P015

Dose-response Evaluation of 9P801, an Engineered AAV Capsid with High BBB Penetration and CNS Transduction in Non-human Primates

M. Nonnenmacher, J. Thompson, N. Pande, M. Hefferan, K. Grant, J. Clement, K. Tyson, J. Laguna-Torres, A. Kulkarni, T. Fiore, J. Natasan, T.C. Carter
Voyager Therapeutics Inc., 64 Sidney Street, Cambridge, MA 02139, USA

voyager THERAPEUTICS

INVESTORS& MEDIA

CAREERS AT VOYAGER

SUMMARY

- TRACER-NHP™ screening platform identified AAV9 variant 9P801 as a lead capsid for primate BBB crossing and CNS transduction following intravenous dosing
- Laboratory-grade 9P801 showed highly promising performance (>2-log increase vs AAV9) in a single-dose study (2e13 VG/kg)
- We applied scalable HEK-293 manufacturing to 9P801 for a descending dose study in NHP
- Transduction properties were similar or better than capsids produced by lab-grade manufacturing, showing widespread distribution in various brain regions with a strong neuronal tropism
- 9P801 showed very widespread CNS transduction at high- and medium-doses, and easily detectable transduction at low dose (≤ 2e12 VG/kg)
- Doses of 2e12 VG/kg and higher were sufficient to achieve supra-physiological mRNA expression of a model transgene

ABSTRACT

Engineering of gene therapy vectors has emerged as an exciting strategy to enable clinically relevant expression levels of a therapeutic gene at vector doses low enough to avoid undesirable events. Using TRACER™ RNA-driven directed evolution of Adeno-Associated Virus (AAV) capsids, we have recently generated 9P801, an AAV capsid variant with an unprecedented capacity for CNS transduction in adult non-human primates (NHPs) following intravenous delivery. Here we attempted to determine the minimal dose of 9P801 vector sufficient for near-physiological expression of a therapeutic payload in the CNS of adult macaques via systemic delivery. A 9P801 vector containing a hemagglutinin-tagged NHP protein under a ubiquitous promoter was injected to male NHPs at various doses spanning a 30-fold range for a duration of 28 days. Widespread transgene protein expression was detected in the spinal cord and the brain of high- and medium-dose animals, especially in the putamen, thalamus, globus pallidus and brainstem. Viral DNA and mRNA were readily detectable in all animals and showed a consistent dose response. Strikingly, the lowest dose of 9P801 allowed higher mRNA and protein expression than a 30-fold higher dose of AAV9. Comparison of transgene mRNA with the matching endogenous transcript indicated that a dose of 2e12VG/kg was sufficient to achieve supraphysiological levels in the CNS, while showing low transduction in the liver and the dorsal root ganglia (DRG). Together, our data suggest that engineered AAV vectors have the potential to achieve a large improvement of their therapeutic index by retaining strong efficacy at low dose.

METHODS

- Cynomolgus macaques (n=3 per dosing group) were dosed intravenously with 9P801 capsid containing a single-stranded primate transgene fused to an HA tag under a CBA promoter.
- At day 28 post-injection, tissues were harvested and split for IHC analysis or bioanalytical readouts.
- IHC was performed with an anti-HA antibody on brain coronal slices and spinal cord transversal sections.
- Vector DNA levels and transgene mRNA expression were analyzed by ddPCR and RT-qPCR, respectively. mRNA was measured with 1) a transgene-specific probe or 2) a CDS probe detecting both transgenic and endogenous mRNA. DNA and mRNA were normalized to housekeeping genes.

Figure 1. **IHC Detection of Transgene Protein in Cynomolgus Macaque**

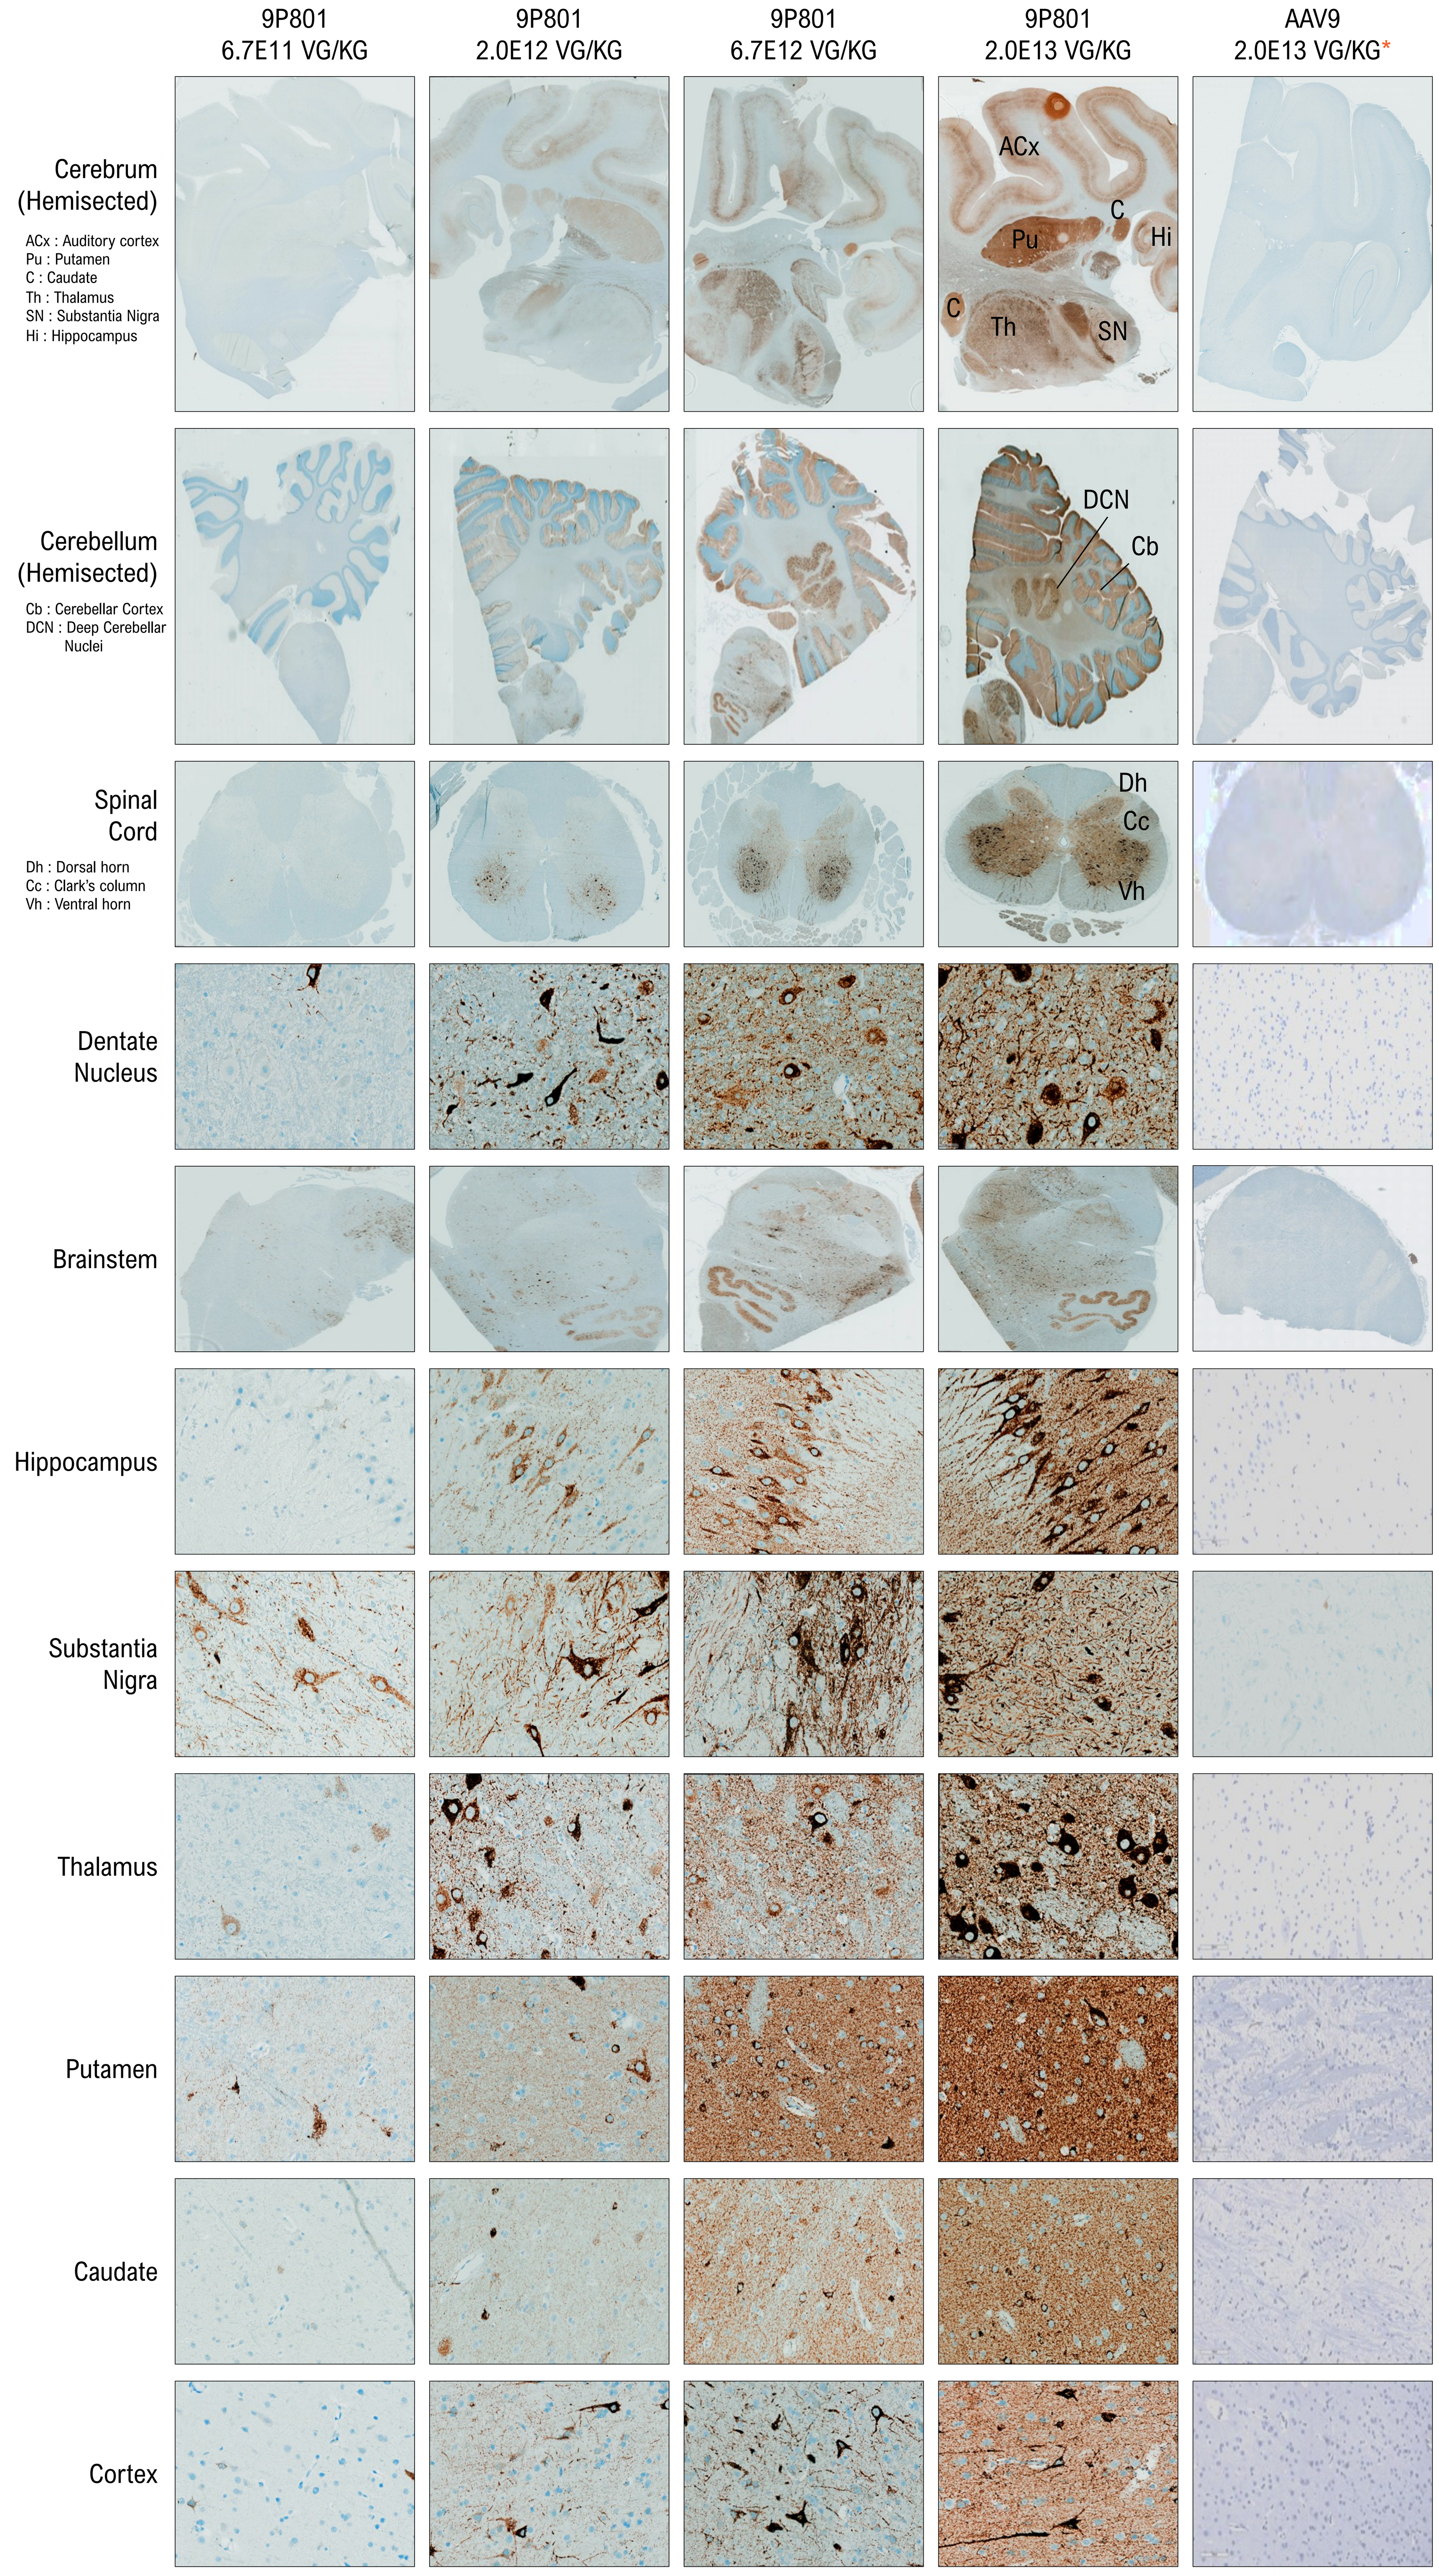

9P801 6.7E11 VG/KG
9P801 2.0E12 VG/KG
9P801 6.7E12 VG/KG
9P801 2.0E13 VG/KG
AAV9 2.0E13 VG/KG*
Cerebrum (Hemisected)
ACx : Auditory cortex
Pu : Putamen
C : Caudate
Th : Thalamus
SN : Substantia Nigra
Hi : Hippocampus
ACx
Pu
C
Hi
Th
SN
Cerebellum (Hemisected)
Cb : Cerebellar Cortex
DCN : Deep Cerebellar Nuclei
DCN
Cb
Spinal Cord
Dh : Dorsal horn
Cc : Clark's column
Vh : Ventral horn
Dh
Cc
Vh
Dentate Nucleus
Brainstem
Hippocampus
Substantia Nigra
Thalamus
Putamen
Caudate
Cortex

*Historical data - 2 weeks post-dosing

Figure 2. **Detection of Viral Genomes and Transgene mRNA**

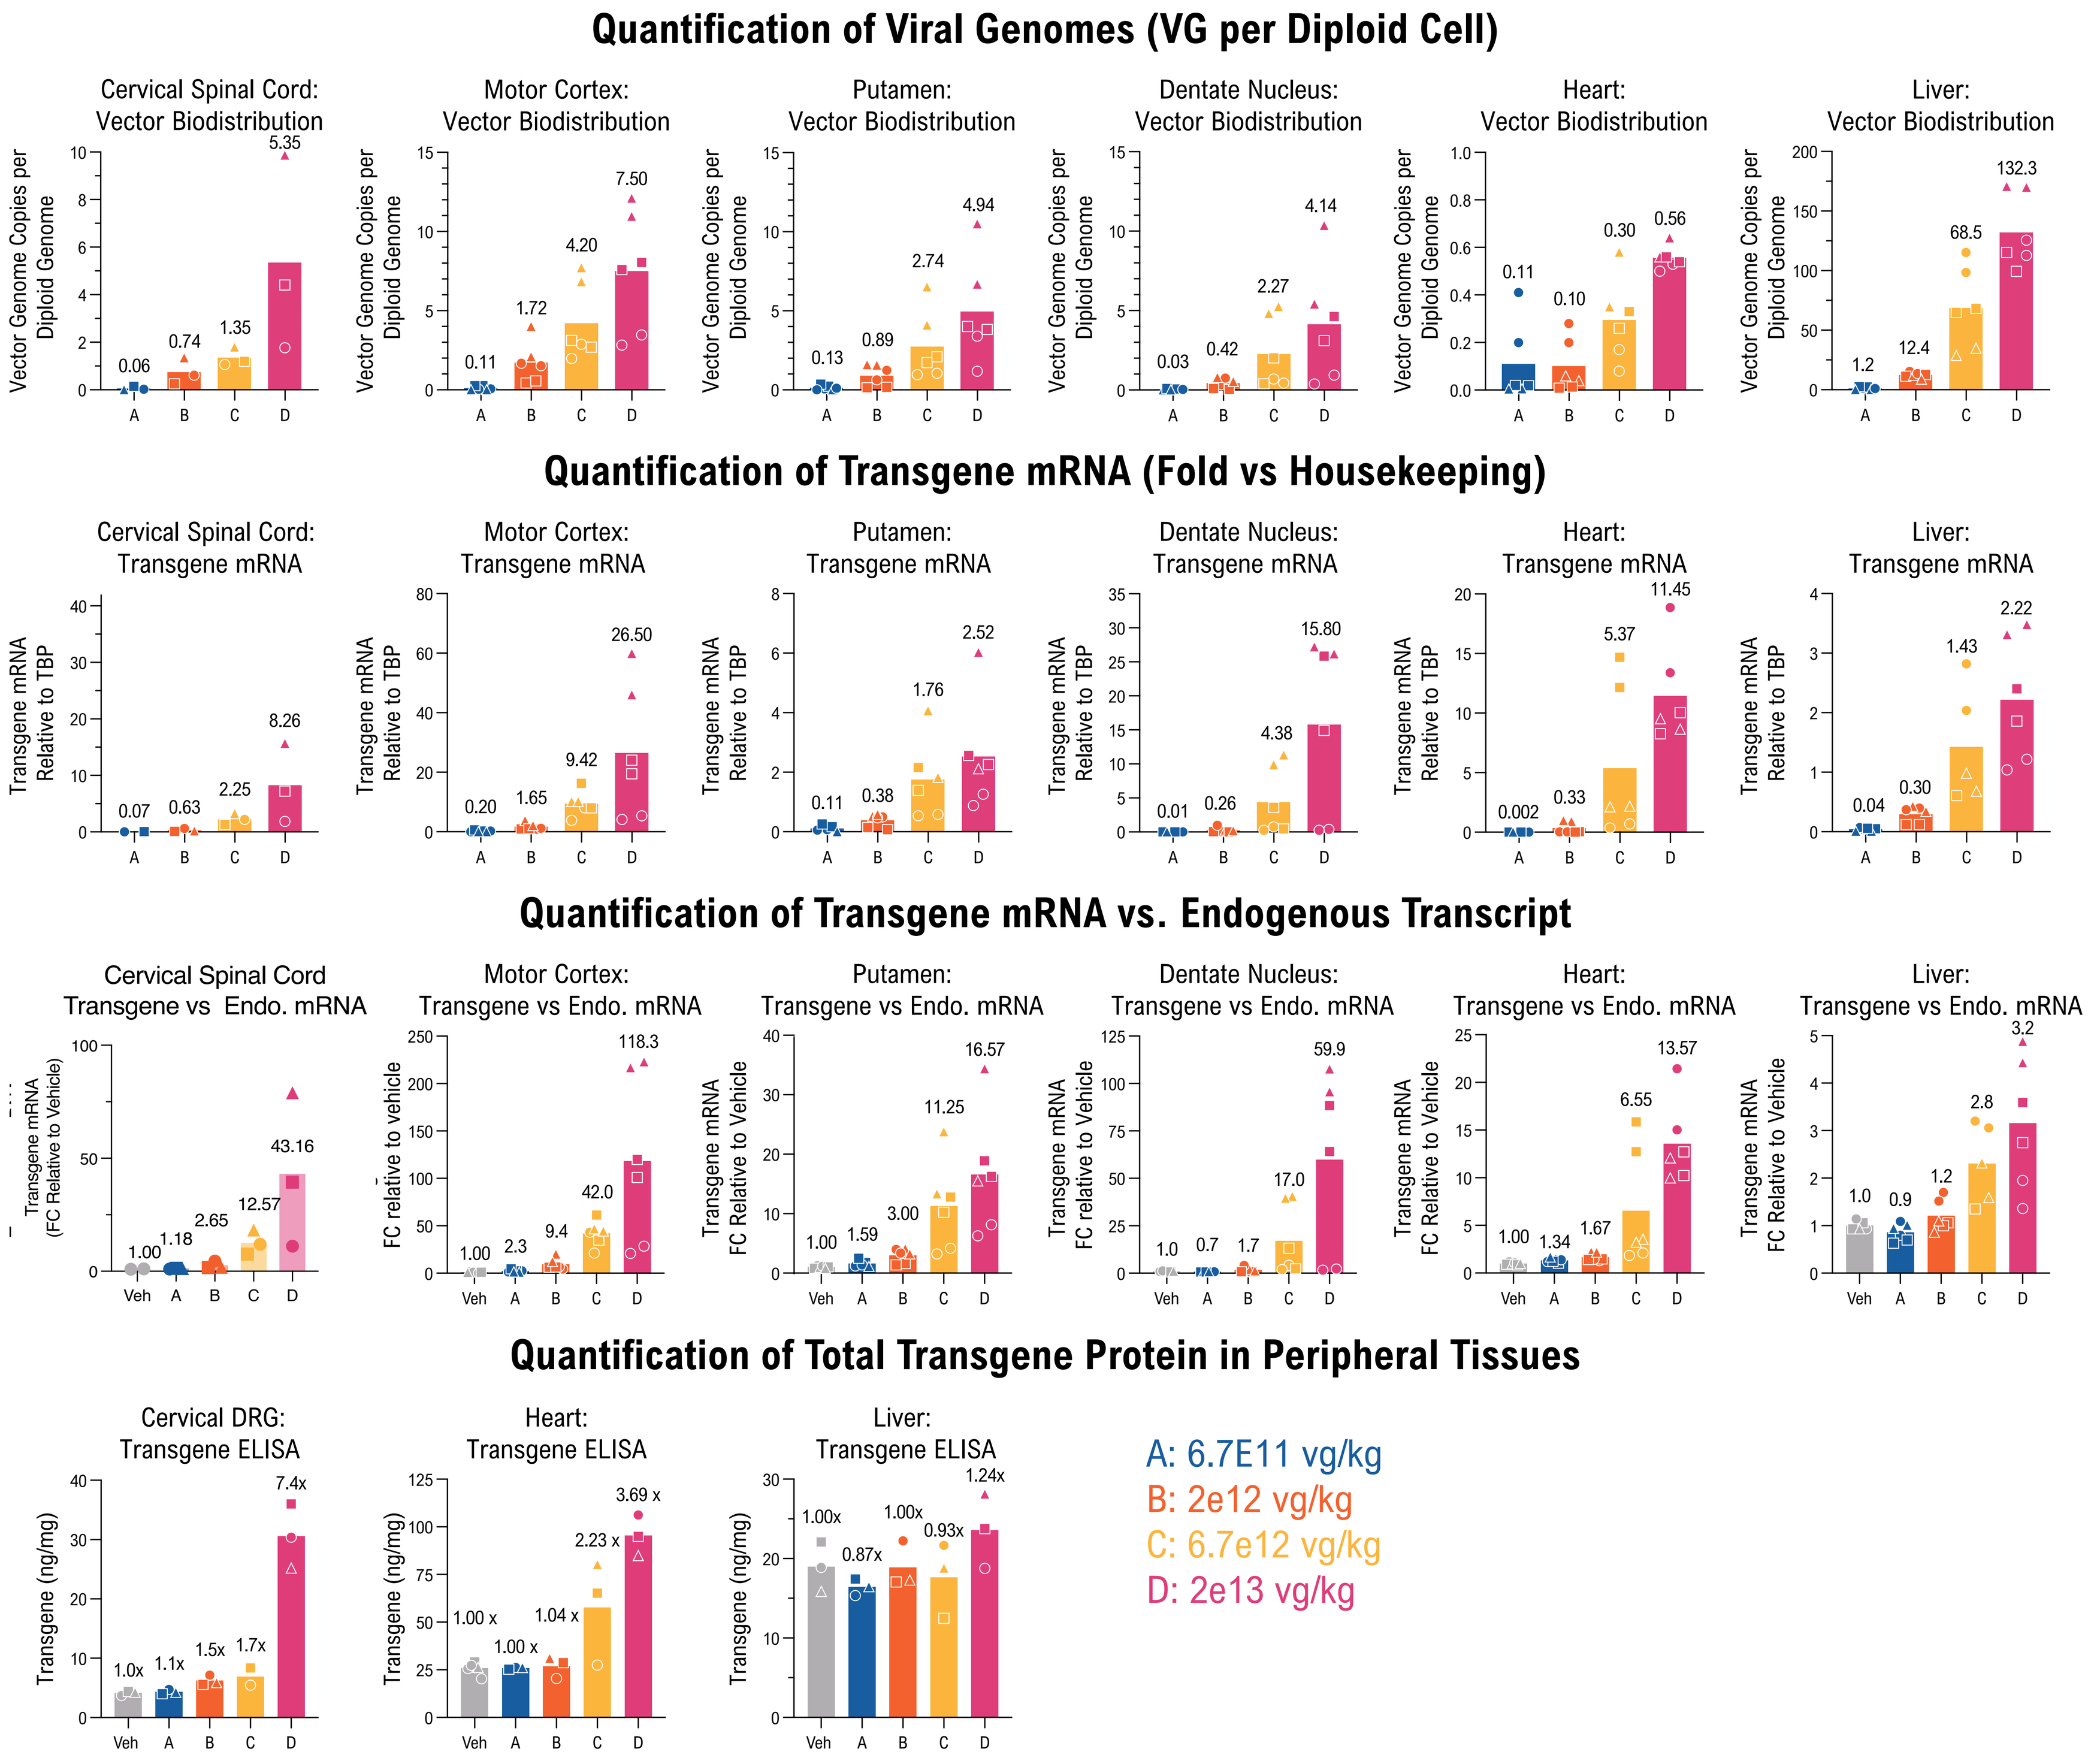

Quantification of Viral Genomes (VG per Diploid Cell)
Cervical Spinal Cord: Vector Biodistribution
0.06 0.74 1.35 5.35
Motor Cortex: Vector Biodistribution
0.11 1.72 4.20 7.50
Putamen: Vector Biodistribution
0.13 0.89 2.74 4.94
Dentate Nucleus: Vector Biodistribution
0.03 0.42 2.27 4.14
Heart: Vector Biodistribution
0.11 0.10 0.30 0.56
Liver: Vector Biodistribution
1.2 12.4 68.5 132.3
Vector Genome Copies per Diploid Genome
Quantification of Transgene mRNA (Fold vs Housekeeping)
Cervical Spinal Cord: Transgene mRNA
0.07 0.63 2.25 8.26
Motor Cortex: Transgene mRNA
0.20 1.65 9.42 26.50
Putamen: Transgene mRNA
0.11 0.38 1.76 2.52
Dentate Nucleus: Transgene mRNA
0.01 0.26 4.38 15.80
Heart: Transgene mRNA
0.002 0.33 5.37 11.45
Liver: Transgene mRNA
0.04 0.30 1.43 2.22
Transgene mRNA Relative to TBP
Quantification of Transgene mRNA vs. Endogenous Transcript
Cervical Spinal Cord Transgene vs Endo. mRNA
1.00 1.18 2.65 12.57 43.16
Motor Cortex: Transgene vs Endo. mRNA
1.00 2.3 9.4 42.0 118.3
Putamen: Transgene vs Endo. mRNA
1.00 1.59 3.00 11.25 16.57
Dentate Nucleus: Transgene vs Endo. mRNA
1.0 0.7 1.7 17.0 59.9
Heart: Transgene vs Endo. mRNA
1.00 1.34 1.67 6.55 13.57
Liver: Transgene vs Endo. mRNA
1.0 0.9 1.2 2.8 3.2
FC Relative to Vehicle
Quantification of Total Transgene Protein in Peripheral Tissues
Cervical DRG: Transgene ELISA
1.0x 1.1x 1.5x 1.7x 7.4x
Heart: Transgene ELISA
1.00 x 1.00 x 1.04 x 2.23 x 3.69 x
Liver: Transgene ELISA
1.00x 0.87x 1.00x 0.93x 1.24x
Transgene (ng/mg)
Veh A B C D
A: 6.7E11 vg/kg
B: 2e12 vg/kg
C: 6.7e12 vg/kg
D: 2e13 vg/kg

Figure 3. **Quantification* and Characterization of Transduced Cells in NHP**

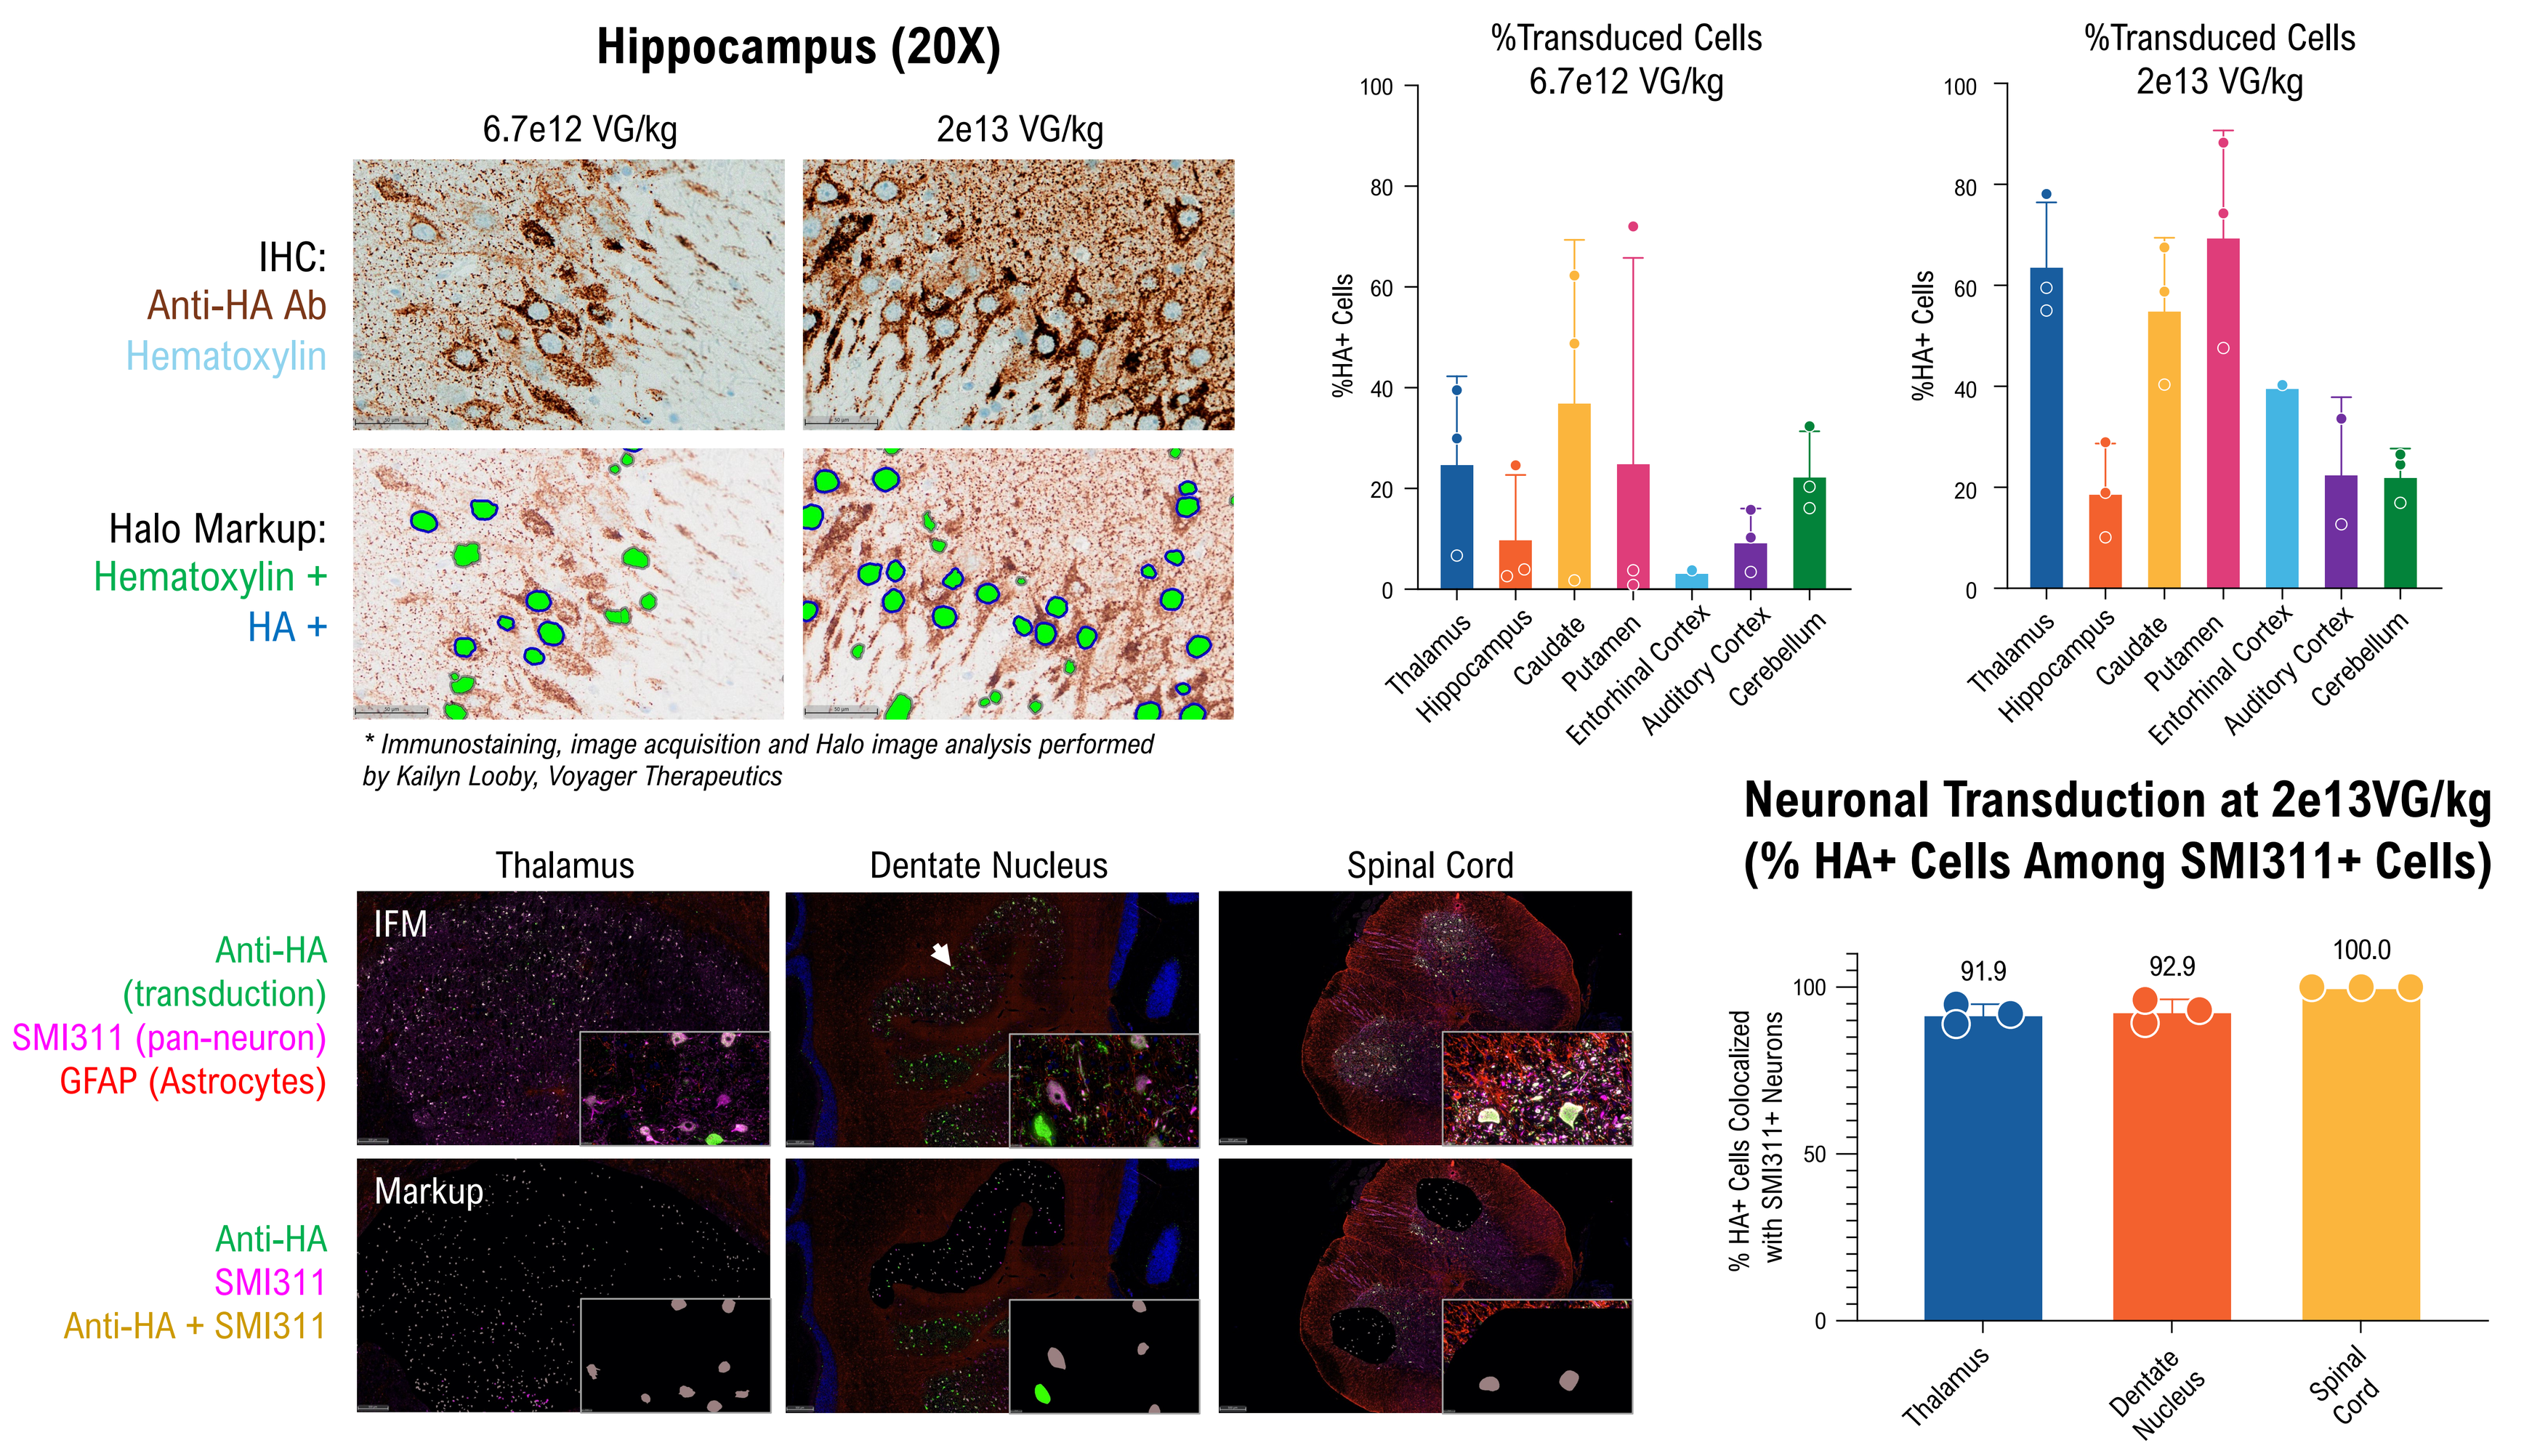

Hippocampus (20X)
6.7e12 VG/kg
2e13 VG/kg
IHC:
Anti-HA Ab
Hematoxylin
Halo Markup:
Hematoxylin +
HA +
%Transduced Cells 6.7e12 VG/kg
%Transduced Cells 2e13 VG/kg
%HA+ Cells
Thalamus
Hippocampus
Caudate
Putamen
Entorhinal Cortex
Auditory Cortex
Cerebellum
Thalamus
Dentate Nucleus
Spinal Cord
Anti-HA (transduction)
SMI311 (pan-neuron)
GFAP (Astrocytes)
IFM
Markup
Anti-HA
SMI311
Anti-HA + SMI311
Neuronal Transduction at 2e13VG/kg (% HA+ Cells Among SMI311+ Cells)
% HA+ Cells Colocalized with SMI311+ Neurons
91.9
92.9
100.0

* Immunostaining, image acquisition and Halo image analysis performed by Kailyn Looby, Voyager Therapeutics

CONCLUSIONS

- The engineered NHP BBB-penetrant capsid 9P801 is compatible with scalable manufacturing.
- Intravenous delivery of 9P801 achieved widespread CNS transduction in adult NHPs at doses of 2e12VG per kg and above.
- Supraphysiological levels of transgene mRNA were achieved at doses of 2e12 VG per kg and above.
- A dose of 2e13 VG/kg was sufficient to transduce **>40% of total cells** in highly permissive brain regions (thalamus, caudate, putamen) and >20% total cells in less permissive regions (entorhinal cortex, auditory cortex, hippocampus).
- A dose of 2e13 VG/kg achieved transduction of **>90% SMI311-positive neurons** in the thalamus, dentate and spinal cord.

voyager THERAPEUTICS 64 Sidney Street, Cambridge, MA 02139, USA
ESGCT - 29th Congress | October 11 – 14, 2022 | Edinburgh