the putamen (mean +/- SD).

Cohort- and Time-Dependent Reduction in Dopaminergic Medication

Consistent with the mechanism of increasing dopamine production, transient increases in dyskinesia generally occurred one month after surgery and resolved with reductions in Parkinson's medication of 35% in Cohort 2 at 12 months.

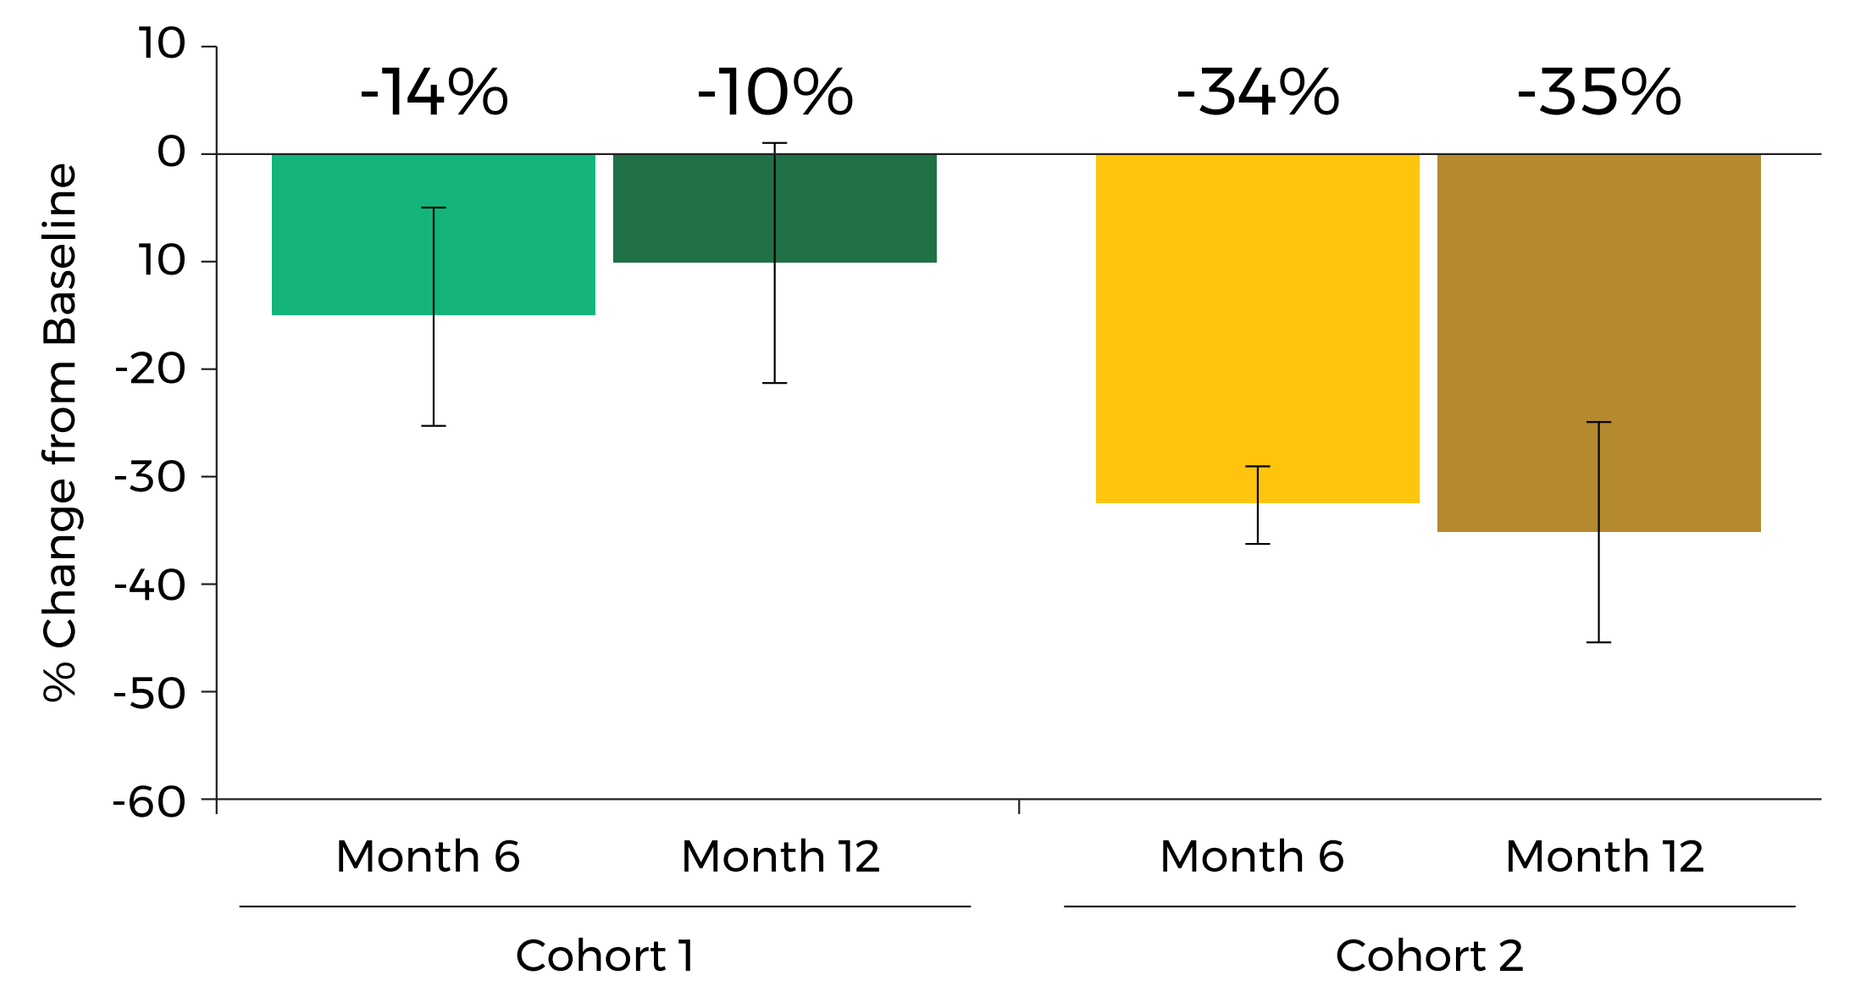

% Change from Baseline
-14%
-10%
-34%
-35%
Month 6
Month 12
Month 6
Month 12
Cohort 1
Cohort 2

Figure 4: Percent change from baseline (BL; mean +/- SE) of levodopa equivalent dose. 6-month data from 5/5 pts (Cohort 2). 12-month data from 5/5 pts (Cohort 1) and 3/5 pts with 12 months of follow-up. 2/5 pts in Cohort 2 had not reached 12-month follow-up.

Enhanced Response to IV Levodopa

Participants showed an increased peak and duration of motor response to a low dose of IV levodopa and an increased duration of response to high dose IV levodopa, as measured by UPDRS III, 6 months after treatment, compared to baseline.

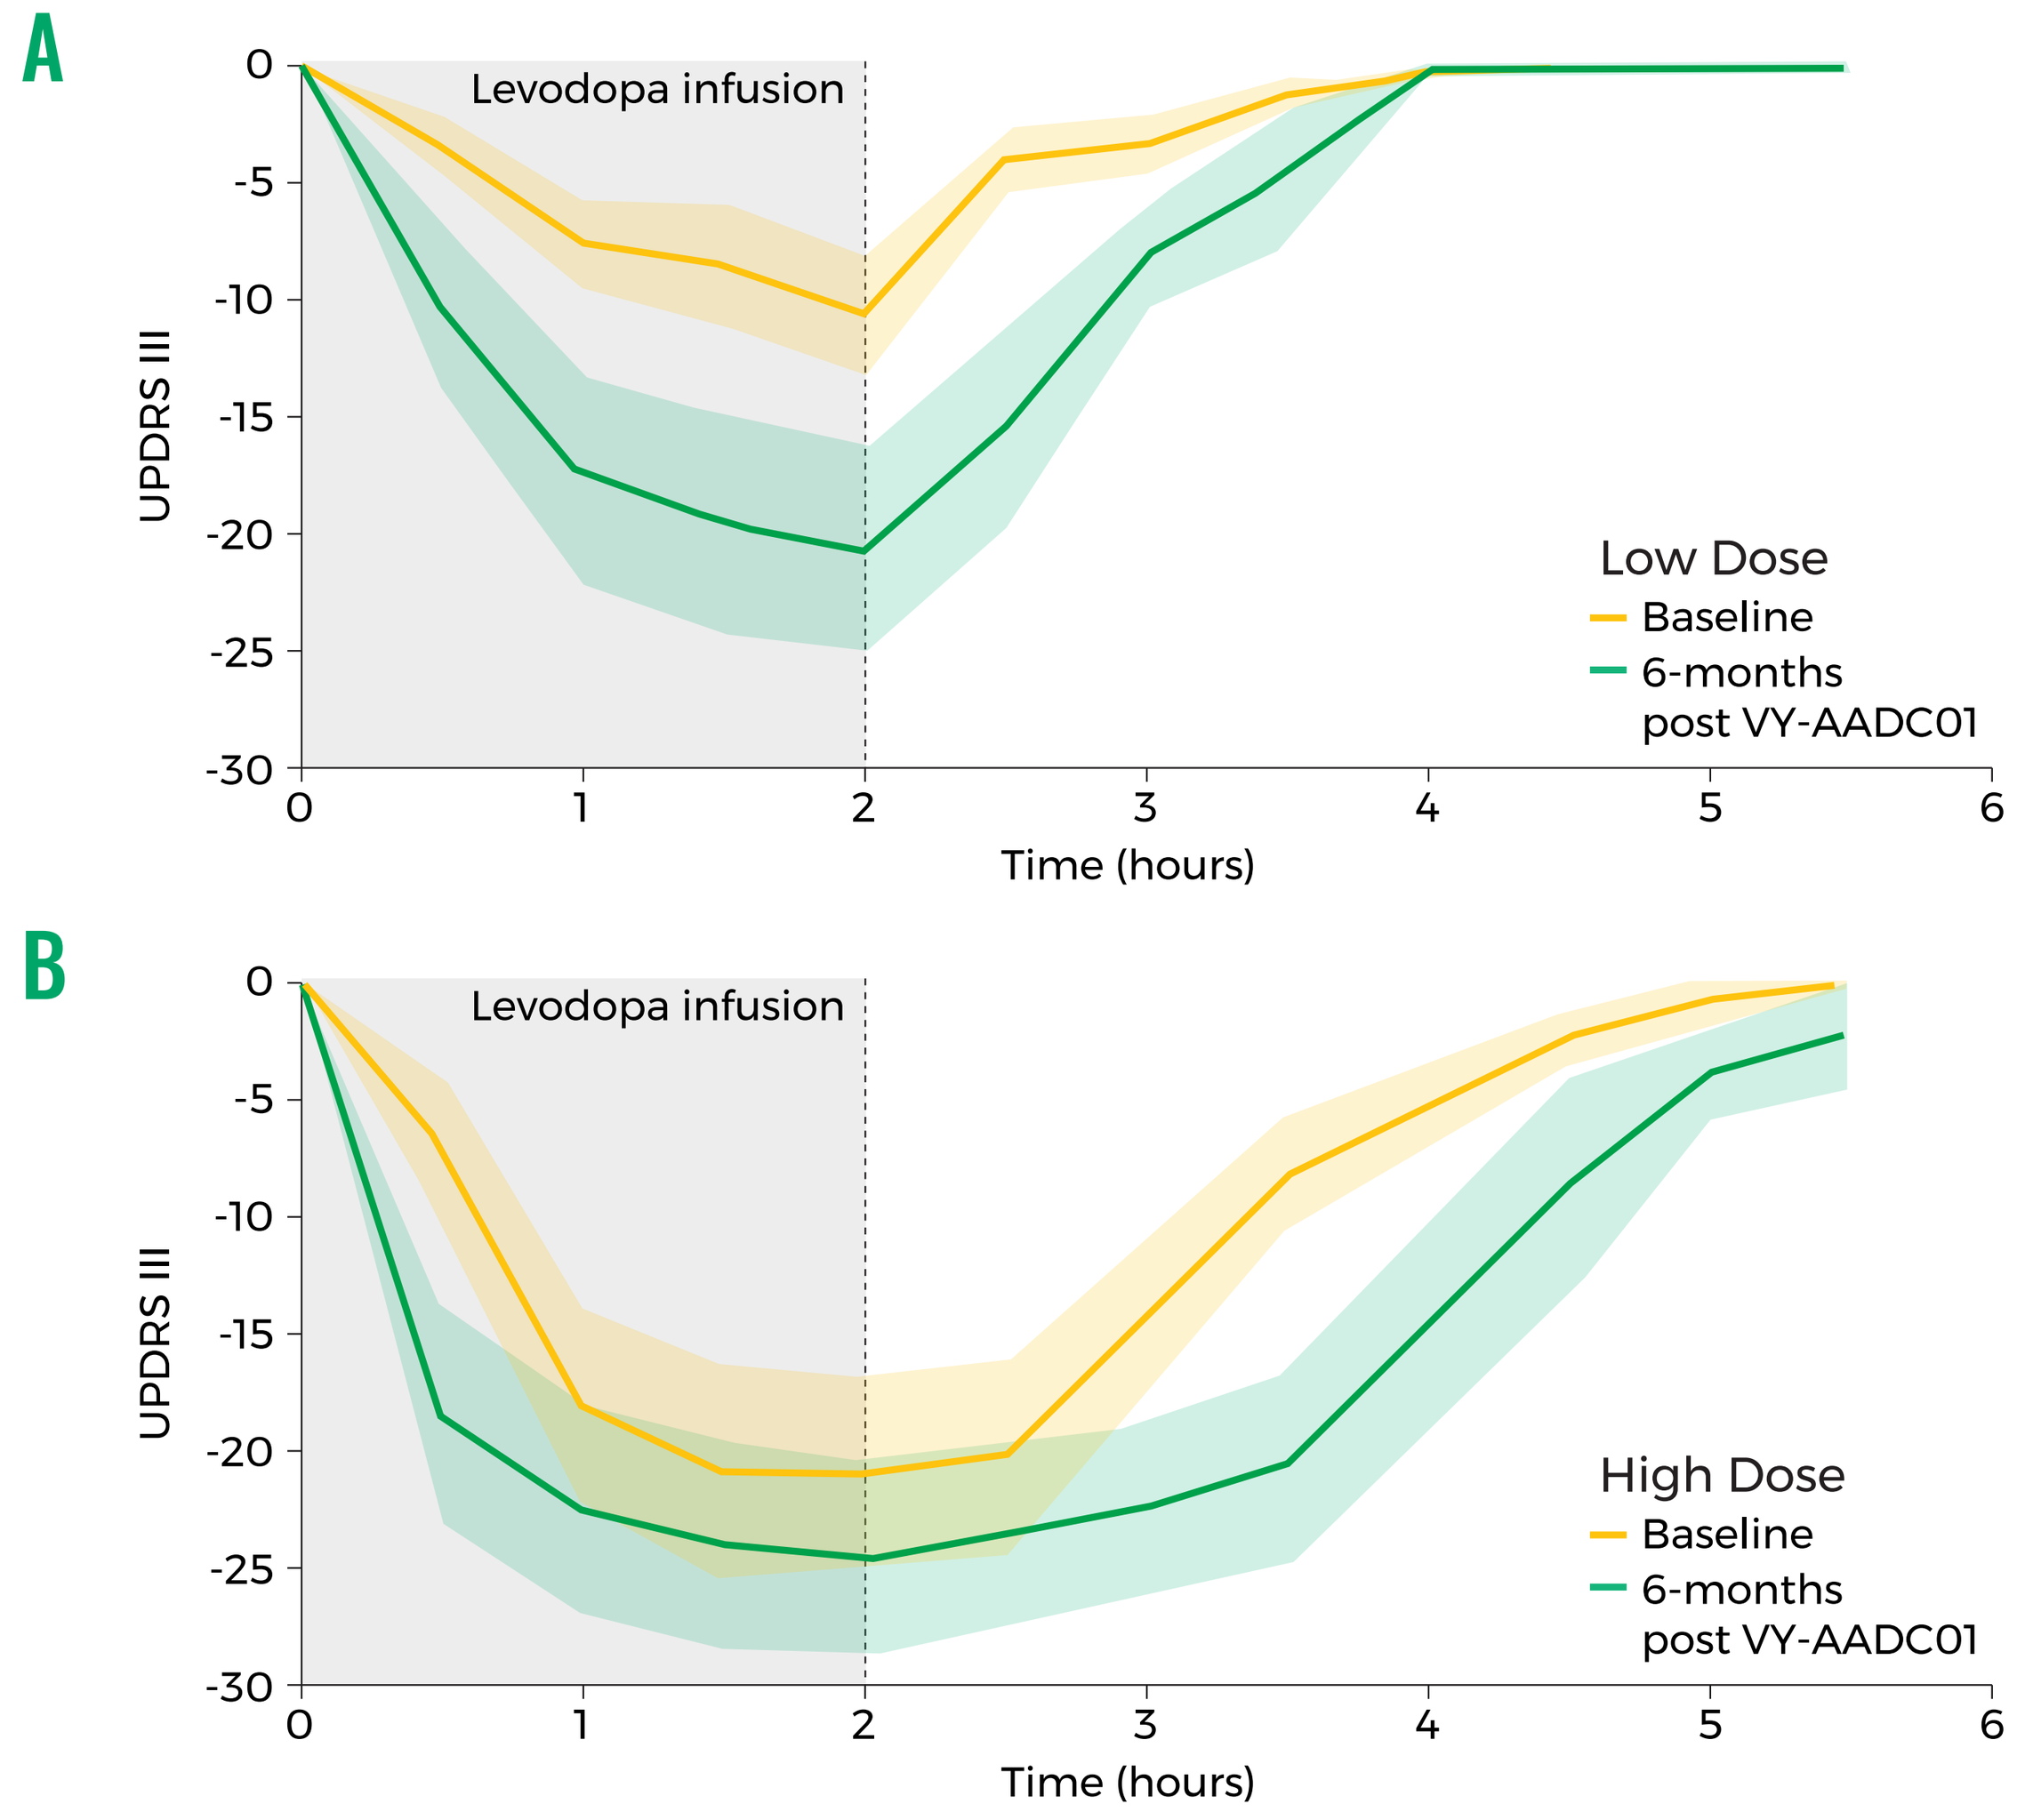

A
Levodopa infusion
UPDRS III
Time (hours)
Low Dose
Baseline
6-months post VY-AADC01
B
Levodopa infusion
UPDRS III
Time (hours)
High Dose
Baseline
6-months post VY-AADC01

Figure 5: Mean change in UPDRS III during and after two-hour infusions of low (**A**) and high (**B**) doses of IV levodopa. Baseline data in yellow, 6-months post VY-AADC01 data in green. Shading represents SE.

Cohort- and Time-Dependent Improvements in Diary Measures

Cohort-and time-dependent increases in diary on-time and decreases in diary off-time were observed. A 2.2 hour (20%) increase in diary on-time (with no dyskinesias or non-troublesome dyskinesias) in Cohort 2 at six months further increased to 4.1 hours (43%) at 12 months. Cohort 1 showed a slight decrease in on-time at six months of 0.3 hours (-3%) and an increase of 1.6 hours (16%) at 12 months (Figure 6A). A 1.1 hour (27%) decrease in diary off-time in Cohort 2 at six months further decreased to 2.2 hours (48%) at 12 months. Decreases in diary off-time in Cohort 2 also occurred in conjunction with a reduction in troublesome dyskinesias. Cohort 1 showed a decrease in diary off-time of 0.8 hours (16%) at six months and 1.4 hours (27%) at 12 months (Figure 6B).

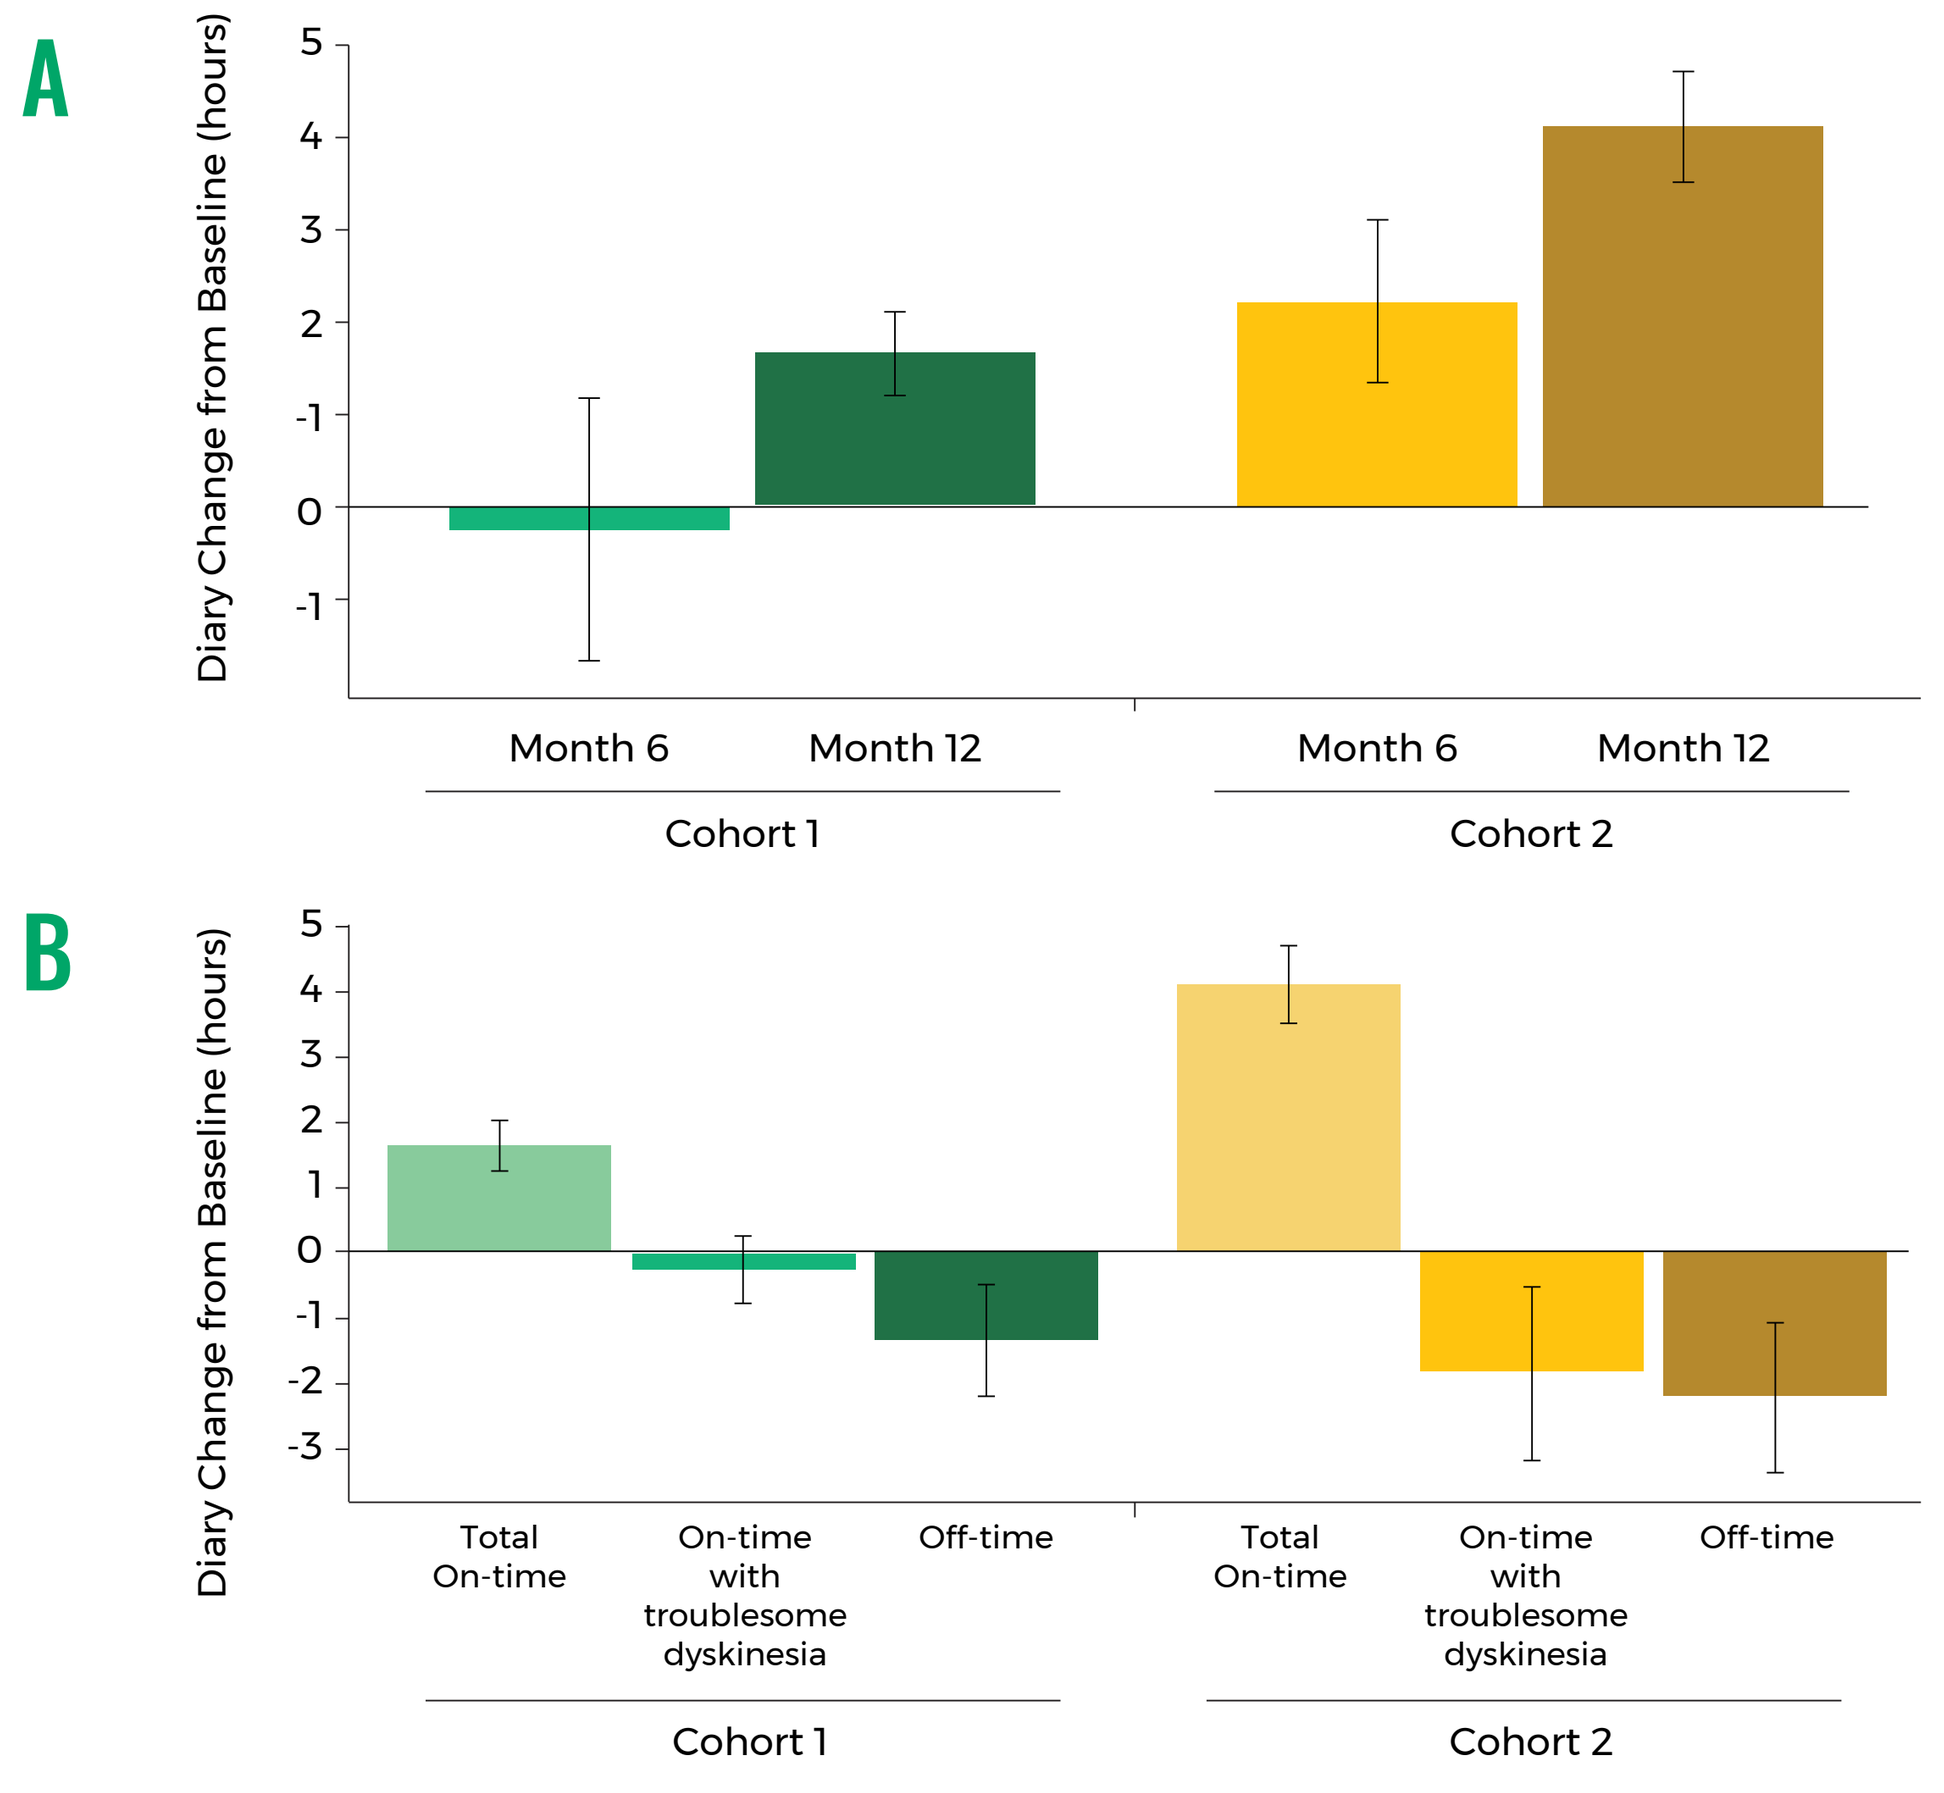

A
Diary Change from Baseline (hours)
Month 6
Month 12
Month 6
Month 12
Cohort 1
Cohort 2
B
Diary Change from Baseline (hours)
Total On-time
On-time with troublesome dyskinesia
Off-time
Total On-time
On-time with troublesome dyskinesia
Off-time
Cohort 1
Cohort 2

Figure 6: Change in patient diary On-time at 6 and 12 months (**A**) and Summary Diary Measures at 12 month (**B**). Change from baseline in Hauser PD Diary Data (mean +/- SE). 6-month data from 5/5 pts in each of Cohorts 1 & 2. 12-month data from 5/5 pts (Cohort 1) and 3/5 pts with 12 months of follow-up. 2/5 pts in Cohort 2 had not reached 12-month follow-up. Diary times normalized to 16- hour waking day.

Cohort-Dependent Improvements in Motor Function, Activities of Daily Living, and Quality of Life

Table 3: Motor function, activities of daily living and quality of life as measured by the UPDRS and PDQ-39 at 12 months.

	UPDRS II Off Medication	UPDRS III On Medication	UPDRS IV Total Score	PDQ-39 Total Score
Cohort 1	0.2 point worsening (SD 3.7)	1.8 point worsening (SD 6.1)	1.2 point improvement (SD 1.9)	1.9 point improvement (SD 5.6)
Cohort 2	4.0 point improvement (SD 1.0)	9.3 point improvement (SD 3.1)	2.7 point improvement (SD 3.1)	9.2 point improvement (SD 5.5)

Data from 5/5 pts (Cohort 1) and 3/5 pts with 12 months of follow-up (Cohort 2). 2/5 pts in Cohort 2 had not reached 12-month follow-up.

CONCLUSIONS

- Administration of up to 900µL/putamen of VY-AADC01 was well-tolerated.
- Coverage of the putamen is dependent on the volume of infusion and ability to monitor the infusions with MRI. Previous gene therapy trials used much lower volumes of infusion and lacked MRI guidance.
- Surgical coverage of the putamen with VY-AADC01 was significantly correlated with change in ^{18}F-DOPA PET signal.
- Larger clinical effects were seen in Cohort 2 which had higher coverage of the putamen than Cohort 1, although the two cohorts had nearly identical surgical procedures.
- We observed consistent, dose-dependent changes in imaging, responsiveness to levodopa, and clinical measures, which have not been seen in prior PD gene therapy trials.
- VY-AADC01 is now being tested via an occipital trajectory. This approach, along the long axis of the putamen, may allow for increased coverage of the putamen with each infusion.

75 Sidney St. Cambridge, MA 02139 USA · info@voyagertherapeutics.com · 857-259-5340

Contact:
Bernard Ravina (bravina@vygr.com)

Acknowledgments:
Funding provided by The Michael J. Fox Foundation for Parkinson's Research (Grant #7266) and Voyager Therapeutics. ClinicalTrials.gov Identifier: NCT01973543.

References:

1. Hadaczek P, et al. Eight years of clinical improvement in MPTP-lesioned primates after gene therapy with AAV2-hAAD. *Mol Ther.* 2010 Aug;18(8):1458-61.
2. Ciesielska A, et al. Depletion of AADC activity in caudate nucleus and putamen of Parkinson's disease patients; implications for ongoing AAV2-AADC gene therapy trial *PLoS One.* 2017 Feb 6;12(2):e0169965.
3. Brodsky, MA, et al. Effects of a dopamine agonist on the pharmacodynamics of Levodopa in Parkinson Disease. *Arch Neurol.* 2010 Jan;67(1):27-32.