Development of HEK293 Cell Line for Optimal Production of Novel Capsids with Enhanced Brain Tropism

Varshini Venkatesan, Zeynep Guillemin, Amanda Ibe-Enwo, Christian Gagnon, Shamik Sharma, Kumar Dhanasekharan

Voyager Therapeutics Inc., Lexington, MA, USA

voyager therapeutics

1035

INTRODUCTION

Voyager has developed novel AAV capsids with significantly enhanced brain tropisms and achieved transgene expression that is superior to conventional AAV serotypes in non-human primates. These capsids have the potential to increase the safety and effectiveness of AAV gene therapy. In order to accelerate clinical studies of therapeutic candidates using novel capsids, rapid process development leading to a cost-effective manufacturing process is the next challenge. HEK293 cells are the most widely used cell-line for rAAV production and Voyager has internally developed a HEK293 cell line to support our portfolio of AAV gene therapy assets. The VYGR-293 cell line was developed by adapting a serum-fed adherent cell line to a serum-free suspension culture. Further, VYGR-293 cells were optimized for maximum productivity throughout the production process from cell seeding to lysis. In this poster, we will discuss the optimization of VYGR-293 cell line using one of our capsids (capsid A) for the following parameters –

1. The upstream process was developed by examining optimal N-1 to N split ratios before production and cell density at transfection.
2. The transfection(txn) process was developed and optimized for parameters such as plasmid to transfection reagent ratio and molar plasmid ratio (Transgene: Rep-Cap: Helper) using Design of Experiment (DOE) studies. Optimal parameters that enabled the highest productivity and %Full capsids were thus selected.
3. In addition, multiple additives and commercial transfection additives were explored. VYGR-293 cells achieved 2-4x increase in volumetric titer after these optimization studies, reaching above 2E11 vg/mL. rAAV obtained from VYGR-293 cell line had good product quality profile including high % full capsids.

In summary, this cost-efficient cell line provides a good foundation for the development of an AAV expression platform to produce drug candidates for neurological disorders.

Figure 1. VYGR-293 Cells Successfully Adapted to Suspension Culture

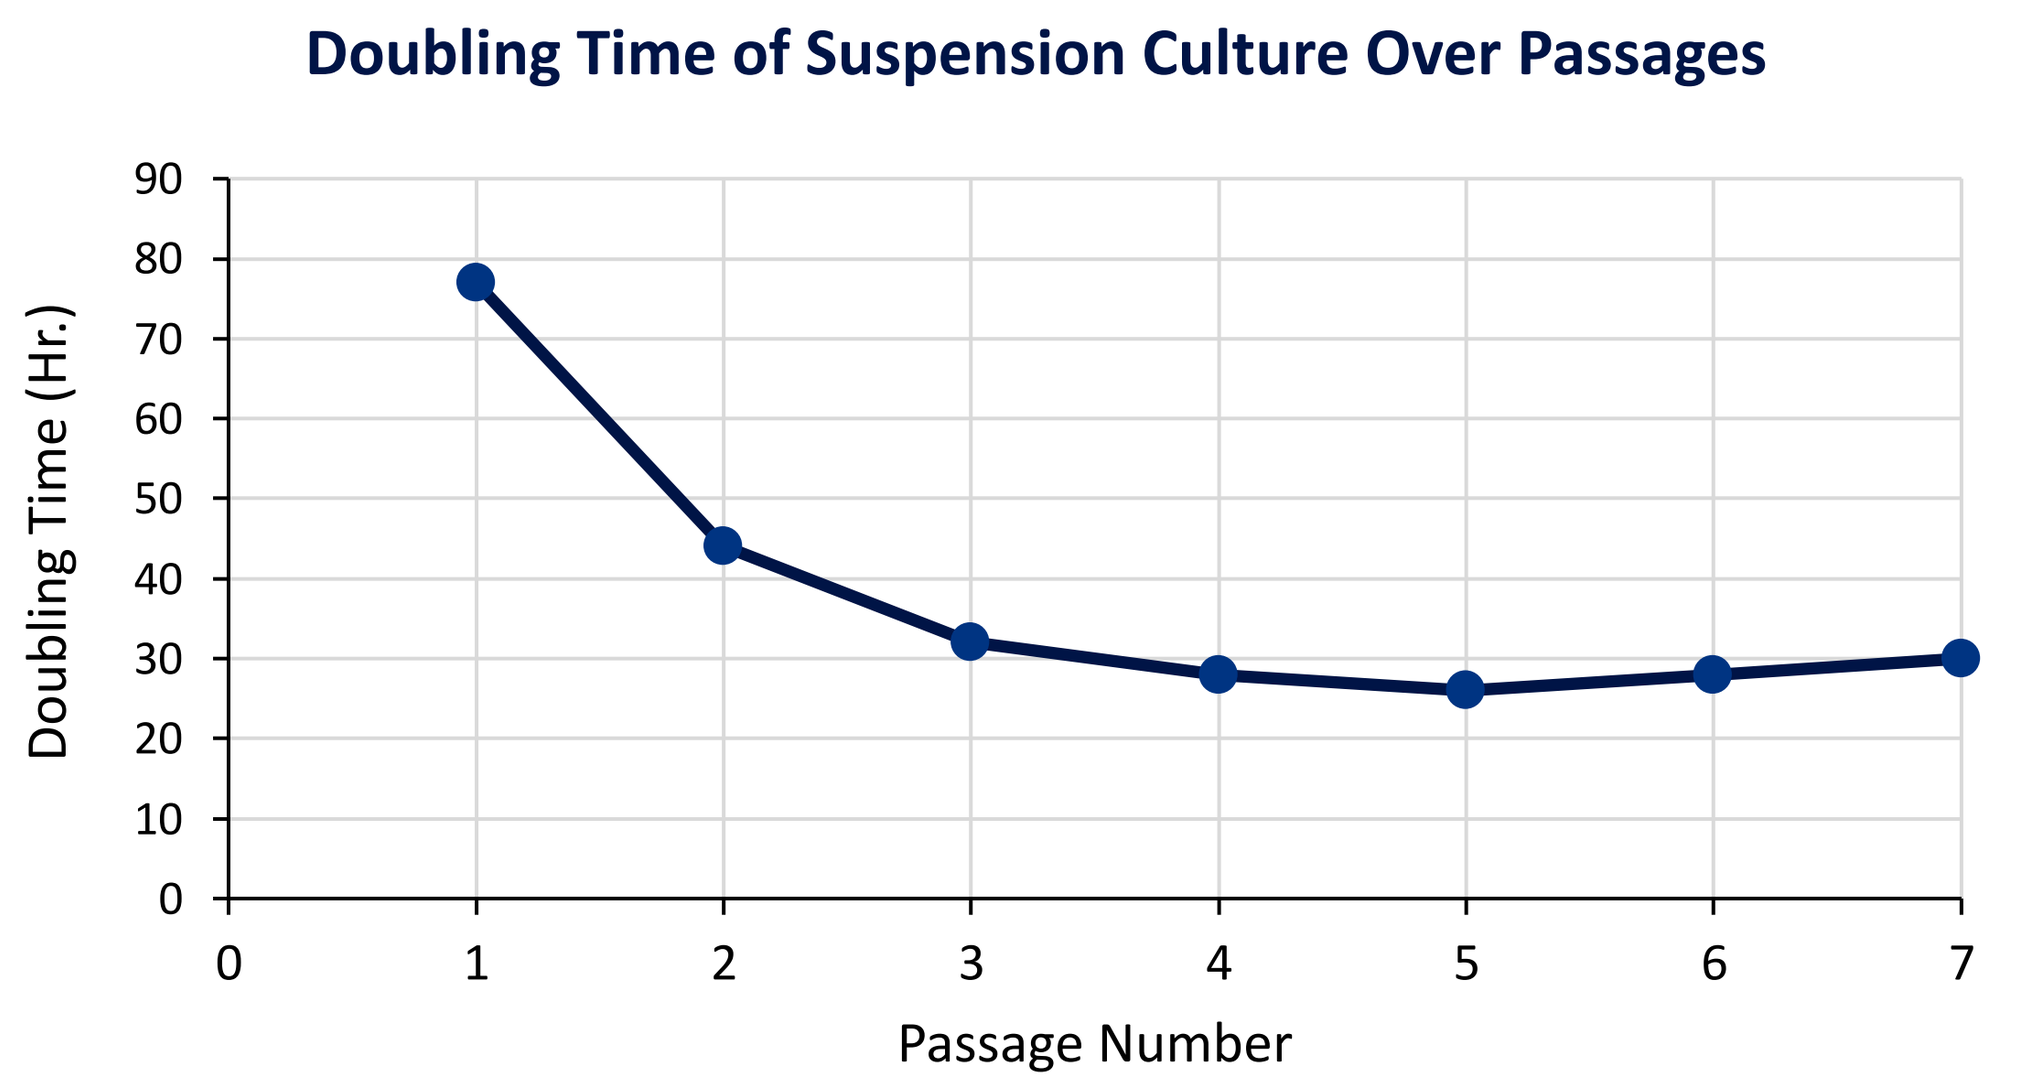
Doubling Time of Suspension Culture Over Passages
Doubling Time (Hr.)
Passage Number

Adherent HEK 293 cells were adapted from adherent serum-fed culture to suspension-based, serum-free culture. The serum-free/suspension-adapted cell line VYGR-293. Cells were banked at 1E7 vc/mL with freezing media.

Figure 2. Voyager Engineered Capsids with Enhanced Brain Tropism

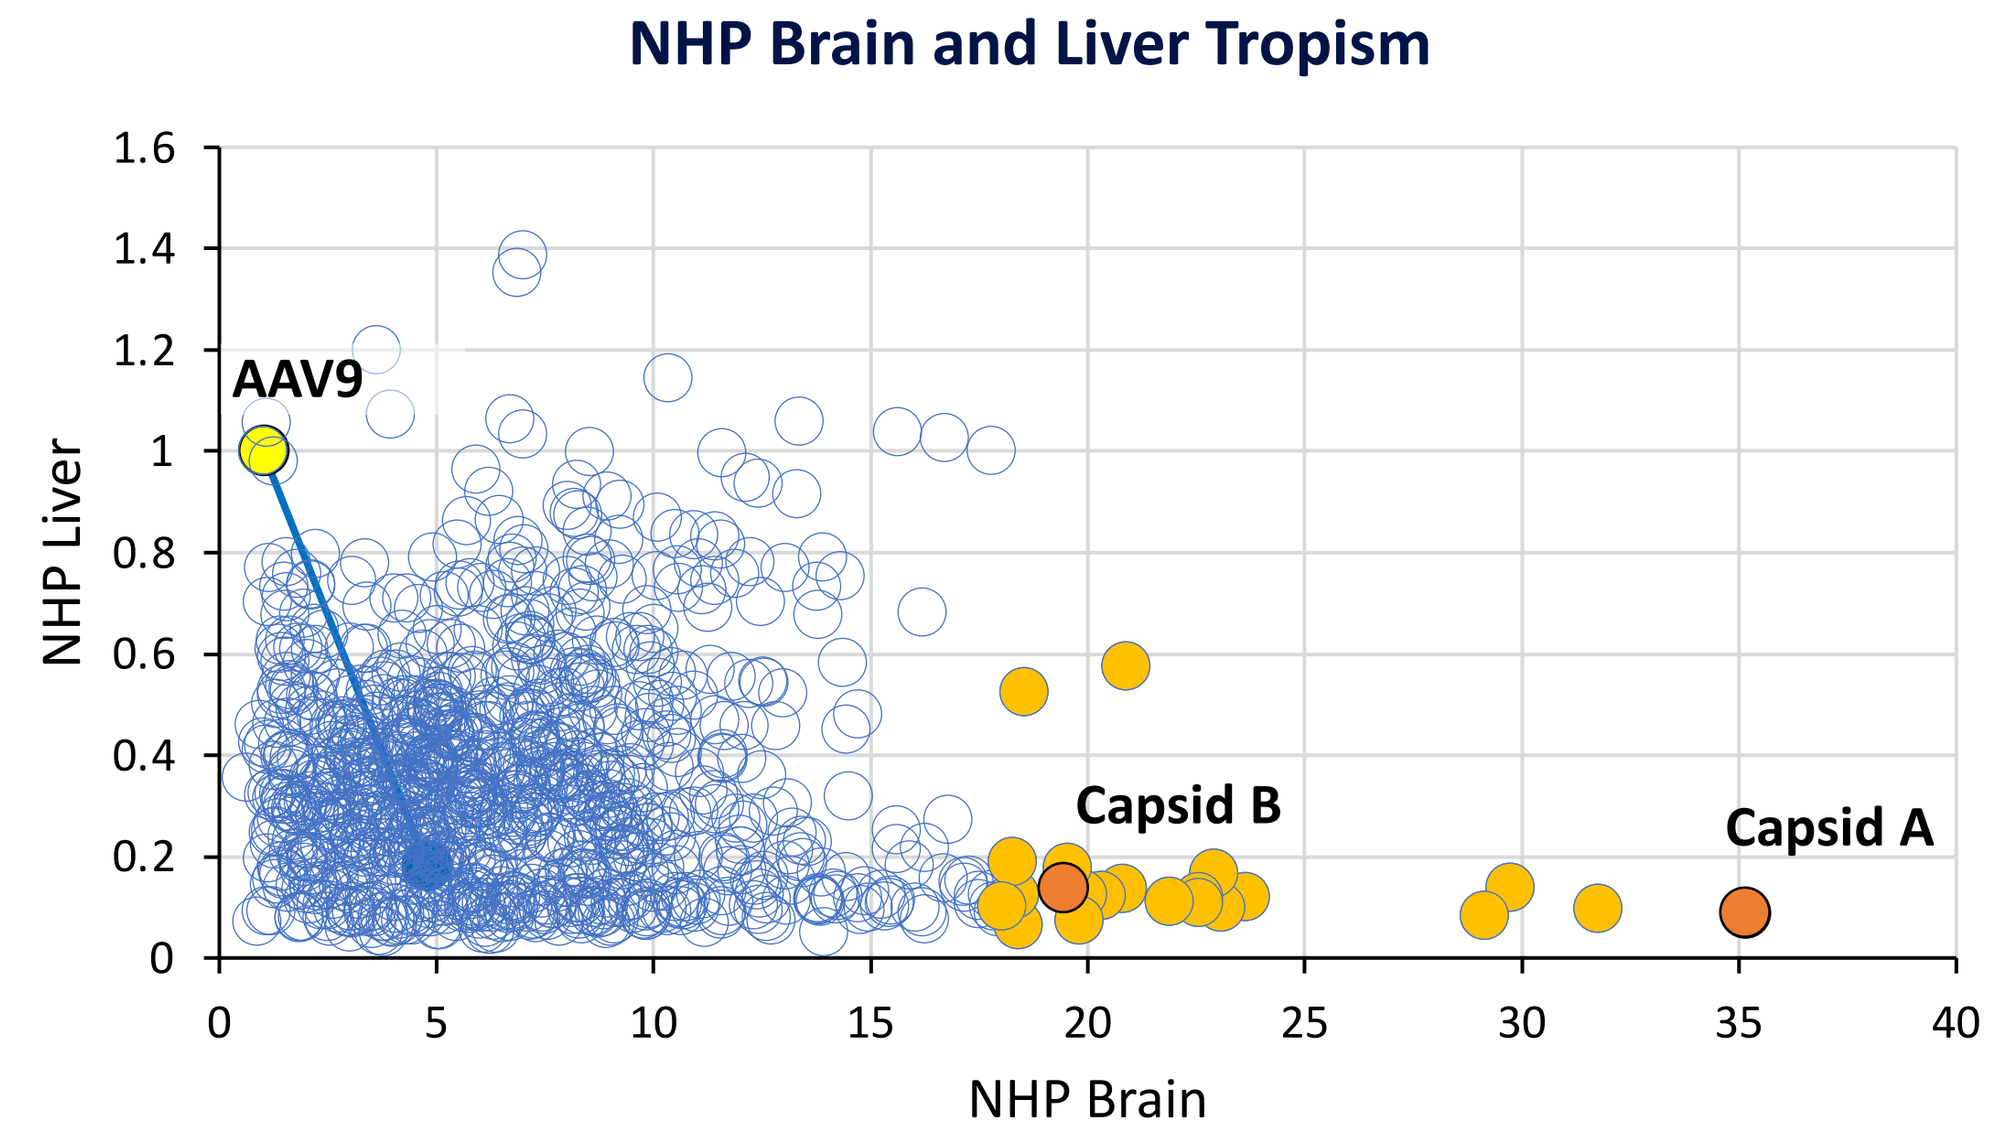
NHP Brain and Liver Tropism
AAV9
Capsid B
Capsid A
NHP Liver
NHP Brain

Multiplexed NGS analysis of RNA enrichment in the brain (X-axis) and liver (Y-axis) of Cynomolgus macaque 28 days after IV dosing of a TRACER library (~1,000 capsid variants). Data indicate the average fold change of normalized RNA reads per million (rpm) relative to AAV9. Internal benchmark capsids AAV9 and VCAP-102 are indicated.

Figure 3. VYGR-293 Upstream Shake Flasks Productivity Using Different Cell Densities at Transfection

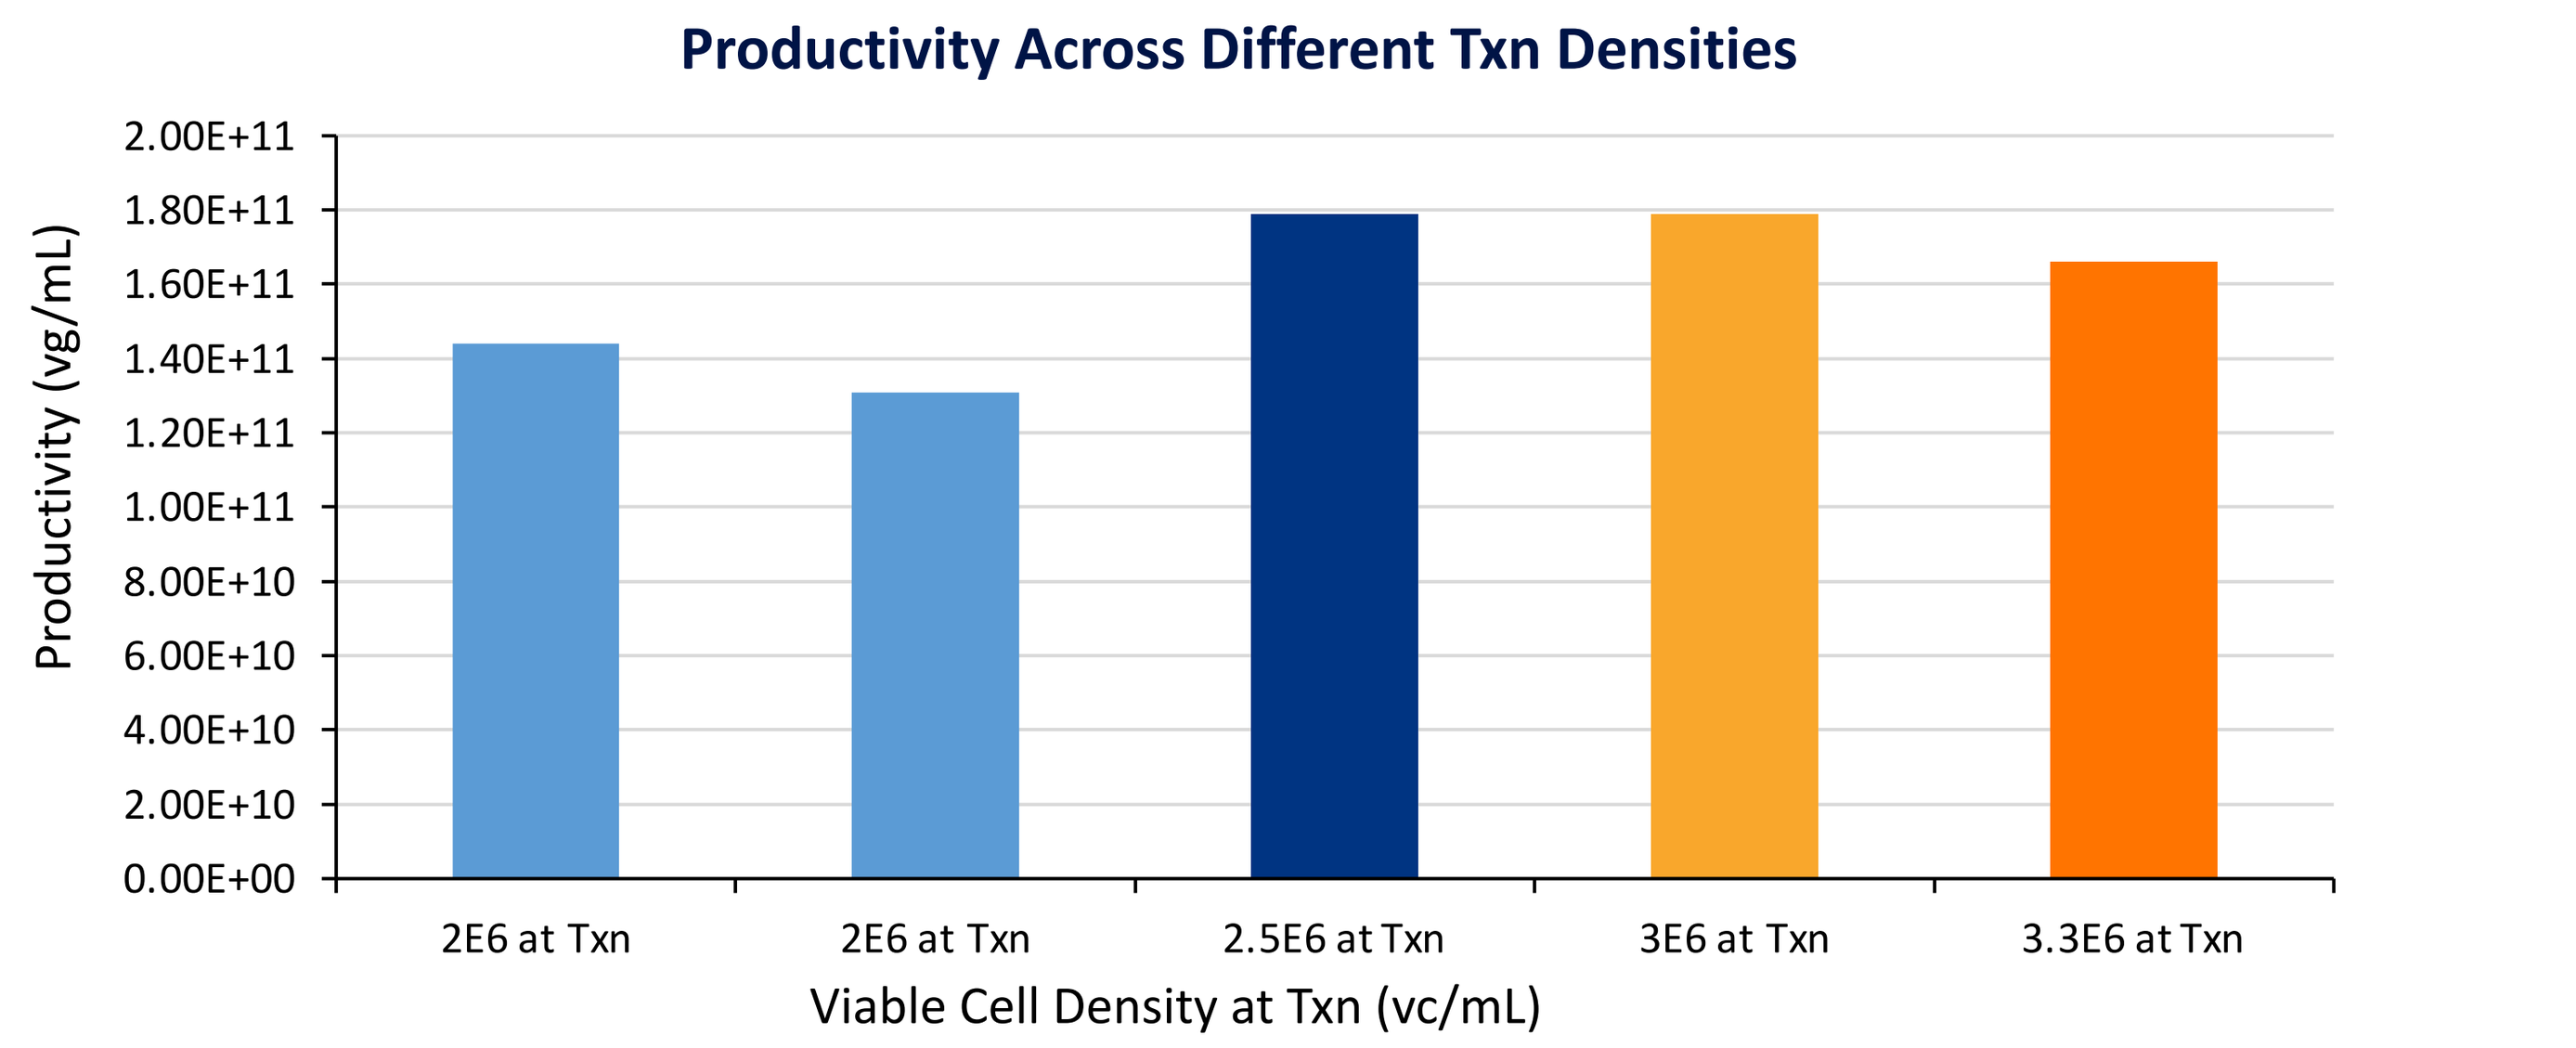
Productivity Across Different Txn Densities
Productivity (vg/mL)
2E6 at Txn
2E6 at Txn
2.5E6 at Txn
3E6 at Txn
3.3E6 at Txn
Viable Cell Density at Txn (vc/mL)

Cells were lysed with Triton-Benzonase and clarified. Clarified lysates were analyzed with ddPCR assay to obtain vector genome/mL productivity.

Figure 4. VYGR-293 Upstream Shake Flasks Productivity Using Different N-1 to N Split Ratios Before Production

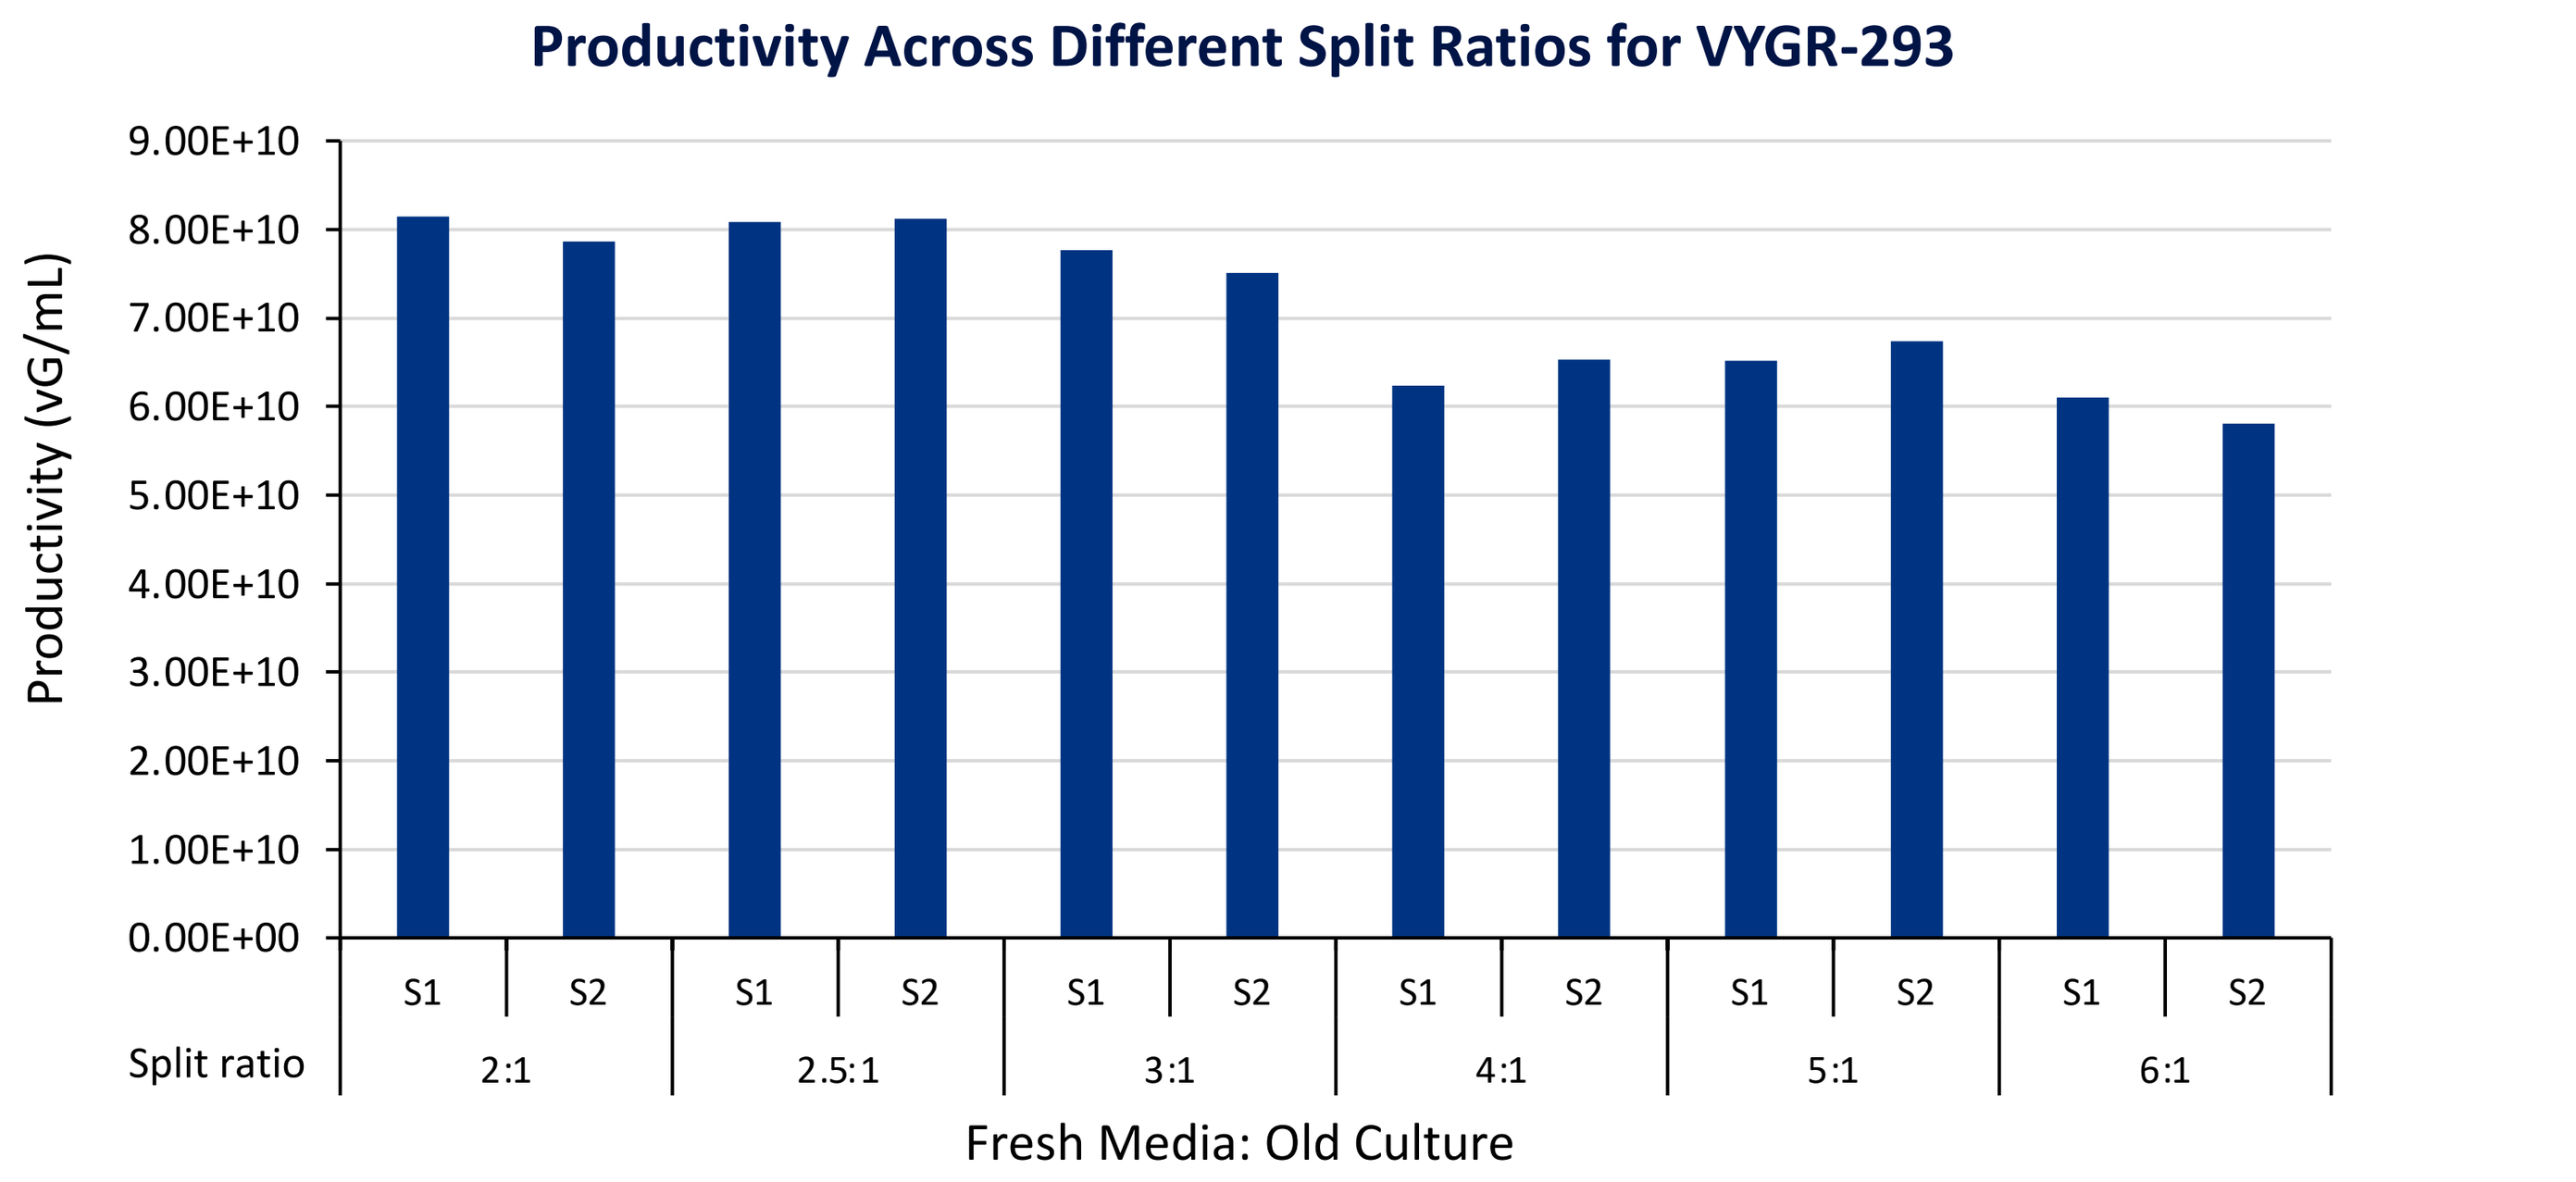
Productivity Across Different Split Ratios for VYGR-293
Productivity (vG/mL)
Split ratio
2:1
2.5:1
3:1
4:1
5:1
6:1
Fresh Media: Old Culture

Cells were lysed with Triton-Benzonase and clarified. Clarified lysates were analyzed with ddPCR assay to obtain vector genome/mL productivity.

Figure 5. DOE to Find Optimal Total Plasmids Amount and Txn Reagent: Total Plasmids Ratio

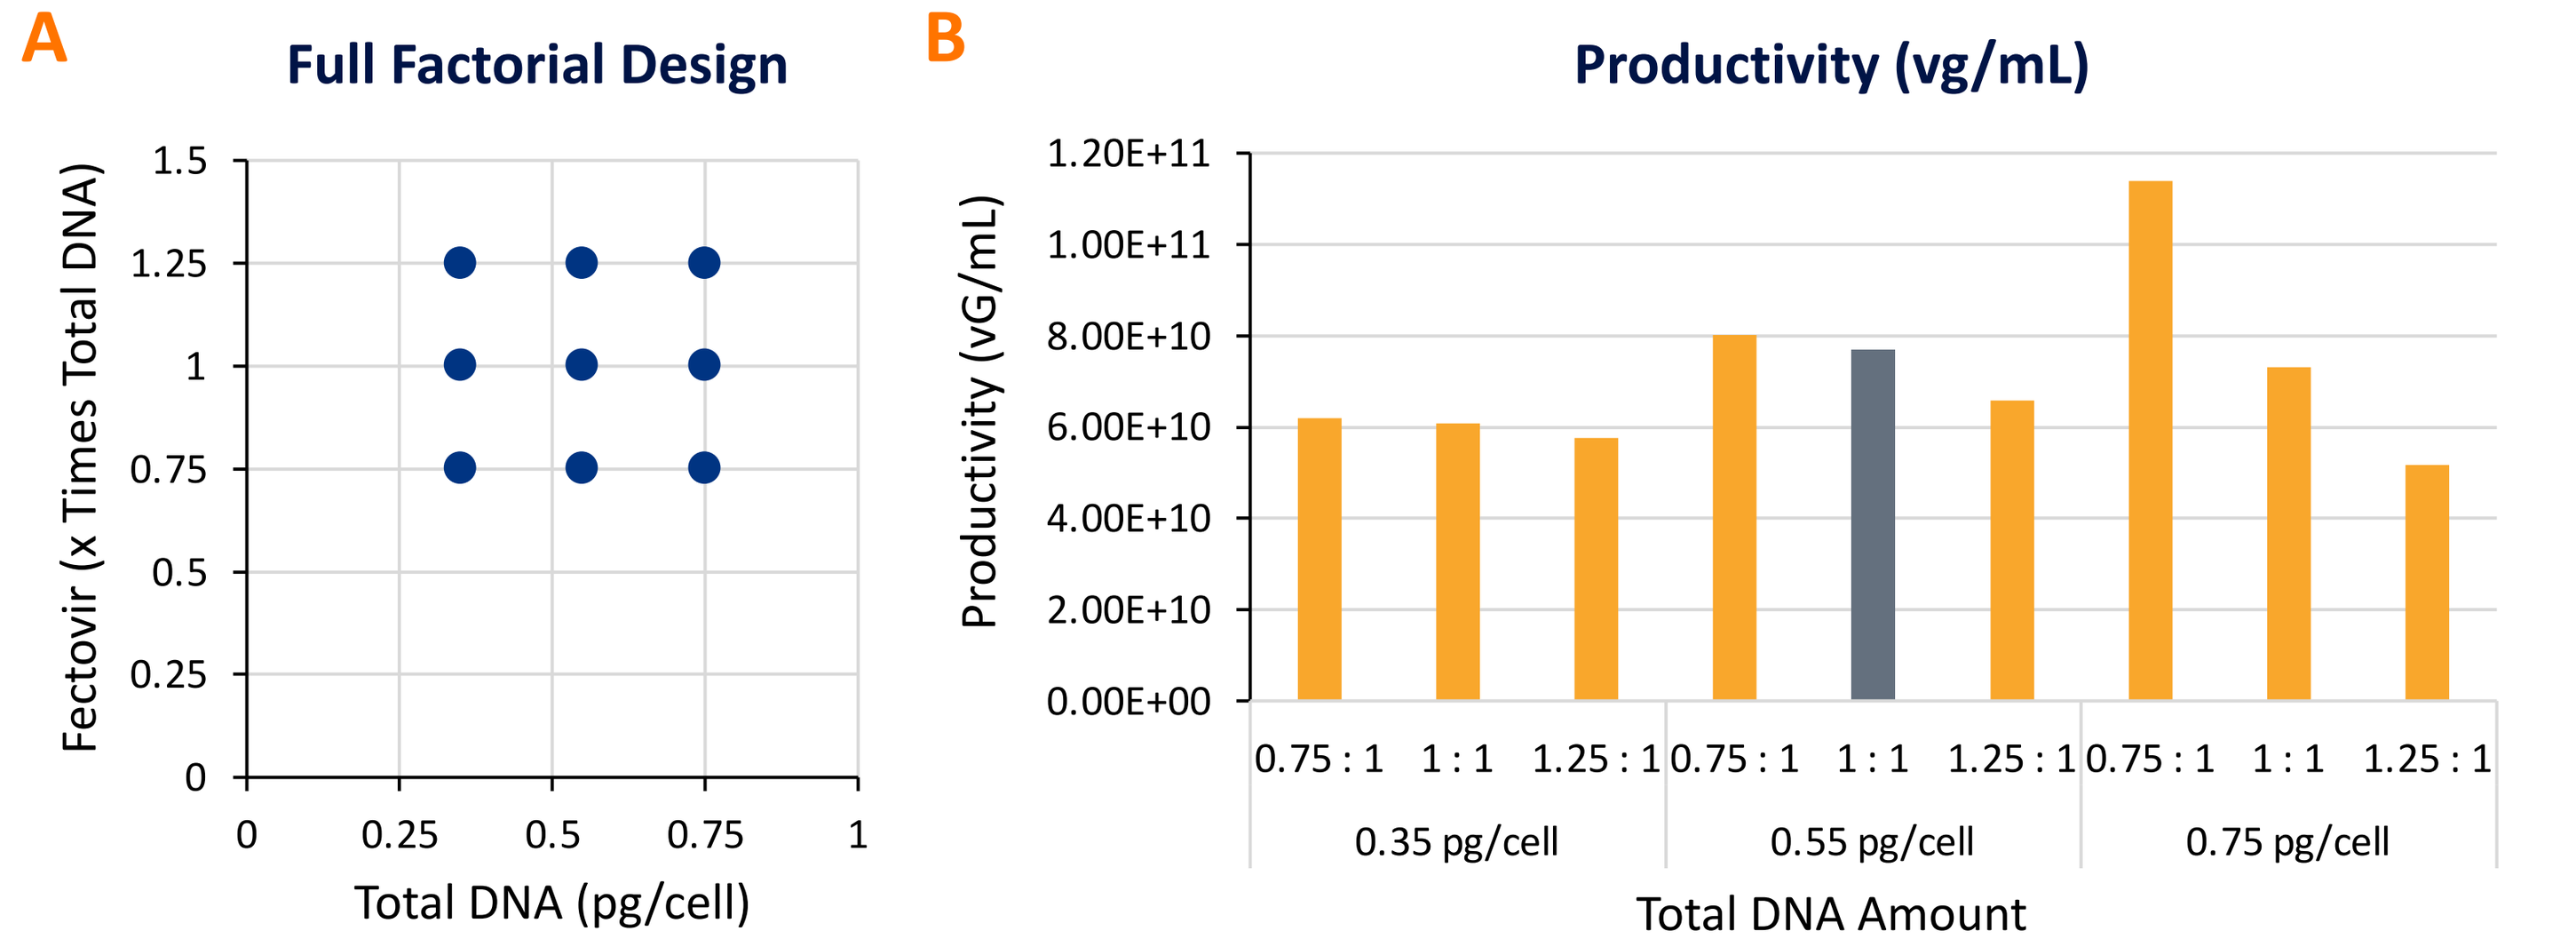
A
Full Factorial Design
Fectovir (x Times Total DNA)
Total DNA (pg/cell)
B
Productivity (vg/mL)
Productivity (vG/mL)
0.35 pg/cell
0.55 pg/cell
0.75 pg/cell
Total DNA Amount

(**A**) Full Factorial design to find optimal total Plasmid amounts (0.25-1 pg./cell) and Txn reagent: Plasmid ratio (0.75-2 times of plasmid amount used) using JMP analysis was performed. (**B**) Cells were lysed with Triton-Benzonase and clarified. Clarified lysates were analyzed with ddPCR assay to obtain vector genome/mL productivity. Clarified lysates were affinity purified by poros capture AAV9 columns. Eluates of affinity purification were utilized for product quality analysis.

Figure 6. Increasing the Total Plasmid Amounts has a Negative Impact on the %full Capsids and the HEK Residual DNA

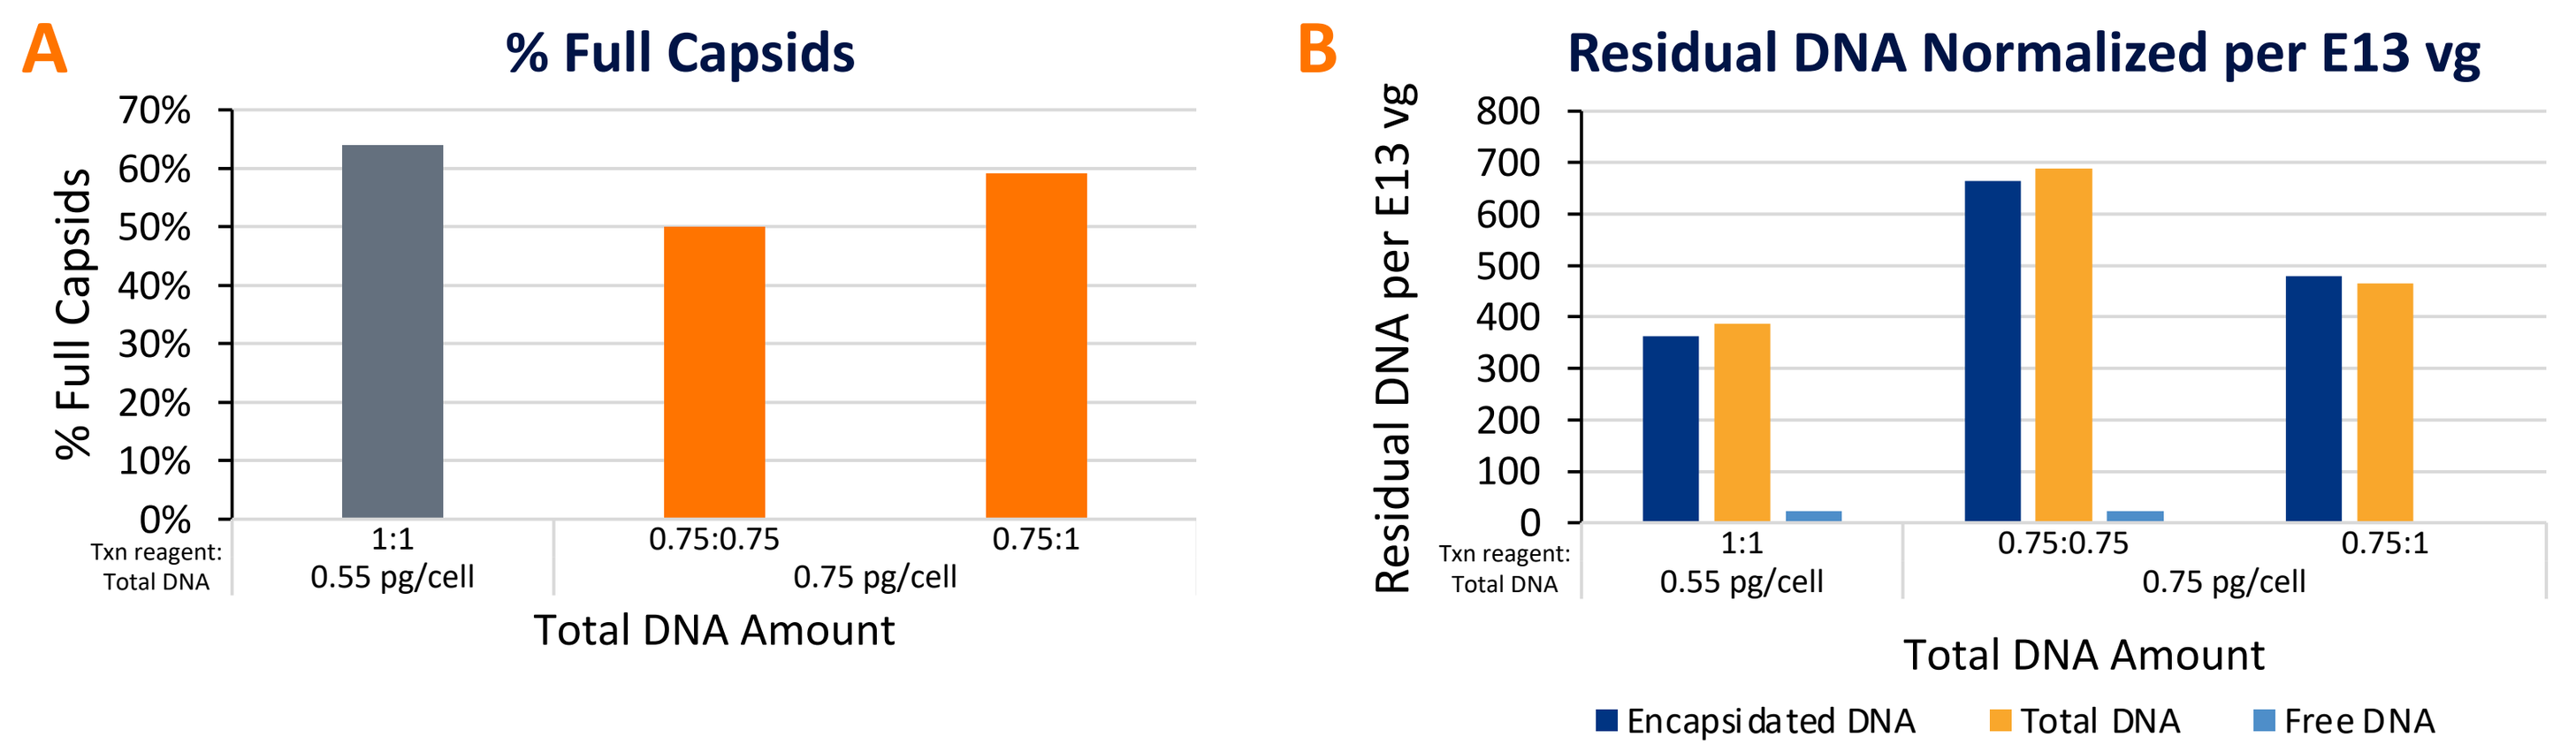
A
% Full Capsids
% Full Capsids
Total DNA Amount
B
Residual DNA Normalized per E13 vg
Residual DNA per E13 vg
Total DNA Amount
Encapsidated DNA
Total DNA
Free DNA

(**A**) Affinity column purified material was analyzed for % Full using size exclusion chromatography (SEC). Online multi angle light-scattering (MALS) combined with refractive index (RI) is used to determine the molar mass, geometry, and protein fraction of the aggregates. This assay measures %Full by deconvoluting the molar mass and determining the contribution from the DNA component of the monomer peak. (**B**) DNA residuals were measured using a commercially available quantitative PCR (qPCR)-based system for the detection of HEK293 host-cell DNA. The kit contains HEK genomic DNA standards and primer probes. During sample processing, DNase digestion was performed to differentiate between the nuclease sensitive and nuclease resistant residual DNA.

Figure 7. Productivity and JMP Analysis of the DOE to Determine Optimal Molar Plasmid Ratios

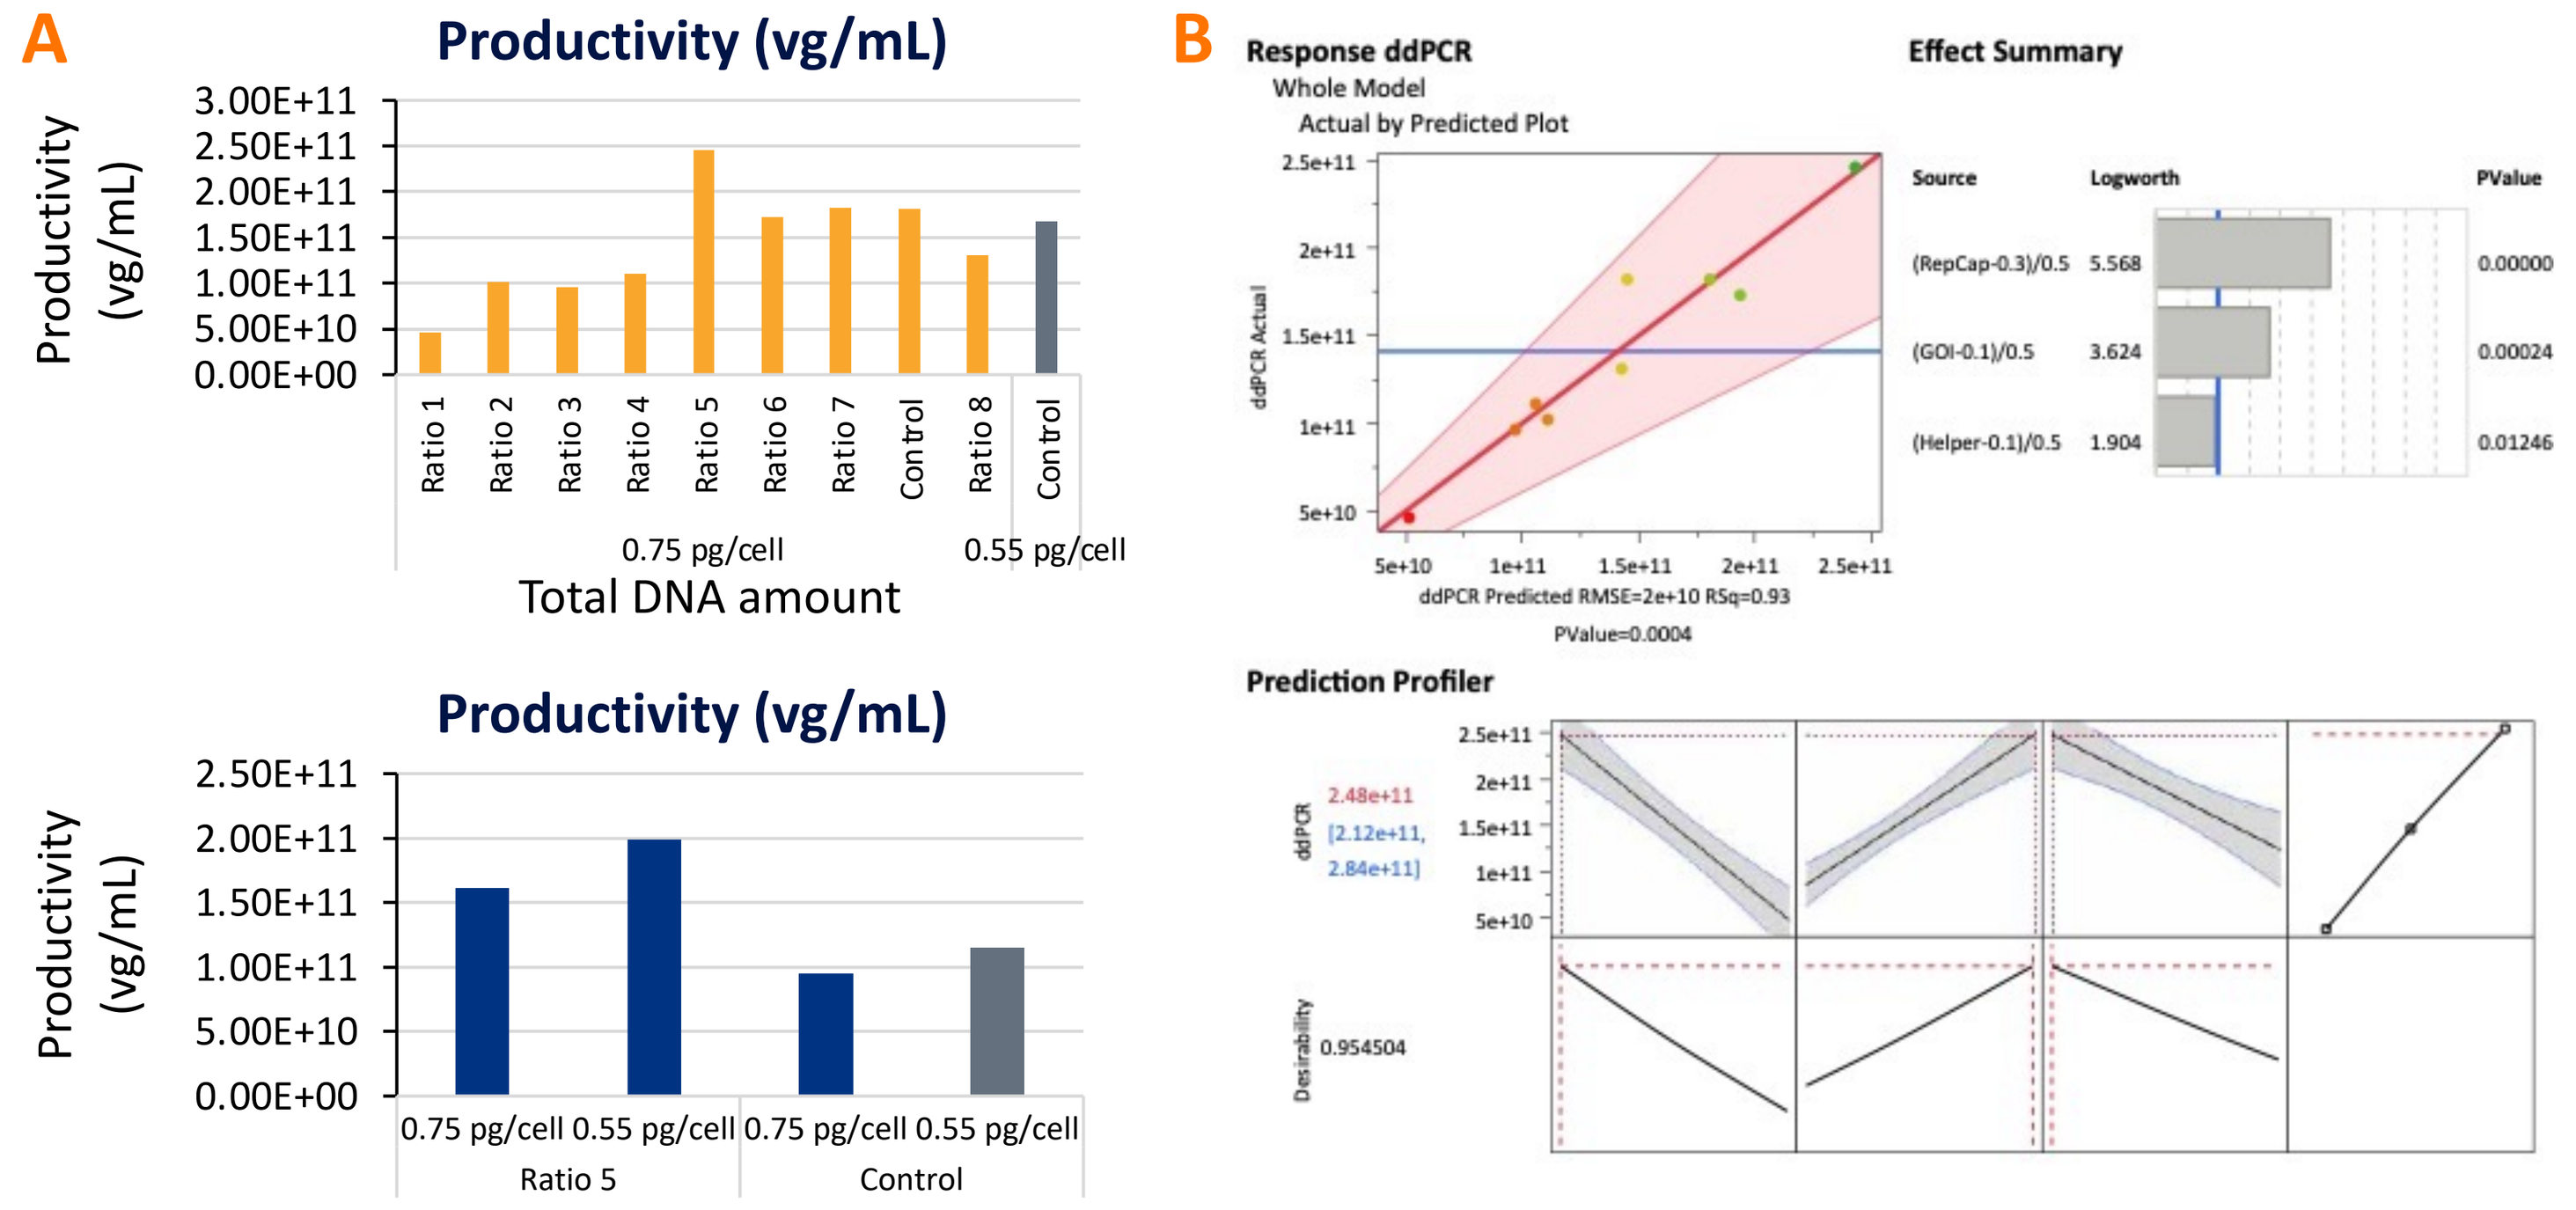
A
Productivity (vg/mL)
Total DNA amount
Productivity (vg/mL)
Ratio 5
Control
B
Response ddPCR
Whole Model
Actual by Predicted Plot
Effect Summary
Prediction Profiler

(**A**) Cells were lysed with Triton-Benzonase and clarified. Clarified lysates were analyzed with ddPCR assay to obtain vector genome/mL productivity. Clarified lysates were affinity purified by poros capture AAV9 columns. Eluates of affinity purification were utilized for product quality analysis. (**B**) JMP analysis performed to determine optimal plasmids ratios shows the ratio 5 is the most optimal based on productivity results.

Figure 8. Effect of Molar Plasmid Ratio and Total Plasmid Amounts on the Product Quality of Capsids

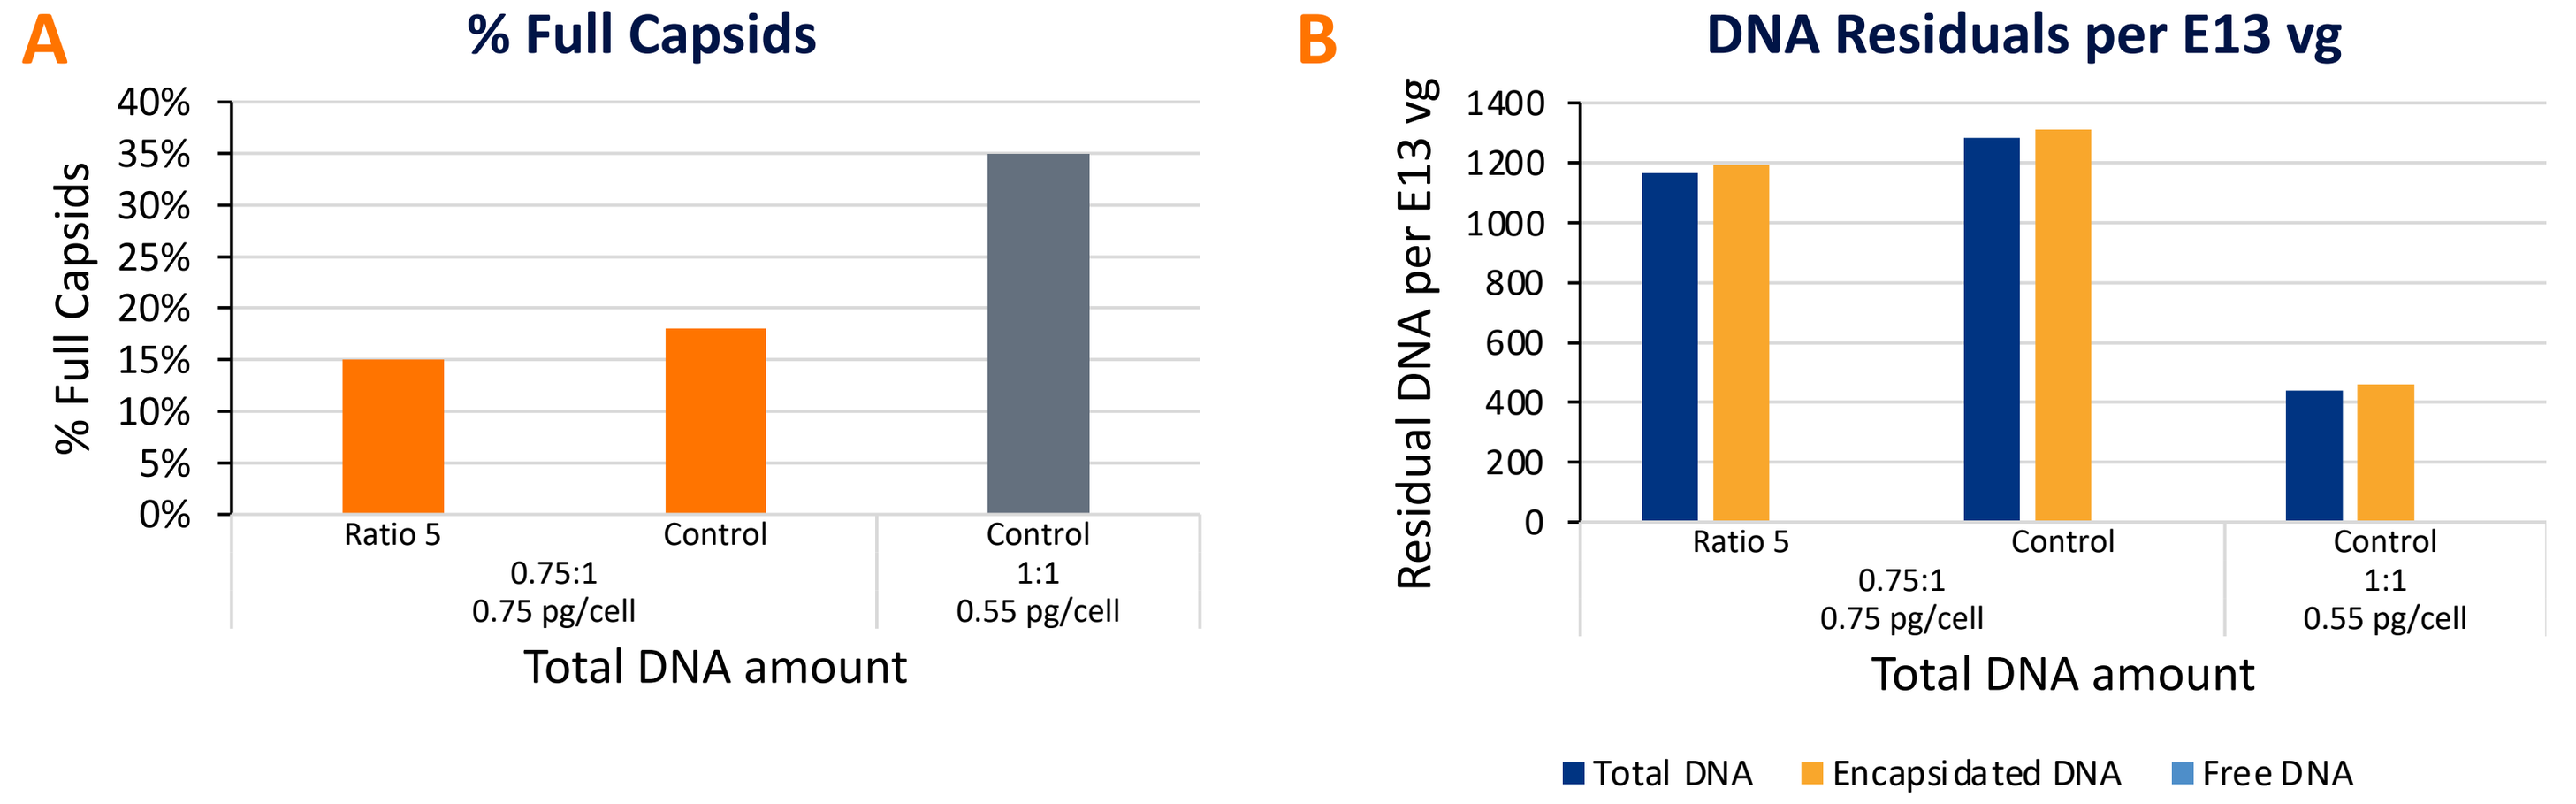
A
% Full Capsids
Total DNA amount
B
DNA Residuals per E13 vg
Residual DNA per E13 vg
Total DNA amount
Total DNA
Encapsidated DNA
Free DNA

(**A**) Affinity column purified material was analyzed for % Full using size exclusion chromatography (SEC-MALS) combined with refractive index (RI). (**B**) DNA residuals were measured using a commercially available quantitative PCR (qPCR)-based system for the detection of HEK293 host-cell DNA. The kit contains HEK genomic DNA standards and primer probes. During sample processing, DNase digestion was performed to differentiate between the nuclease sensitive and nuclease resistant residual DNA.

Figure 9. VYGR-293 Productivity Optimization with the Use of Additives in the Production Process

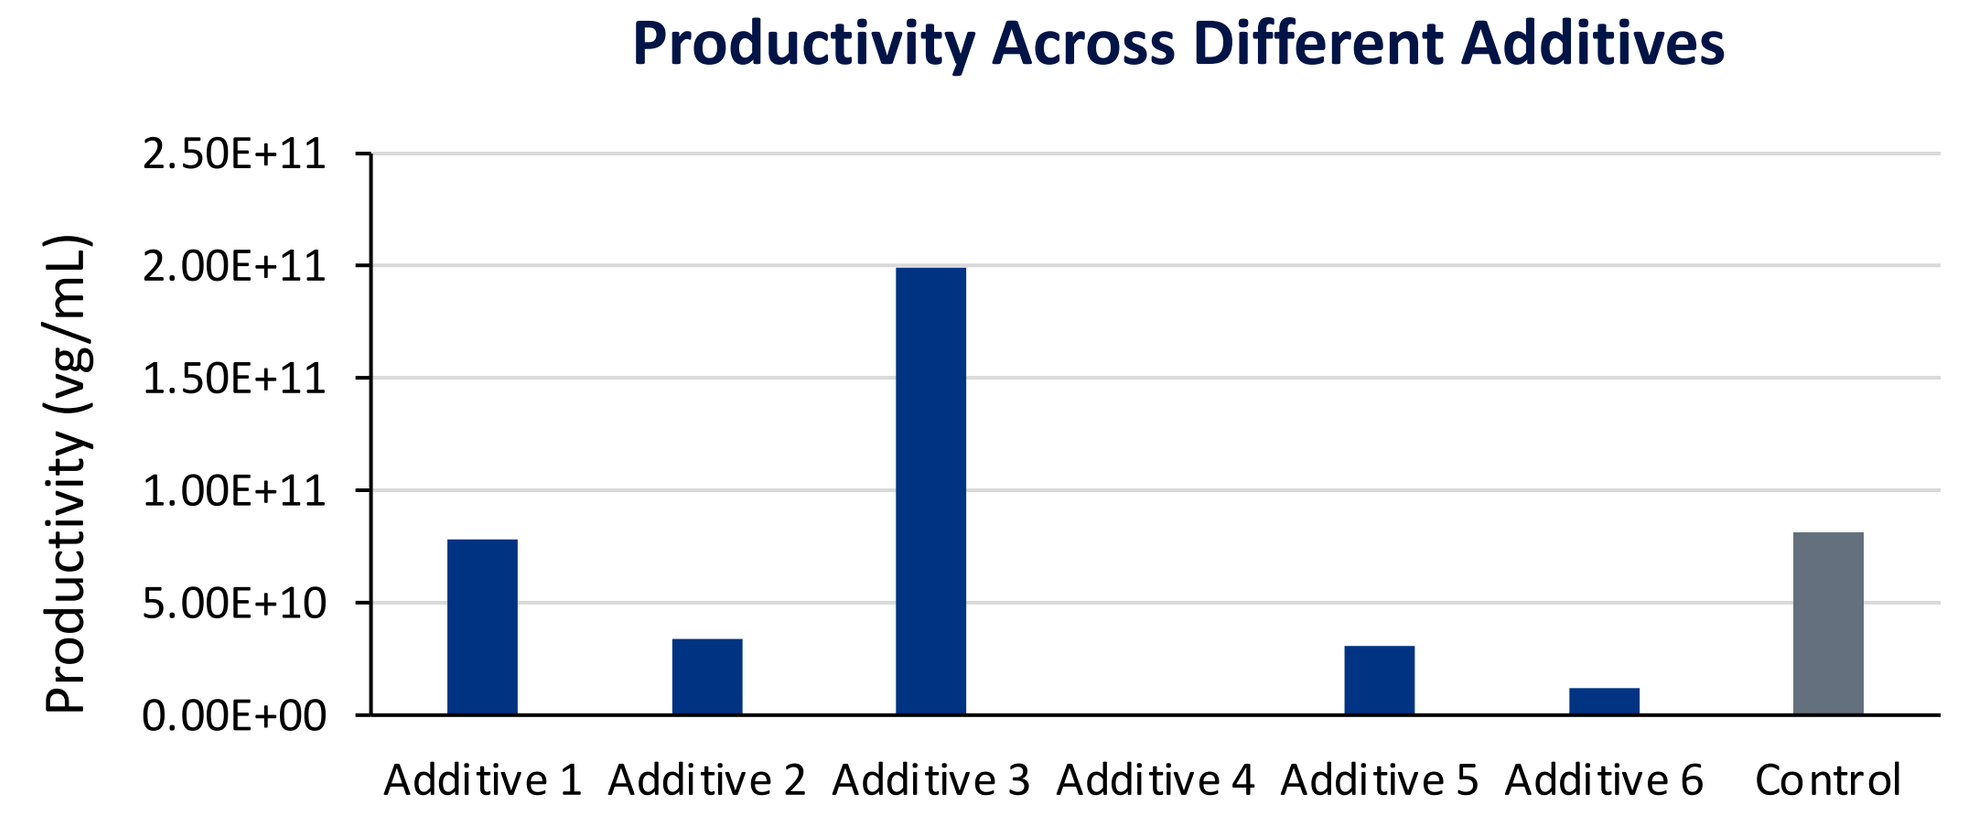
Productivity Across Different Additives
Productivity (vg/mL)
Additive 1
Additive 2
Additive 3
Additive 4
Additive 5
Additive 6
Control

Cells were lysed with Triton-Benzonase and clarified. Clarified lysates were analyzed with ddPCR assay to obtain vector genome/mL productivity.

Figure 10. Bioreactor Confirmation Post Upstream Optimization Strategies

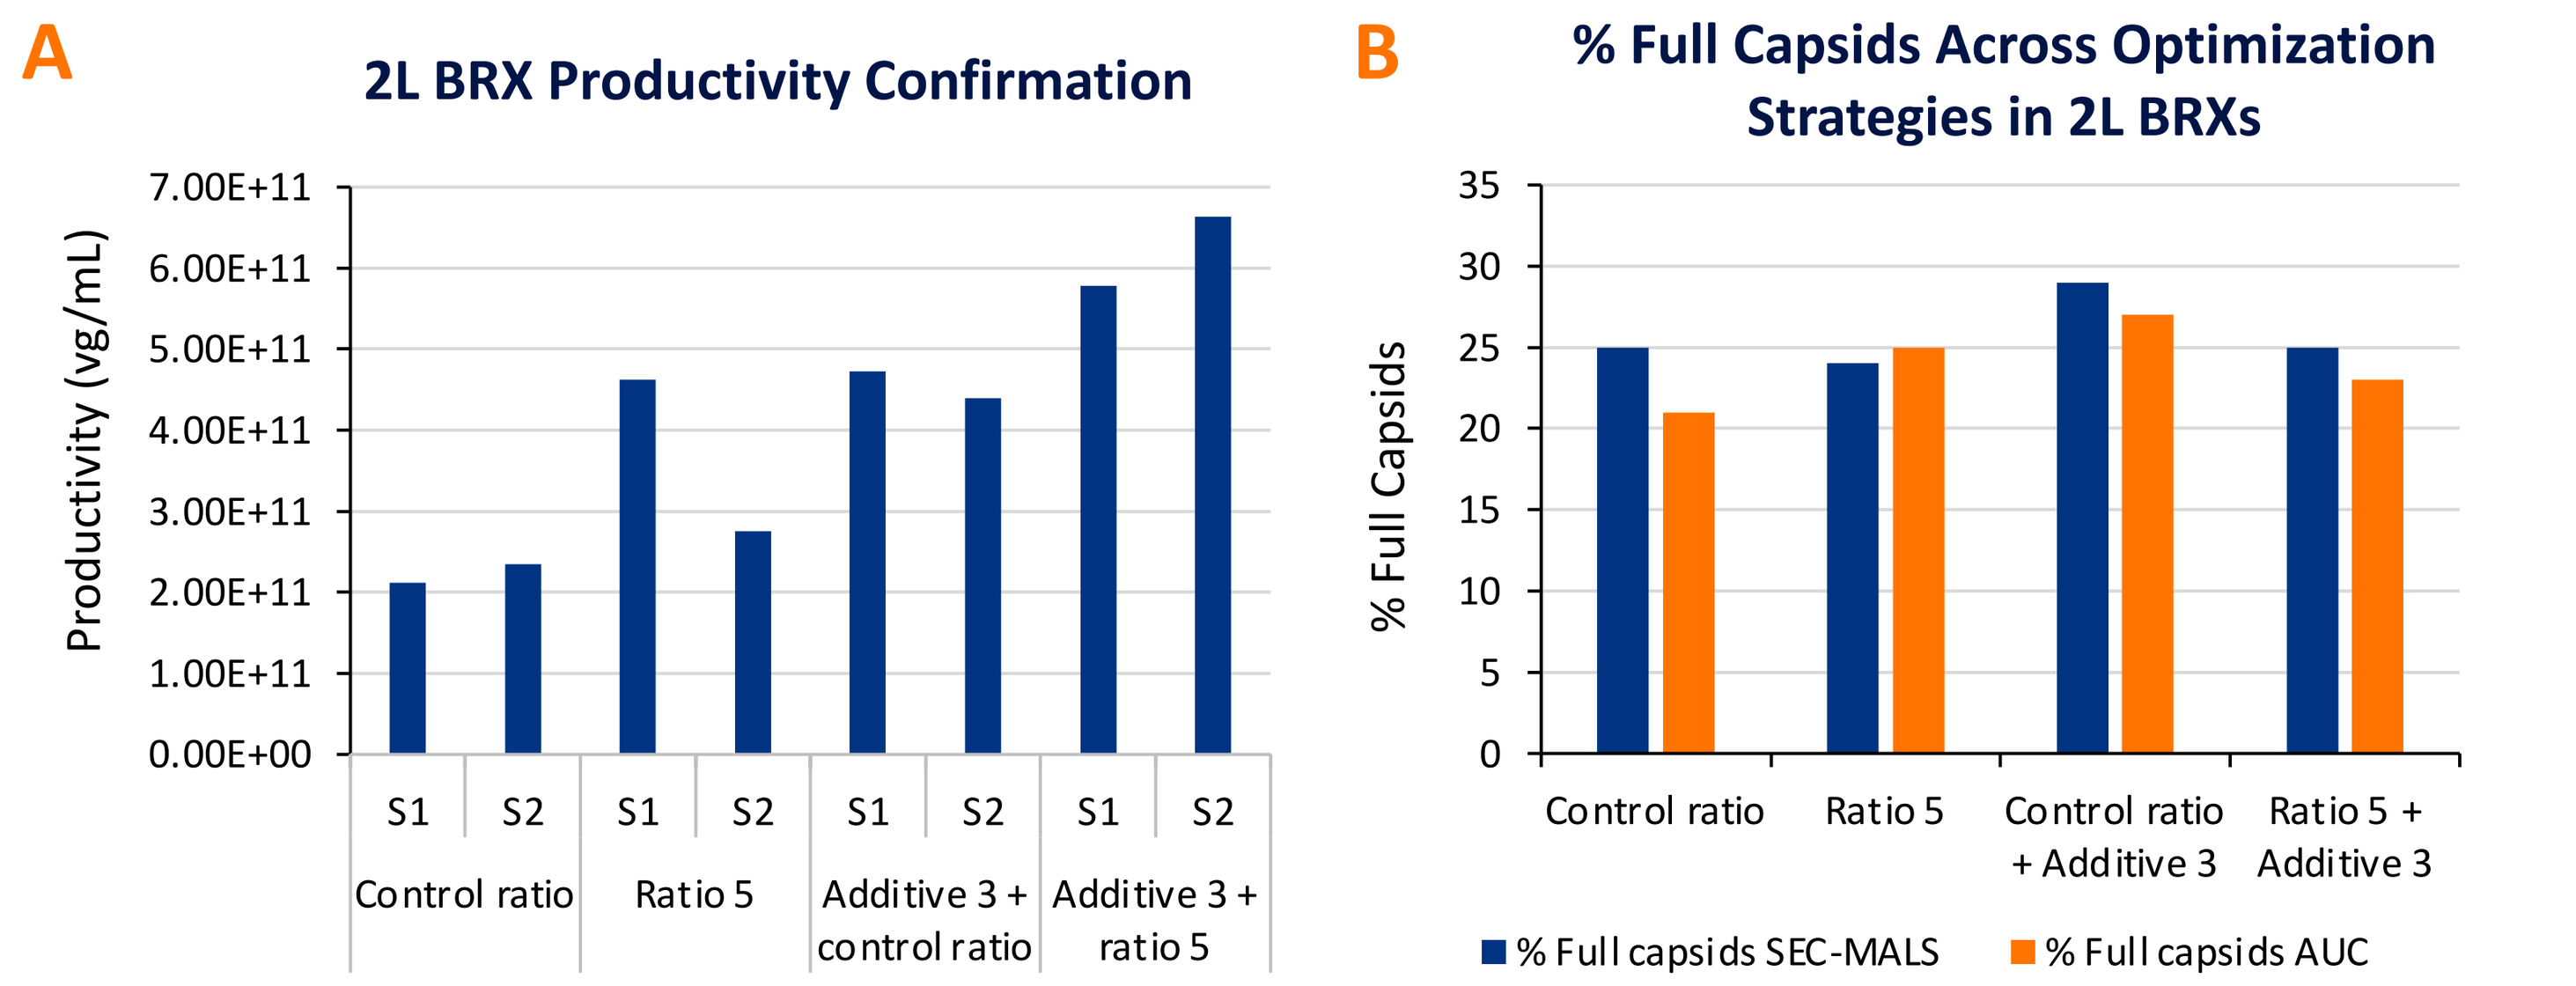
A
2L BRX Productivity Confirmation
Productivity (vg/mL)
Control ratio
Ratio 5
Additive 3 + control ratio
Additive 3 + ratio 5
B
% Full Capsids Across Optimization Strategies in 2L BRXs
% Full Capsids
Control ratio
Ratio 5
Control ratio + Additive 3
Ratio 5 + Additive 3
% Full capsids SEC-MALS
% Full capsids AUC

(**A**) Cells were lysed with Tween - Denarase and clarified. Clarified lysates were analyzed with ddPCR assay to obtain vector genome/mL productivity. (**B**) Affinity column purified material was analyzed for % Full using size exclusion chromatography (SEC-MALS) combined with refractive index (RI).

CONCLUSIONS

- A HEK293 (VYGR-293) cell line was successfully adapted from an adherent serum-fed culture to suspension-based, serum-free culture. The suspension culture had a consistent doubling time passages and a cell bank was created.
- Upstream productivity was optimized by evaluating multiple viable cell densities at transfection.
- The upstream process was further developed by examining optimal N-1 to N split ratios.
- Total plasmid amounts were optimized using DOE studies and it was observed that increasing the total plasmid amounts from 0.55 pg/cell to 0.75 pg/cell has a negative impact on % full capsids and residual HEK DNA.
- Molar plasmid ratios were optimized to improve productivity 2x higher than the control process with little to no negative impact on product quality.
- Upstream productivity was further improved up to 2x higher than control by optimization of adding additives to the production process.
- VYGR-293 2L bioreactor runs showed up to 3x increase in productivity post upstream optimizations with consistent product quality results.
- In conclusion, VYGR-293 cell line was successfully optimized for productivity and product quality results demonstrating that its suitability for the development of an AAV expression platform to produce drug candidates for neurological disorders.
- Further development such as identifying the best clonal selection for the cell line is currently ongoing to improve and optimize the overall productivity.

voyager therapeutics | 75 Hayden Avenue, Lexington, MA 02421 | www.voyagertherapeutics.com

ASGCT 27th Annual Meeting 2024 | May 7 – 11, 2024 | Baltimore, MD