P024

Identification of a Cell Surface Receptor Utilized by an Engineered BBB-Penetrant Capsid Family with Enhanced Brain Tropism in Non-Human Primates and Mice

B.A. Hoffman, T. Knox, T. Moyer, I. Shah, R. Kaur, M.A. Child, D.R. Laks, M. Nonnenmacher

Voyager Therapeutics Inc., 64 Sidney Street, Cambridge, MA 02139, USA

voyager THERAPEUTICS

INVESTORS& MEDIA

CAREERS AT VOYAGER

SUMMARY

- VCAP-101/102 engineered capsid class exhibits ~50-fold increased BBB penetrance in both macaques and mouse relative to AAV9
- We have identified a highly conserved membrane protein, designated Receptor X, as the primary attachment surface receptor for the VCAP-101/102 capsid family.
- Conserved expression of Receptor X on brain endothelial cells in mice and humans suggests a role in meditating the enhanced BBB penetrance exhibited by this capsid family.
- Receptor identification will provide confidence in the transferability of our novel capsids to humans and facilitate prediction of capsid behavior based on receptor expression patterns.

INTRODUCTION

The blood-brain barrier (BBB) represents a significant limitation to AAV-mediated gene therapies for CNS indications. Natural AAVs typically cross the BBB with low efficiency, requiring high systemic doses or invasive direct injection into the CNS for therapeutic efficacy. Thus, considerable efforts have been undertaken to engineer capsids with enhanced BBB-crossing properties. These efforts have recently yielded several AAV capsid variants with enhanced CNS tropism in non-human primates (NHP). The mechanisms utilized by these engineered capsids to cross the BBB are still unknown. We have previously reported the generation of an engineered capsid class exhibiting 50-fold increased BBB penetrance in both macaques and mouse. Here we report the identification of a surface receptor specifically bound by this engineered capsid class. Ectopic overexpression of the human isoform of this receptor in cultured cells led to a significant increase in capsid binding and transduction while no difference was observed with the parental capsid, AAV9. Direct capsid-receptor interaction was confirmed by additional biochemical analyses, which strongly supports a role as a primary attachment receptor. Immunostaining and single-cell RNA-seq experiments performed in mouse brain suggest that the receptor expression pattern and capsid tropism are correlated, further supporting a role in the transport of these cross-species capsids across the BBB. Our data provide mechanistic insights into the enhanced brain transduction by these novel BBB-penetrant capsids and raise exciting possibilities for the prediction of capsid behavior in humans based on receptor cross-species conservation and expression pattern.

Figure 1. TRACER Biopanning of an AAV9 Variable Loop 4 Peptide Insertion Library Identifies VCAP-101/102 Capsid Class With Enhanced BBB-Penetrance in Both NHP and Mouse

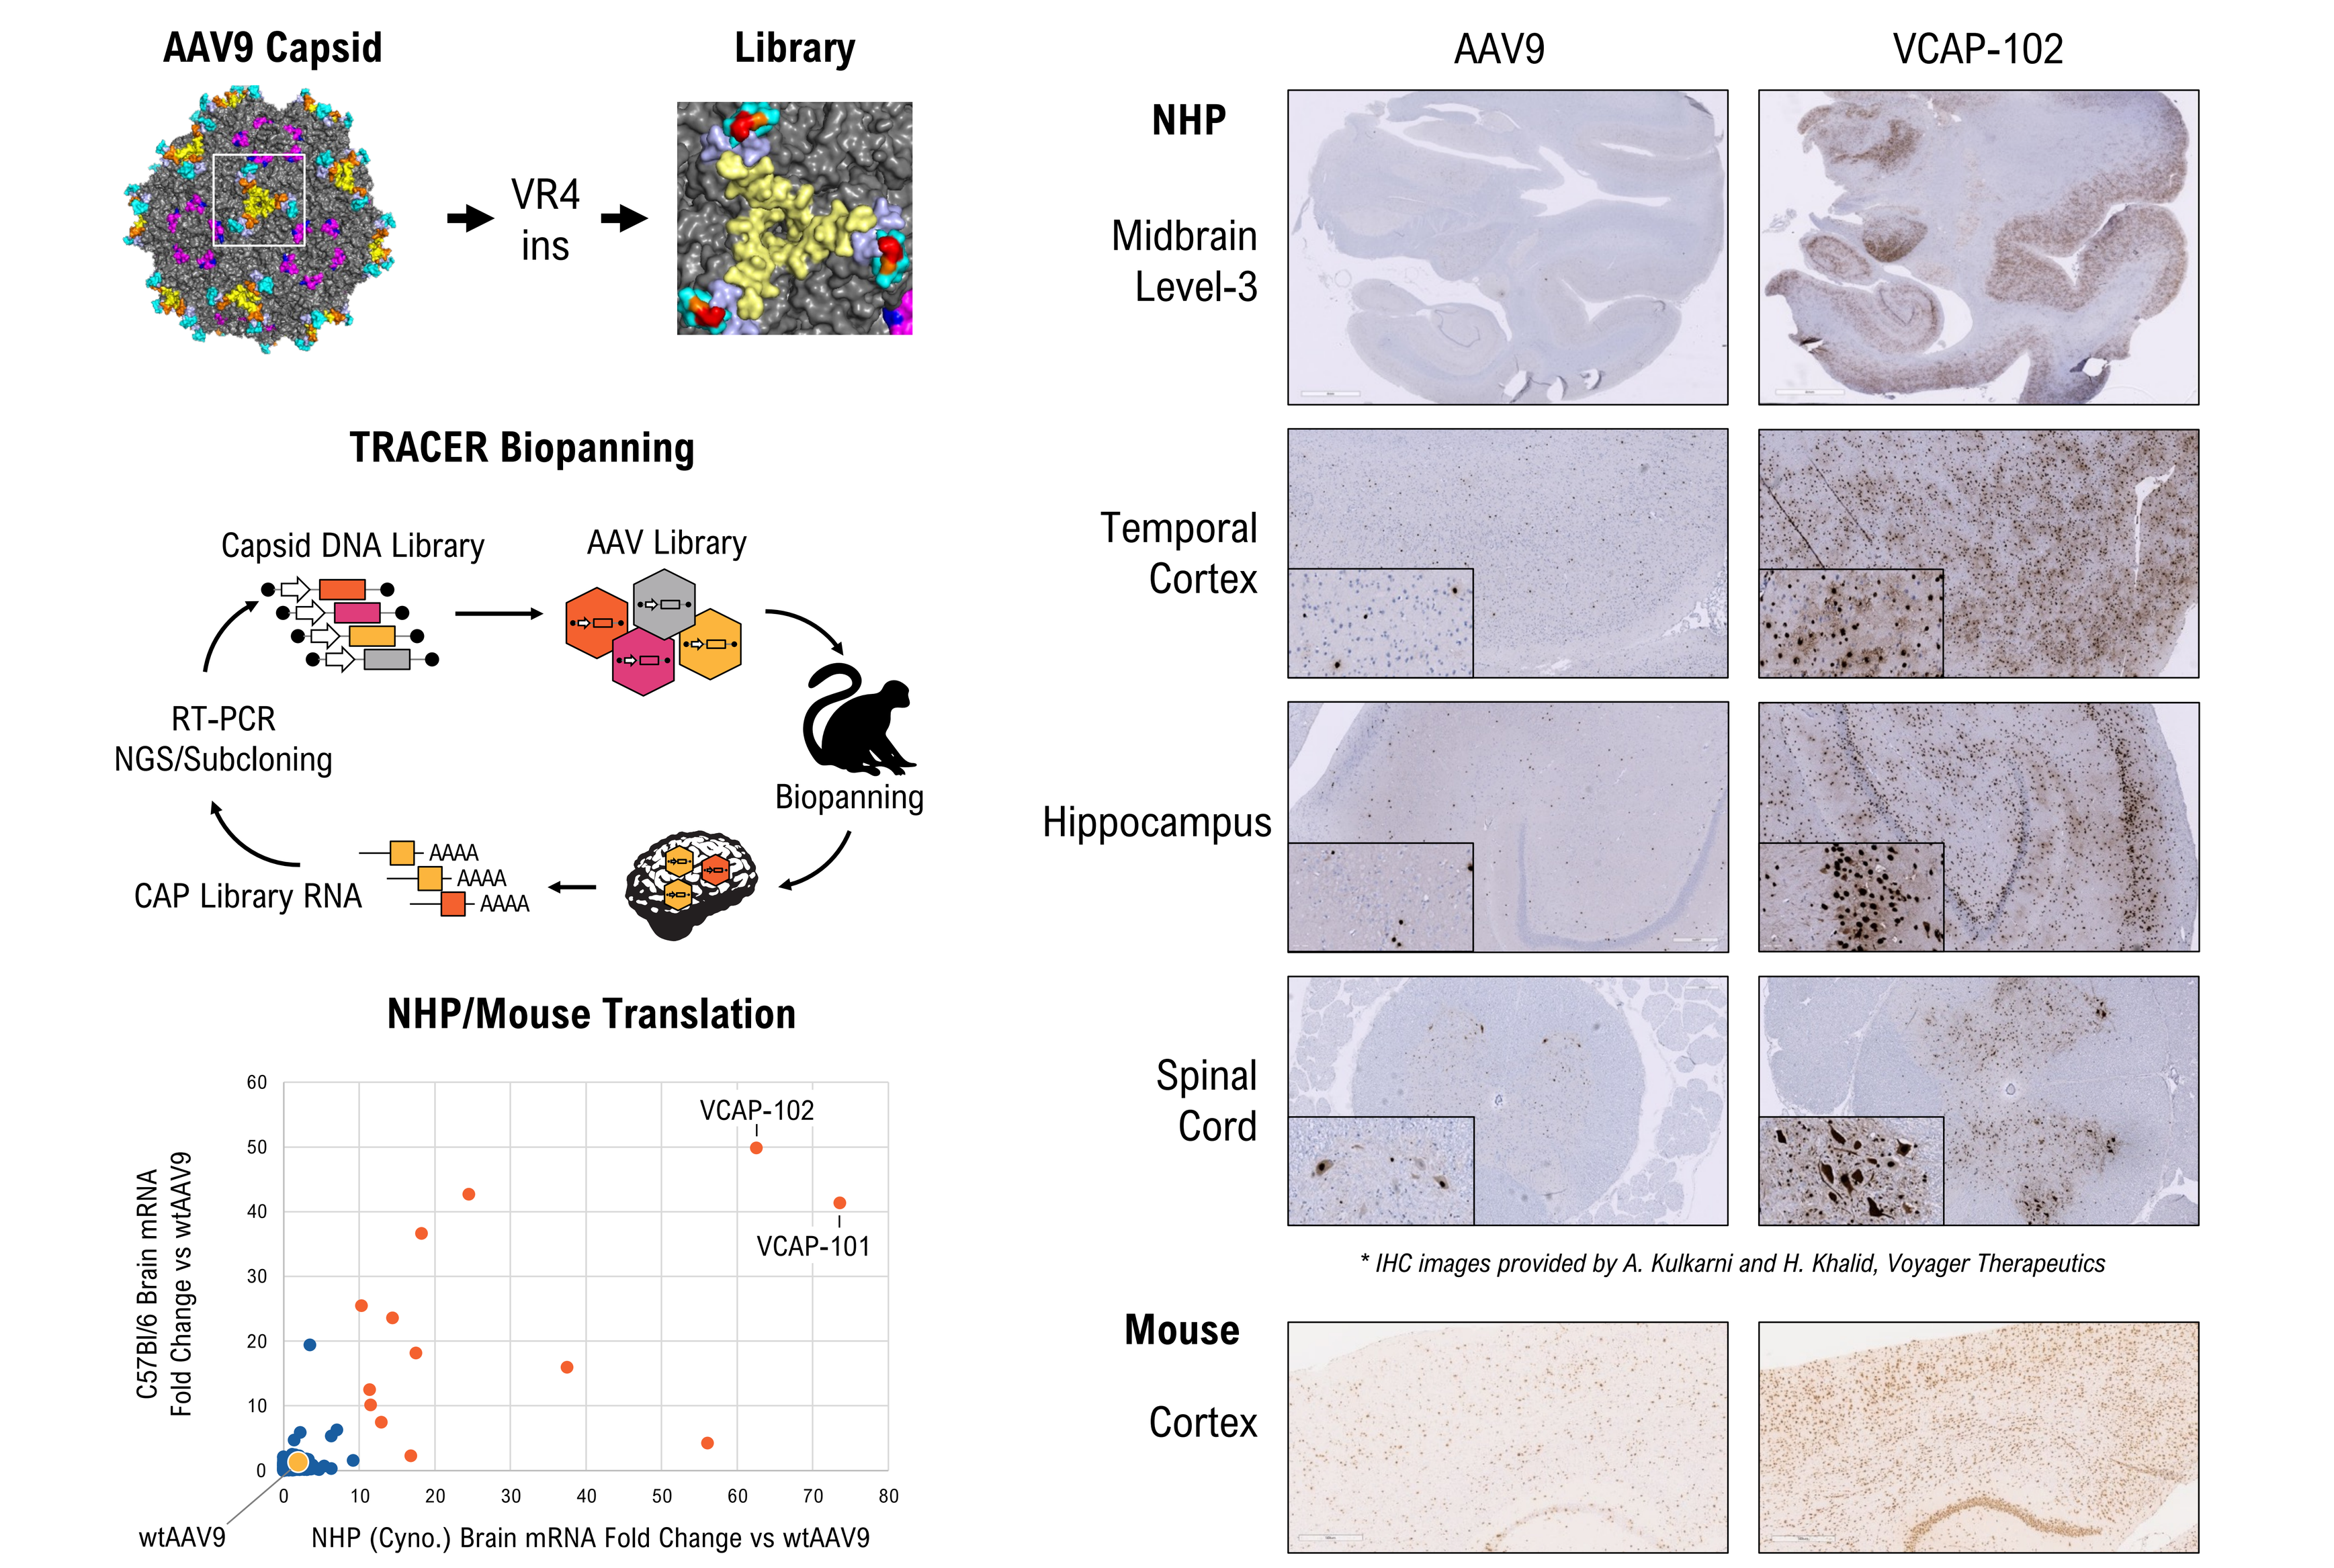
AAV9 Capsid
VR4 ins
Library
TRACER Biopanning
Capsid DNA Library
AAV Library
Biopanning
RT-PCR NGS/Subcloning
CAP Library RNA
AAAA
NHP/Mouse Translation
C57Bl/6 Brain mRNA Fold Change vs wtAAV9
NHP (Cyno.) Brain mRNA Fold Change vs wtAAV9
wtAAV9
VCAP-102
VCAP-101
AAV9
VCAP-102
NHP
Midbrain Level-3
Temporal Cortex
Hippocampus
Spinal Cord
Mouse
Cortex

* IHC images provided by A. Kulkarni and H. Khalid, Voyager Therapeutics

Figure 2. Cryo-EM Structure of VCAP-102

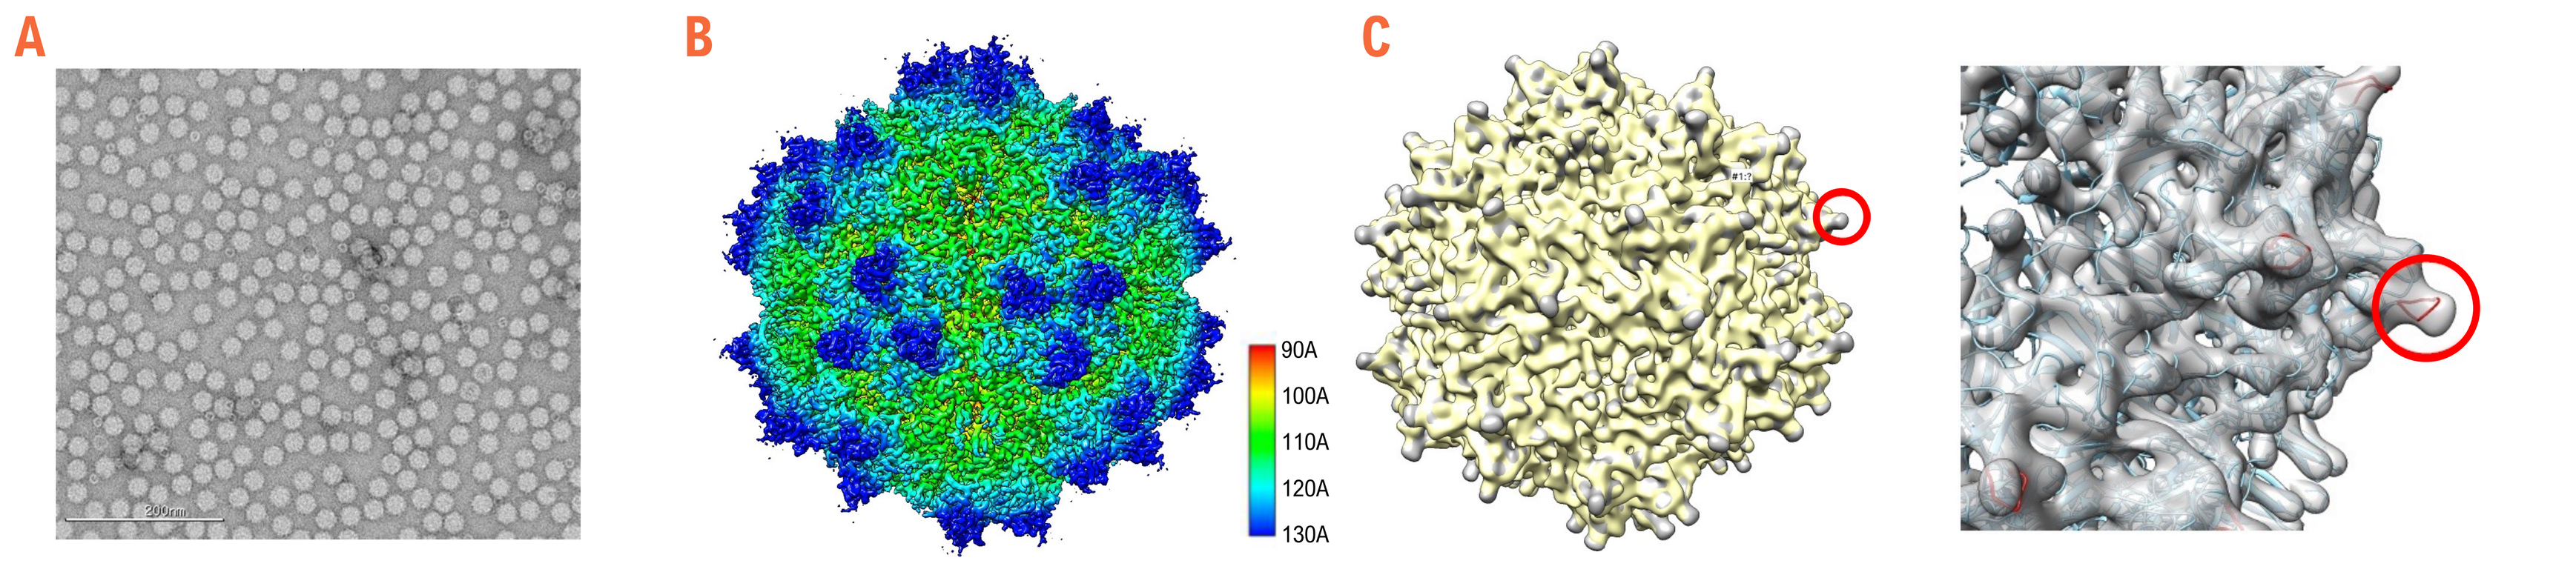
A
B
C
90A
100A
110A
120A
130A

A) Negative staining image of VCAP-102 produced by triple transfection of HEK293T cells and purified by iodixanol gradient centrifugation. B) Cryo-EM structure of VCAP-102 at 3.4 Å resolution. C) Overlay of VCAP-102 map (grey) and AAV9 map (yellow). A difference within a surface loop is highlighted.

Figure 3. Human Receptor Screen Identifies an Interaction Between VCAP-102 and a Membrane Protein Designated Receptor X

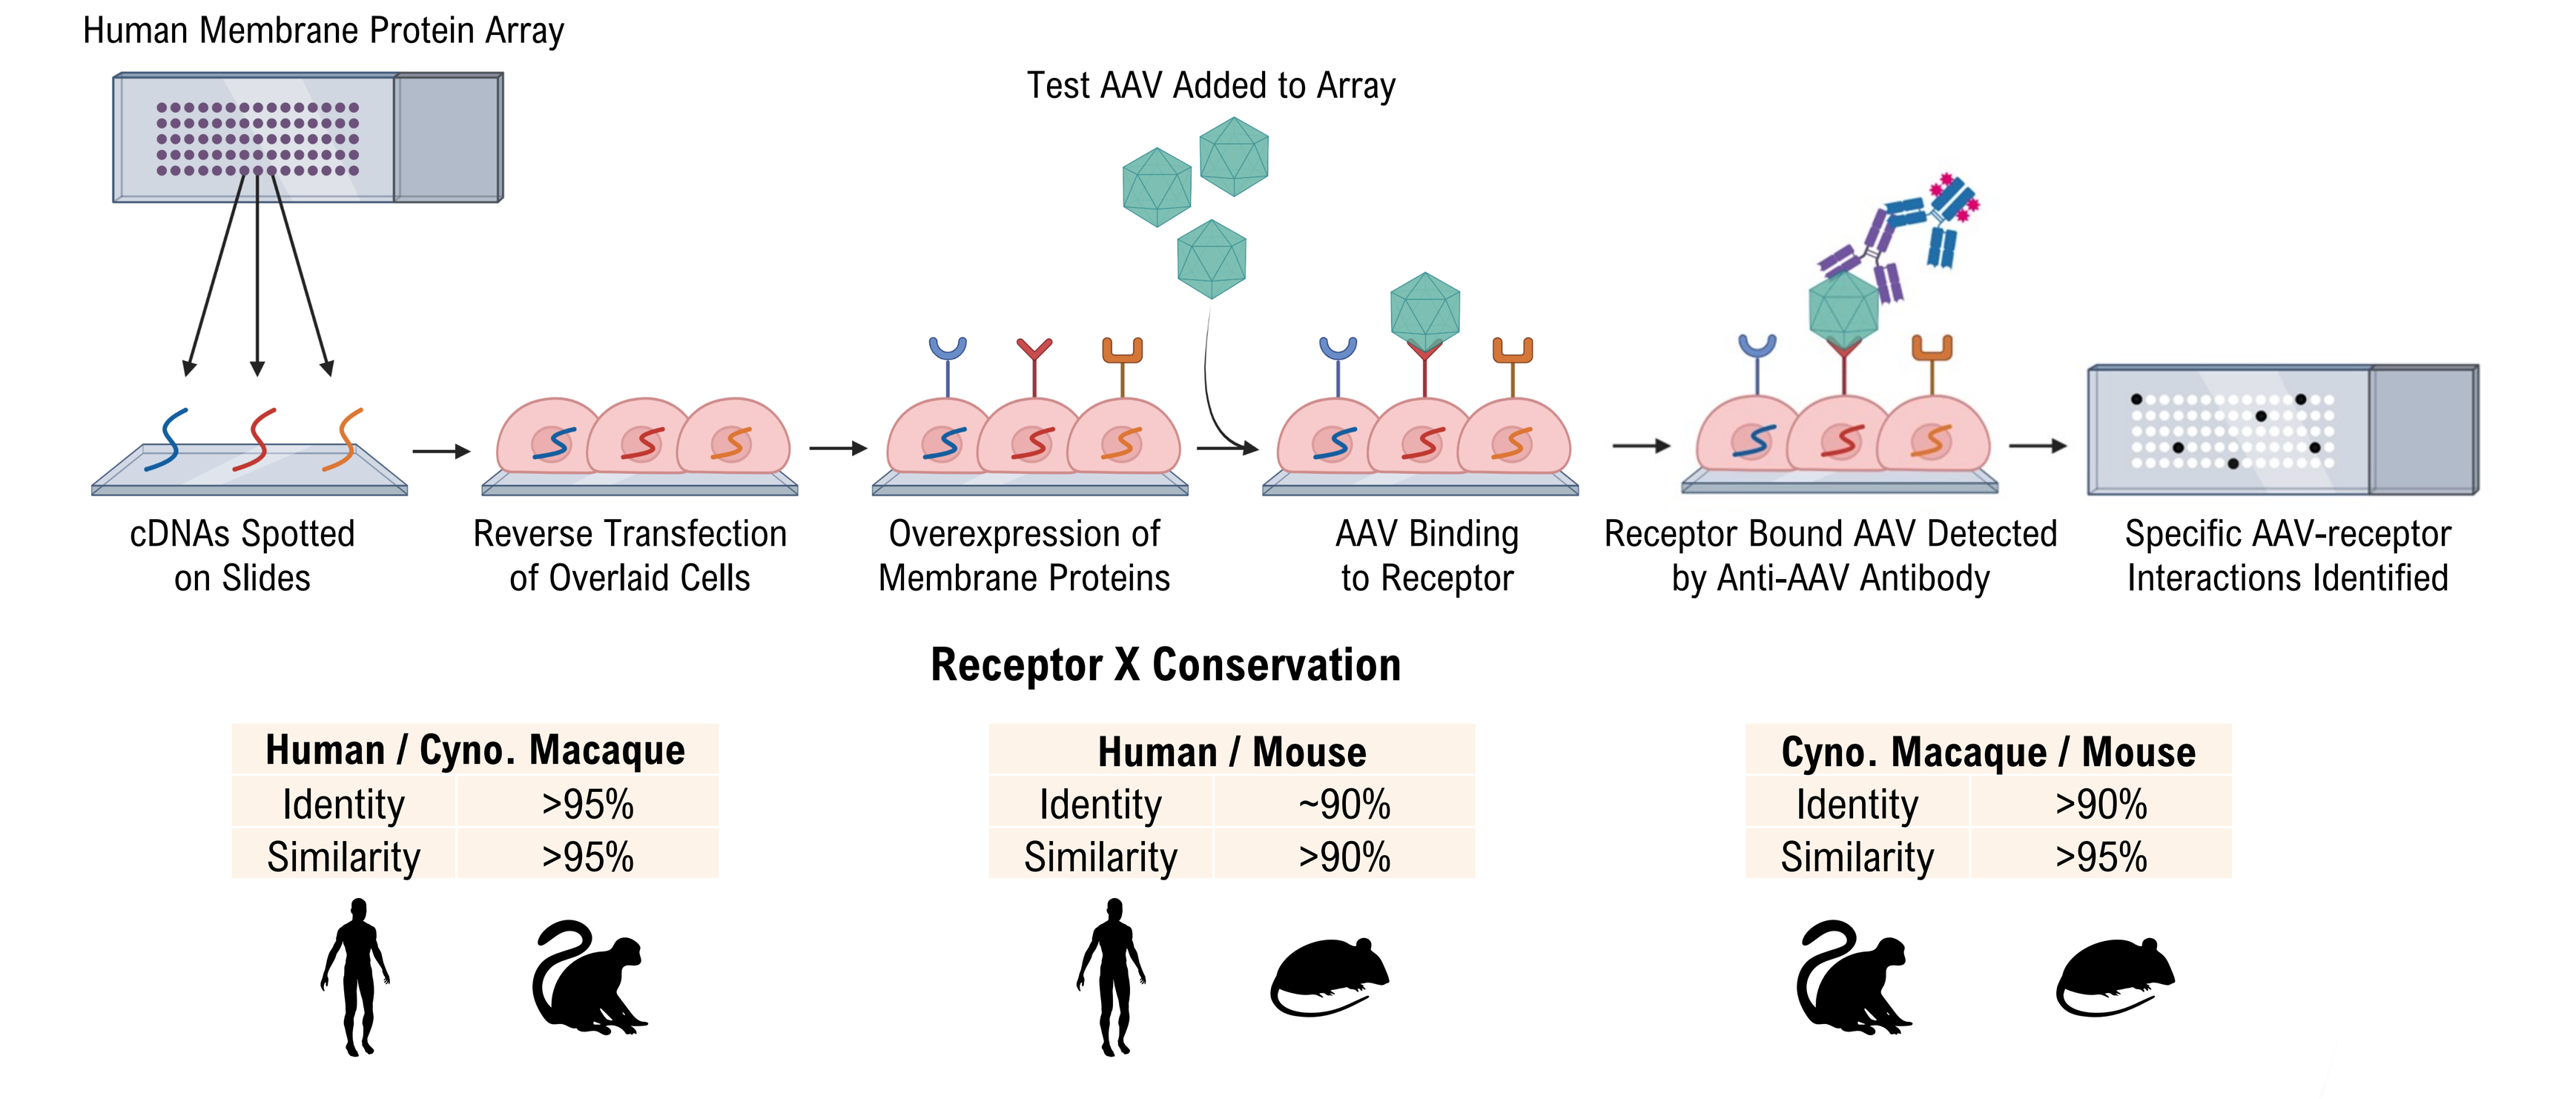
Human Membrane Protein Array
Test AAV Added to Array
cDNAs Spotted on Slides
Reverse Transfection of Overlaid Cells
Overexpression of Membrane Proteins
AAV Binding to Receptor
Receptor Bound AAV Detected by Anti-AAV Antibody
Specific AAV-receptor Interactions Identified
Receptor X Conservation
Human / Cyno. Macaque
Identity >95%
Similarity >95%
Human / Mouse
Identity ~90%
Similarity >90%
Cyno. Macaque / Mouse
Identity >90%
Similarity >95%

Overview of human receptor screen that identified Receptor X as a binding partner of VCAP-102. Receptor X is highly conserved across multiple species as shown by nucleotide identity and similarity.

Figure 4. VCAP-101/102 Transduction Directly Correlates with Human / Mouse Receptor X Expression in Overexpression and Knockdown Assays

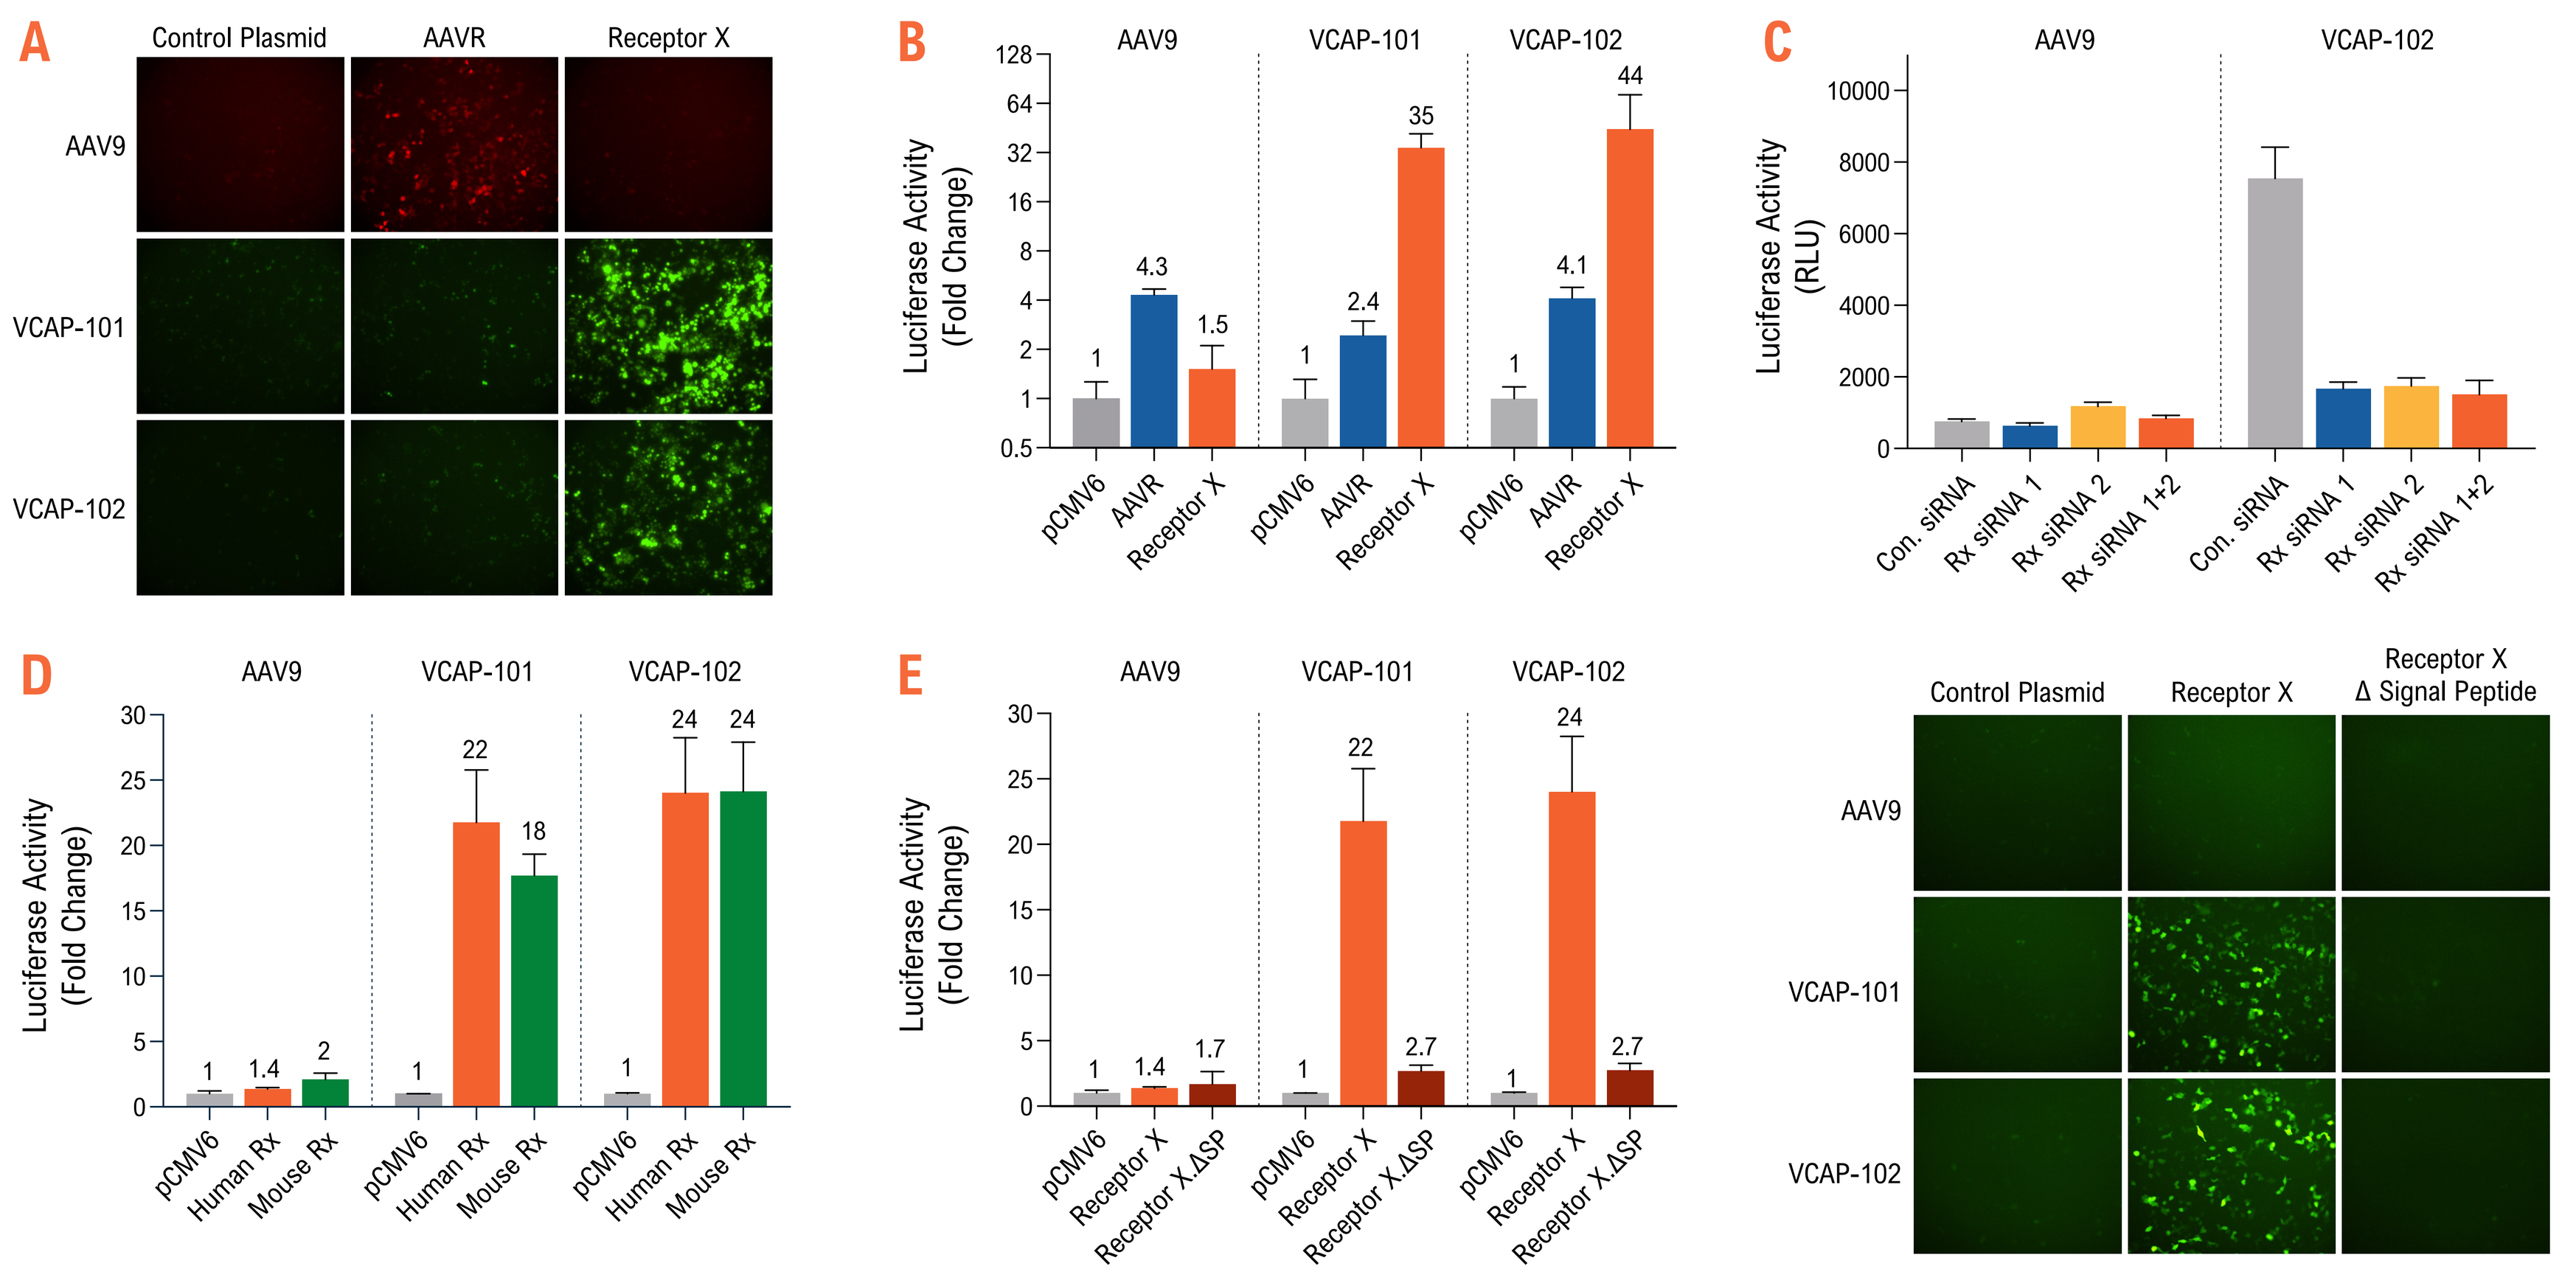
A
Control Plasmid
AAVR
Receptor X
AAV9
VCAP-101
VCAP-102
B
Luciferase Activity (Fold Change)
1
4.3
1.5
1
2.4
35
1
4.1
44
pCMV6
AAVR
Receptor X
C
Luciferase Activity (RLU)
Con. siRNA
Rx siRNA 1
Rx siRNA 2
Rx siRNA 1+2
D
1
1.4
2
1
22
18
1
24
24
Human Rx
Mouse Rx
E
1
1.4
1.7
1
22
2.7
1
24
2.7
Receptor X.ΔSP
Receptor X Δ Signal Peptide

A-E) Transduction assay on HEK293T cells performed 24 hours after transfection with indicated expression plasmids (A, B, D, E) or 48 hours after transfection with siRNAs against Receptor X or scrambled siRNA (C). Cells were transduced with 1E4 VG/cell of the indicated AAV capsid containing the following transgenes: A) CBA-GFP.WPRE / CBA-mCherry.WPRE, B,C,D,E) CBA-Luc2-T2A-GFP). Number above bars indicates average fold change relative to control (n=3). D) Receptor X.ΔSP lacks the ER signal peptide required for surface localization. Unless otherwise stated the human Receptor X ortholog is used.

Figure 5. Direct and Specific Interaction Between Receptor X and VCAP-102 Demonstrated by SPR

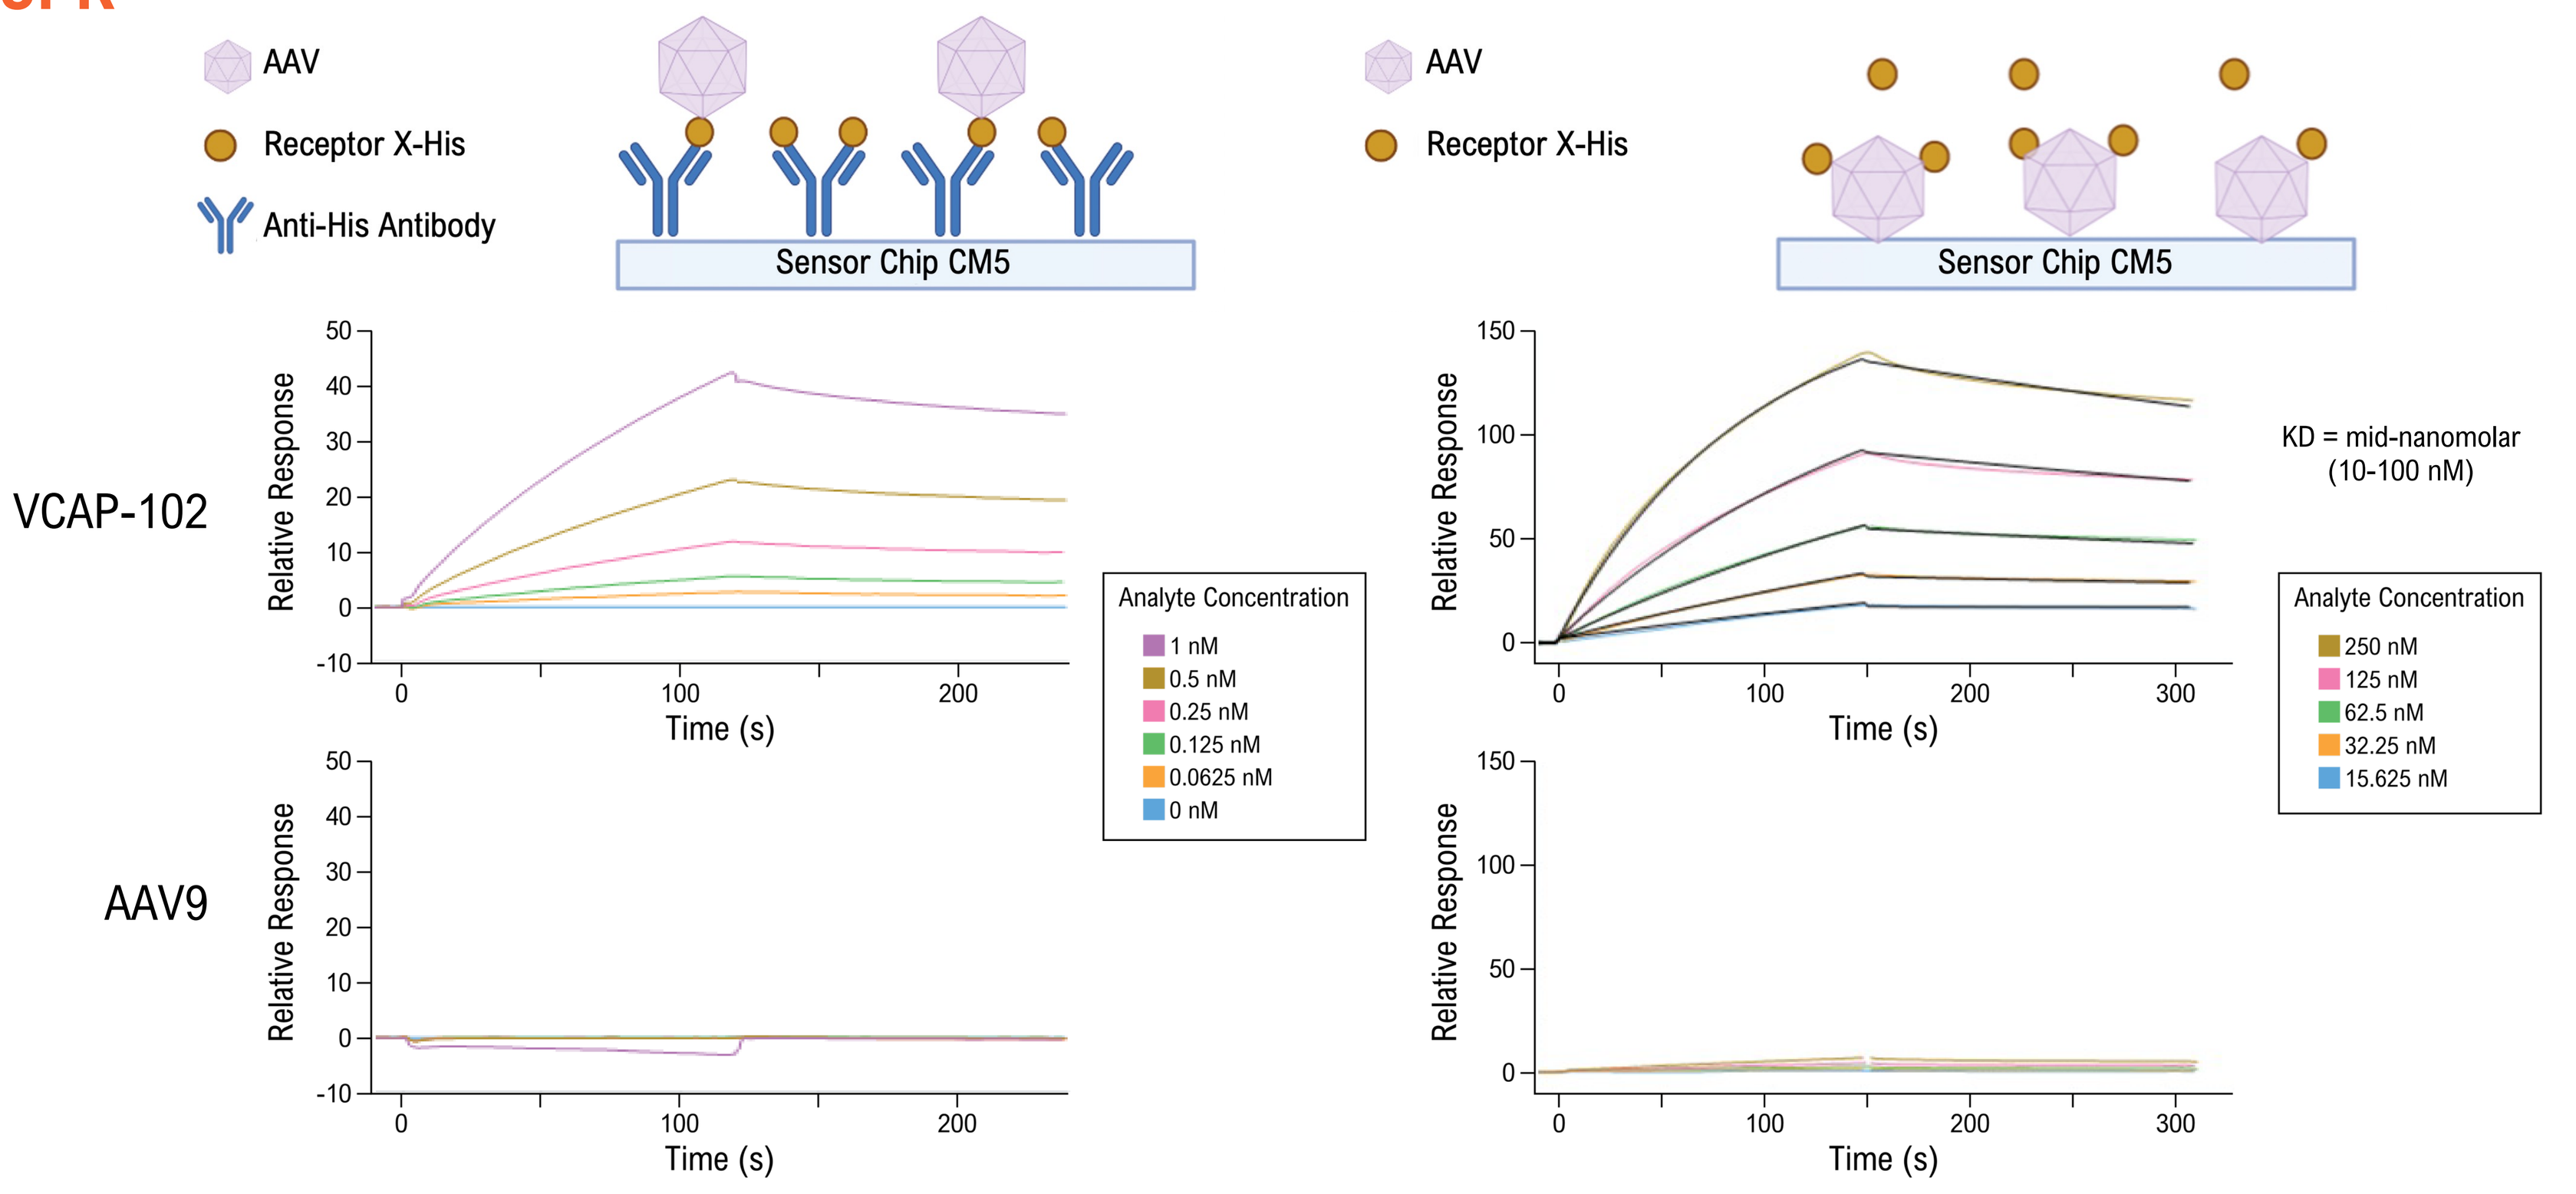
AAV
Receptor X-His
Anti-His Antibody
Sensor Chip CM5
VCAP-102
AAV9
Relative Response
Time (s)
Analyte Concentration
1 nM
0.5 nM
0.25 nM
0.125 nM
0.0625 nM
0 nM
KD = mid-nanomolar (10-100 nM)
250 nM
125 nM
62.5 nM
32.25 nM
15.625 nM

Figure 6. Correlation Between Expression of Receptor X and VCAP-102 Cellular Tropism in Mice

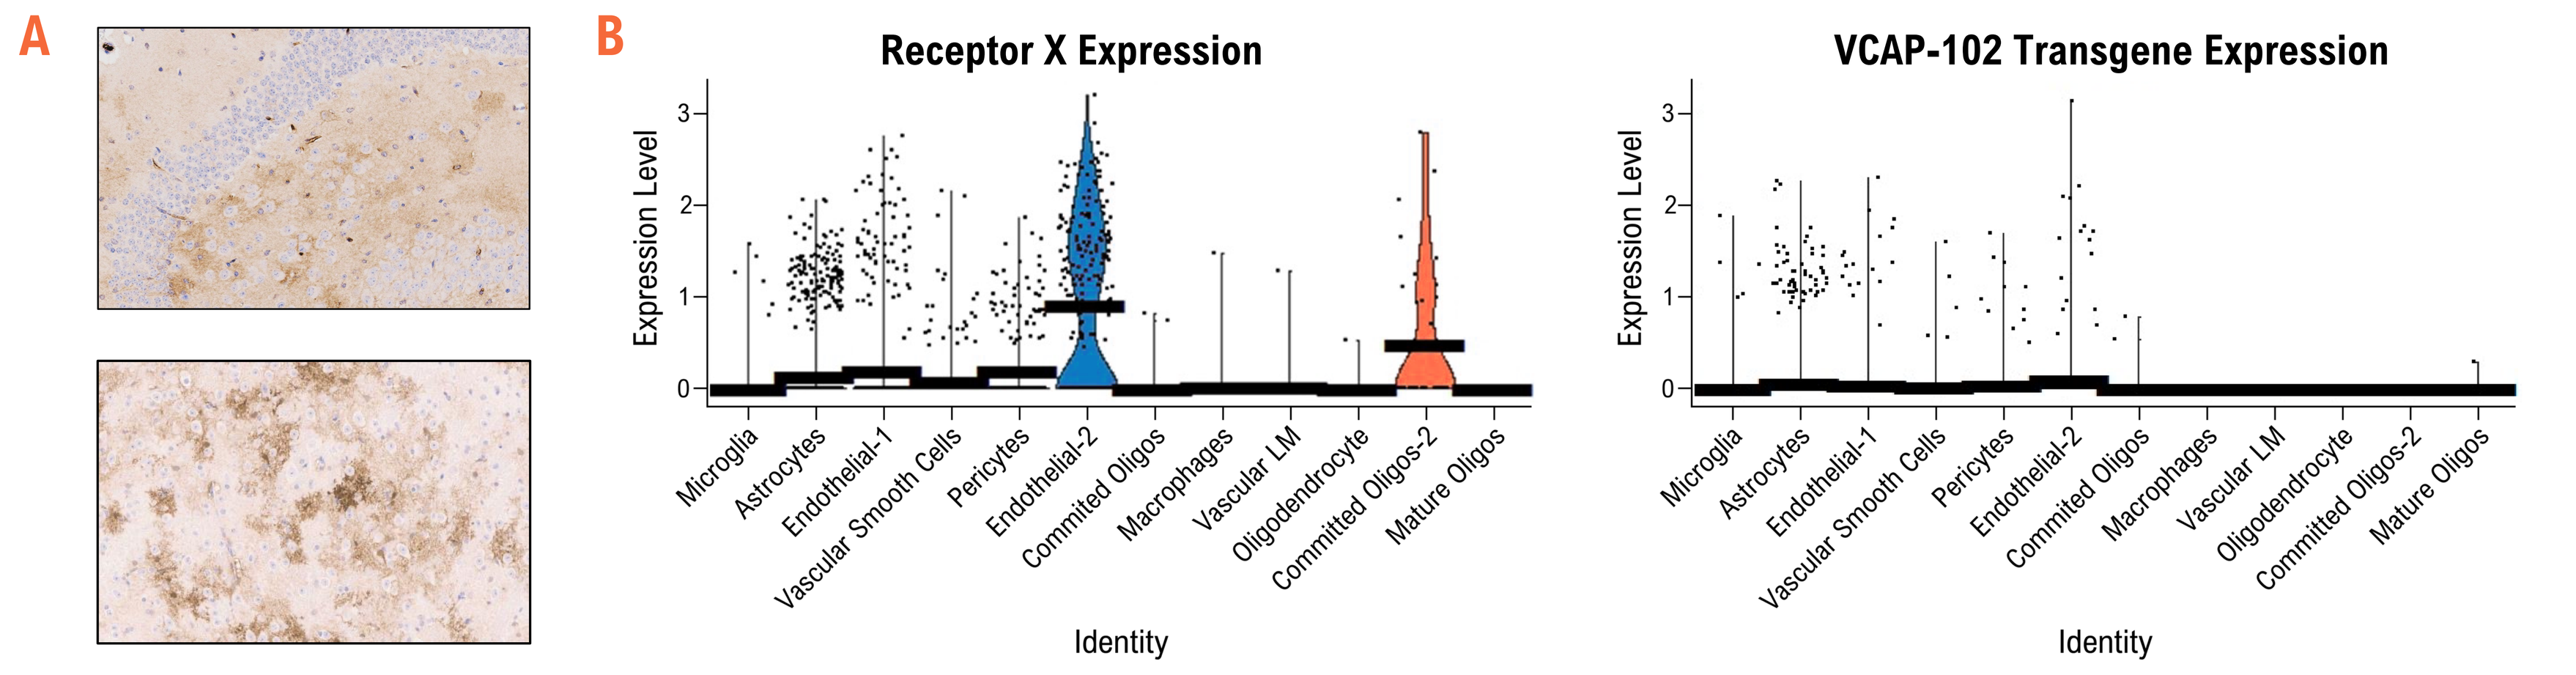
A
B
Receptor X Expression
VCAP-102 Transgene Expression
Expression Level
Identity
Microglia
Astrocytes
Endothelial-1
Vascular Smooth Cells
Pericytes
Endothelial-2
Commited Oligos
Macrophages
Vascular LM
Oligodendrocyte
Committed Oligos-2
Mature Oligos

A) Top panel - Receptor X staining in naïve mouse brain. Bottom panel - GFP staining in mouse brain 28 days after IV dosing of 2.5E13 vg/kg VCAP-102 (CBA-Luc2-T2A-GFP). B) scRNA-seq data from mouse midbrain 28 days post IV dosing of 2.5E13 vg/kg VCAP-102 (CBA-Luc2-T2A-GFP).

Figure 7. VCAP-101/102 Retain Galactose Binding

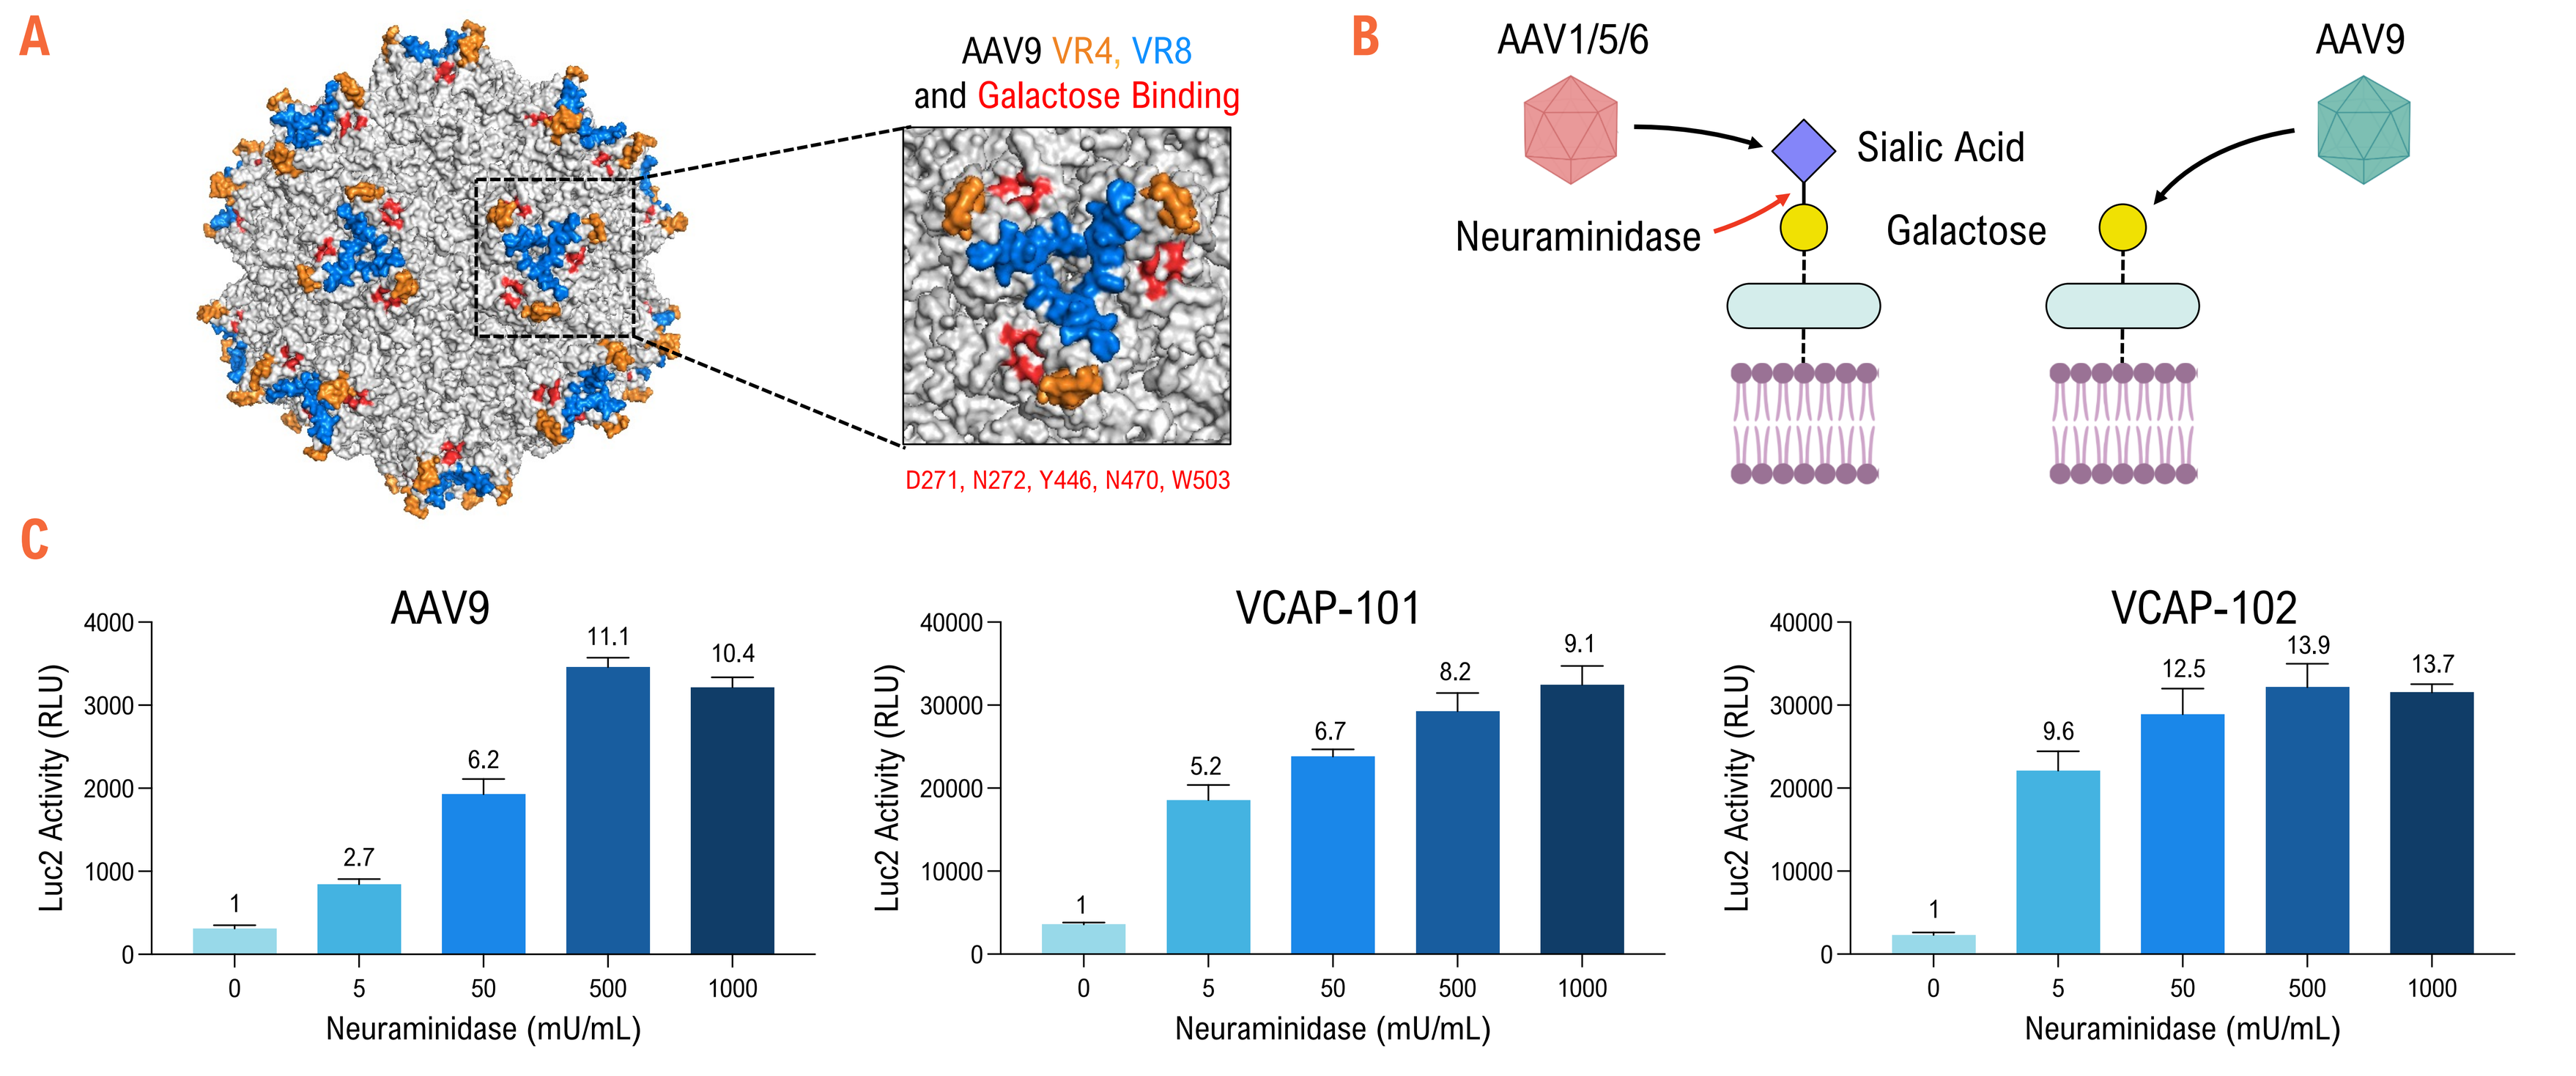
A
AAV9 VR4, VR8 and Galactose Binding
D271, N272, Y446, N470, W503
B
AAV1/5/6
AAV9
Sialic Acid
Neuraminidase
Galactose
C
AAV9
1
2.7
6.2
11.1
10.4
VCAP-101
1
5.2
6.7
8.2
9.1
VCAP-102
1
9.6
12.5
13.9
13.7
Luc2 Activity (RLU)
Neuraminidase (mU/mL)

A) Location of surface residues implicated in galactose binding are highlighted in red. B) Treatment with neuraminidase removes terminal sialic acid resulting in increased levels of terminal galactose. C) HEK293T cells were transduced with 1E4 VG/cell of the indicated AAV capsid (CBA-Luc2-T2A-GFP) following 4hr neuraminidase pre-treatment at the indicated concentration. Number above bars indicates average fold change relative to control (n=3).

CONCLUSIONS

- We have identified the primary attachment surface receptor used by the VCAP-101/102 BBB-penetrant engineered capsid class designated Receptor X.
- Overexpression, knockdown and direct binding experiments strongly support a role of Receptor X as a primary receptor for VCAP-101/102 and confirm cross-species properties of these capsids.
- Expression pattern of Receptor X shows some correlation with VCAP-102 cellular tropism in mice.
- Data suggests VCAP-101/VCAP-102 retain galactose and AAVR binding properties.
- In addition to the cross-species nature of this capsid family, our data provide strong evidence for direct and functional interaction with a human receptor. It is therefore anticipated that the BBB-penetrating properties of VCAP-101 and VCAP-102 will translate to humans.

voyager THERAPEUTICS 64 Sidney Street, Cambridge, MA 02139, USA

ESGCT - 29th Congress | October 11 – 14, 2022 | Edinburgh