541

One-time Delivery of a Vectorized Anti-amyloid Antibody for Increased and Sustained CNS Expression and Target Engagement

Cassandra Retzlaff, Nivanthika Wimalasena, Roberto Calitri, Dave Rappoli, Alexa Tsolias, Xiaoqin Ren, Ishan Shah, Jordan O'Brien, Jeron Chen, Mathieu Nonnenmacher, Charlotte Chung, Rajeev Sivasankaran

Voyager Therapeutics Inc., Lexington, MA, USA

INTRODUCTION

Alzheimer's disease (AD) is characterized by the accumulation of amyloid plaques, leading to cognitive decline. Clinically, passive administration of anti-amyloid antibodies has shown promise in reducing amyloid plaque burden and improving cognitive function in patients with mild cognitive impairment. However, these therapies are limited by poor blood-brain barrier (BBB) penetration and require frequent administration, which can be burdensome for patients and care partners. Here we show that the use of an adeno-associated viral (AAV) vector to deliver a transgene encoding an anti-amyloid antibody can enable sustained expression of the antibody in the brain after a single injection, especially if the vector is delivered via a BBB-penetrant capsid.

METHODS

***In vivo* Studies:**

- Passive and vectorized administration of an anti-amyloid antibody in WT animals (Figure 1): WT mice were dosed (IP) weekly with a passive anti-amyloid antibody (24mg/kg) for 4 weeks. CNS tissues and plasma were collected at various time points (as indicated below) to assess IgG levels of exposure. An additional cohort of WT mice were injected once (IV) with a vectorized anti-amyloid antibody (2E13vg/kg). CNS and liver tissues and plasma were collected after 4 weeks and assessed for IgG expression.
- Vectorized anti-amyloid antibody administration in a mouse model of Alzheimer's Disease (AD) (Figures 2-4): Transgenic mice were injected one-time (IV) with a vectorized anti-amyloid antibody at a high (4E13vg/kg) or low (2E13vg/kg) dose. Tissues and plasma were collected at either 1, 2, or 4 months (as indicated) and assessed for VG and IgG expression.
- Anti-amyloid antibody was vectorized in a blood barrier penetrating capsid, VOY101.

Quantification of Vector Genomes (VG): Tissue levels of VG copies were assessed by digital droplet PRC (ddPCR) after DNA isolation. The number of VG copies are recorded per diploid cell (VG/DC).

Quantification of IgG Expression: Immunoassay (IgG ELISA) was used to determine the expression levels in soluble tissue homogenates and plasma.

Immuno-histochemical or fluorescent Imaging: CNS tissues were fixed, embedded, sectioned and stained for IgG or amyloid-beta. Immunohistochemical images were stained using DAB chromogenic staining methods, serial sections were stained individually for either IgG or amyloid-beta. Immunofluorescent images were achieved by co-staining IgG and amyloid-beta antibodies within the same section.

Figure 1. Vectorization of an Anti-amyloid Antibody via a BBB-penetrant Capsid Enhances Brain Uptake as Compared to Passive Administration

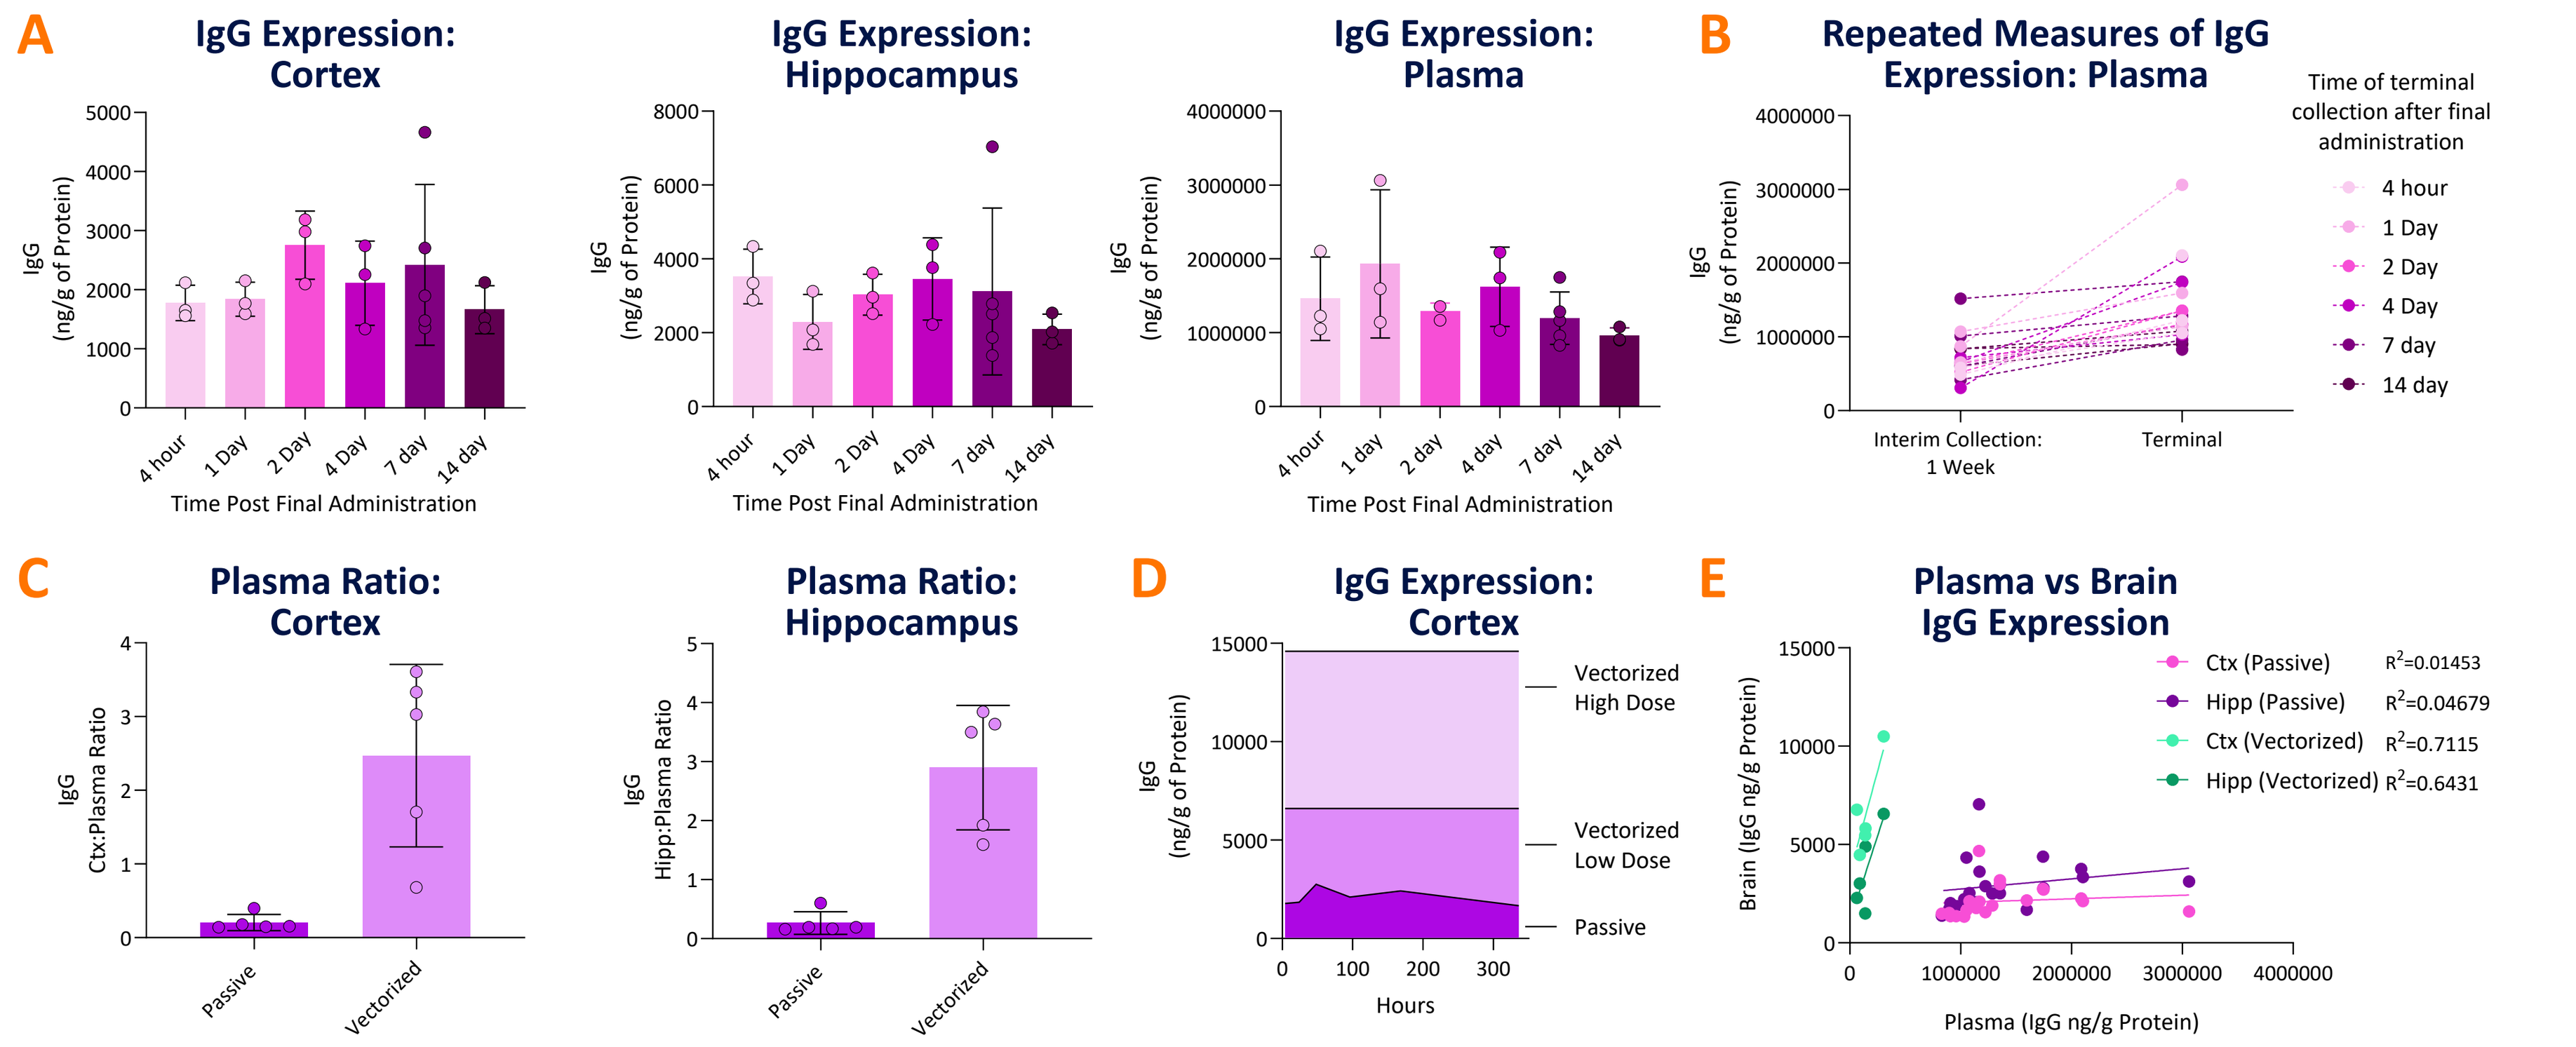
A
IgG Expression: Cortex
IgG Expression: Hippocampus
IgG Expression: Plasma
Time Post Final Administration
B
Repeated Measures of IgG Expression: Plasma
Time of terminal collection after final administration
4 hour
1 Day
2 Day
4 Day
7 day
14 day
Interim Collection: 1 Week
Terminal
C
Plasma Ratio: Cortex
Plasma Ratio: Hippocampus
Passive
Vectorized
D
IgG Expression: Cortex
Vectorized High Dose
Vectorized Low Dose
Passive
Hours
E
Plasma vs Brain IgG Expression
Ctx (Passive) R²=0.01453
Hipp (Passive) R²=0.04679
Ctx (Vectorized) R²=0.7115
Hipp (Vectorized) R²=0.6431
Plasma (IgG ng/g Protein)

A passively administered anti-amyloid antibody was injected weekly for 4 weeks in WT mice before terminal collection of plasma and brain tissues at 4 hours, 1, 2, 4, 7, and 14 Days after final administration. (**A**) IgG expression in cortex, hippocampus, and plasma of passively administered anti-amyloid antibody. (**B**) IgG expression in the plasma from all groups 1 week post the initial injection compared to terminal collection. Elevation of IgG is observed in all groups after 4 weeks of passive administration. (**C**) IgG brain (cortex and hippocampus) to plasma ratios are over 15-fold greater after single dose of the AAV vectorized anti-amyloid antibody payload. Vectorized data are from 4-weeks post a single IV administration. (**D**) Passive administration IgG expression over time compared to 2 doses of vectorized anti-amyloid antibody. Data show higher consistent expression with vectorization. (**E**) Correlation plots from cortex and hippocampus of passively administered or vectorized anti-amyloid antibody.

Figure 2. Broad Biodistribution and IgG Expression in Brain Tissue 4-weeks After a Single IV Injection of Vectorized Anti-amyloid Antibody in a Mouse Model of AD

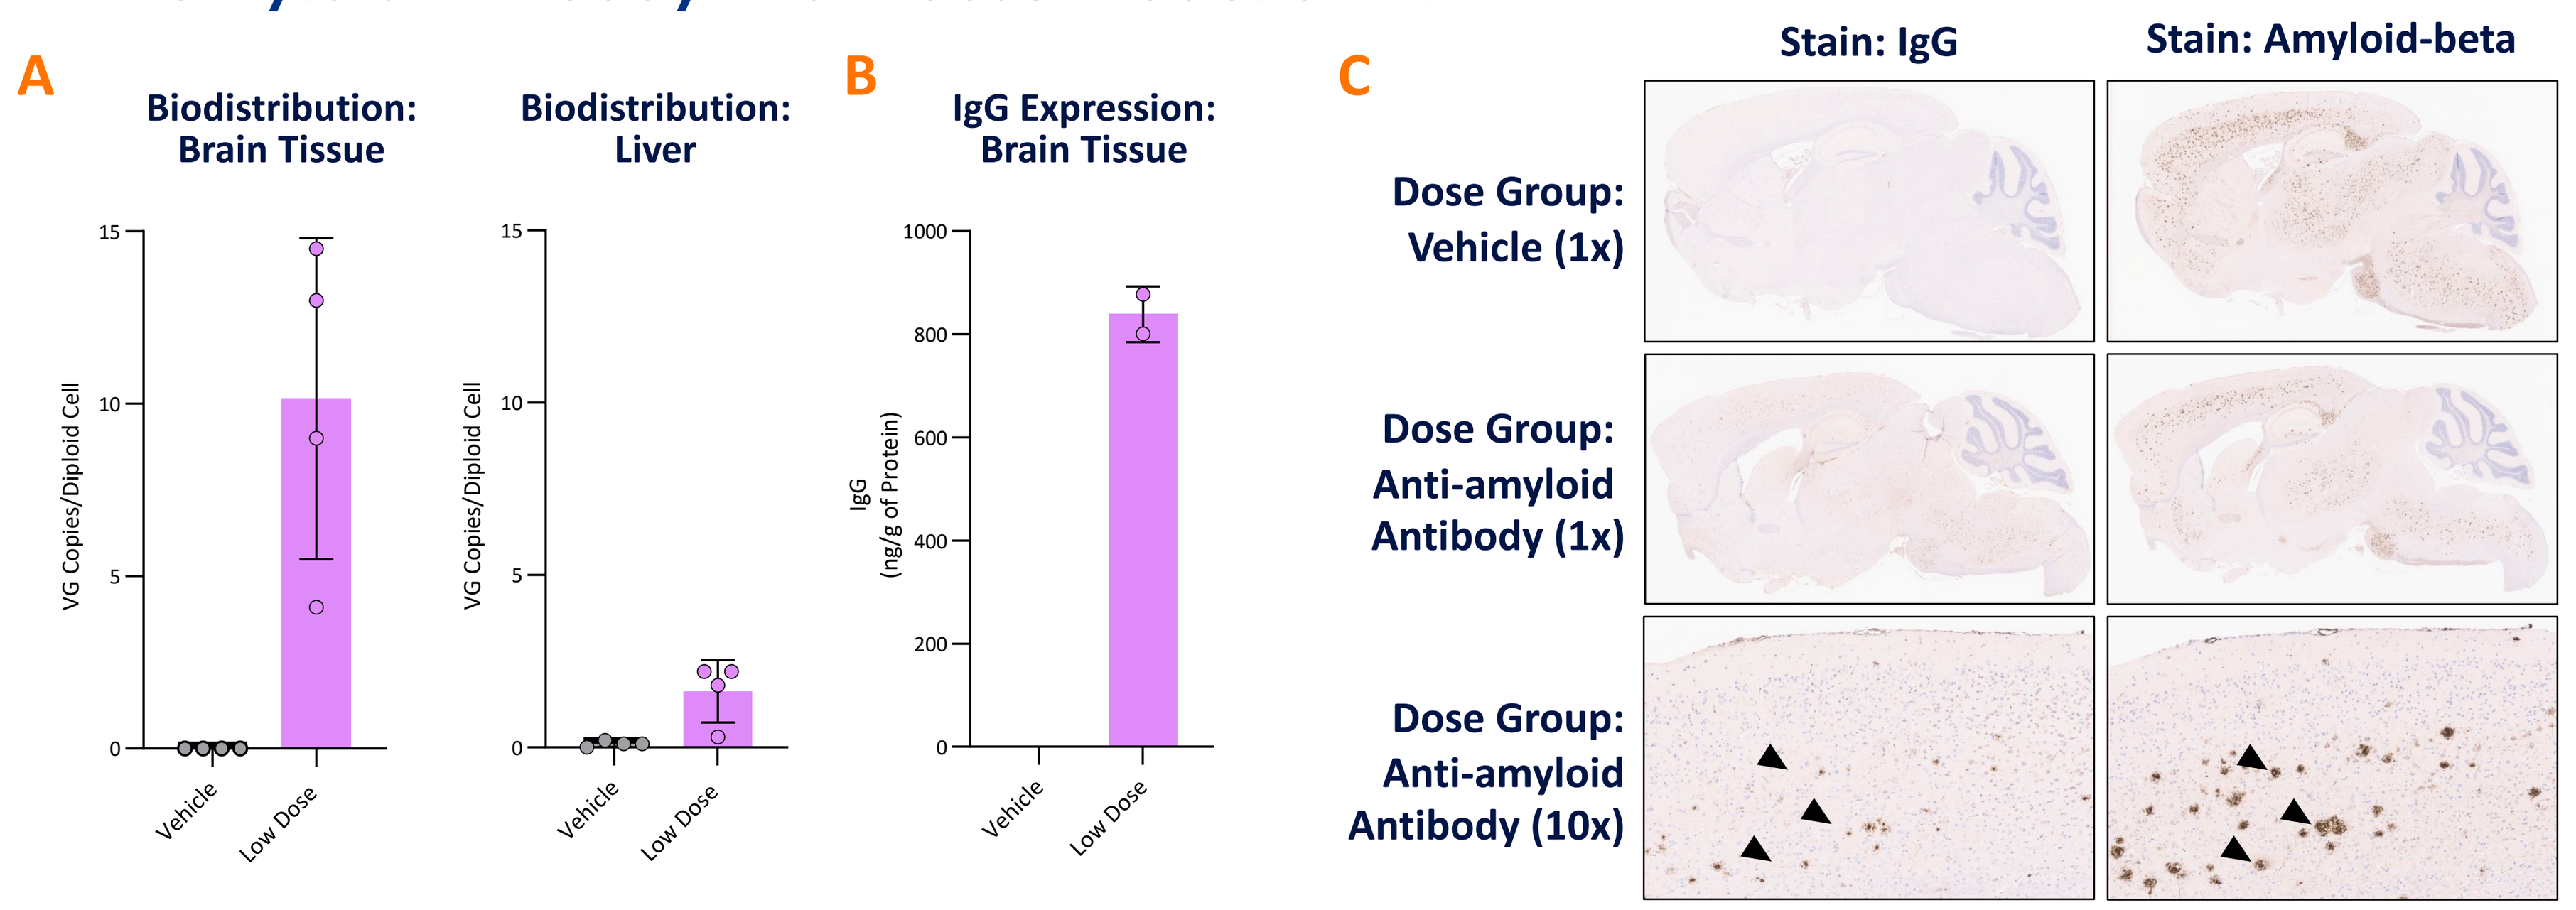
A
Biodistribution: Brain Tissue
Biodistribution: Liver
VG Copies/Diploid Cell
Vehicle
Low Dose
B
IgG Expression: Brain Tissue
C
Stain: IgG
Stain: Amyloid-beta
Dose Group: Vehicle (1x)
Dose Group: Anti-amyloid Antibody (1x)
Dose Group: Anti-amyloid Antibody (10x)

Transgenic animals were injected with a single low dose of a vectorized anti-amyloid antibody to evaluate biodistribution (VG/DC) and IgG protein expression. (**A**) Brain and liver biodistribution (VG/DC) as determined by ddPCR. (**B**) IgG protein expression in brain as determined by ELISA. (**C**) Immunohistochemistry on fixed brains shows overlapping expression of IgG and amyloid-beta in serial sections. This suggests target engagement of the delivered anti-amyloid antibody payload with amyloid-beta plaques. Arrows indicate representative points of overlapping expression of IgG and amyloid-beta.

Figure 3. IV Delivery of Vectorized Anti-amyloid Antibody Shows Sustained Expression of IgG in the Brain After 4 Months

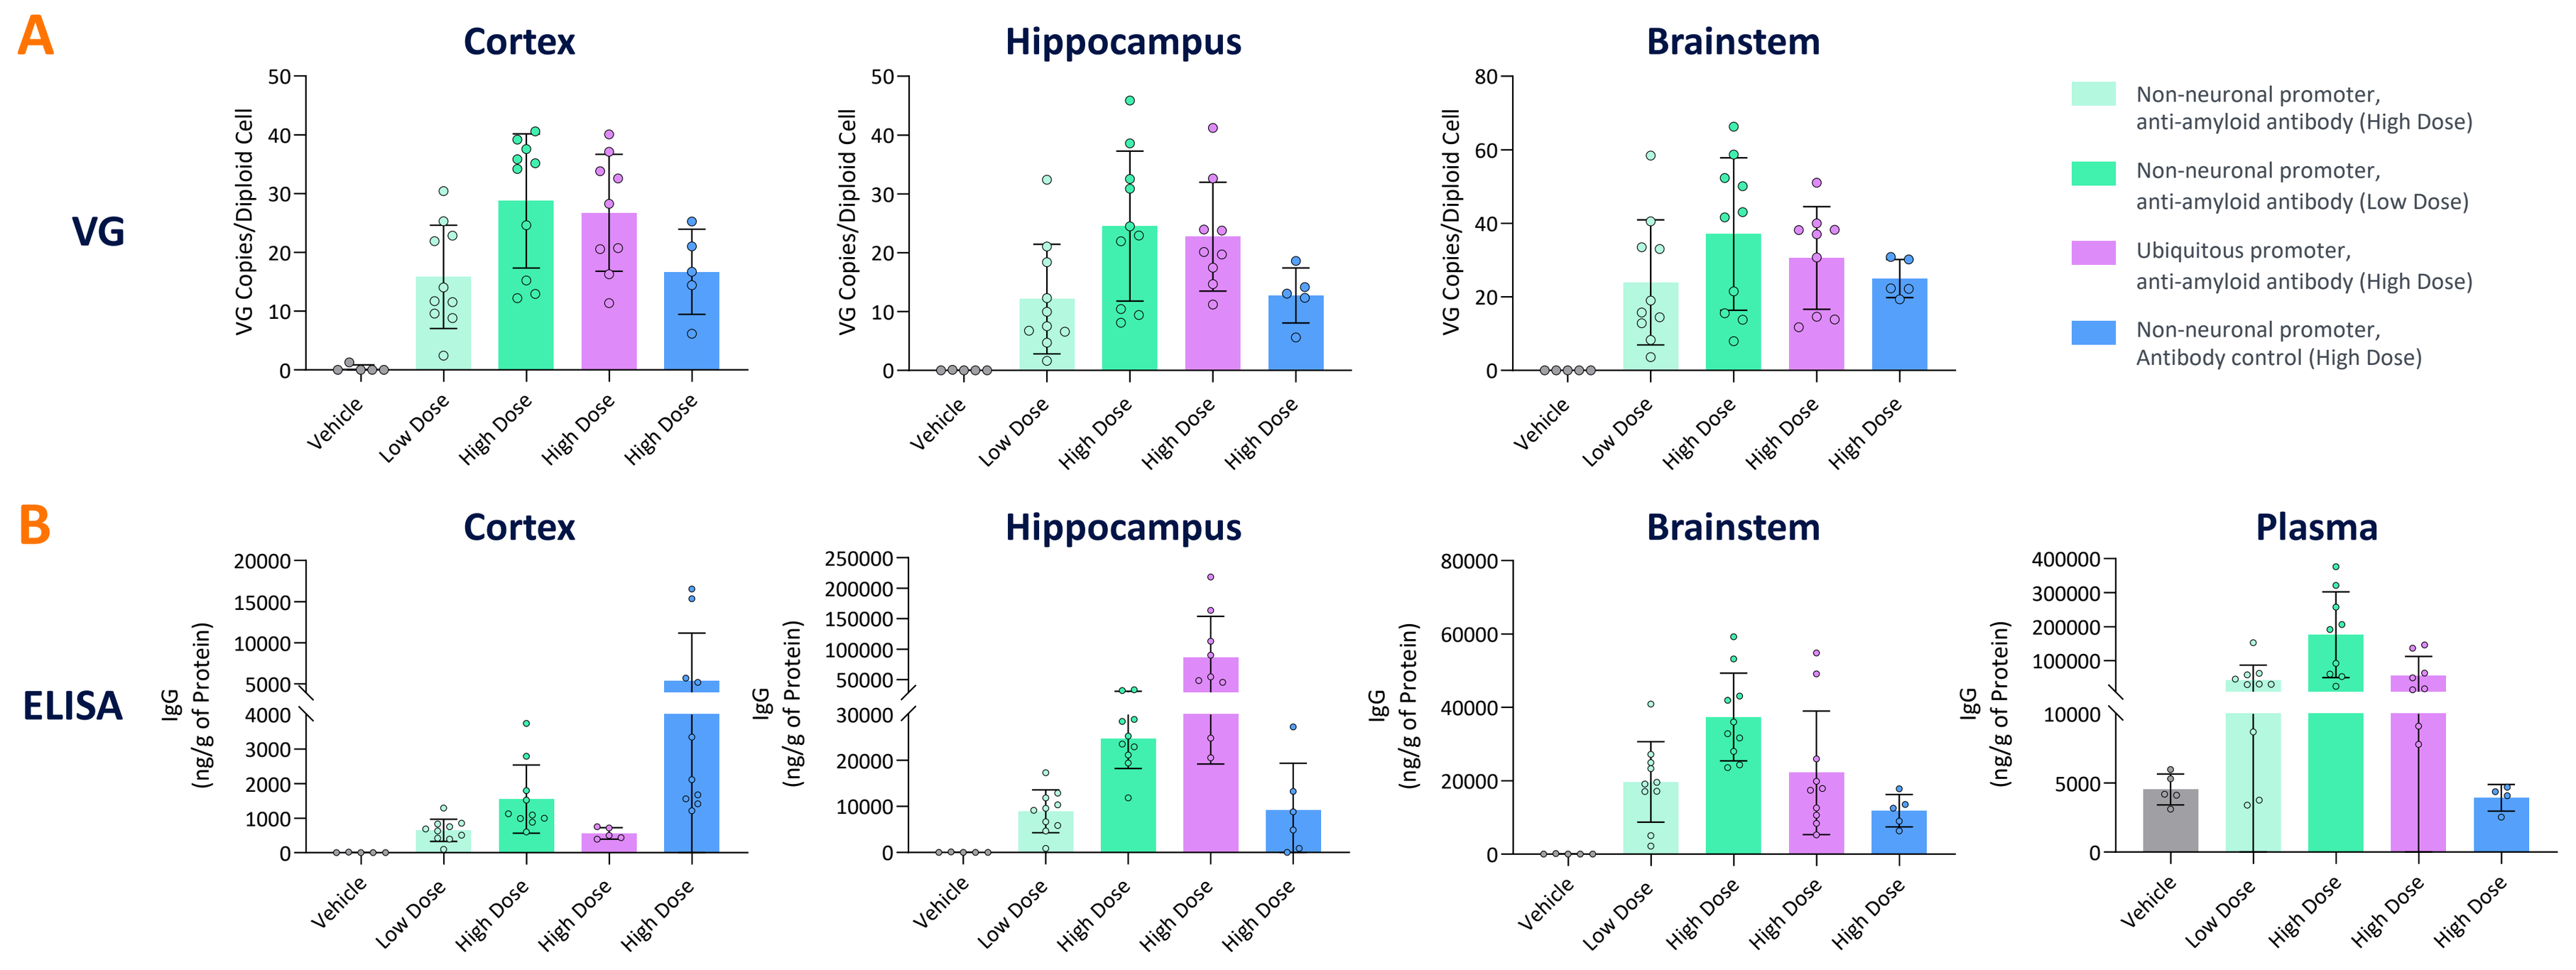
A
VG
Cortex
Hippocampus
Brainstem
B
ELISA
Plasma
Vehicle
Low Dose
High Dose
Non-neuronal promoter, anti-amyloid antibody (High Dose)
Non-neuronal promoter, anti-amyloid antibody (Low Dose)
Ubiquitous promoter, anti-amyloid antibody (High Dose)
Non-neuronal promoter, Antibody control (High Dose)

Sustained biodistribution and IgG protein expression up to 4 months post a single IV administration of vectorized anti-amyloid antibody in a mouse model of Alzheimer's Disease. Animals were injected IV in four dose groups: vehicle, vectorized anti-amyloid antibody under either a ubiquitous or non-neuronal promoter (2 doses), and a vectorized antibody control. (**A**) Cortex, hippocampus, and brainstem show trends towards dose dependent biodistribution (VG/DC) after 4 months in-life. (**B**) IgG expression in cortex, hippocampus, brainstem, and plasma show trends toward dose dependent expression of anti-amyloid antibody after 4 months in-life.

Figure 4. IV Delivery of Vectorized Anti-amyloid Antibody Shows Qualitative Reduction in Amyloid-beta After 2 and 4 Months in a Mouse Model of Alzheimer's Disease

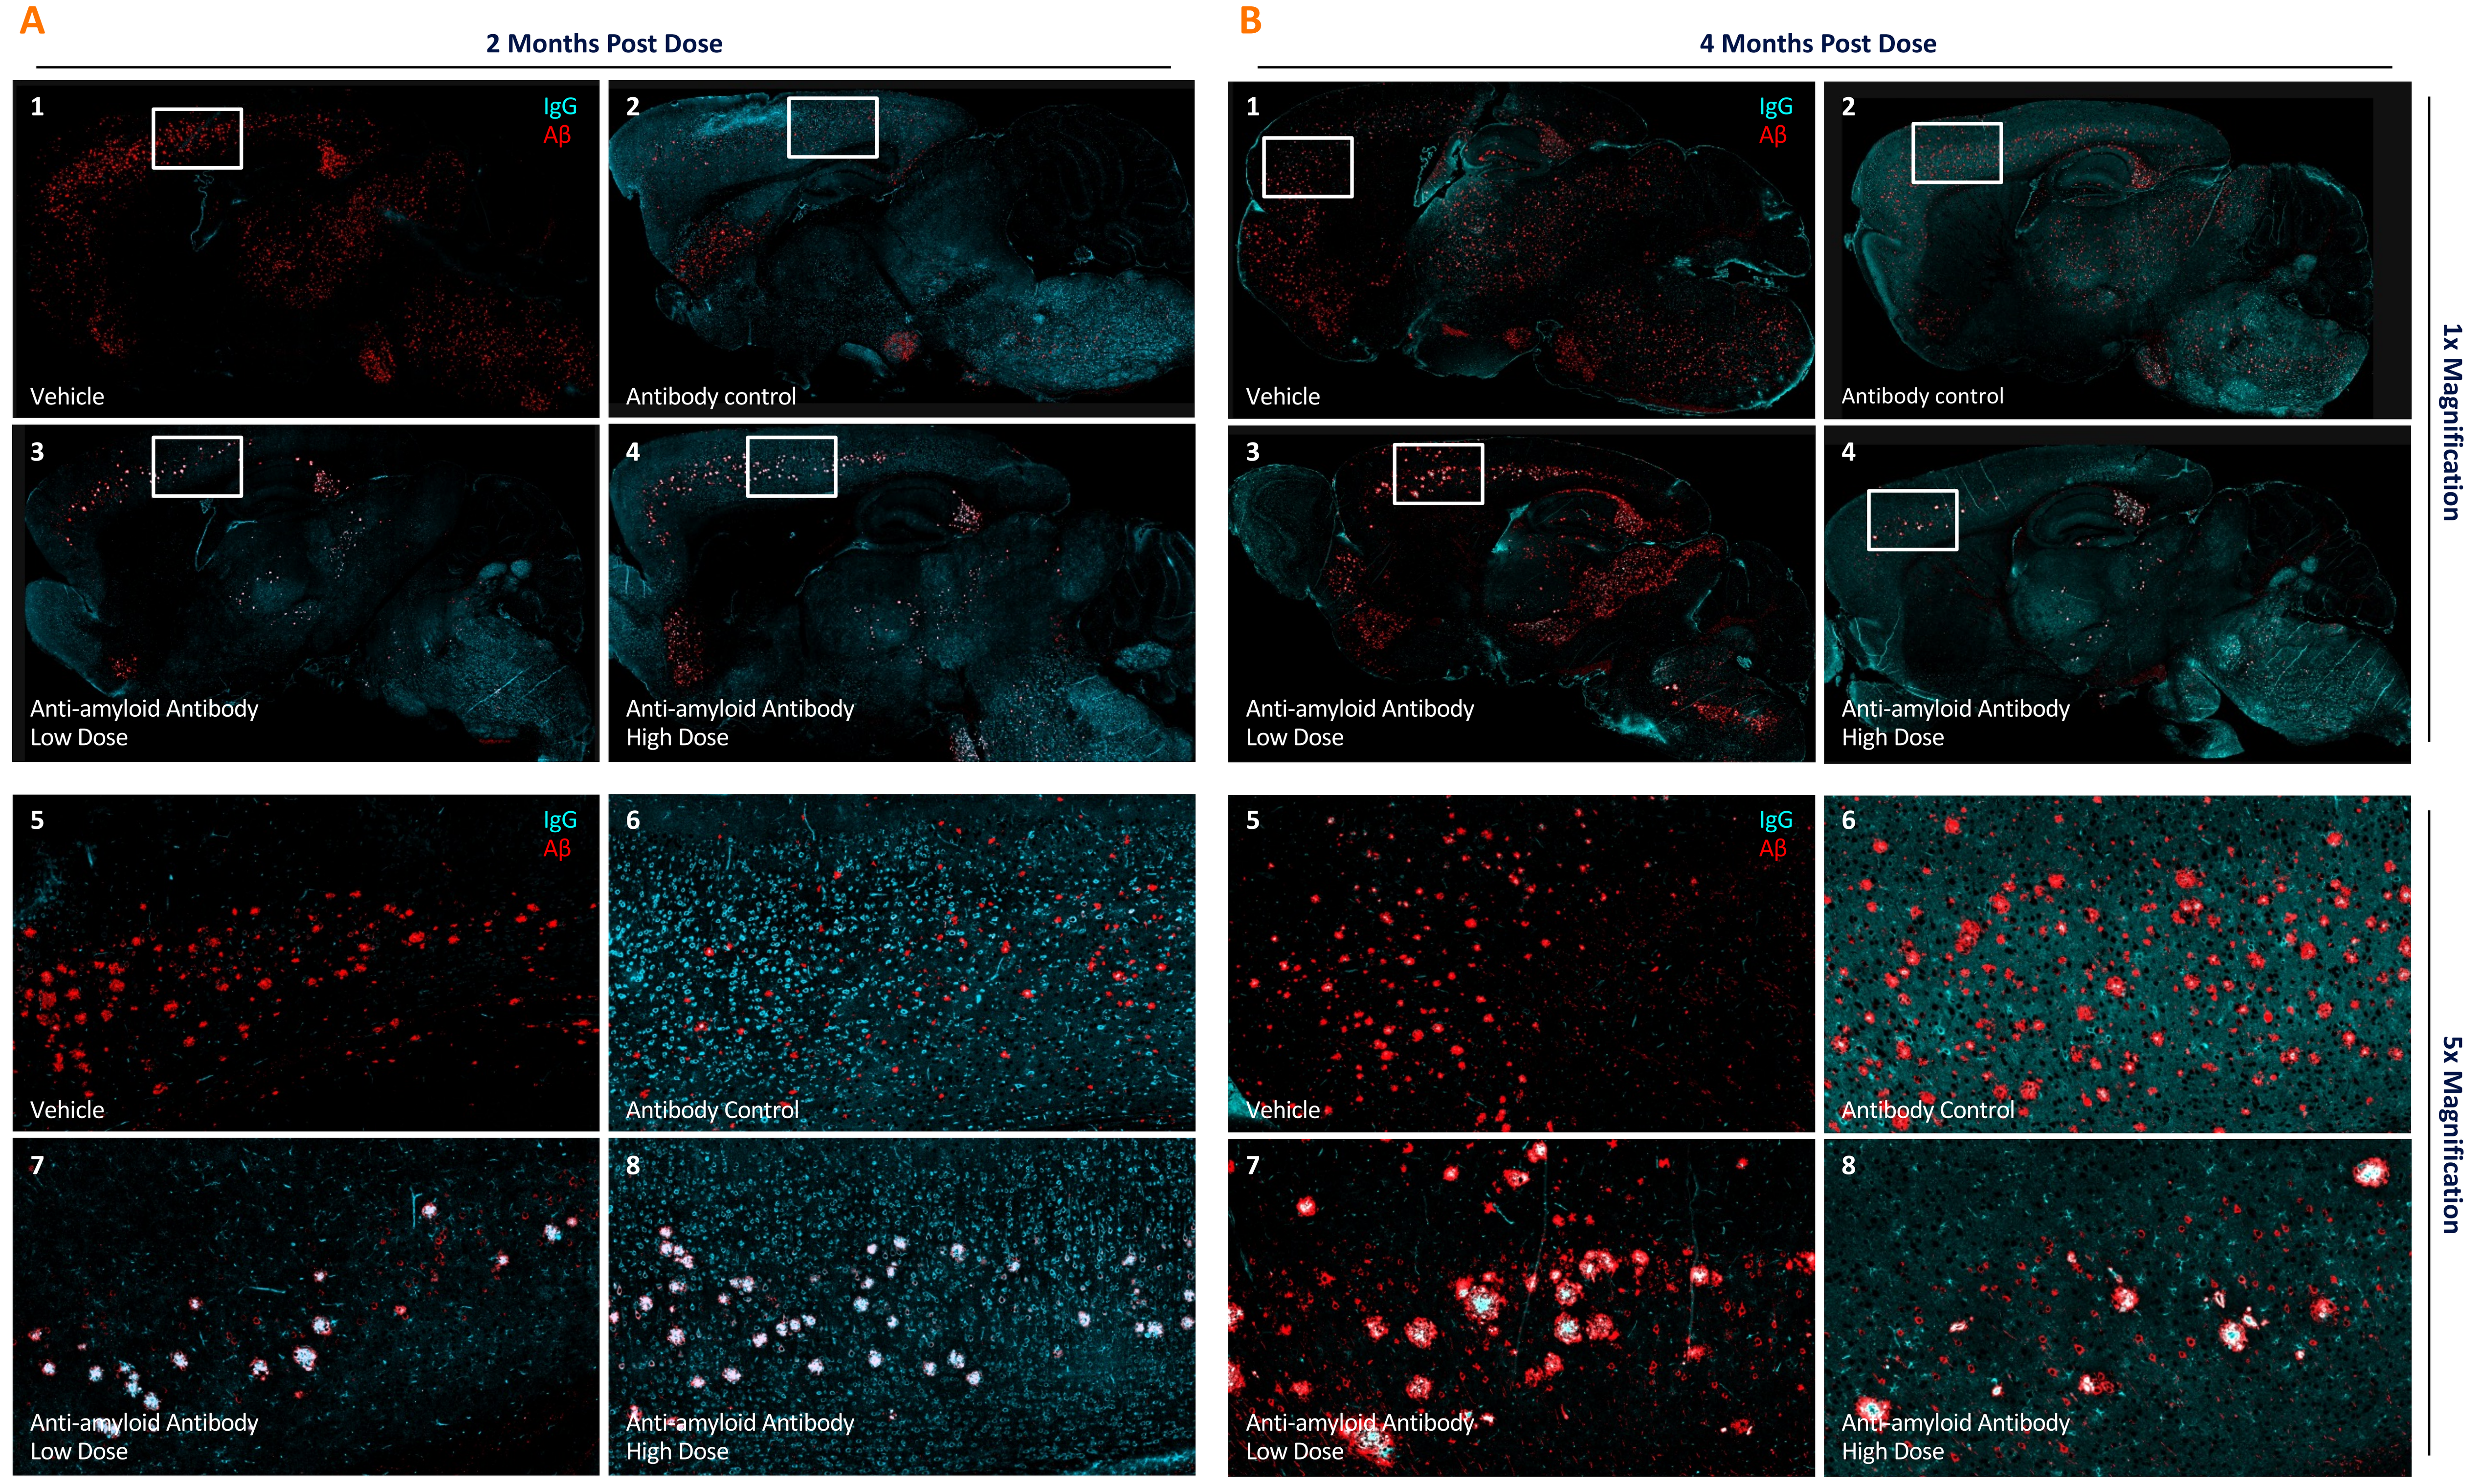
A
2 Months Post Dose
B
4 Months Post Dose
IgG
Aβ
Vehicle
Antibody control
Anti-amyloid Antibody Low Dose
Anti-amyloid Antibody High Dose
1x Magnification
5x Magnification

Immunofluorescence reveals qualitative reduction in amyloid-beta up to 4 months post single IV injection of vectorized anti-amyloid antibody in a mouse model of Alzheimer's Disease. All dose groups (except vehicle) received a vectorized antibody with expression driven by a non-neuronal promoter. (**A**) Immunofluorescence images of IgG and amyloid-beta at two months post administration. Images 1-4 show whole brain sagittal slices at 1x magnification. Images 5-8 show 5x magnification of the cortex. The magnified images highlight the apparent reduction and target engagement in amyloid-beta detection after administration of a vectorized anti-amyloid antibody. (**B**) Immunofluorescence images of IgG and amyloid-beta at four months post administration. Images 1-4 show whole brain sagittal slices at 1x magnification. Images 5-8 show 5x magnification of the cortex. The magnified images highlight further apparent reduction at 4 months in amyloid-beta detection after a single administration of a vectorized anti-amyloid antibody.

CONCLUSIONS

- In wildtype mice a vectorized anti-amyloid antibody packaged in a BBB penetrant capsid has over 15-fold greater brain:plasma ratios as compared to a passively administered antibody over 4 weeks.
- In a transgenic mouse model of AD, there is broad biodistribution of vector genomes and sustained antibody expression in the brain after a 1-month in-life period. Immunohistochemical evidence in the cortex and hippocampus reveal that the vectorized antibody co-localizes with amyloid-beta plaques, indicating successful target engagement.
- In a transgenic mouse model of AD, there is sustained biodistribution of vector genomes and antibody expression in the brain over a 4-month period. Immunofluorescence analysis of the cortex and hippocampus reveal high degree of co-localization of the vectorized antibody and amyloid-beta. Qualitatively, there is a notable reduction in amyloid-beta staining after a 1-time administration of a vectorized anti-amyloid beta antibody.
- These results demonstrate that vectorization of an anti-amyloid antibody using a BBB penetrant capsid offers a promising strategy for sustained and enhanced, brain-wide antibody expression, overcoming the limitations of passive antibody therapies in AD.

75 Hayden Avenue, Lexington, MA 02421 | www.voyagertherapeutics.com

ASGCT 28th Annual Meeting 2024 | May 13 – 17, 2025 | New Orleans, LA