Intravenous Delivery of AAV Gene Therapy for the Treatment of SOD1-ALS Provides Broad SOD1 Lowering in NHP

Michael D. Grannan, Tyler Moyer, Margaret R. Dalrymple, Ambreen Sayed-Zahid, Xiao Ren, Chiso Egbuchulam, Amy Bruce, Anupriya Kulkarni, Wenya Yang, Joe Clement, Jeyashree Natasan, Dhiral Shah, Ruohong Zhou, Dillon Kavanagh, Kyle Grant, Jeffrey Thompson, Nilesh Pande, Beth Mellen, Naomi Muse, Crystal Taveras-Cui, Vikram Arora, Chanchal Sadhu, Dinah W.Y. Sah, Mathieu Nonnenmacher, Rajeev Sivasankaran, Todd Carter

Voyager Therapeutics Inc., Lexington, MA, USA

1647

SUMMARY

- Mutations in SOD1 are responsible for a toxic gain of function and cause familial SOD1 ALS
- Tofersen is an intrathecally delivered SOD1 ASO approved for the treatment of SOD1 ALS (USA), validating SOD1 reduction as a therapeutic target
- Using a capsid suitable for IV delivery in rodents, we have demonstrated robust knockdown of SOD1 in all levels of the spinal cord resulting in significant improvements in motor performance and survival in a mouse model of SOD1 ALS
- In primates, VY9323, an AAV gene therapy combining a highly potent pri-amiRNA against SOD1 with an intravenous (IV)-delivered, blood-brain barrier-penetrant TRACER™ capsid allowing for one time administration, provided substantial vector genome delivery to the spinal cord and motor cortex
- An IND submission is expected in 2025

INTRODUCTION

Mutations in superoxide dismutase 1 (SOD1) result in progressive motor neuron loss through toxic gain-of-function properties and are responsible for up to 20% of familial amyotrophic lateral sclerosis (ALS), or approximately 2% of all ALS patients in the U.S. Studies evaluating different strategies for SOD1 reduction have demonstrated reduced neuropathology, improved motor behavior, and extension of survival in transgenic mice expressing mutant SOD1. The recent FDA approval of an antisense oligonucleotide targeting SOD1 has further validated SOD1 as a therapeutic target. While approaches to reduce SOD1 have demonstrated varying degrees of efficacy, they rely on direct CNS administration that fails to achieve the broad, CNS-wide SOD1 reduction that may be necessary for maximal therapeutic benefit. We previously reported the results from a series of *in vitro* and *in vivo* studies demonstrating SOD1 reduction following IV delivery of an AAV gene therapy targeting SOD1. In the G93A mouse model of disease, we demonstrated robust SOD1 knockdown throughout the rostral-caudal extent of the spinal cord, significant improvements in motor performance, and survival extension beyond what has previously been reported with intraparenchymal, intrathecal, or intracisternal delivery. In the current studies, we combined a highly potent siRNA against SOD1 with an IV-delivered, blood-brain barrier-penetrant TRACER™ capsid for evaluation in NHPs. Following a 2-month in-life period, we observed favorable biodistribution to the spinal cord and motor cortex which resulted in significant reduction of SOD1 mRNA. The enhanced BBB-penetrance and natural peripheral tissue detargeting inherent to the novel capsid enabled efficacy at a lower dose than typically used in intravenous AAV delivery, resulting in a favorable safety profile. These results demonstrate that the combination of a potent SOD1 RNAi transgene with a novel TRACER™ capsid produces significant reduction of SOD1 mRNA in critical spinal cord and brain regions impacted in ALS and support its continued development and advancement into the clinic.

Figure 1. SOD1 Knockdown Rationale and SOD1 siRNA Selection

Mutant SOD1 is Causal for Disease:

- >180 mutations in SOD1 gene linked to human disease
- Mutant SOD1 forms toxic aggregates that impair multiple cell functions, and result in dysfunction and degeneration of motor neurons

Lowering of SOD1 Provides Therapeutic Benefit in Humans:

- Partial lowering of SOD1 provides potential therapeutic benefit and is well-tolerated in animal models, providing a useful tool to test our therapeutic modality
- Reduction of SOD1 by tofersen results in NfL reduction and potential therapeutic benefit in humans. NfL is a marker of neuron and axonal degeneration and is prognostic for survival and function in ALS

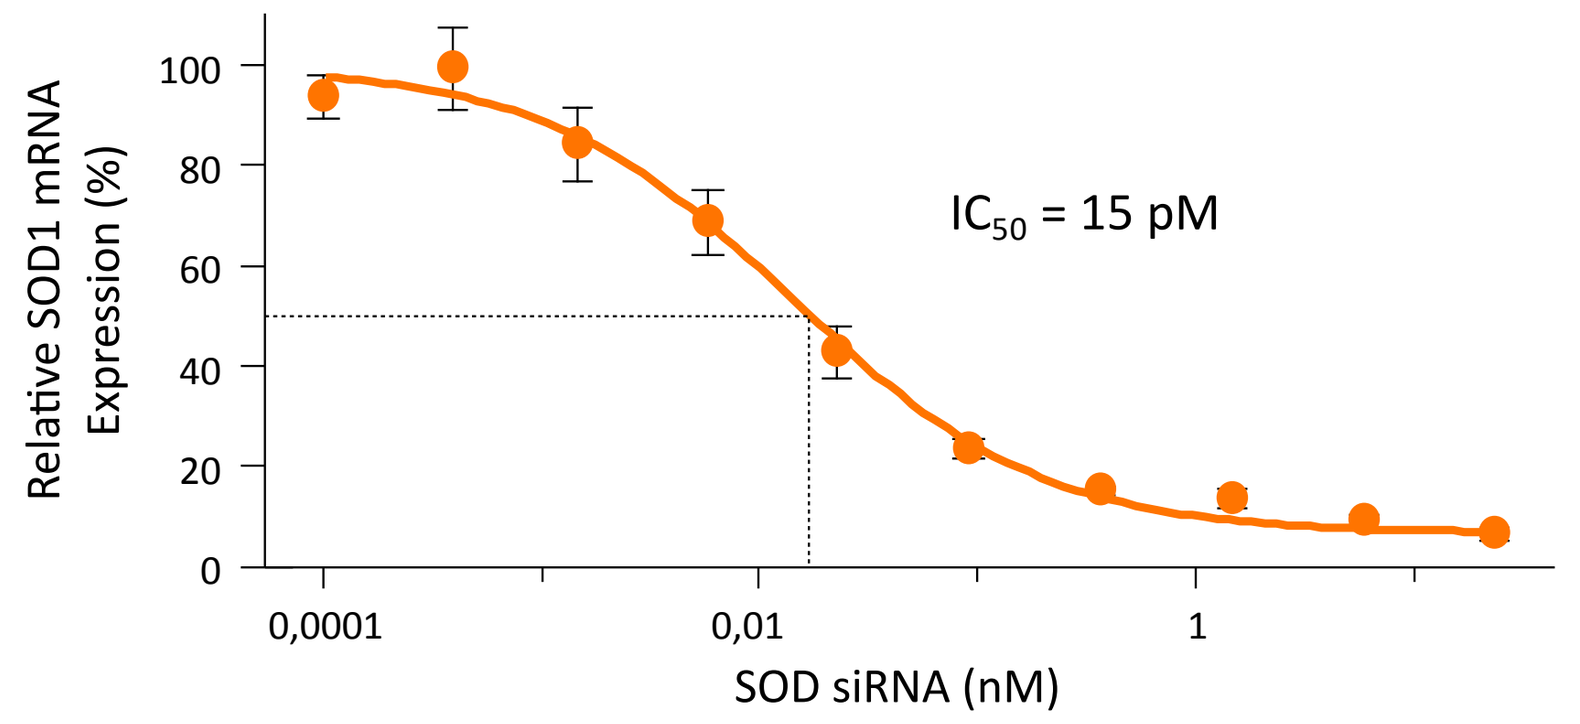
Relative SOD1 mRNA Expression (%)
100
80
60
40
20
0
IC_{50} = 15 pM
0,0001
0,01
1
SOD siRNA (nM)

SOD1 mRNA silencing by a potent siRNA *in vitro*. 169 sequences selectively targeting SOD1 were designed, synthesized and evaluated in HeLa cells. 24 hr after transfection of 100 pM SOD1 siRNA, cells were harvested, and SOD1 and GAPDH mRNA were quantified by RT-qPCR. Dose response curve of the lead candidate is depicted. Error bars indicate SD.

SOD1 G93A Efficacy Study

Mice: ~56-day old B6SJL-Tg(SOD1*G93A)1Gur/J (male and female)

Vector: VOY101.SOD1 siRNA

Dose: 100µl, IV administration 2e12, 6.3e12, and 2e13 vg/kg

Endpoints: Motor performance, body weight, survival, SOD1 knockdown in spinal cord

SOD1 Pharmacology Study in NHP

NHP: 2-5yo Male & Female Cynomolgus monkey (Macaca fascicularis); ~4 kg

Vector: VCAP-2nd Generation.SOD1 siRNA

Dose: IV administration, 3e13 vg/kg

Endpoints: Biodistribution, SOD1 reduction, tolerability

SOD1 G93A Efficacy Study

Figure 2. Broad Vector Genome Biodistribution and SOD1 Knockdown in Mouse Spinal Cord

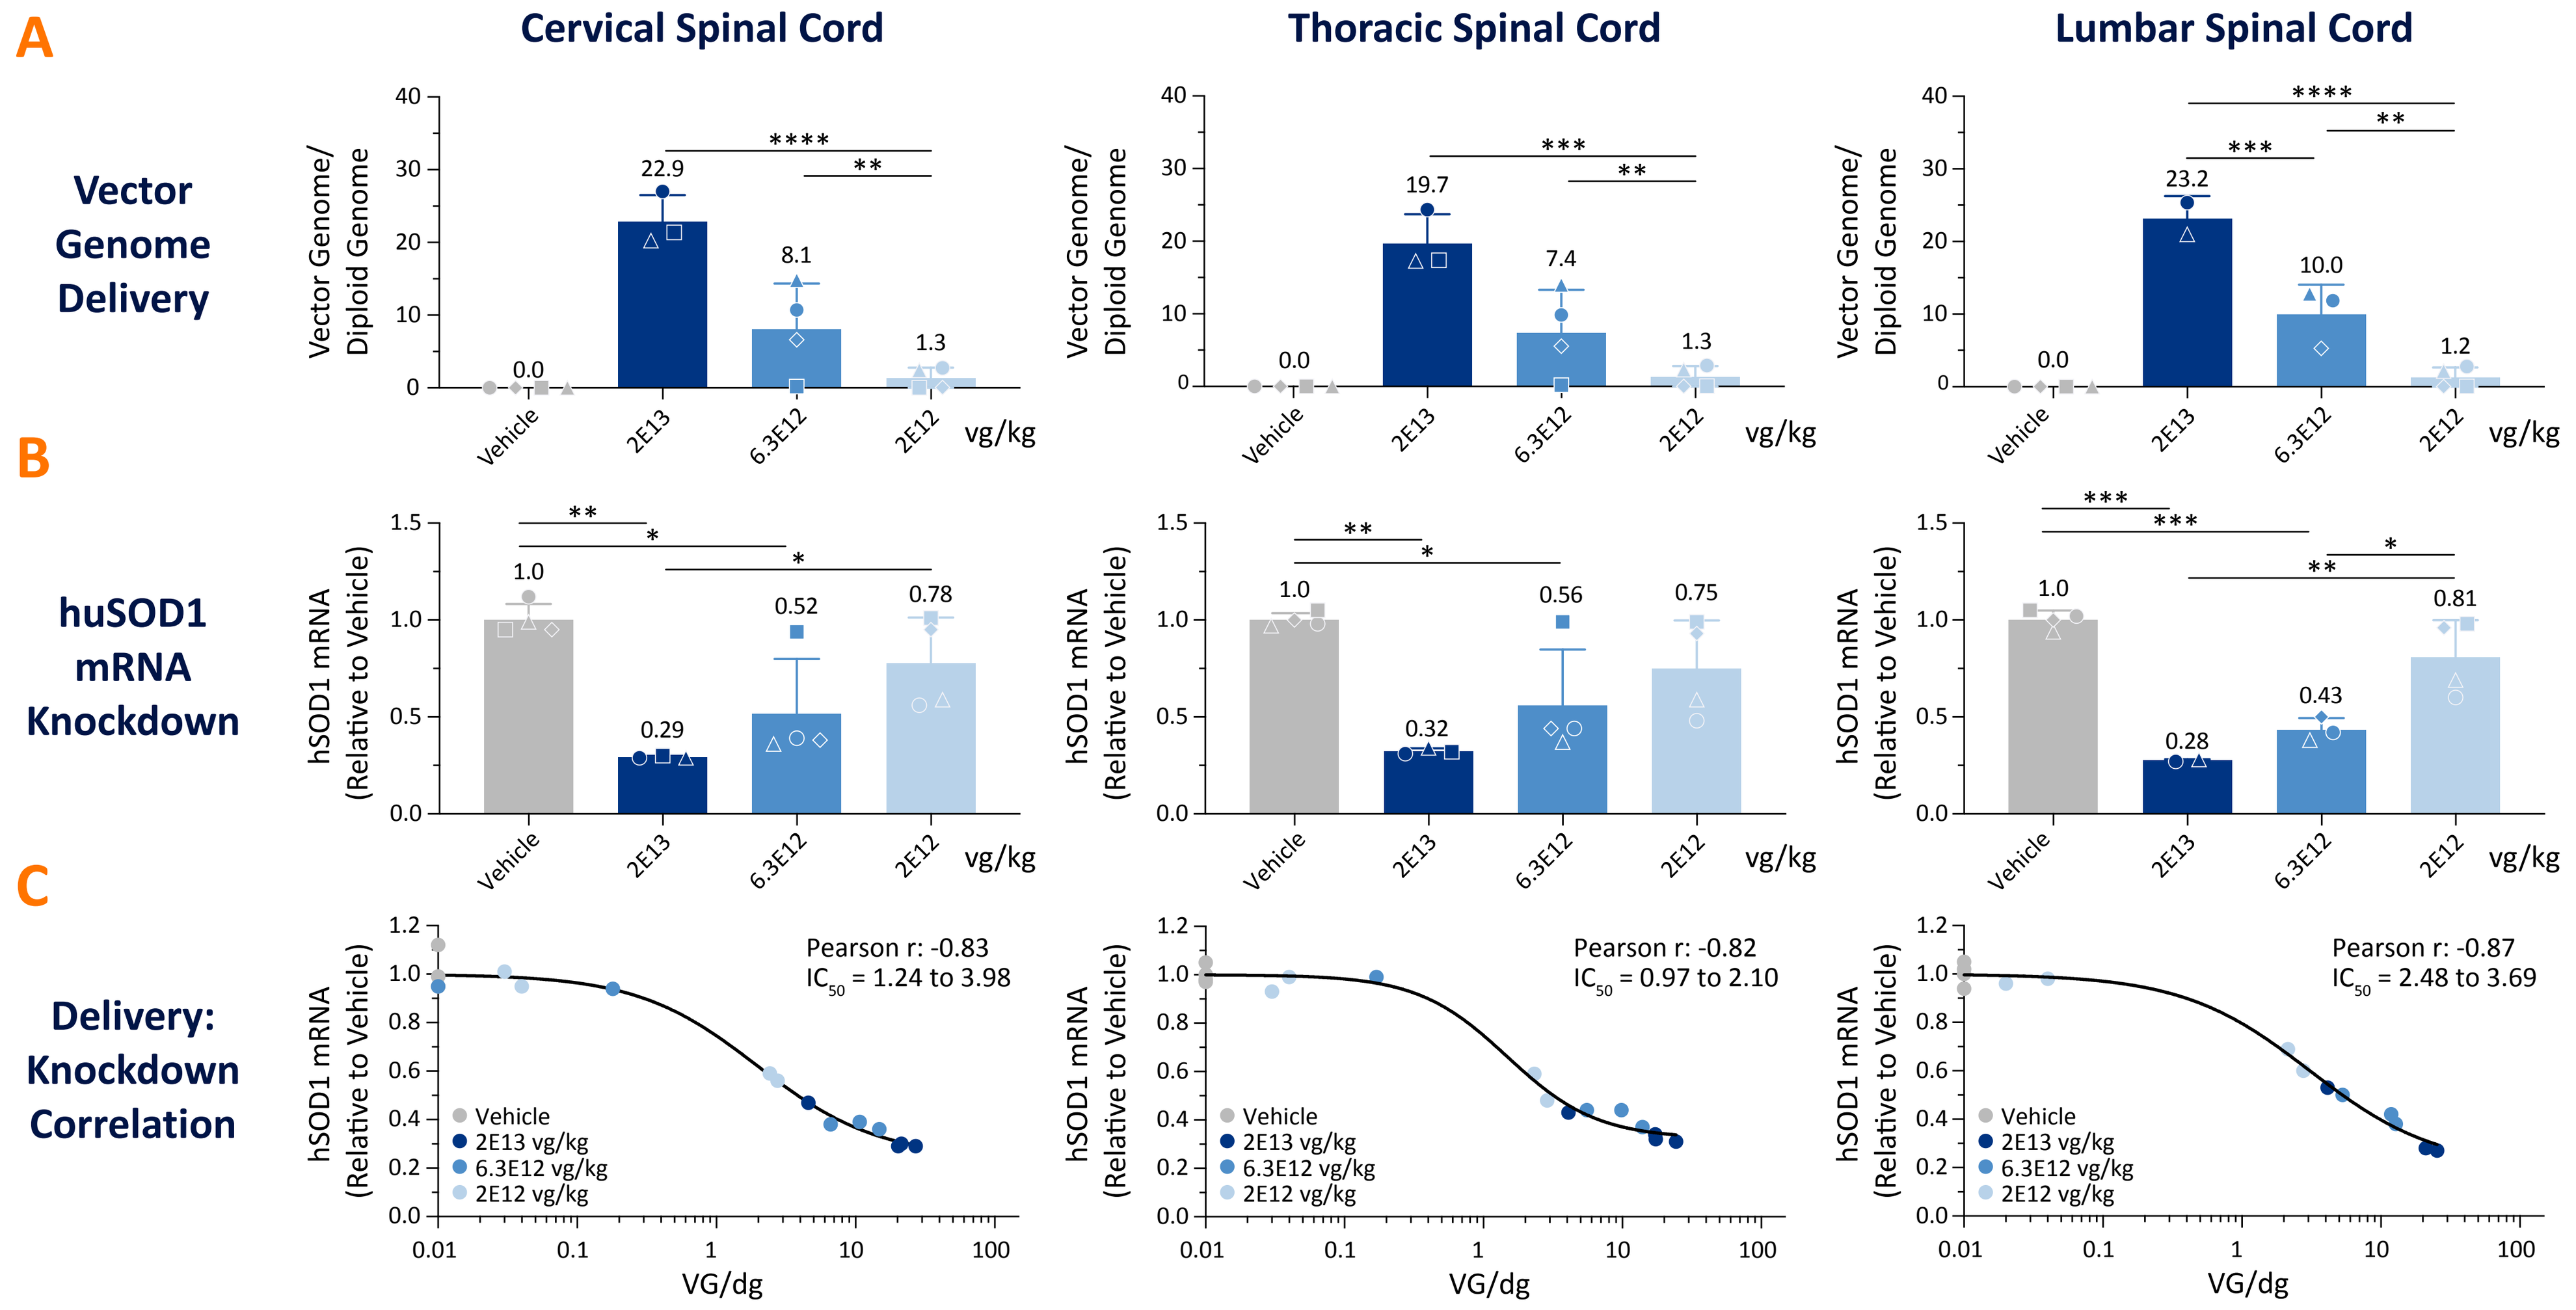
A
B
C
Cervical Spinal Cord
Thoracic Spinal Cord
Lumbar Spinal Cord
Vector Genome Delivery
huSOD1 mRNA Knockdown
Delivery: Knockdown Correlation
Vector Genome/ Diploid Genome
0.0
22.9
8.1
1.3
19.7
7.4
23.2
10.0
1.2
Vehicle
2E13
6.3E12
2E12
vg/kg
hSOD1 mRNA (Relative to Vehicle)
1.0
0.29
0.52
0.78
0.32
0.56
0.75
0.28
0.43
0.81
Pearson r: -0.83
IC_{50} = 1.24 to 3.98
Pearson r: -0.82
IC_{50} = 0.97 to 2.10
Pearson r: -0.87
IC_{50} = 2.48 to 3.69
Vehicle
2E13 vg/kg
6.3E12 vg/kg
2E12 vg/kg
VG/dg

Vector genome distribution and hSOD1 knockdown in the cervical, thoracic, and lumbar spinal cord of G93A mice 32 days following AAV administration. (**A**) Vector genome distribution was analyzed using a multiplex ddPCR assay against transgene and host (mouse) targets in multiple regions of the spinal cord. (**B**) hSOD1 expression measured using multiplexed RT-qPCR with hSOD1 expression level normalized to 2 host reference gene transcripts with vehicle control group as the comparator. (**C**) Correlation of vector genome to hSOD1 knockdown in the mouse spinal cord. *p<0.05, **0.01, ***0.001, 1-way ANOVA with Tukey's Multiple Comparisons.

Figure 3. Increase in Survival in G93A Mice

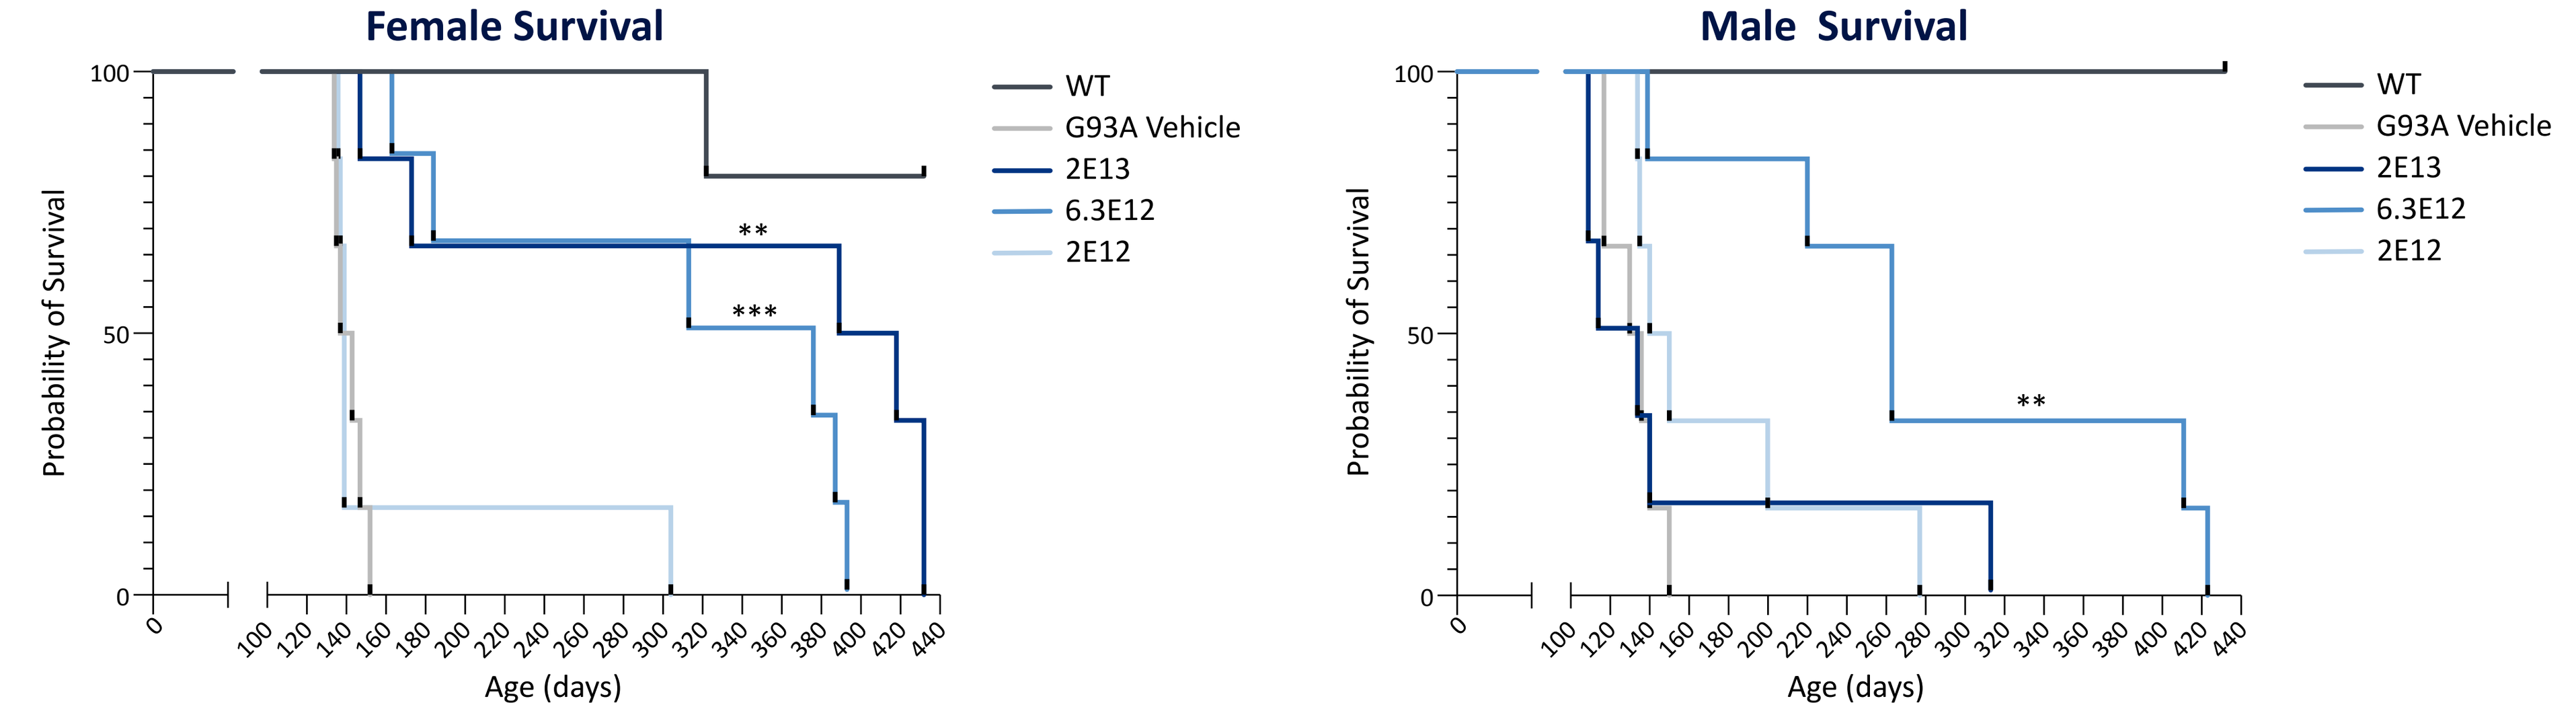
Female Survival
Male Survival
Probability of Survival
Age (days)
WT
G93A Vehicle
2E13
6.3E12
2E12

Female	Median Survival (days)	Improvement From Vehicle (days)	Significance (p=)
G93A Vehicle	140	N/A	N/A
2E12 vg/kg	139	-1	ns
6.3E12 vg/kg	345	205	0.0005
2E13 vg/kg	404	264	0.0022

Male	Median Survival (days)	Improvement From Vehicle (days)	Significance (p=)
G93A Vehicle	133	N/A	N/A
2E12 vg/kg	145	12	ns
6.3E12 vg/kg	263	133	0.003
2E13 vg/kg	124	-9	ns

Reduction of spinal cord SOD1 results in increased survival in G93A mice. Median survival was 140 days for vehicle treated female mice and 139 (n.s.) days for low dose animals. Median survival for moderate and high doses were 345 and 404 days, respectively. Median survival was 133 days for vehicle treated males, 124 (n.s.) and 145 (n.s.) days for the high and low dose, respectively, and 263 (**p = 0.003) days for the mid dose group. Statistical analysis was performed using log-rank (Mantel-Cox) test.

Biodistribution and Pharmacology Studies in NHP

Figure 4. Broad Vector Genome Biodistribution, SOD1 Knockdown, and Motor Neuron Transduction in NHP Spinal Cord

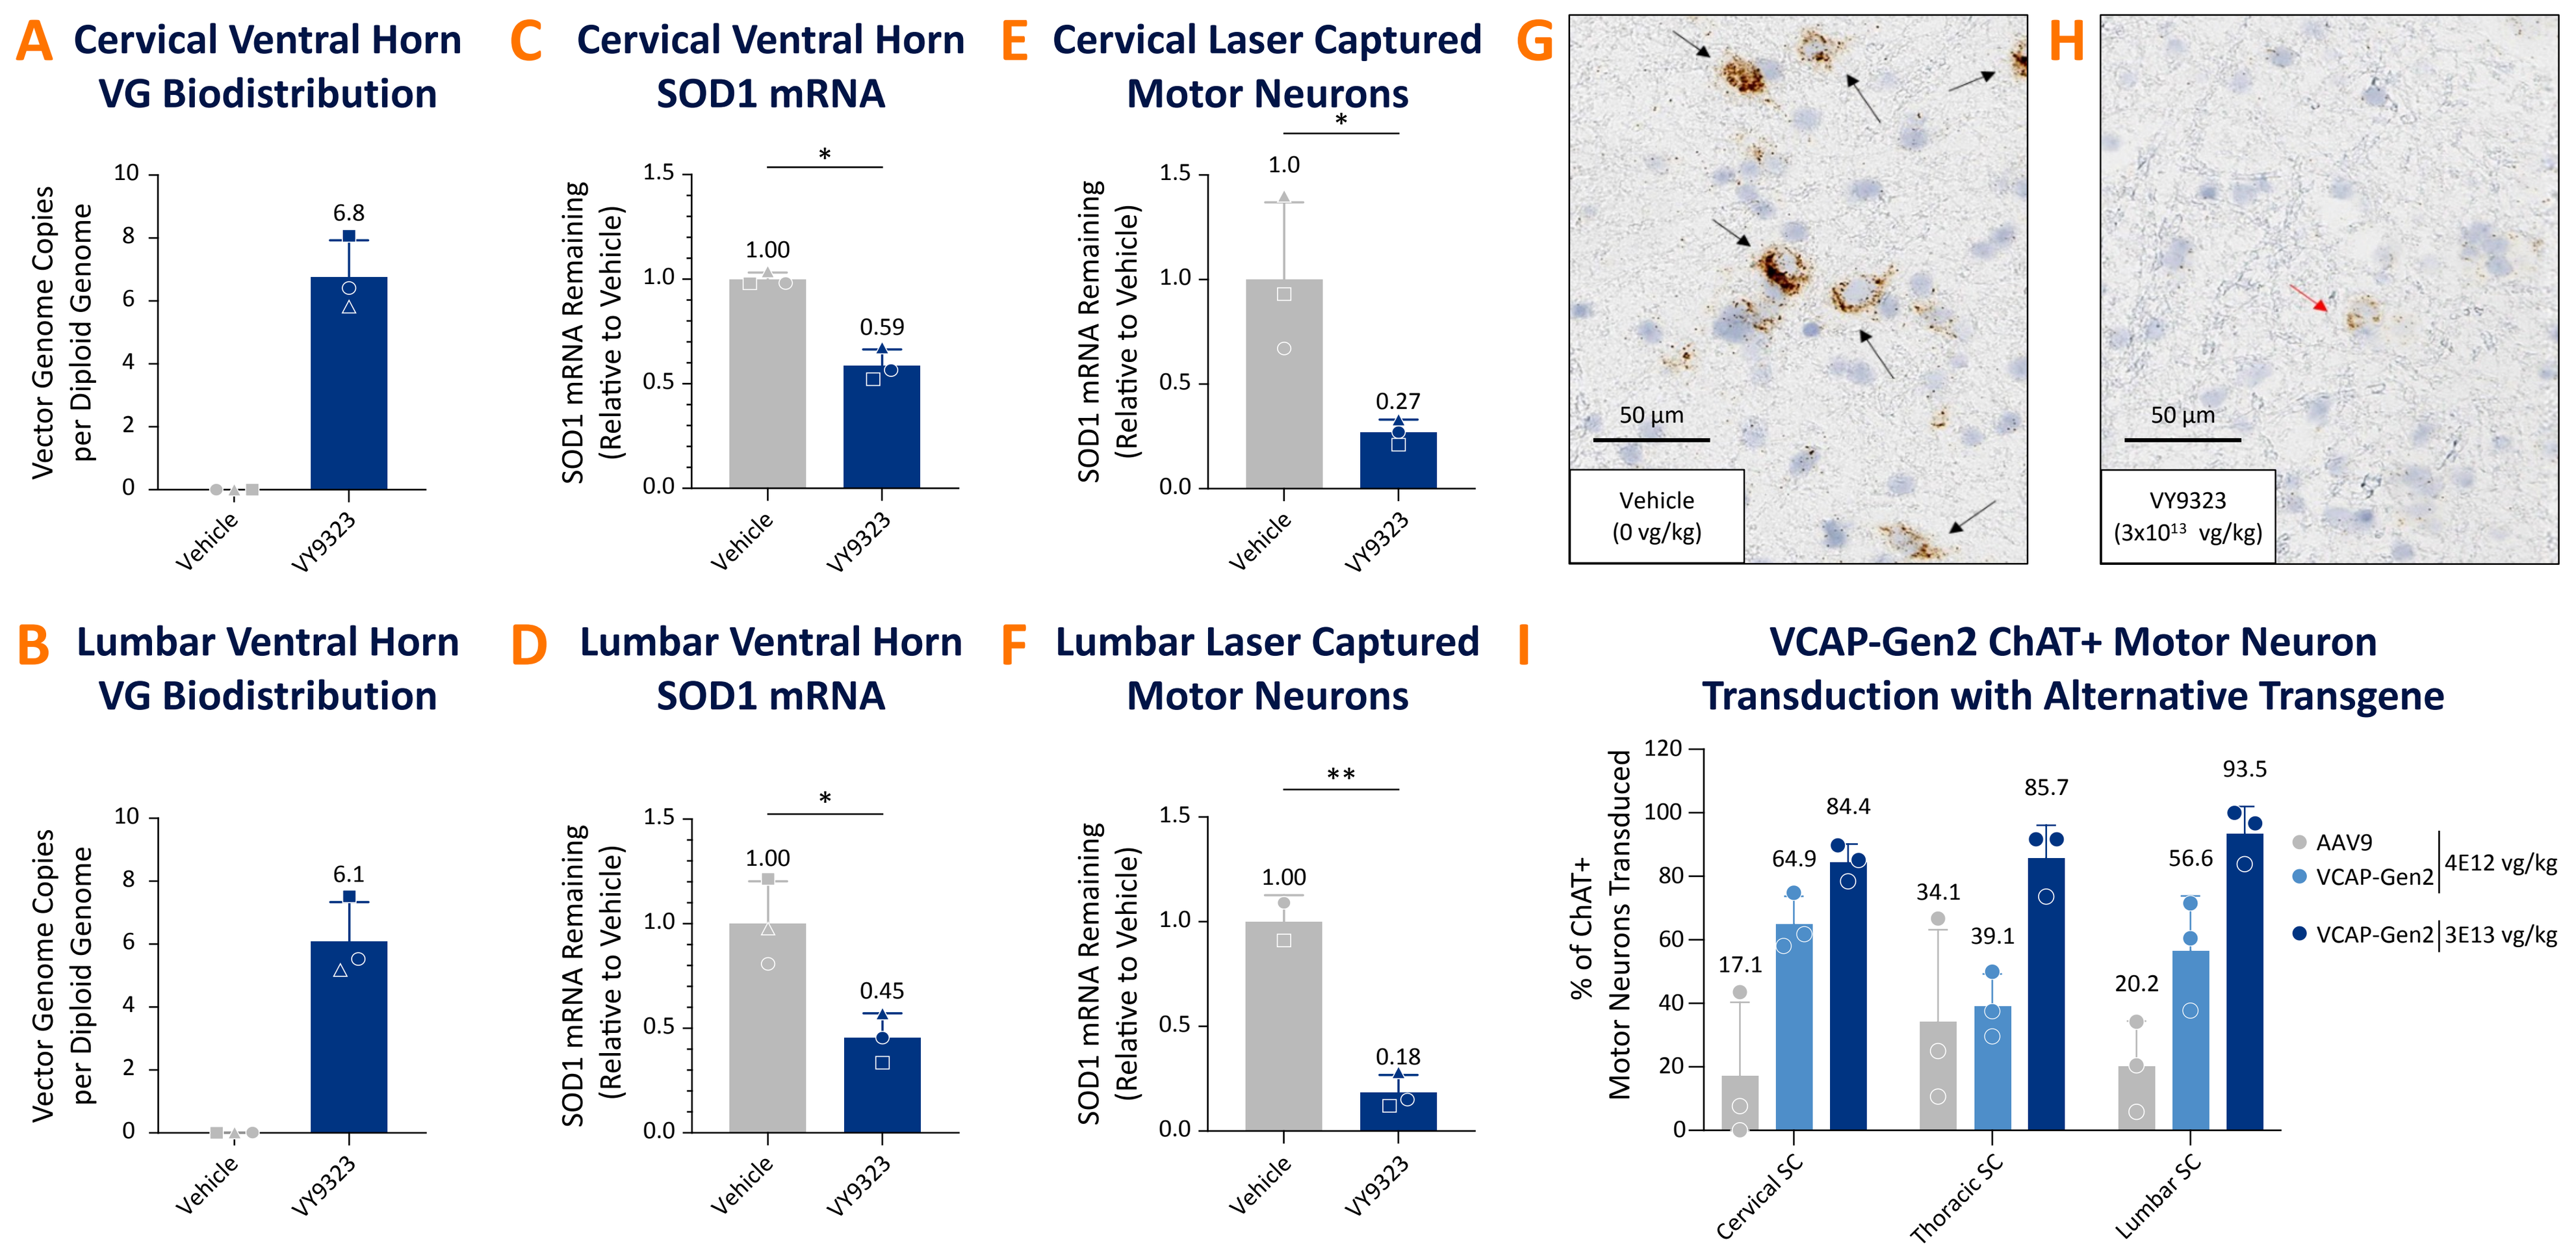
A Cervical Ventral Horn VG Biodistribution
B Lumbar Ventral Horn VG Biodistribution
C Cervical Ventral Horn SOD1 mRNA
D Lumbar Ventral Horn SOD1 mRNA
E Cervical Laser Captured Motor Neurons
F Lumbar Laser Captured Motor Neurons
G
H
I VCAP-Gen2 ChAT+ Motor Neuron Transduction with Alternative Transgene
Vector Genome Copies per Diploid Genome
SOD1 mRNA Remaining (Relative to Vehicle)
6.8
6.1
1.00
0.59
0.45
1.0
0.27
0.18
Vehicle
VY9323
50 µm
Vehicle (0 vg/kg)
VY9323 ($3x10^{13}$ vg/kg)
% of ChAT+ Motor Neurons Transduced
17.1
64.9
84.4
34.1
39.1
85.7
20.2
56.6
93.5
Cervical SC
Thoracic SC
Lumbar SC
AAV9
VCAP-Gen2
4E12 vg/kg
VCAP-Gen2 3E13 vg/kg

Intravenous administration of VY9323 in cynomolgus monkeys results in substantial vector genome delivery (**A**, **B**) in the cervical and lumbar spinal cord and produces significant SOD1 mRNA reduction in cervical and lumbar spinal cord ventral horn tissue punches (**C**, **D**) and laser captured motor neurons (**E**, **F**). VY9323 substantially reduces SOD1 mRNA in cervical spinal cord when evaluated using RNAscope **(G, H)**. Black arrows represent cells expressing many copies of SOD1 mRNA, and red arrows indicate cells that express fewer copies. (**I**) Motor neuron transduction, determined by ChAT co-staining, was evaluated using an alternative transgene for AAV9 and the VY9323 capsid at 4E12 vg/kg, and for the VY9323 capsid at 3E13 vg/kg. *p<0.05, ** p< 0.01, (Unpaired t-test) Error shown as standard deviation.

Figure 5. Biodistribution in Motor Cortex and Peripheral Tissues

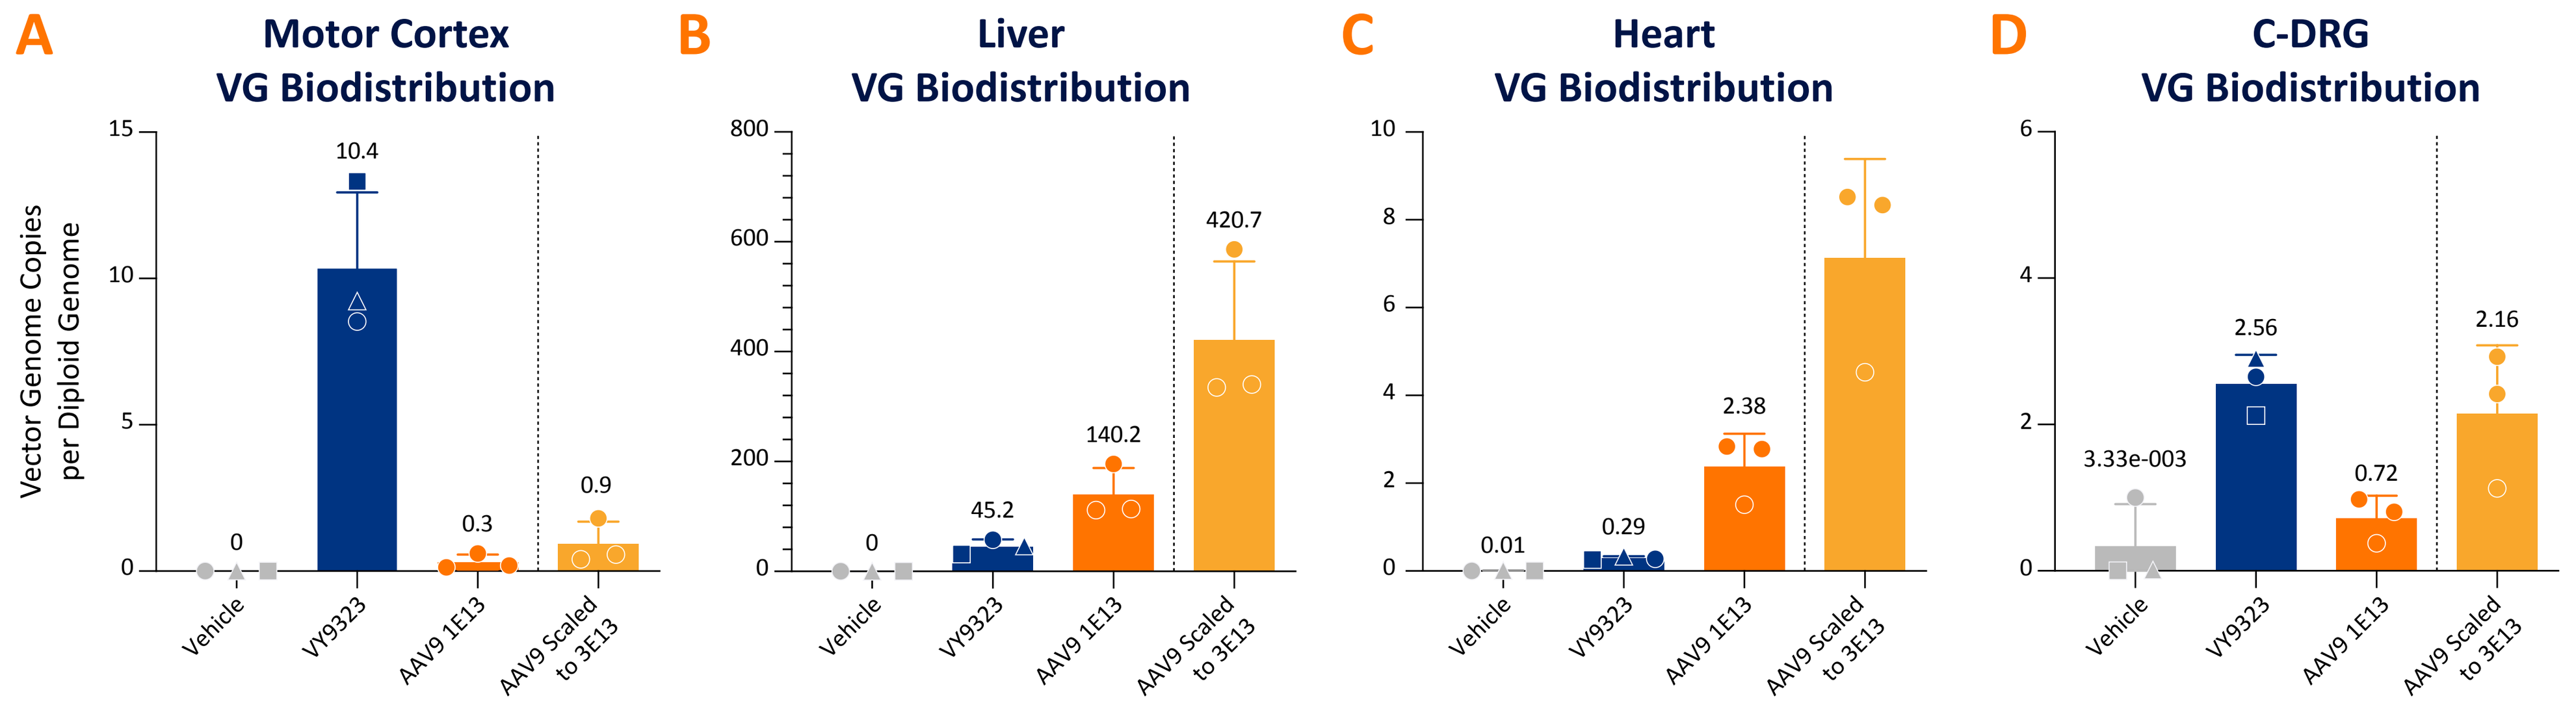
A Motor Cortex VG Biodistribution
B Liver VG Biodistribution
C Heart VG Biodistribution
D C-DRG VG Biodistribution
Vector Genome Copies per Diploid Genome
0
10.4
0.3
0.9
45.2
140.2
420.7
0.01
0.29
2.38
3.33e-003
2.56
0.72
2.16
Vehicle
VY9323
AAV9 1E13
AAV9 Scaled to 3E13

(**A**) Intravenous administration of VY9323 in cynomolgus monkeys results in substantial vector genome delivery in the motor cortex while exhibiting peripheral tissue detargeting (**B**-**D**) compared to historical WT-AAV9 data expressing an alternative transgene at a 1E13 vg/kg dose scaled to 3E13 vg/kg. C-DRG = Cervical Dorsal Root Ganglia. Error bars represent standard deviation.

CONCLUSIONS

- Reduction of SOD1 produces significant survival benefit in the G93A mouse model of SOD1 ALS
- In primates, robust knockdown of SOD1 in all levels of the spinal cord and motor cortex is observed following IV administration of our novel TRACER™ capsid and SOD1 targeting pri-amiRNA, while exhibiting a favorable profile in peripheral tissues
- These data support the continued development of IV delivered RNAi using a novel BBB-penetrant capsid for use in the clinic

75 Hayden Avenue, Lexington, MA 02421 | www.voyagertherapeutics.com

ASGCT 27th Annual Meeting 2024 | May 7 – 11, 2024 | Baltimore, MD