Identification and Characterization of Novel Anti-tau Antibodies that Inhibit Tau-seed Mediated Pathology in a P301S Tauopathy Mouse Model of Alzheimer's Disease and Tauopathies

Wencheng Liu, Vinodh Kurella, Li Liu, Ishan Shah, Dillon Kavanagh, Maneesha Paranjpe, Jerrah Holth, Charlotte Chung, Alex Powers, Xiao-Qin Ren, Yanqun Shu, Allen Capili, Jay Hou, Bernardino Ghetti, Kelly Bales, Steve Paul, Todd Carter

Voyager Therapeutics Inc., 64 Sidney Street, Cambridge, MA 02139, USA

INVESTORS & MEDIA

CAREERS AT VOYAGER

#62878

SUMMARY

- Diverse starting pool of proprietary Ab's targeting pathological tau from human AD brain generated and screened
- 4 Ab's selected (Ab01, Ab03, Ab04 and Human Ab5), with novel sequences and epitopes that fit the target profile based on selectivity, functional inhibition *in vitro* and *in vivo*, and developability
- These Ab's targeting mid-domain and C-terminus have passed the selectivity, functional inhibition *in vitro/ in vivo* and developability criteria
- Ab01 and Human Ab5 demonstrated efficacy in the seeding model

INTRODUCTION

The current hypothesis for the progression of tau pathology in Alzheimer's disease (AD) and tauopathies (for example, Progressive supranuclear palsy (PSP)), is based on mechanisms involving seeding and propagation of pathologic tau via cell-to-cell transmission, including trans-synaptic propagation, a mechanism which provides the opportunity for anti-tau immunotherapy as a logical and promising therapy for these diseases. Here, we describe the characterization of 113 anti-tau antibodies based on their biochemical and biophysical properties, including affinity for PHF-tau, immunohistochemical selectivity for AD/PSP in human brain sections, functional inhibition of PHF seeding *in vitro*, developability, as well as epitope mapping. 10 antibodies that met the target profile described above were selected for *in vivo* efficacy screening in the P301S seeding-propagation tauopathy mouse model. Epitope mapping for these 12 antibodies demonstrates that these antibodies target phospho-tau epitopes in diverse locations throughout the tau protein, with eight antibodies targeting the mid-domain region, three targeting the C-terminal region, and one targeting the N-terminal/mid-domain region. We have evaluated 10 antibodies for their ability to block PHF seeding/propagation in a P301S mouse seeding model and demonstrated substantial reduction of induced tau pathology. Additional antibodies are currently under evaluation.

Figure 1. Voyager Anti-Tau Antibody Discovery

GOAL: Identify anti-tau antibodies that block tau-seed mediated propagation of pathology

APPROACH:

Focus on antibodies that target pathological tau species

Generate a diverse starting pool of anti-tau Ab's

Immunization Campaigns

- Host animals: WT mouse, Tau-KO mouse for murine Ab's; human mouse for human Ab's
- Immunogens: Paired Helical Filamentous tau (ePHF or sarkosyl insoluble fraction) from AD brain

Screen and prioritize anti-tau Ab's based on:

- Biochemical selectivity for pathological tau
- IHC selectivity for AD/PSP vs WT human brain
- Functional inhibition of PHF seeding *in vitro* and *in vivo*
- Developability based on low polyspecificity and lack of aggregation

Characterize additional attributes

- Sequence
- Epitope
- Western blot binding to AD, PSP vs WT human brain

Mouse immunization strategy and candidates' selection profiles for identifying efficacious anti-tau candidates.

STATUS:

- Epitopes mapped to **12** regions, including **4** new epitopes
- Anti-tau Ab's sequenced, revealing **64** unique sequence families

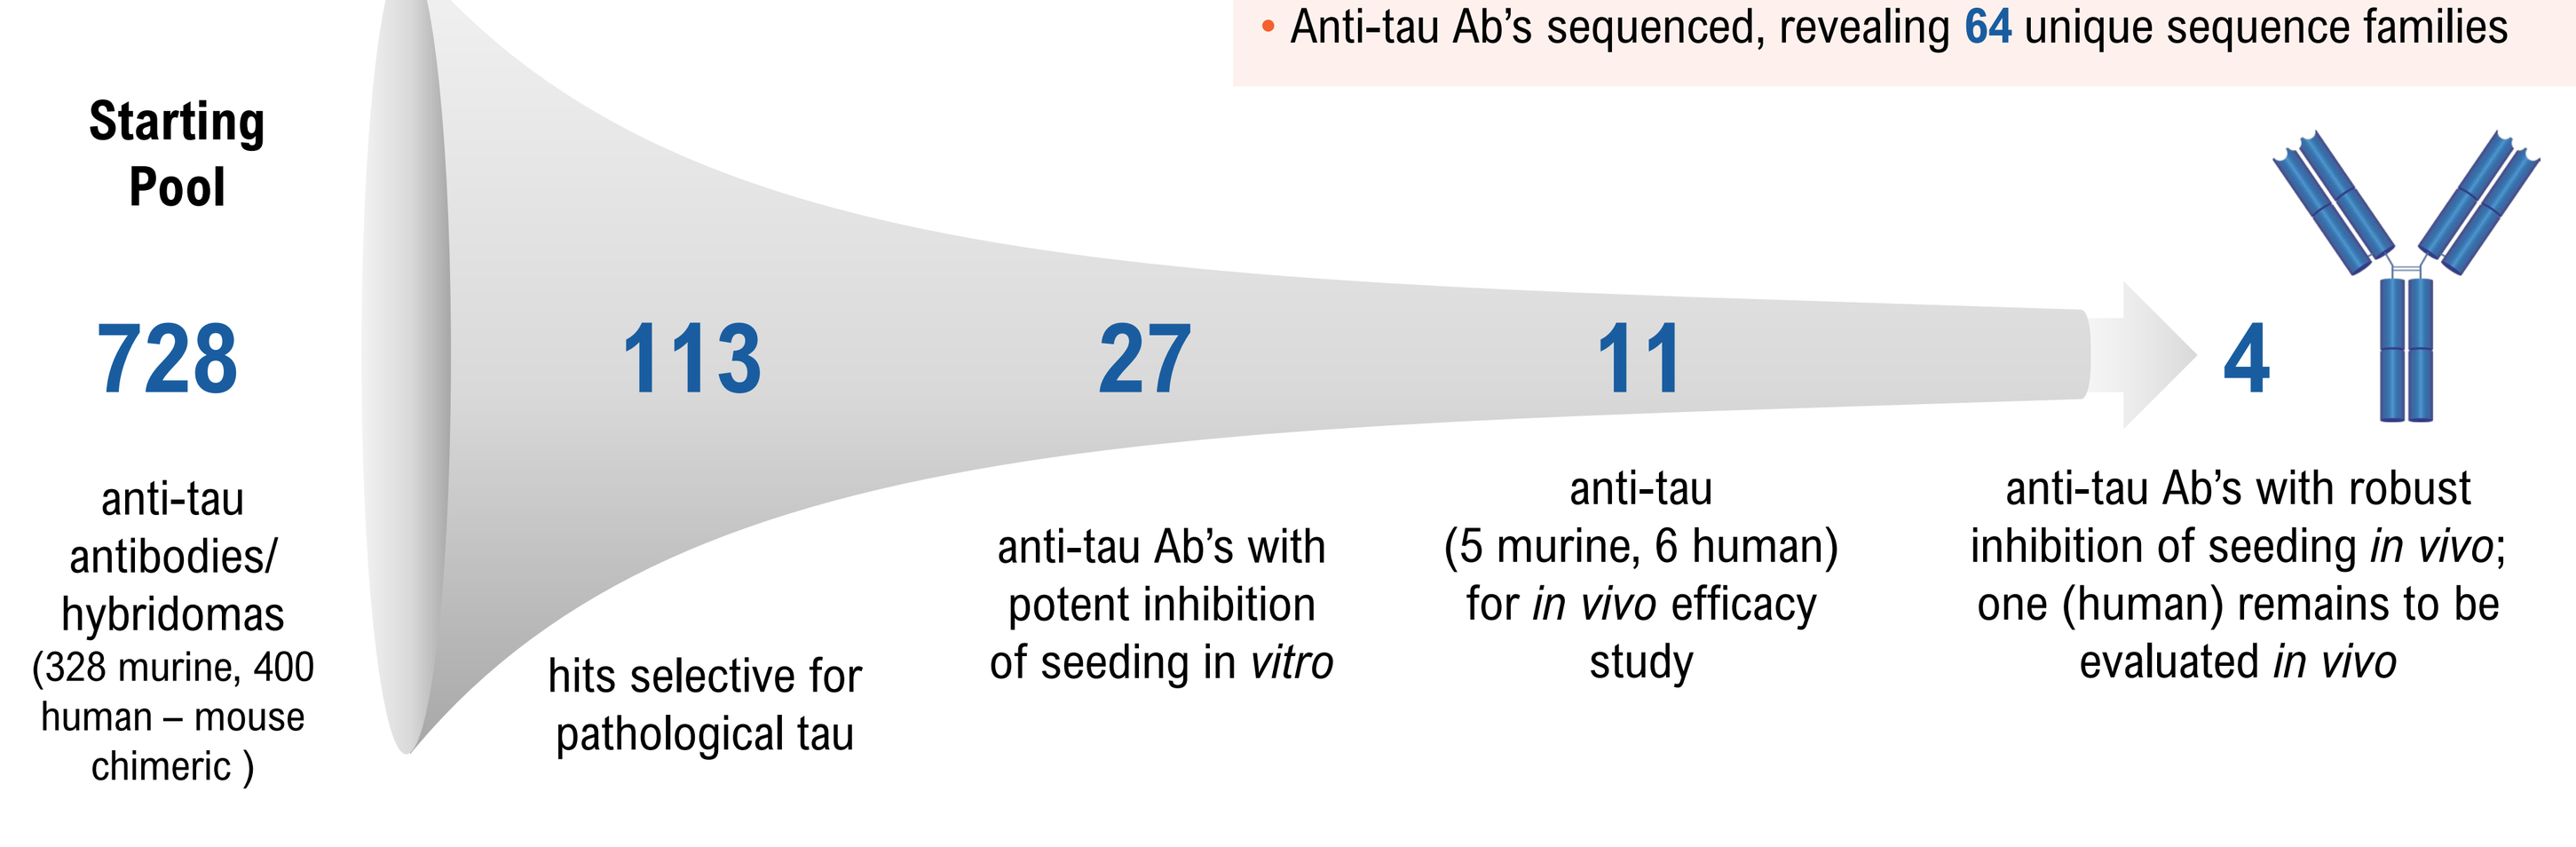

Starting Pool
728
anti-tau antibodies/ hybridomas (328 murine, 400 human – mouse chimeric)
113
hits selective for pathological tau
27
anti-tau Ab's with potent inhibition of seeding in vitro
11
anti-tau (5 murine, 6 human) for in vivo efficacy study
4
anti-tau Ab's with robust inhibition of seeding in vivo; one (human) remains to be evaluated in vivo

Overview of Voyager ant-tau antibodies discovery that leads to identify 4 anti-tau demonstrating robust inhibition of seeding *in vivo*.

Figure 2. Biophysical Properties of Voyager Anti-Tau Antibodies that Met the Target Profile of Selectivity, Functional in Inhibition and Developability

A Voyager Anti-tau Antibodies Target Various Epitopes Located Throughout Full-length Tau

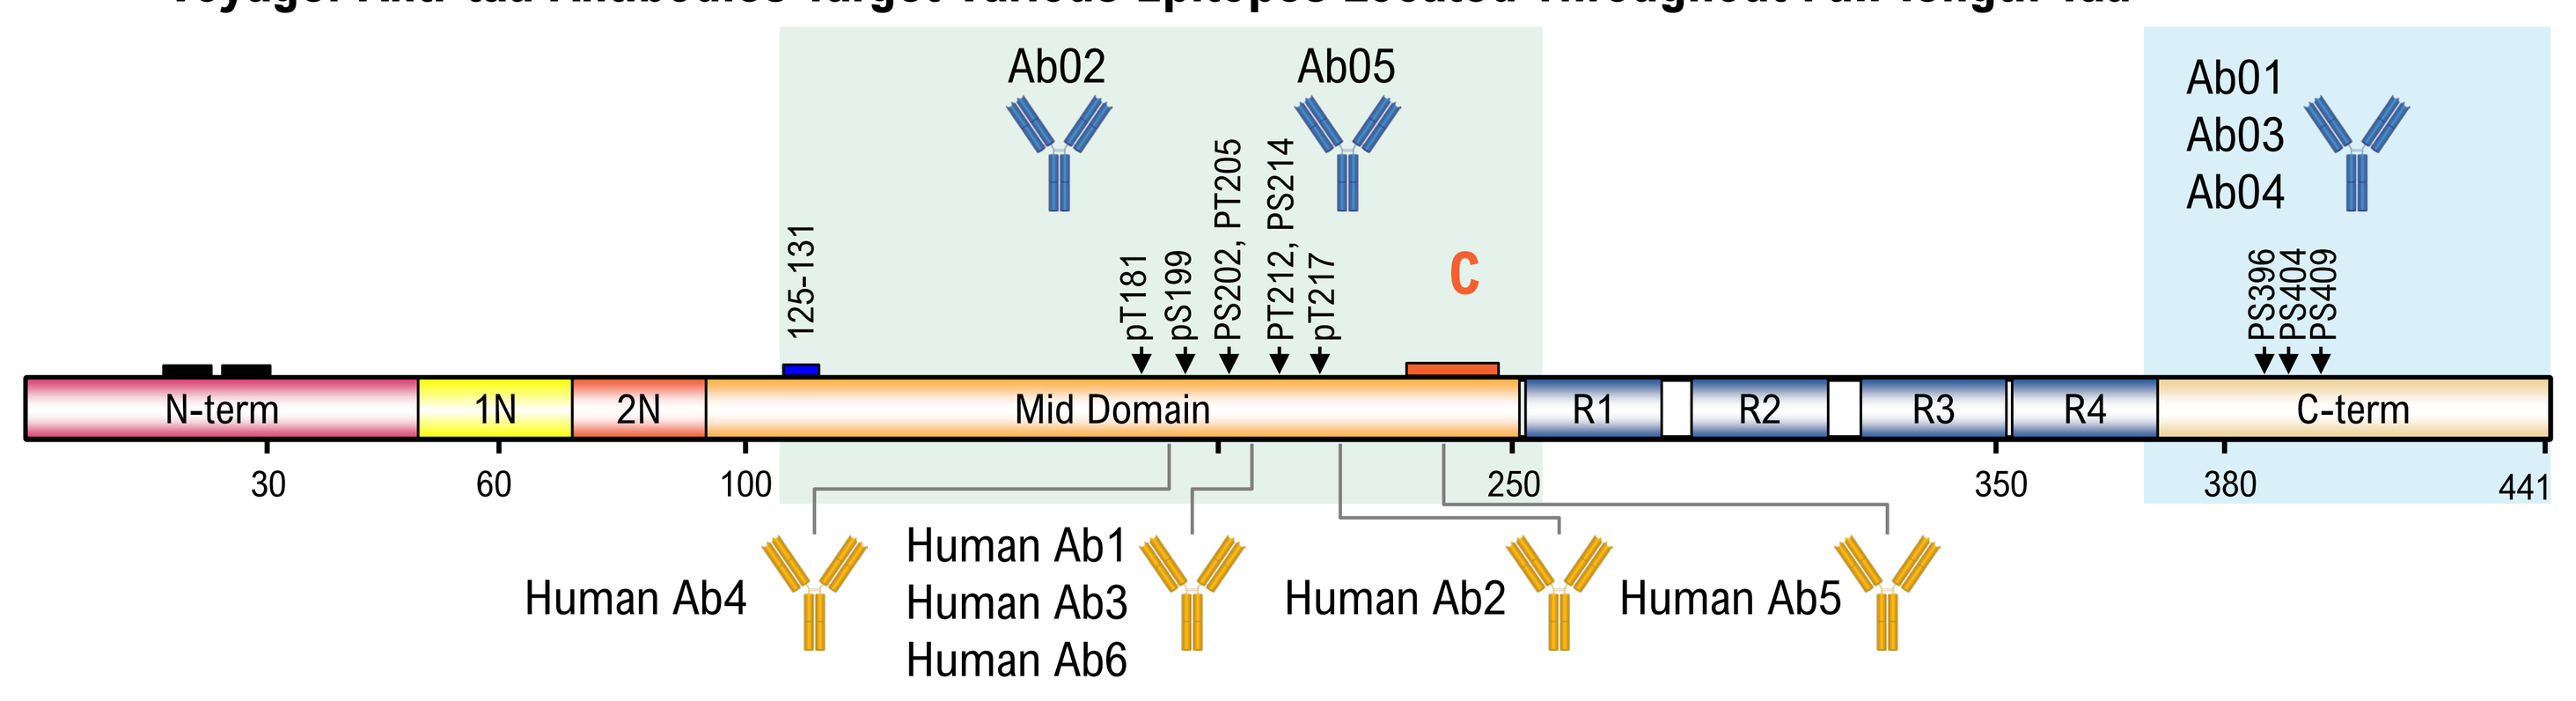

Ab02
Ab05
Ab01
Ab03
Ab04
125-131
pT181
pS199
PS202, PT205
PT212, PS214
pT217
C
PS396
PS404
PS409
N-term
1N
2N
Mid Domain
R1
R2
R3
R4
C-term
30
60
100
250
350
380
441
Human Ab4
Human Ab1
Human Ab3
Human Ab6
Human Ab2
Human Ab5

B Binding Affinity of VYGR's Murine and Human Abs

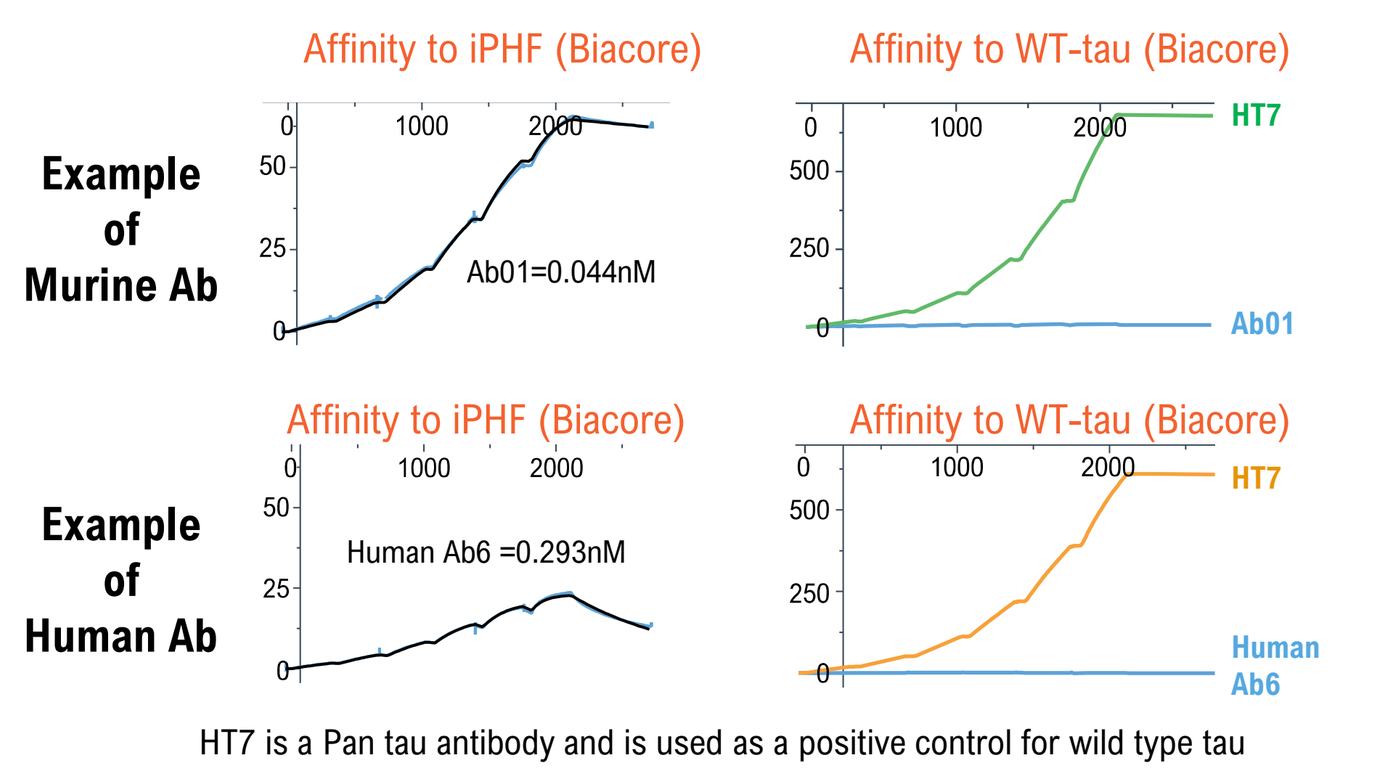

Affinity to iPHF (Biacore)
Affinity to WT-tau (Biacore)
Example of Murine Ab
Ab01=0.044nM
HT7
Ab01
Example of Human Ab
Human Ab6 =0.293nM
HT7
Human Ab6

HT7 is a Pan tau antibody and is used as a positive control for wild type tau

	Kinetic parameters from 1:1 binding model (Mean values from duplicate measurements)		
	KD (pM)	ka (1/Ms)	kd (1/s)
CONTROL ANTIBODY			
Ref Ab1	5.2	1.87E+06	9.66E-06
MURINE			
Ab01	43.9	1.70E+06	7.09E-05
Ab02	296.5	2.92E+06	7.87E-04
Ab03	57.8	5.22E+05	3.00E-05
Ab04	124.5	5.89E+05	7.32E-05
Ab05	4465 (KD1), 2640 (KD2)	3.0E+07, 6.6E+04	1.3E-01, 1.8E-04
HUMAN			
Human Ab1	596	1.03E+07	6.10E-03
Human Ab2	11650	1.82E+06	2.14E-02
Human Ab3	356	1.04E+08	3.69E-02
Human Ab4	238	5.45E+06	1.30E-03
Human Ab5	6060	3.93E+05	2.38E-03
Human Ab6	293	3.84E+06	1.13E-03

C Voyager Anti-tau Ab's Prevent PHF Seeding *in vitro*

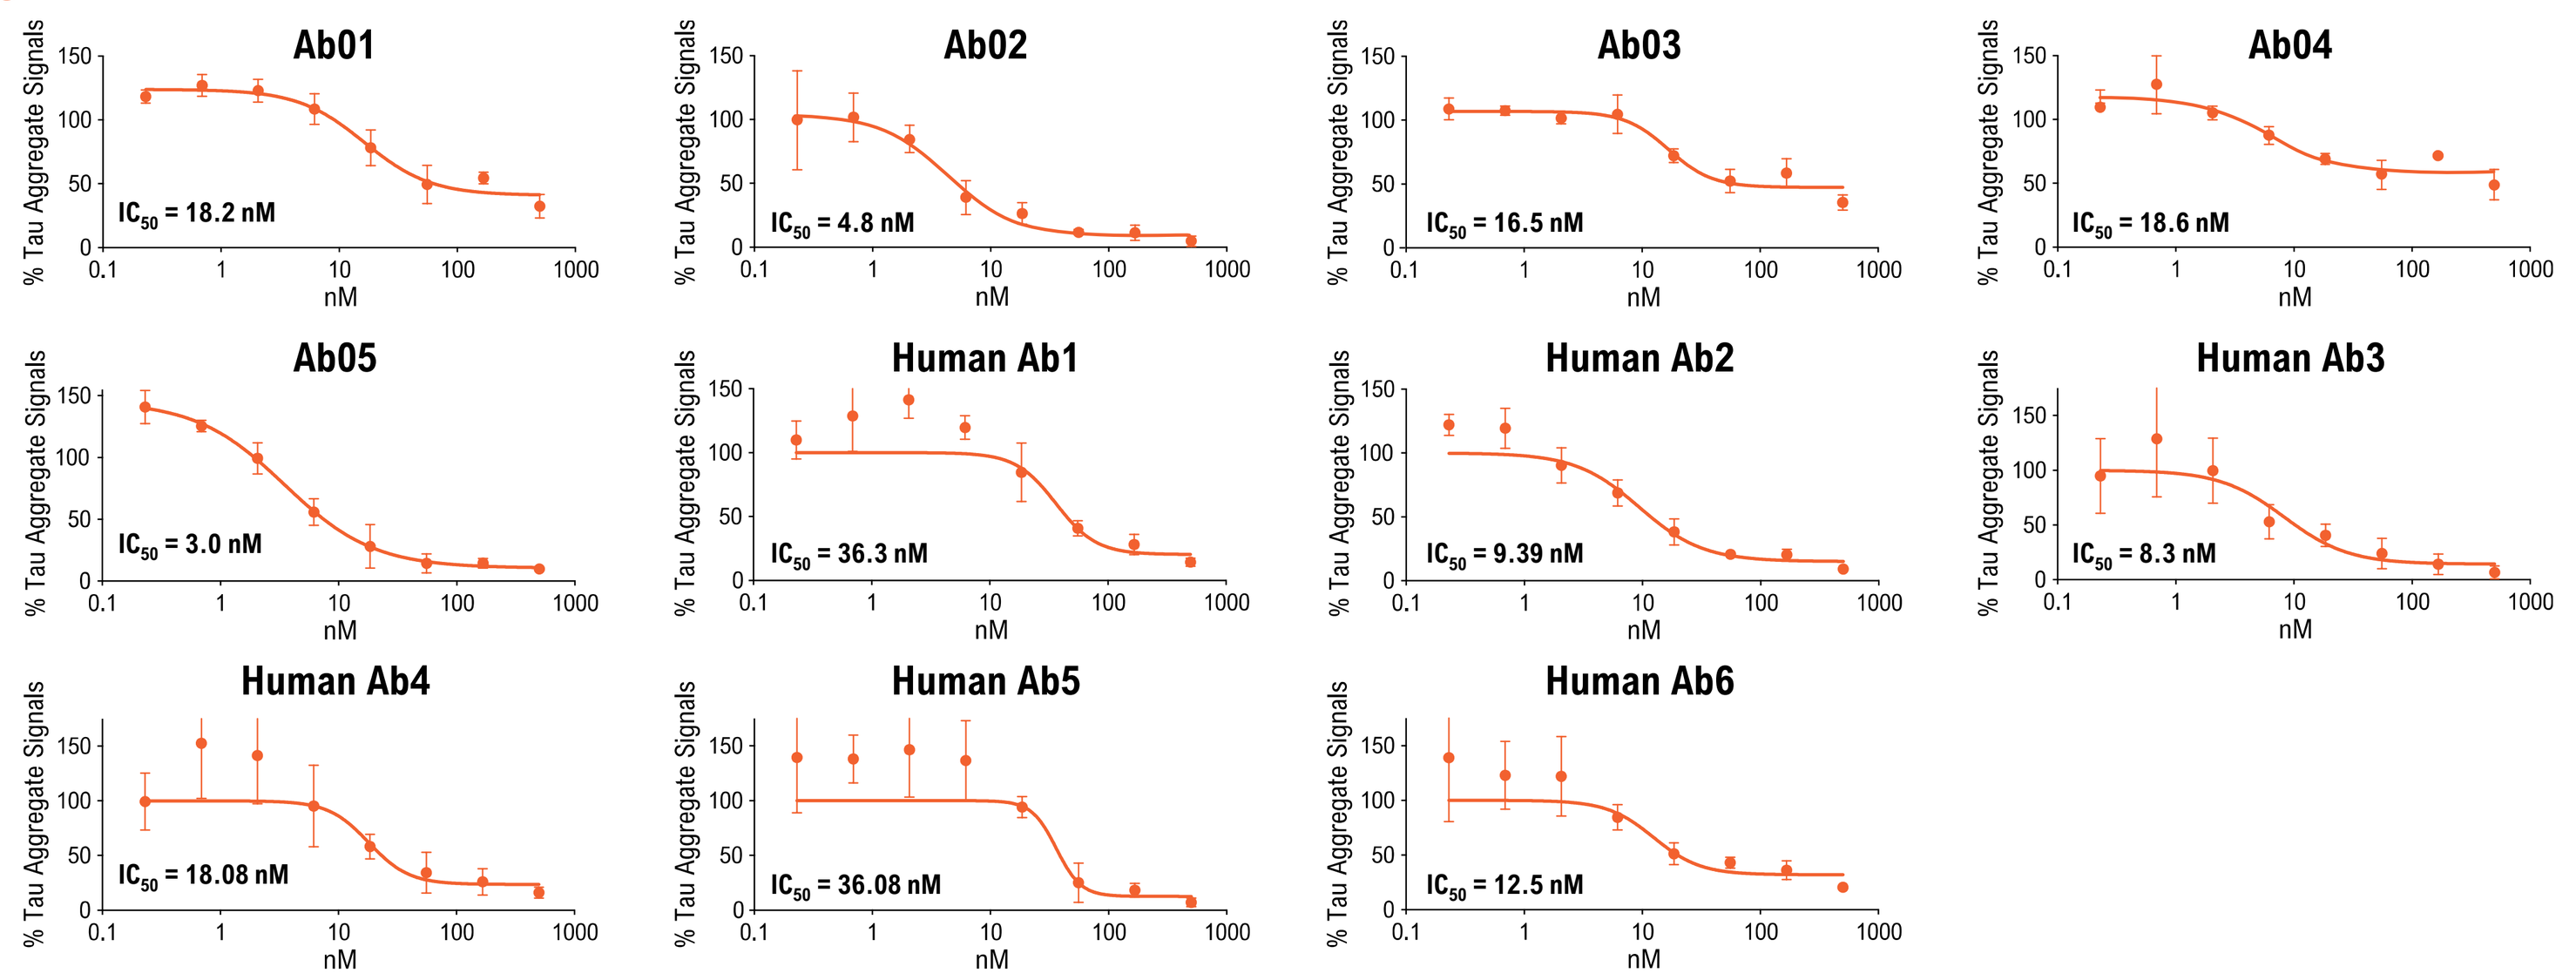

Vehicle
Ab01
IC50 = 18.2 nM
Ab02
IC50 = 4.8 nM
Ab03
IC50 = 16.5 nM
Ab04
IC50 = 18.6 nM
Ab05
IC50 = 3.0 nM
Human Ab1
IC50 = 36.3 nM
Human Ab2
IC50 = 9.39 nM
Human Ab3
IC50 = 8.3 nM
Human Ab4
IC50 = 18.08 nM
Human Ab5
IC50 = 36.08 nM
Human Ab6
IC50 = 12.5 nM
% Tau Aggregate Signals
nM

D Voyager Anti-tau Abs Bind Specifically to Tau Pathology

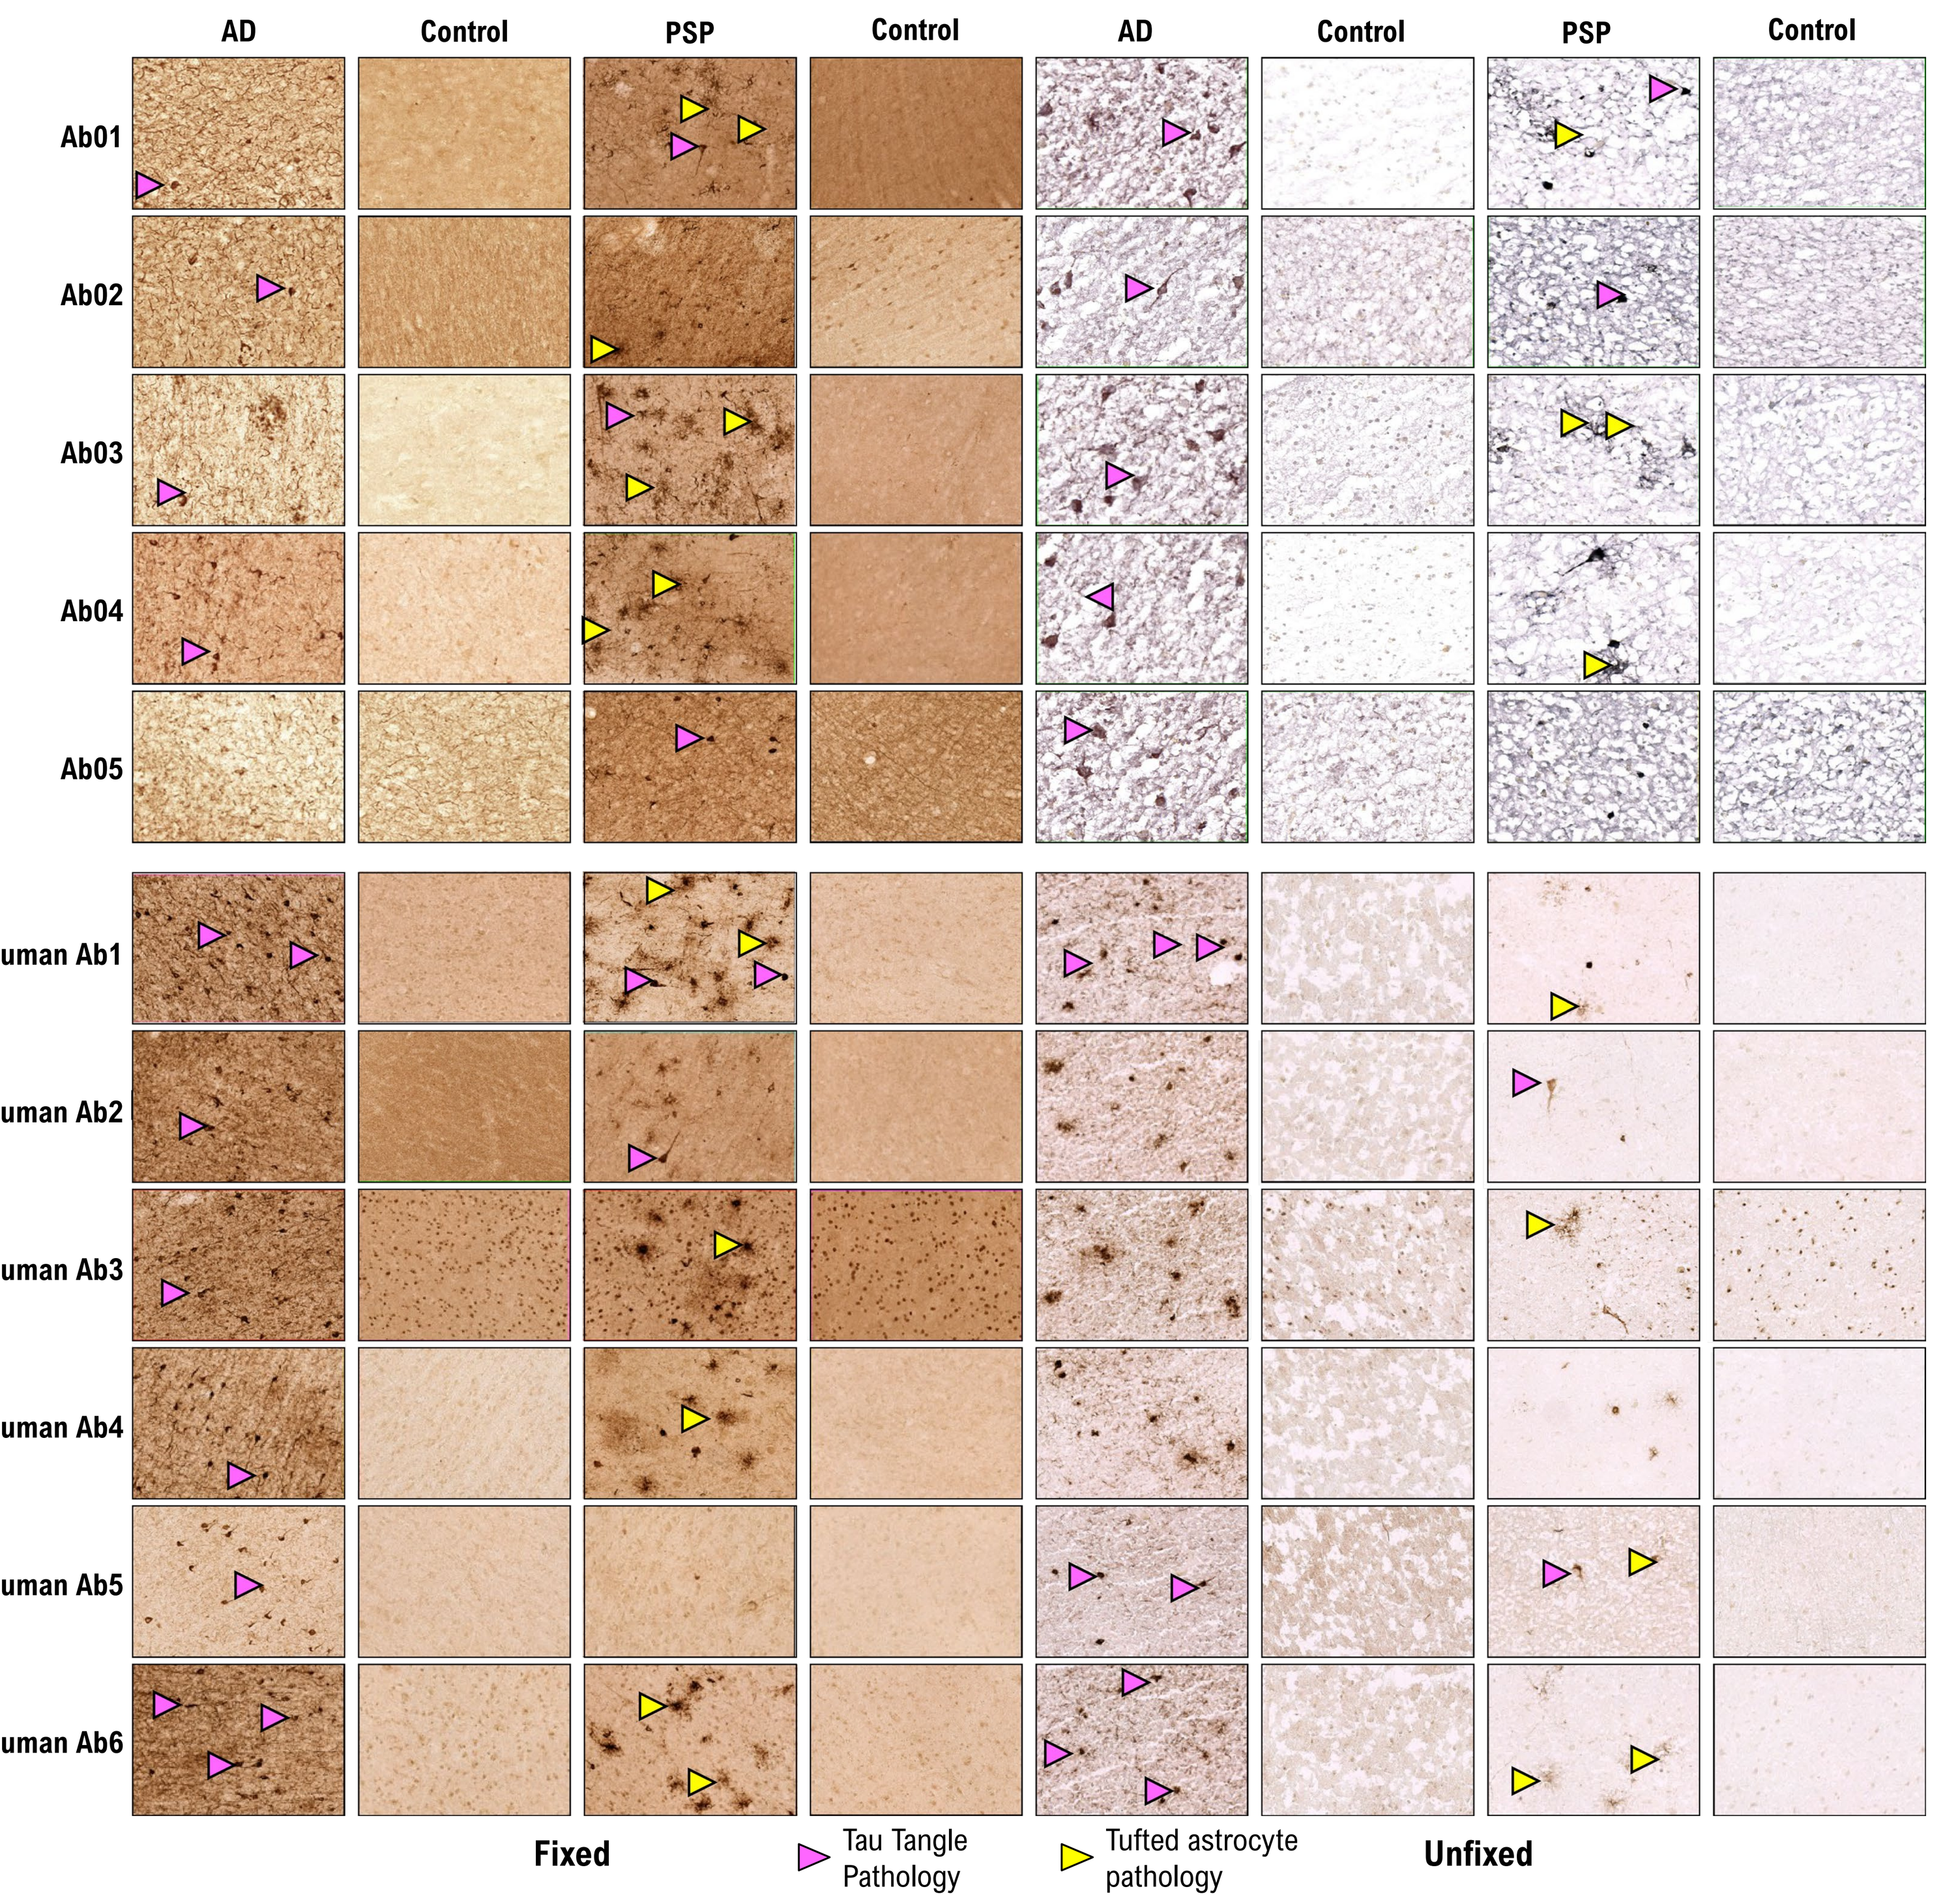

AD
Control
PSP
Control
AD
Control
PSP
Control
Ab01
Ab02
Ab03
Ab04
Ab05
Human Ab1
Human Ab2
Human Ab3
Human Ab4
Human Ab5
Human Ab6
Fixed
Tau Tangle Pathology
Tufted astrocyte pathology
Unfixed

Biophysical characteristics of 11 candidates that were subjected to *in vivo* efficacy study. **A**) Epitope position mapped against 2N4R-human tau Isoform (441 residues). The 11 anti-tau Abs candidates, that were selected for *in vivo* efficacy studies, target diverse location of full-length tau including 6 in mid-domain and 3 in C-terminus Epitope position mapped against 2N4R-human tau isoform (441 residues). **B**) Voyager anti-tau antibodies bind selectively to immunopurified PHF tau with high affinity but not to wild type tau. Antibody was flown over immobilized immunopurified PHF (iPHF) or WT Tau to measure affinity using Surface Plasmon Resonance (SPR) on Biacore 8K instrument, except for Human Ab5 where iPHF was flown over captured antibody. Ab01 and human Ab6 demonstrated high affinity to iPHF but did not bind to wild type tau; Affinity (K_D), including k_{on} and k_{off}, for each of murine and human Abs are listed in the table. **C**) Voyager Abs demonstrated functional inhibition of ePHF-seeding activity *in vitro* with desirable IC_{50}. Cortical sections from patient AD and PSP patients. **D**) Voyager Abs bind specifically to tau pathology on the cortical sections of AD, PSP but not non-AD, non-tauopathy. Note that AD/non-AD or PSP/non-tauopathy cortical sections were purchased from Banner Sun Health Research Institute, Sun City, AZ.

Table 1. Biophysical Properties of Voyager Anti-tau Antibodies

		MURINE ABS					HUMAN ABS					
Property	Criteria[a]	Ab01	Ab02	Ab03	Ab04	Ab05	Human Ab1	Human Ab2	Human Ab3	Human Ab4	Human Ab5	Human Ab6
Selectivity: iPHF:WT rec. Tau*	≥ 50-fold	>838*	>1450*	>54.5*	>80*	>396*	>109*	>382*	>145 *	>108*	>73*	>30*
Selectivity: ePHF:WT rec. Tau*	≥ 50-fold	>222*	>415*	>140*	>123*	Not selective**	>23*	>59*	>40*	>137*	Not selective**	>181*
IHC Fixed - Human AD Brain	positive	Positive	Positive	Positive	Positive	Weak	Positive	Positive	Positive	Positive	Positive	Positive
IHC Fixed - Human Ctl Brain	negative	Negative	Negative	Negative	Negative	Negative	Negative	Negative	Negative (Nu)	Negative	Negative	Negative
IHC Fixed - Human PSP	-	Positive	Positive (Wk)	Positive	Positive	Positive	Positive	Positive	Positive	Positive	Wk Positive	Positive (Nu)
IHC-Fixed – Human Ctl Brain	-	Negative	Negative	Negative	Negative	Negative	Negative	Negative	Negative (Nu)	Negative	Negative	Negative
IHC Frozen - Human AD Brain	-	Positive	Positive	Positive	Positive	Positive	Positive	Positive	Positive	Positive	Wk Positive	Positive
IHC Frozen - Human PSP	positive	Positive	Positive	Positive	Positive	Positive	Positive	wk Positive	Positive	Positive	Wk Positive	Positive
IHC Frozen - Human Ctl Brain	negative	Negative	Negative	Negative	Negative	weak	Negative	Negative	Negative (Nu)	Negative	Negative	Negative
Inhibition of ePHF seeding in Biosensor Cells	≤ 20 nM IC_{50}	18.2	4.8	16.5	18.6	3.0	36.3	9.39	8.29	18.1	36	12.5
Low Polyspecificity (using BVP ELISA)	in range of comp. Abs	√	√	√	√	√	√	√	√	√	√	√
Solution and Colloidal Stability at >10 mg/mL	95% pure by SEC, no particulates	√ b	√ b	√ b	√ b	√ b	√ b	√ b	√ b	√ b	√ b	√ b

* No binding to highest concentration tested; ** not selective, 1:1 binding ; a: dash indicates not a criteria ; b: tested at 1 mg/mL

Summary of biophysical characteristics including selectivity, *in vitro* functional inhibition and developability of Voyager Ab's.

Figure 3. AD-PHF Seeding Model in P301S

A **P301S Hippocampal Seeding Model**

- FTD Mutant Tau TG Animals
- AD derived PHF Seeding into Hippocampus
- Experiment terminated +6 weeks
- Critical Readout: AT8 Immunoreactivity ELISA

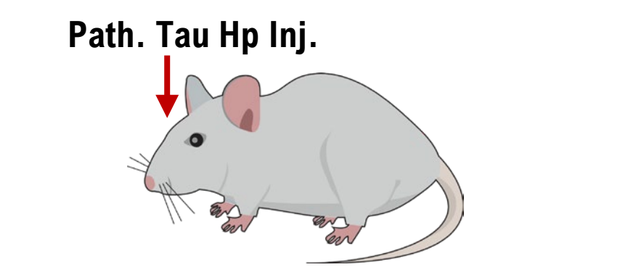

Path. Tau Hp Inj.

B

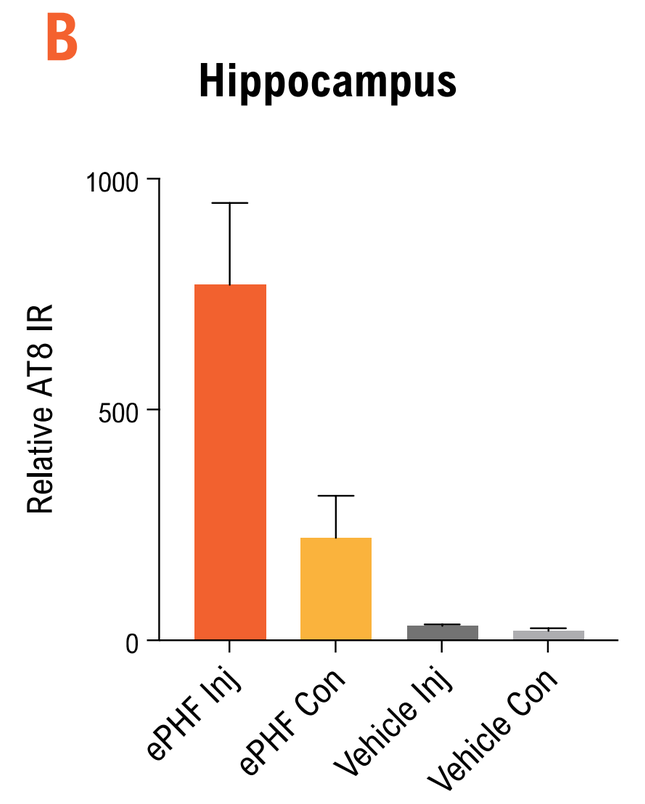

Hippocampus
Relative AT8 IR
ePHF Inj
ePHF Con
Vehicle Inj
Vehicle Con

C

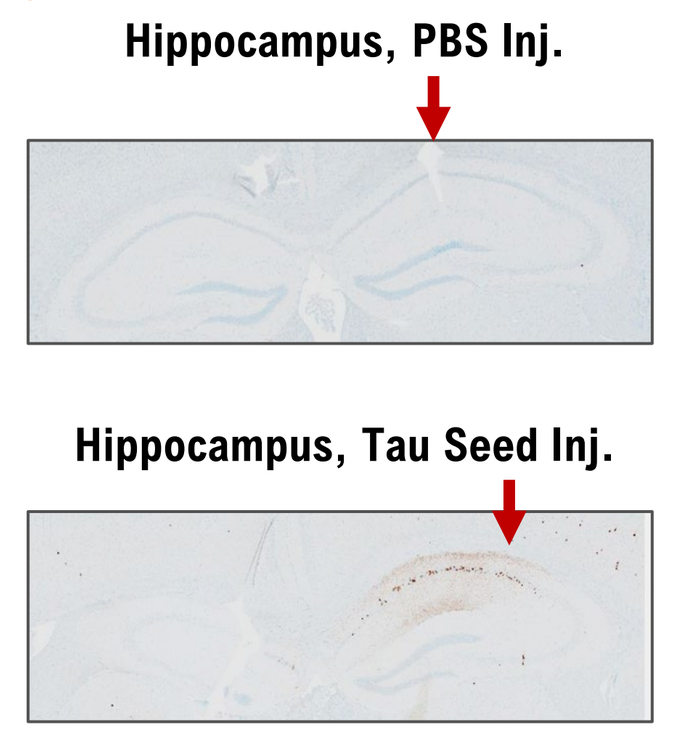

Hippocampus, PBS Inj.
Hippocampus, Tau Seed Inj.

AD-PHF seeded hippocampal P301S model that is used for *in vivo* efficacy study. **A**) An experimental design that is used to generate a seeding model is outlined here. Briefly, AD brain-derived PHFs is injected into the left hippocampus of 8-weeks-old P301S mice. 6 weeks after injection, mice are sacrificed to examine PHF induced tau pathology. **B**) Tau pathology can be detected by AT8 ELISA using brain lysate. Note that a significant increase of AT8 immunoreactivity (IR) is detected in the PHF injected Ipsilateral site but not in vehicle injected site. In the Tau seed injected mice, tau pathology can also be detected in the contralateral site to a lesser extent, indicating tau pathology induced by injected PHF can spread across hippocampus. **C**) Tau pathology can also be detected by IHC staining with AT100 anti-tau antibody (bottom panel, Tau seed inj.). As shown here, a significant number of CA neurons of ipsilateral site are with tau pathology (AT100 positives).

Figure 4. P301S Seeding Model Treated with Murine Antibody Candidates

A

PROPERTY	CRITERIA	Ab01	Ab02	Ab03	Ab04	Ab05
Affinity to iPHF (Biacore)	nM	0.044	0.3	0.058	0.12	<4.4
Affinity to iPHF (Octet)	nM	0.26	0.15	4.01	2.75	0.55
Affinity to WT rec. Tau (Octet)*	(nM)	>218*	>218*	>218*	>218*	>218*
Inhibition of seeding in biosensor Cells with ePHF	≤ 20 nM IC_{50}	18.2*	4.8	16.5*	18.6*	3.0

*No binding on highest concentration tested

B **C**

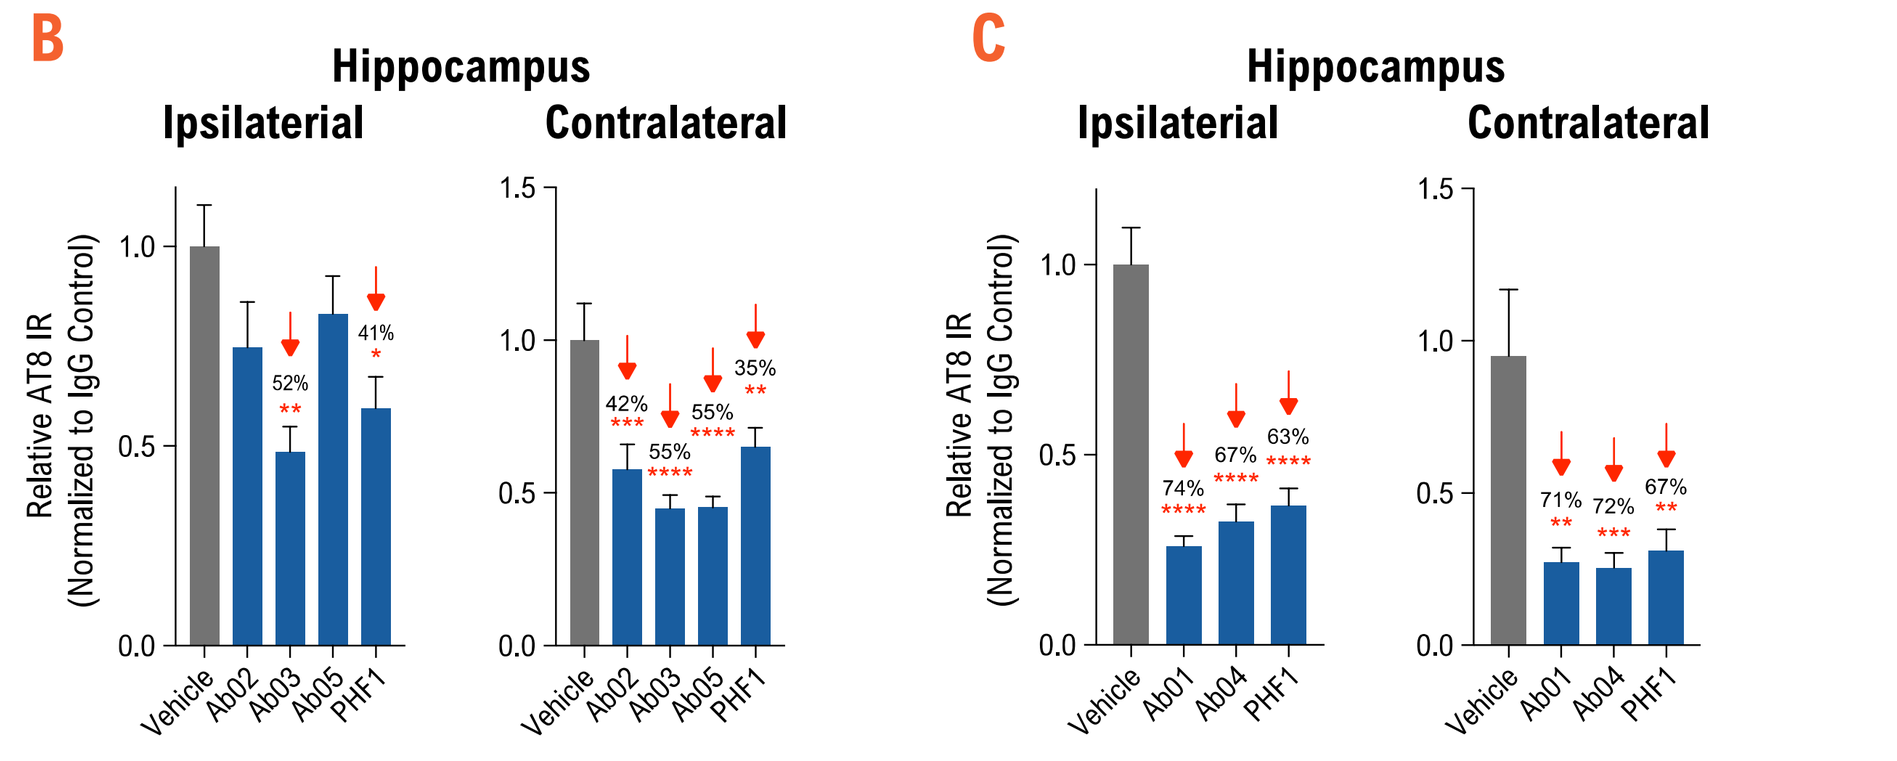

Hippocampus
Ipsilaterial
Contralateral
Relative AT8 IR (Normalized to IgG Control)
52%
41%
42%
55%
55%
35%
Vehicle
Ab02
Ab03
Ab05
PHF1
Hippocampus
Ipsilaterial
Contralateral
74%
67%
63%
71%
72%
67%
Vehicle
Ab01
Ab04
PHF1

Efficacy of 5 murine Abs were examined in the P301S seeding model described in Figure 4. To test efficacy of a given Ab, the treatment started one week before surgery (2 doses). There were 5 additional doses that were applied weekly after surgery. At the end of six week after surgery, hippocampi of each animal were isolated for AT8 ELISA. **A**) Biophysical properties of 5 murine antibodies are listed on the top. **B**) 3 murine antibodies, Ab02, Ab03 and Ab05, were tested for their activity in the seeding model. Tau pathology from each treatment group was normalized to vehicle control. As shown here, there is a significant reduction of tau pathology in both ipsil- and contra-lateral site of hippocampi of Ab03-treated group. **C**) Two other murine antibodies, Ab01 and Ab04, were also tested for their activity in the seeding model. There is a significant reduction of tau pathology in both ipsil- and contra-lateral site of hippocampi from mice treated with Ab01 or Ab04. Note that PHF1 was used as a positive control for both experiments.

Figure 5. Reduction of Pathological Tau Spreading from Ipsilateral to Contralateral Hippocampus is Observed in the Mice Treated with Ab01, Ab03 and Ab04

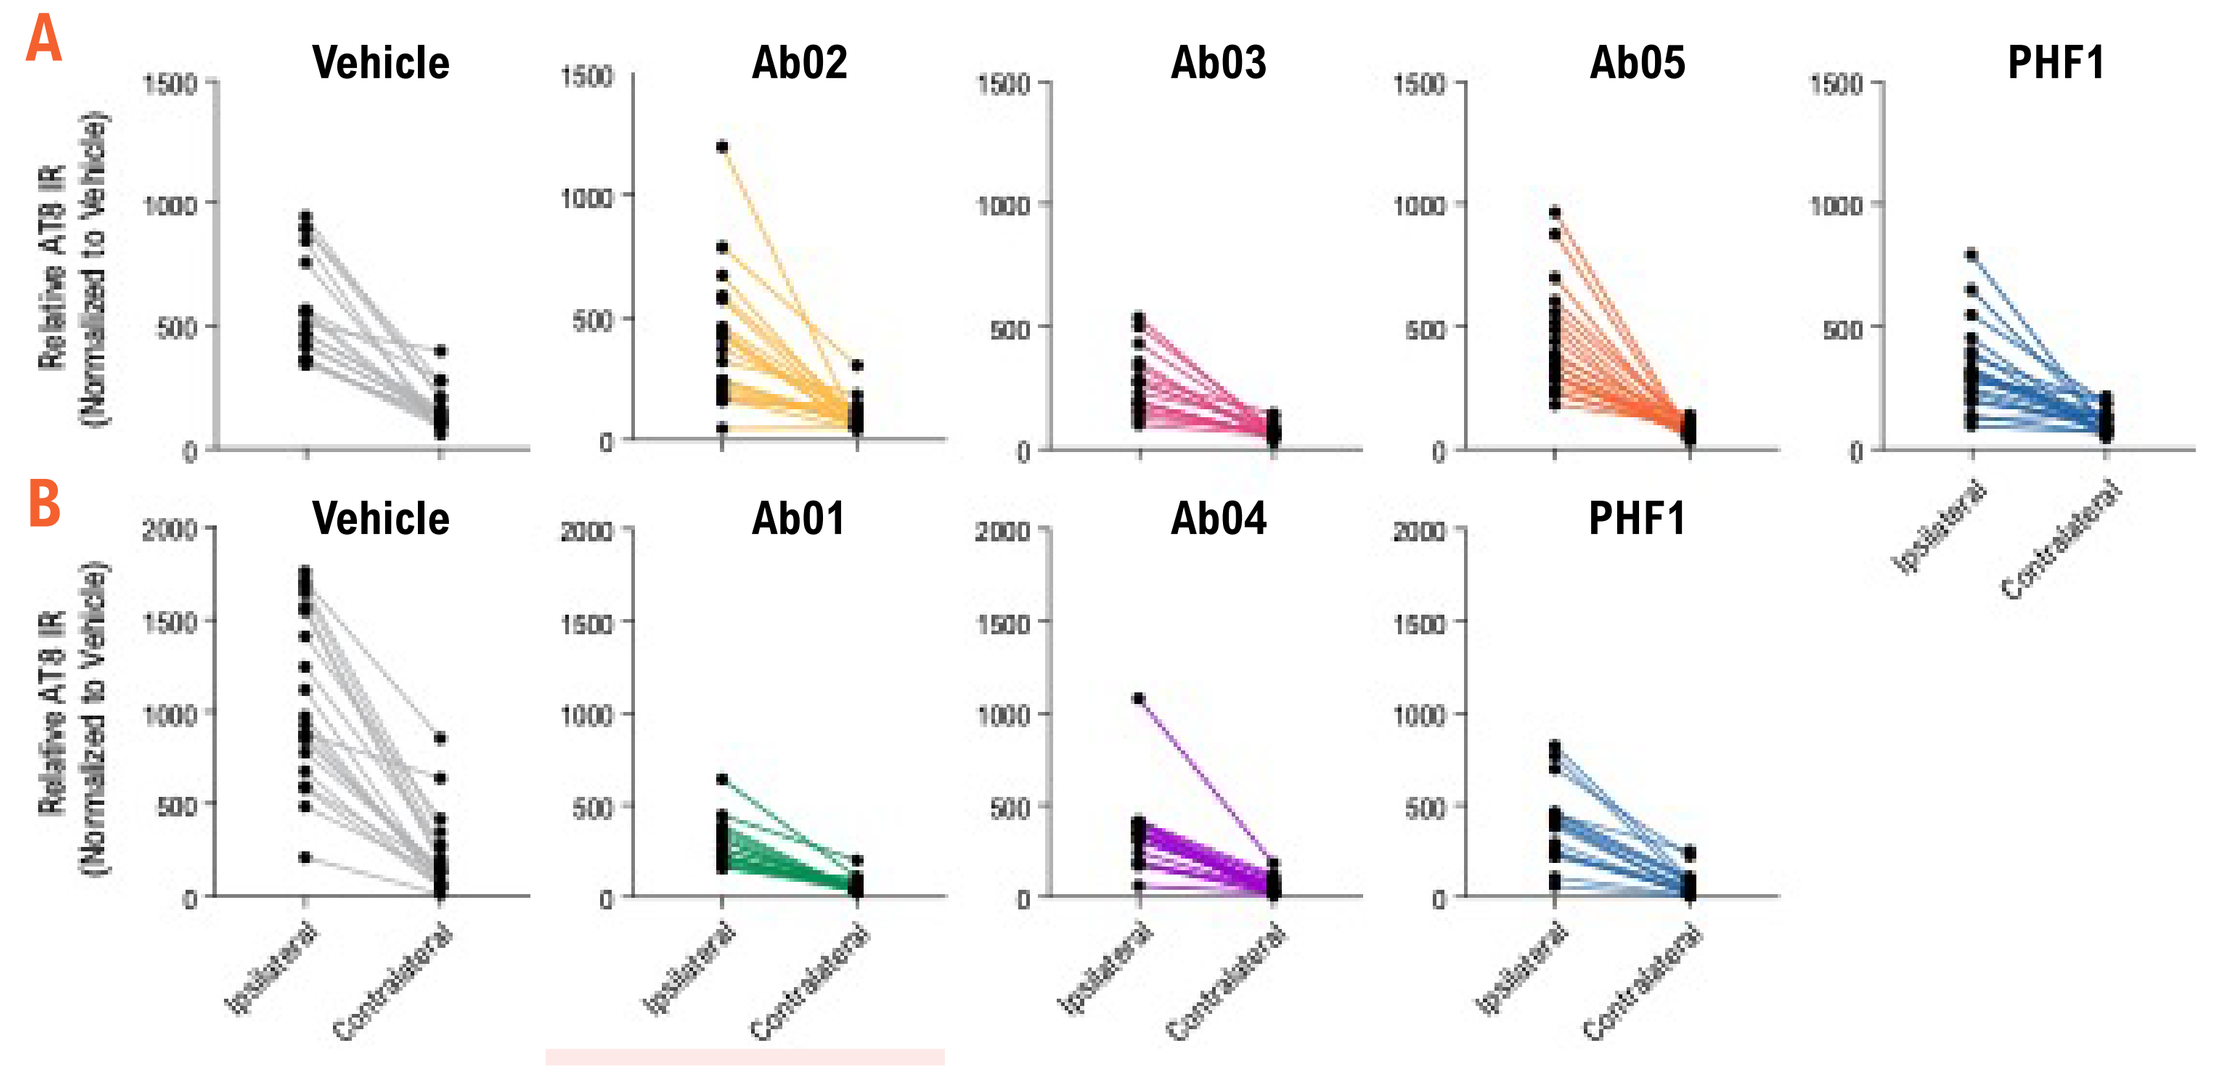

A
Vehicle
Ab02
Ab03
Ab05
PHF1
B
Vehicle
Ab01
Ab04
PHF1
Relative AT8 IR (Normalized to Vehicle)
Ipsilateral
Contralateral

Spaghetti plots of tau pathology from ipsilateral to contralateral hippocampus were made using data derived from Figure 4 B (**A**) and Figure 4 C (**B**). Data demonstrated that there is robust reduction of tau pathology spreading across hippocampus from the mice treated with Ab01, Ab03, Ab04 and Ab05 with Ab01 demonstrating best efficacy.

Figure 6. P301S Seeding Model Treated with Human Antibody Candidates

A

PROPERTY	CRITERIA	HUMAN Ab1	Ab2	Ab3	Ab4	Ab5
Affinity to iPHF (Biacore, nM)		0.6	11.6	0.36	0.24	TBD
Affinity to iPHF (Octet, nM)		2	0.57	1.5	2	3
Affinity to WT rec. Tau (Octet, nM)*		>218*	>218*	>218*	>218*	>218*
Inhibition of seeding in biosensor Cells with ePHF	≤ 20 nM IC_{50}	36.3	9.4	8.3	18.1	36

*No binding on highest concentration tested

B **C**

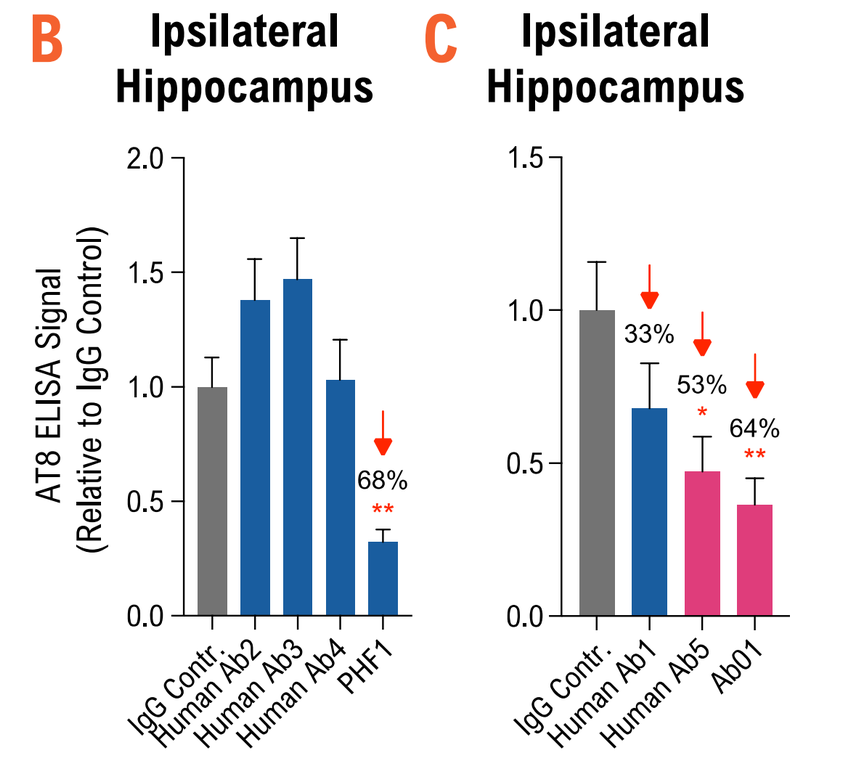

Ipsilateral Hippocampus
AT8 ELISA Signal (Relative to IgG Control)
68%
IgG Contr.
Human Ab2
Human Ab3
Human Ab4
PHF1
Ipsilateral Hippocampus
33%
53%
64%
IgG Contr.
Human Ab1
Human Ab5
Ab01

Efficacy of 5 human Abs were examined in the P301S seeding model using the protocol described in Figure 4. **A**) Biophysical properties of human antibodies are listed on the top. **B**) 3 human antibodies, Human Ab2, Human Ab3 and Human Ab4, were tested for their activity in the seeding model. Tau pathology from each treatment group was normalized to the IgG control. As shown here, there is no significant reduction of tau pathology detected in the ipsilateral site of hippocampus of these three treatment groups. **C**) Two other human antibodies, Human Ab1 and Human Ab5, were tested for their activity in the seeding model. There is a significant reduction of tau pathology in the ipsilateral site of hippocampus from mice treated with Human Ab5. Additionally, there is a trend of reduction of tau pathology (33%) in mice treated with human Ab1. Note that Ab01 was used as a positive control for both experiments.

Table 2. Summary of Voyager Anti-tau Ab's - Activity/efficacy *in vivo*

ANTIBODY	SPECIES	AFFINITY BY BIACORE (NM)	IPSILATERAL EFFICACY (REDUCTION OF AT8 IR VS VEHICLE OR IGG CONTR)	CONTRALATERAL EFFICACY (REDUCTION OF AT8 IR VS VEHICLE)
Ab01	Mouse	<0.04	74% or 64-70%*	71%*
Ab02	Mouse	0.3	no	43%*
Ab03	Mouse	<0.06	52%*	55%*
Ab04	Mouse	0.12	67%*	72%*
Ab05	Mouse	<5	no	55%*
Human Ab1	Human	0.6	33%	TBD
Human Ab2	Human	11	no	no
Human Ab3	Human	0.36	no	no
Human Ab4	Human	0.24	no	no
Human Ab5	Human		53%*	TBD
Human Ab6	Human	0.3	TBD	TBD
PHF1	Mouse	0.16	47-68%*	35-67%*

10 voyager antibodies' performance in inhibiting tau seeding and spreading *in vivo*.

CONCLUSION

We have generated a set of potent and specific anti-tau antibodies targeting a diverse set of phospho-tau epitopes that are candidates for the treatment of Alzheimer's disease and/or other tauopathies.

64 Sidney Street, Cambridge, MA 02139, USA

Alzheimer's Association International Conference 2022 | July 31 – August 4, 2022 | San Diego, CA