Intravenous Delivery of VY1706, a CNS Penetrant AAV Gene Therapy for Tauopathies, Provides Broad Tau Lowering in Mouse Models of Tauopathy and Non-Human Primates

Rajeev Sivasankaran, Hechen Bao, Maneesha A. Paranjpe, Cansu Colpan, Jeffrey Thompson, Ching-Lien Fang, Yasuhiro Ito, Alexandra Tsolias, Nilesh Pande, Anupriya Kulkarni, Munachiso Egbuchulam, Joydip Kundu, Su Jing Chan, Bridget Shaughnessy, Shiron J. Lee, Joe Clement, Camila Arce, Rebecca Spellman, Wenya Yang, Rong Zhao, Amy Bruce, Jiachen Liu, Manonmani Kumar, Gretchen Rivera, Tyler Moyer, Dillon Kavanagh, Jordan Routhier, Jack Studer, Dhiral Shah, Kyle Grant, Matteo Placidi, Weitong Chen, Johnny Yao, Mathieu Nonnenmacher, Dinah W.Y. Sah, Todd Carter, Wencheng Liu

Voyager Therapeutics Inc., Lexington, MA, USA

voyager therapeutics

INTRODUCTION

Tau pathology is a hallmark of tauopathies, including Alzheimer's disease (AD) and progressive supranuclear palsy (PSP). In AD, the progression of tau pathology in the brain is strongly correlated with cognitive decline. PSP is a rare, progressive neurodegenerative movement disorder, neuropathologically defined by the accumulation of subcortical tau tangles in neurons and glia, within the frontal cortex, basal ganglia, and brainstem. Despite differences in regional vulnerability and clinical presentation, both AD and PSP share a common pathogenic mechanism driven by aberrant tau aggregation and propagation. Antisense oligonucleotide (ASO) approaches targeting tau silencing have shown promise in the clinic but require repeated intrathecal or intraventricular administration.

We have developed VY1706, a blood-brain barrier-penetrant TRACER™-derived Gen2-capsid carrying a potent tau-targeting siRNA. In the present study, we evaluated whether VY1706 could serve as a therapeutic candidate for both AD and PSP based on biodistribution and pharmacological activity in disease-relevant brain regions in NHPs following a single IV dose.

Methods: Quantitation of VG levels by ddPCR, Tau mRNA by qPCR and Tau protein by MSD and AlphaLISA. BaseScope™ chromogenic duplex staining.

Results: The results demonstrated a dose-dependent increase in biodistribution with concomitant tau mRNA and protein knockdown, in AD and PSP relevant brain regions of NHP treated with a single IV dose of VY1706. Additionally, VY1706 shows broad CNS cellular distribution to neurons and glia making it well suited for both AD and PSP.

Summary: The extensive biodistribution of VY1706 into AD and PSP relevant brain regions, together with its robust pharmacological activity, supports its potential as a promising one-time, IV treatment strategy for these Tauopathies.

Figure 1. On-target Potency, *in vitro* Activity of siRNA1

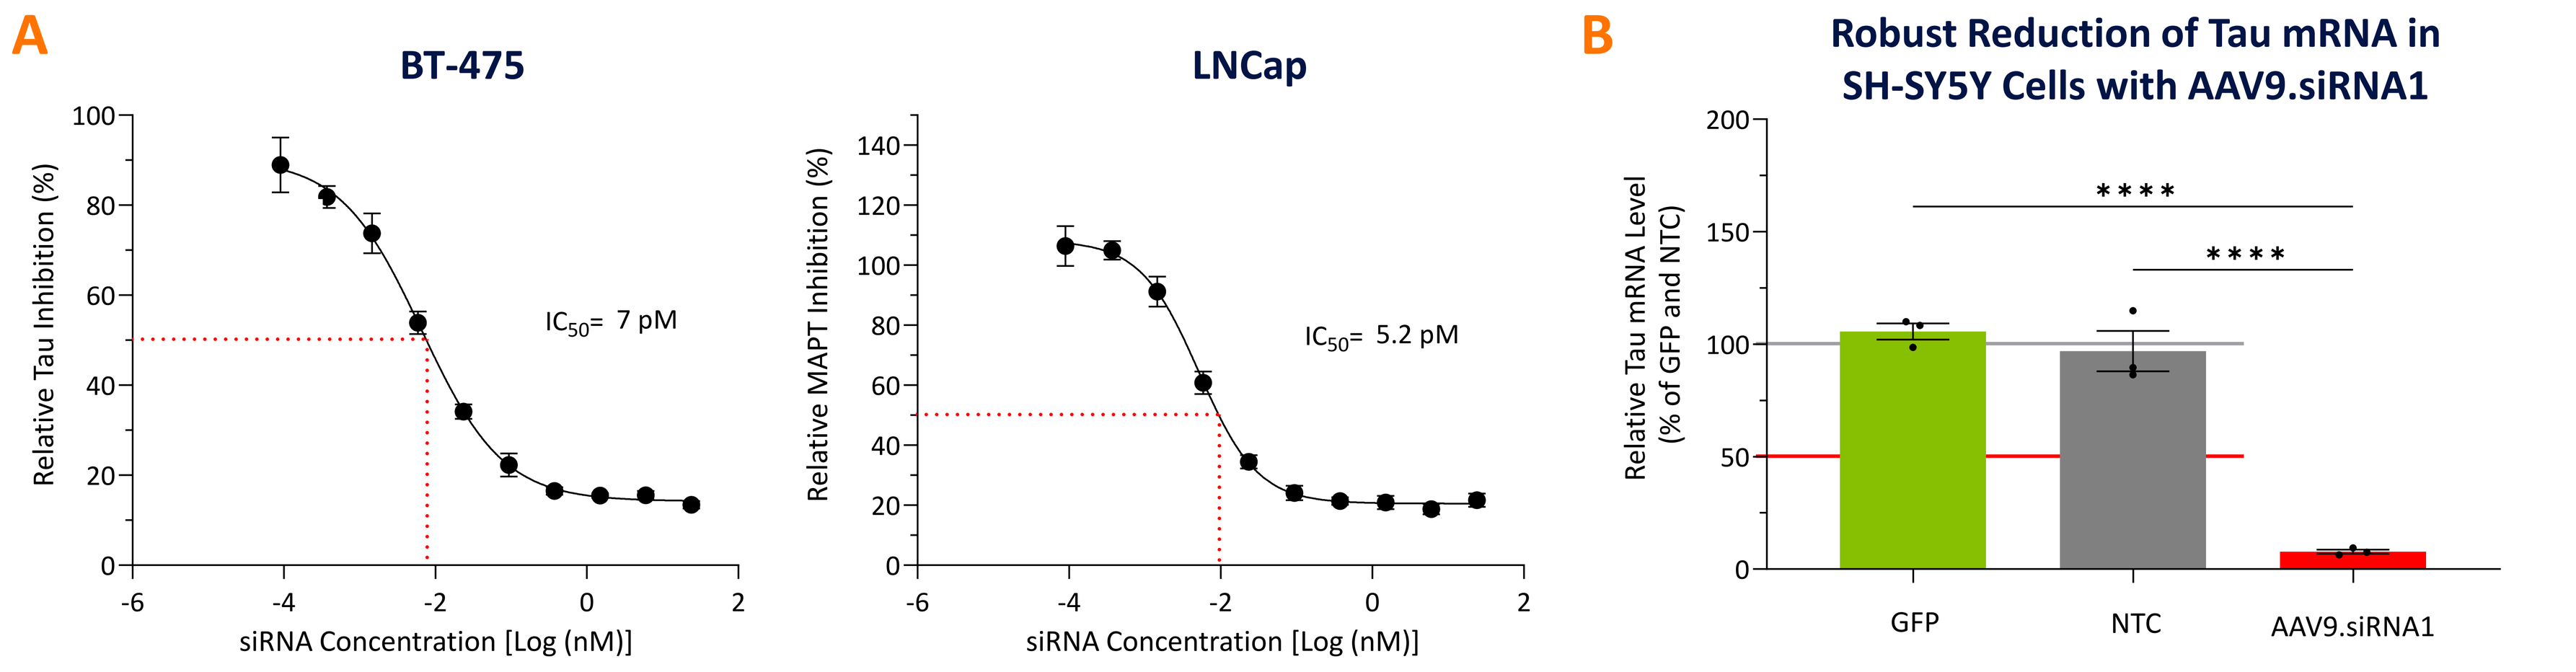
A
BT-475
LNCap
Relative Tau Inhibition (%)
Relative MAPT Inhibition (%)
siRNA Concentration [Log (nM)]
IC_{50}= 7 pM
IC_{50}= 5.2 pM
B
Robust Reduction of Tau mRNA in SH-SY5Y Cells with AAV9.siRNA1
Relative Tau mRNA Level (% of GFP and NTC)
GFP
NTC
AAV9.siRNA1

siRNA1 demonstrated robust potency in tau mRNA knockdown in various cell lines. (**A**) A ten-point dose-response experiment was conducted with siRNA1 in the BT-474 cells (Top), and LNCap (bottom). The results demonstrated dose-dependent lowering of Tau mRNA with an IC_{50} ranging from 5-7 pM and maximal reduction by ~90%. Tau mRNA levels were normalized to GAPDH mRNA levels and then further normalized to the mock control. (**B**) Robust 92% reduction of Tau mRNA was observed in SH-SY5Y cells with vectorized siRNA1 delivered with AAV9 (1E5 MOI). Tau mRNA levels were normalized to TATA-box binding protein (TBP) mRNA levels and then further normalized to the GFP and NTC controls. Statistical significance was evaluated with a one-way ANOVA and Tukey's multiple comparisons post-hoc test; *, **, ***, and **** indicate p < 0.05, 0.005, 0.0005 and 0.0001, respectively. Data are shown as the group mean ± SEM. N=6 per group.

Figure 2. Dose Response Pharmacology and Efficacy of VY1706 in Mouse Models

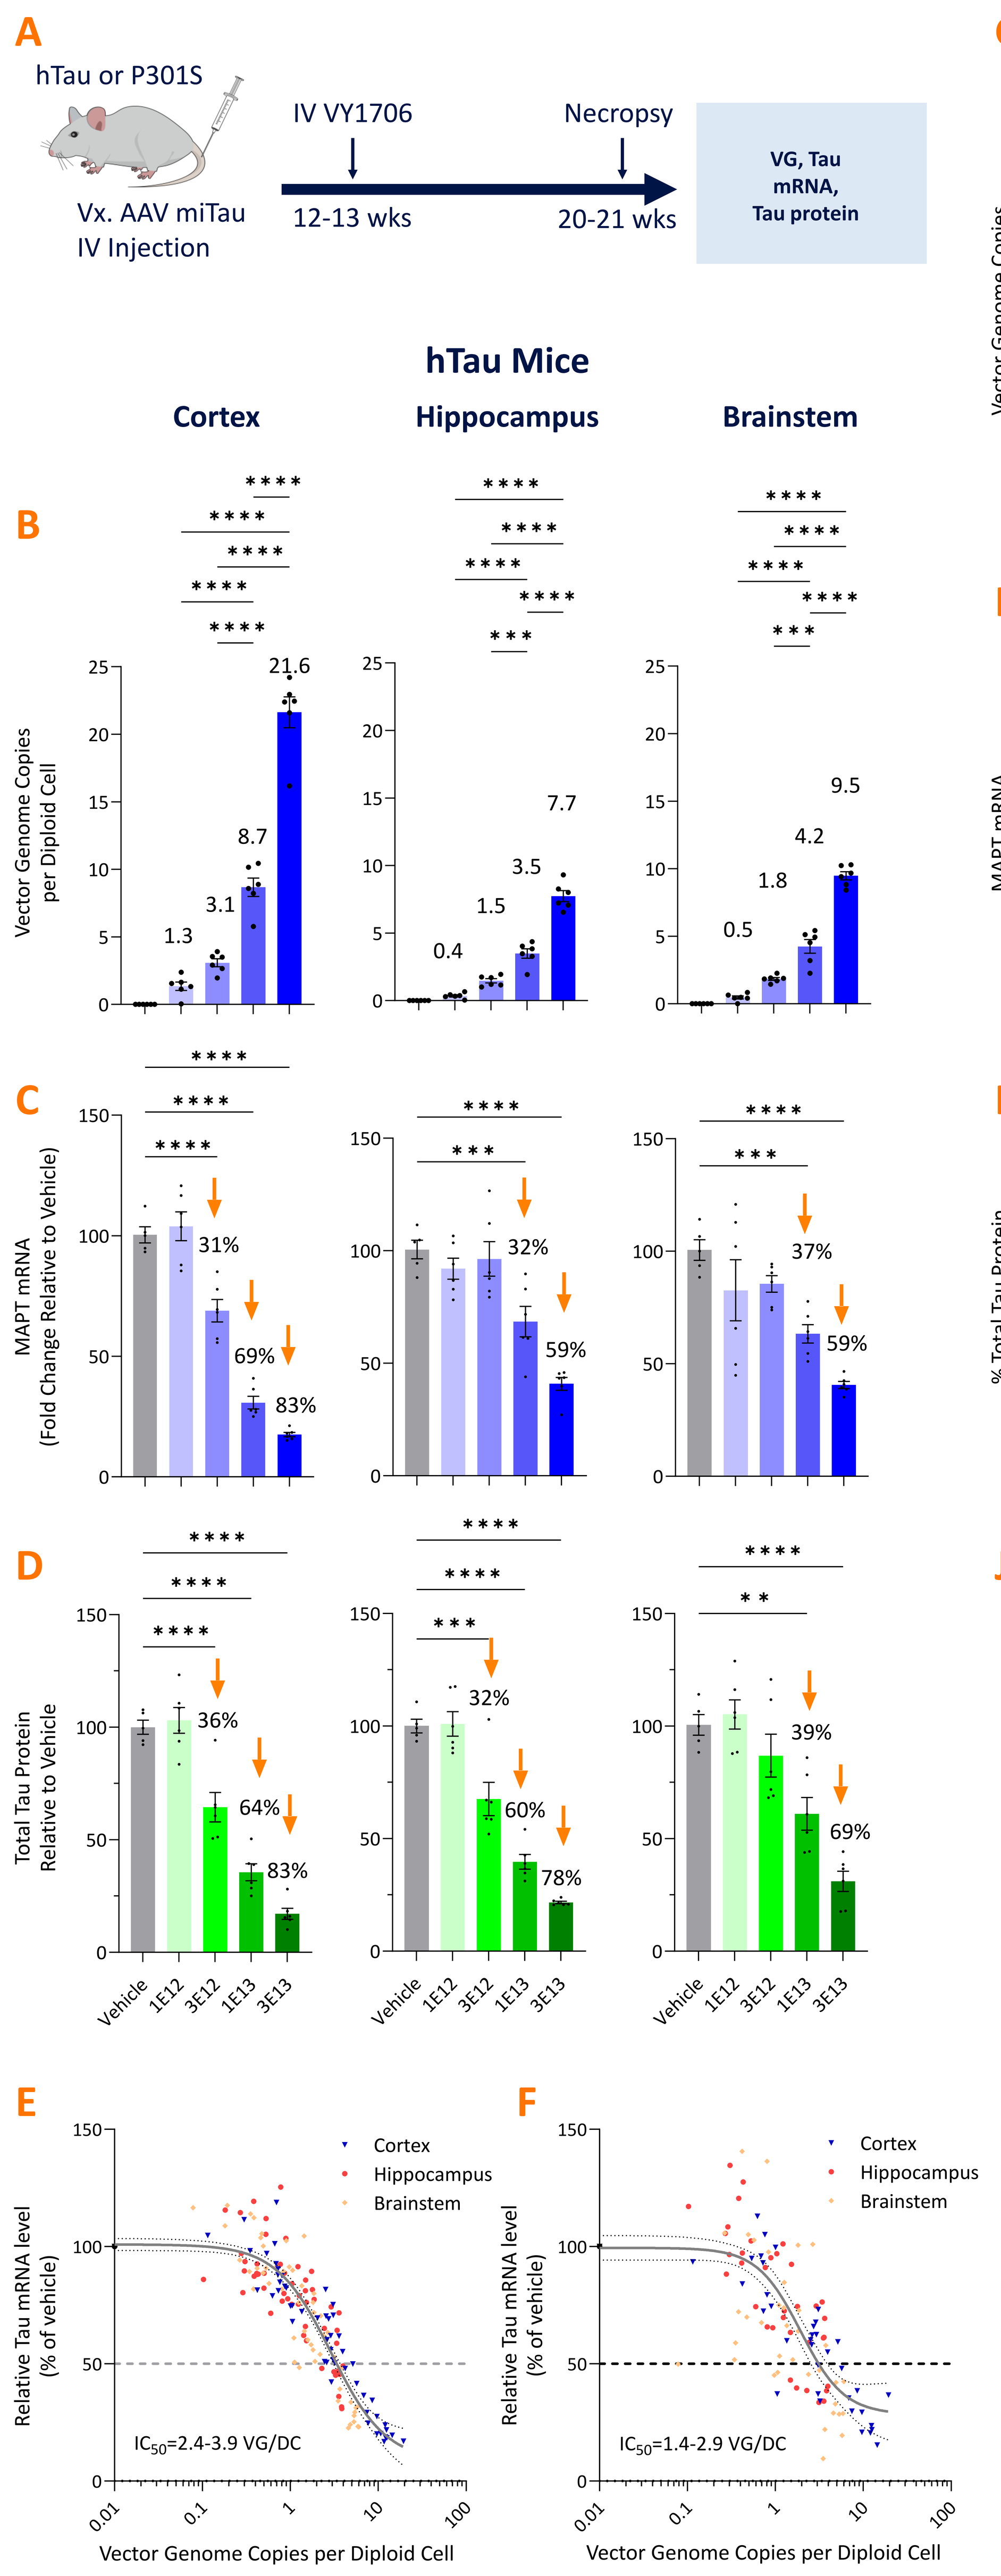
A
hTau or P301S
Vx. AAV miTau
IV Injection
IV VY1706
Necropsy
12-13 wks
20-21 wks
VG, Tau mRNA, Tau protein
hTau Mice
Cortex
Hippocampus
Brainstem
B
Vector Genome Copies per Diploid Cell
C
MAPT mRNA (Fold Change Relative to Vehicle)
D
Total Tau Protein Relative to Vehicle
Vehicle
1E12
3E12
1E13
3E13
E
F
Cortex
Hippocampus
Brainstem
Relative Tau mRNA level (% of vehicle)
IC_{50}=2.4-3.9 VG/DC
IC_{50}=1.4-2.9 VG/DC
Vector Genome Copies per Diploid Cell

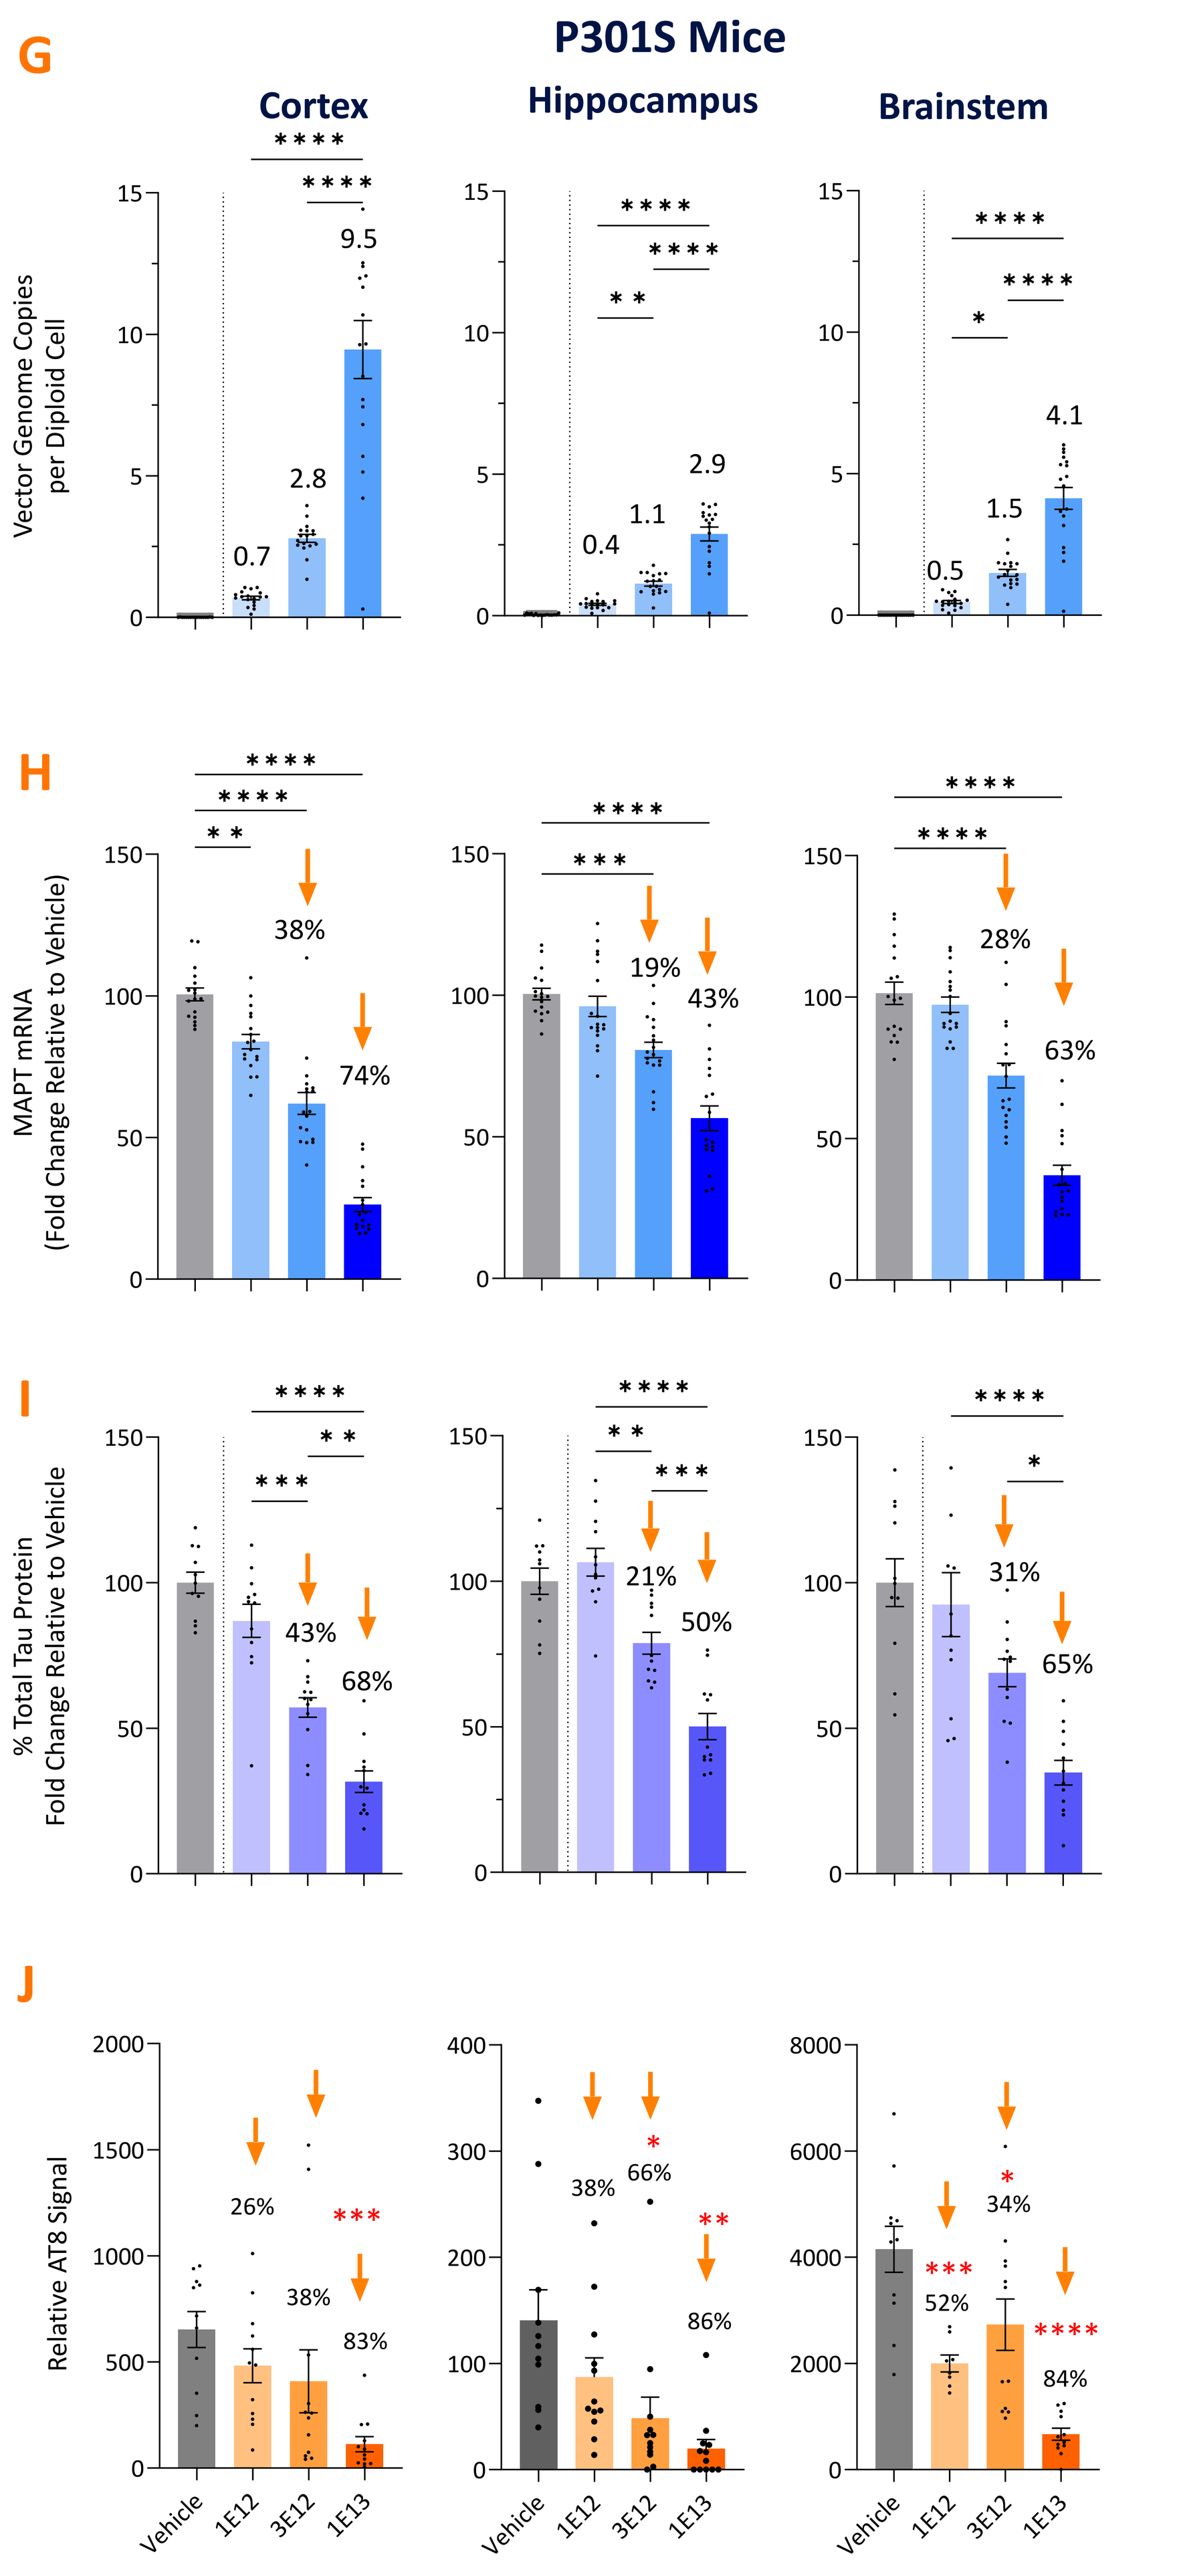
G
P301S Mice
Cortex
Hippocampus
Brainstem
Vector Genome Copies per Diploid Cell
H
MAPT mRNA (Fold Change Relative to Vehicle)
I
% Total Tau Protein Fold Change Relative to Vehicle
J
Relative AT8 Signal
Vehicle
1E12
3E12
1E13

(**A**) Study design. Twelve- to thirteen-week-old hTau or P301S mice were injected intravenously (IV) with VY1706. Eight weeks later, the cortex, hippocampus, and brainstem were harvested to quantify vector genome, Tau mRNA and protein levels. Dose dependent increase in VG, was accompanied by corresponding dose dependent reduction of tau mRNA/protein in multiple brain regions of hTau mice (**B**-**D**) or of P301S mice (**G**-**I**) were observed. Furthermore, there was a strong inverse correlation between VG levels and and Tau protein reduction with IC_{50} at 2.4-3.9 VG/DC (95% CI) in all the analyzed CNS regions of hTau mice (**E**) and IC_{50} at 1.4-2.9 VG/DC (95%CI) in all the analyzed CNS regions of P301S mice (**F**). A dose dependent, robust, and significant reduction of pathological tau protein levels was observed in cortex, hippocampus, and brainstem of P301S mice dosed with VY1706. Pathological hyperphosphorylated human tau levels were quantified using AT8 ELISA and normalized to the vehicle control. Statistical significance was evaluated with a one-way ANOVA and Tukey's multiple comparisons post-hoc test; *, **, ***, and **** indicate p < 0.05, 0.005, 0.0005 and 0.0001, respectively. Data are shown as the group mean ± SEM. N=6 per group except N=3 for (**J**). Percentage reductions are indicated above each bar.

Figure 3. VY1706 Treatment in Non-Human Primates (NHPs)-Biodistribution, Tau mRNA and Protein Knockdown

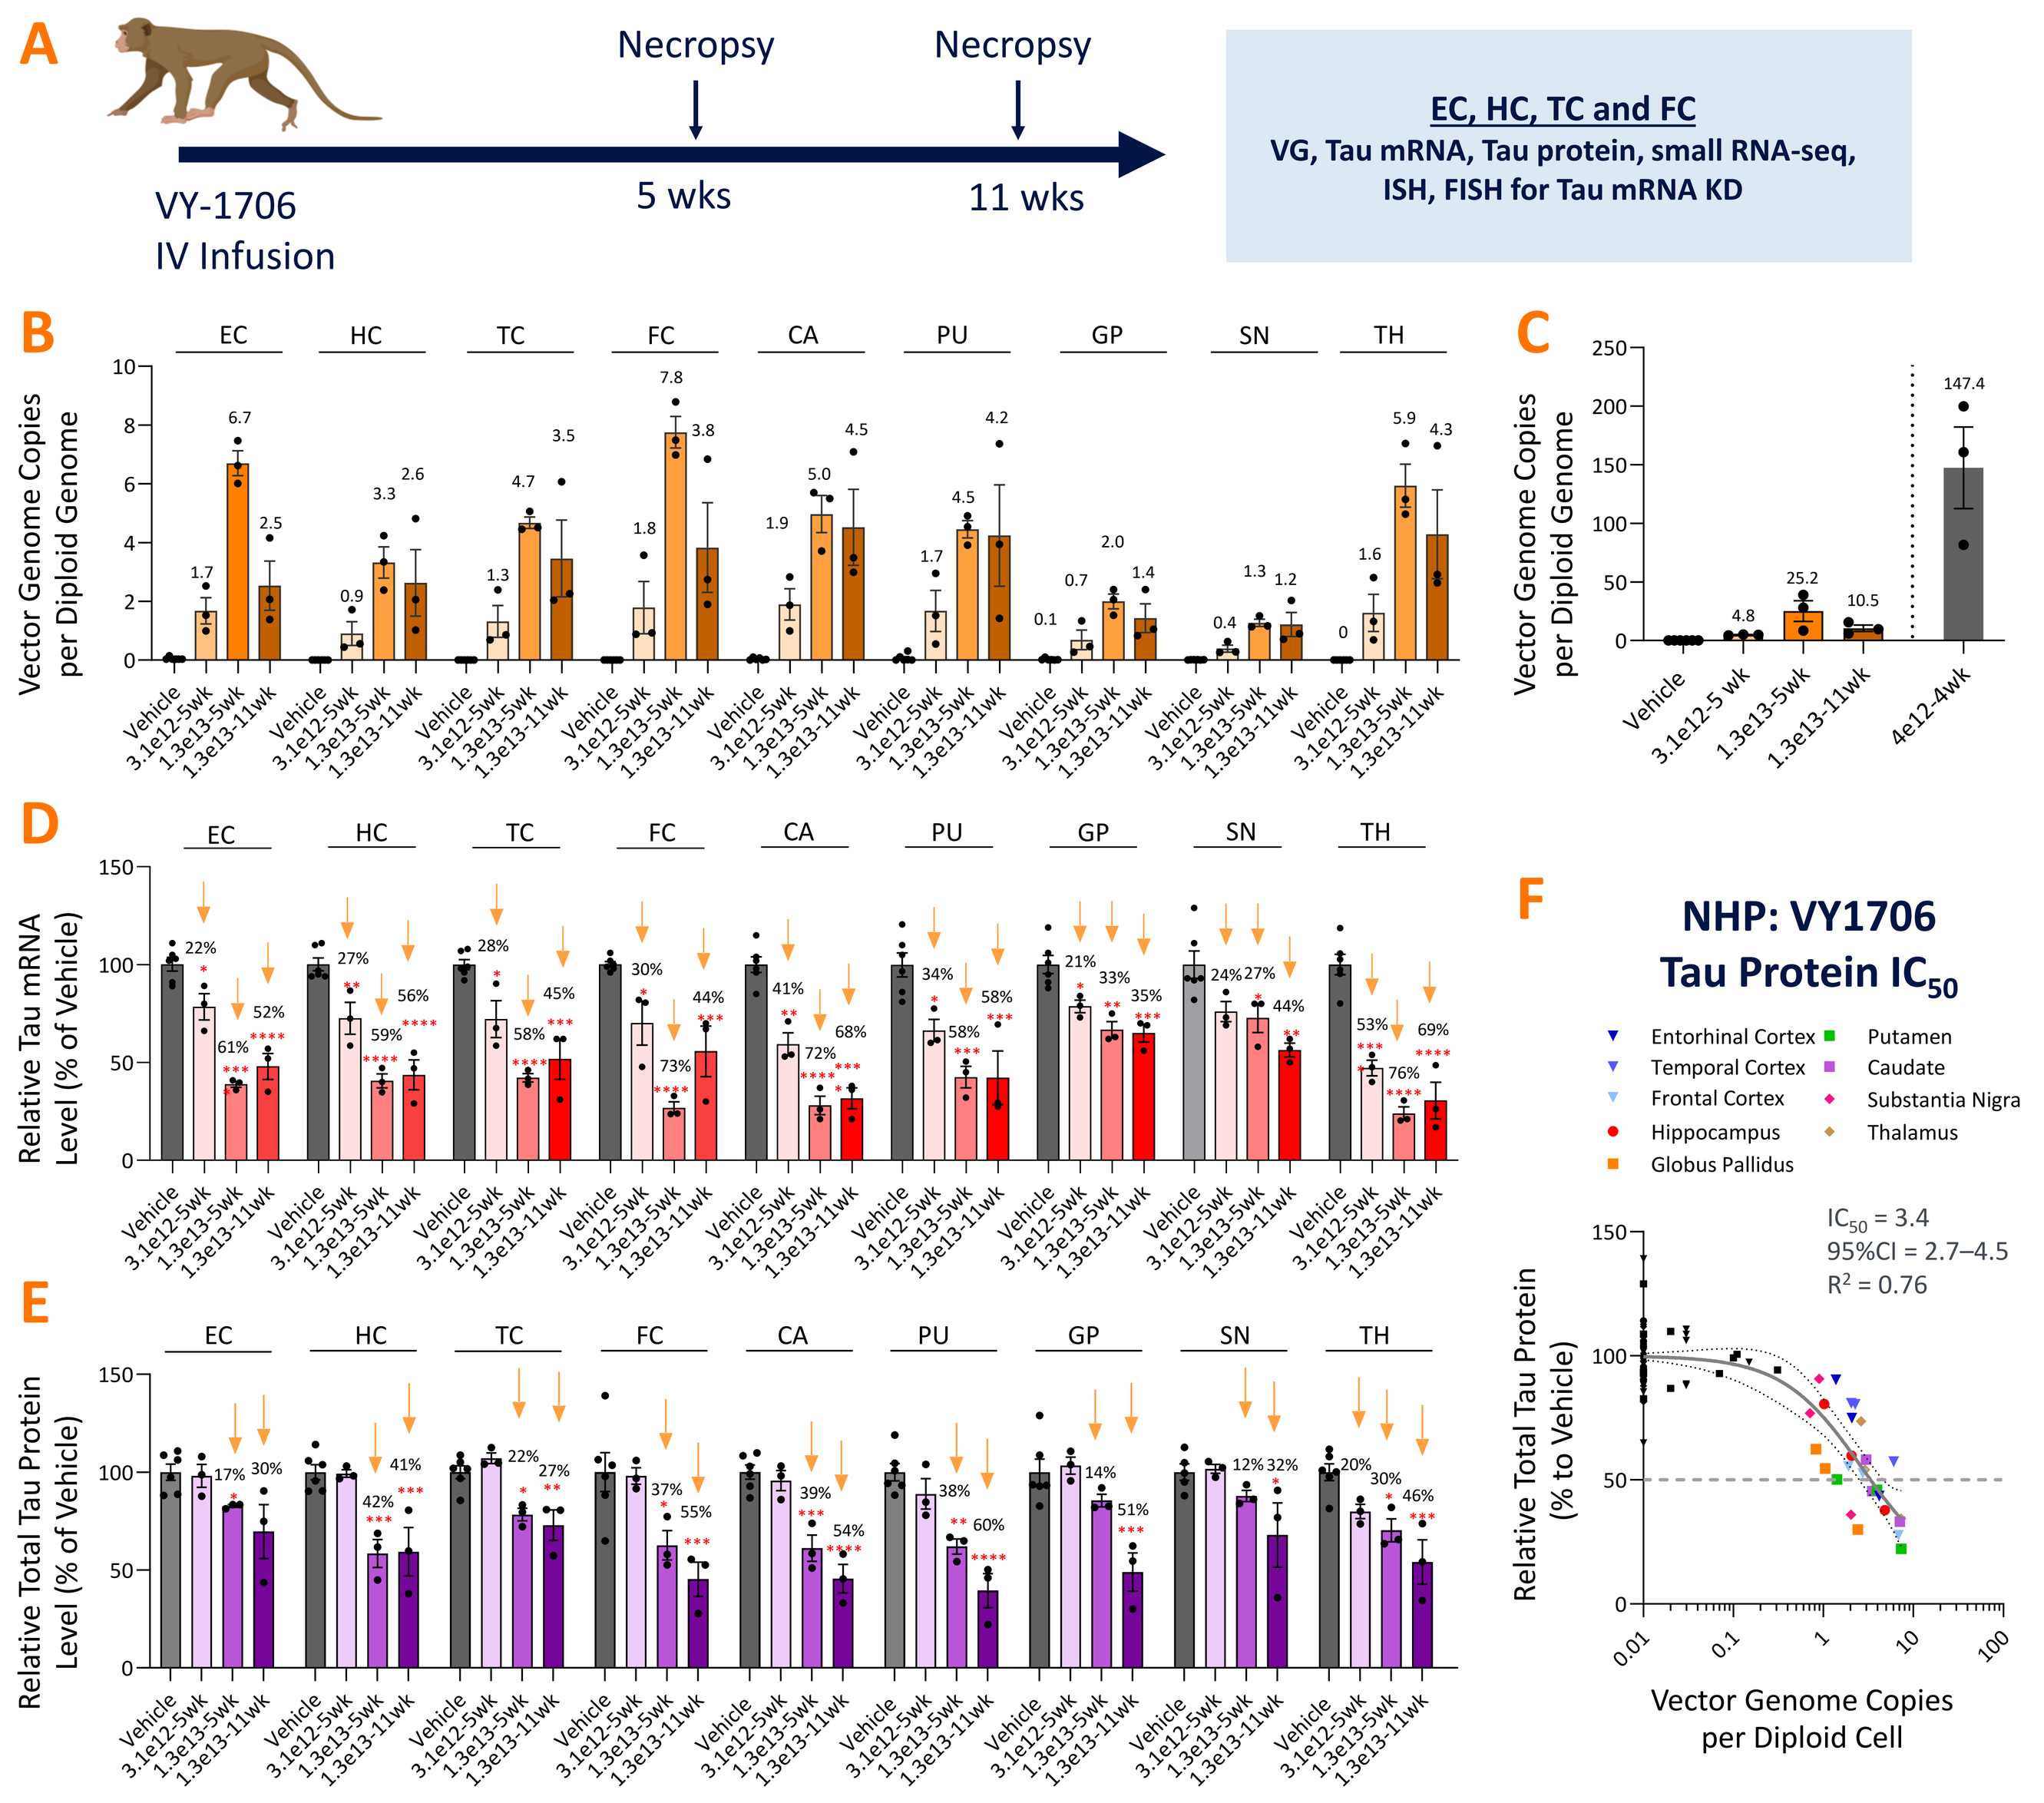
A
Necropsy
Necropsy
VY-1706
IV Infusion
5 wks
11 wks
EC, HC, TC and FC
VG, Tau mRNA, Tau protein, small RNA-seq, ISH, FISH for Tau mRNA KD
B
Vector Genome Copies per Diploid Genome
EC HC TC FC CA PU GP SN TH
C
4e12-4wk
D
Relative Tau mRNA Level (% of Vehicle)
E
Relative Total Tau Protein Level (% of Vehicle)
Vehicle
3.1e12-5wk
1.3e13-5wk
1.3e13-11wk
F
NHP: VY1706
Tau Protein IC_{50}
Entorhinal Cortex
Temporal Cortex
Frontal Cortex
Hippocampus
Globus Pallidus
Putamen
Caudate
Substantia Nigra
Thalamus
IC_{50} = 3.4
95%CI = 2.7–4.5
R^2 = 0.76
Relative Total Tau Protein (% to Vehicle)
Vector Genome Copies per Diploid Cell

Broad biodistribution and robust tau mRNA and protein reduction in key Alzheimer's and Primary Tauopathy related brain regions of NHP. (**A**) Study design. 2–3-year-old Non-human primates were injected intravenously (IV) with VY1706. Five or eleven weeks later, the hippocampus (HC), entorhinal (EC), temporal (TC), frontal cortex (FC), Caudate (CA), putamen (PU), globus pallus (GP), substantia nigra (SN), and thalamus (TH) were harvested to quantify vector genome, Tau mRNA and protein levels. Small RNA-seq was also performed on these four CNS regions, dorsal root ganglion, and liver. (**B**) A dose-dependent increase in vector genome (VG) levels was observed in all 9 CNS regions of 5-week-, and 11-week- NHP administered with VY1706. The mean VG levels for each dose group are indicated. (**C**) VY1706 exhibited liver detargeting compared to historical WT-AAV9 data expressing an alternative transgene at a 1E13 vg/kg dose. (**D**) A dose-dependent decrease in Tau mRNA was evident in each of the nine CNS regions of the VY1706-treated NHPs that were 5-week or 11-week in-life, indicating a sustained pharmacologic activity up to 11 weeks. Tau mRNA levels were normalized to cyno XPNPEP-1 mRNA levels and then further normalized to the vehicle control. (**E**) A dose-dependent decrease in Tau protein was evident in each of the analyzed CNS regions of the VY1706-treated NHP that had 5-week or 11-week in-life. Tau protein levels were normalized to total protein levels and then further normalized to the vehicle control. (**F**) There was a strong inverse correlation between Tau protein and VG levels in all the analyzed CNS regions with IC_{50} at 3.4 VG/DG.

Figure 4. Robust Tau mRNA Reduction in Neurons of AD- and Primary Tauopathy-Related Brain Regions of NHP Dosed with High Dose of VY1706

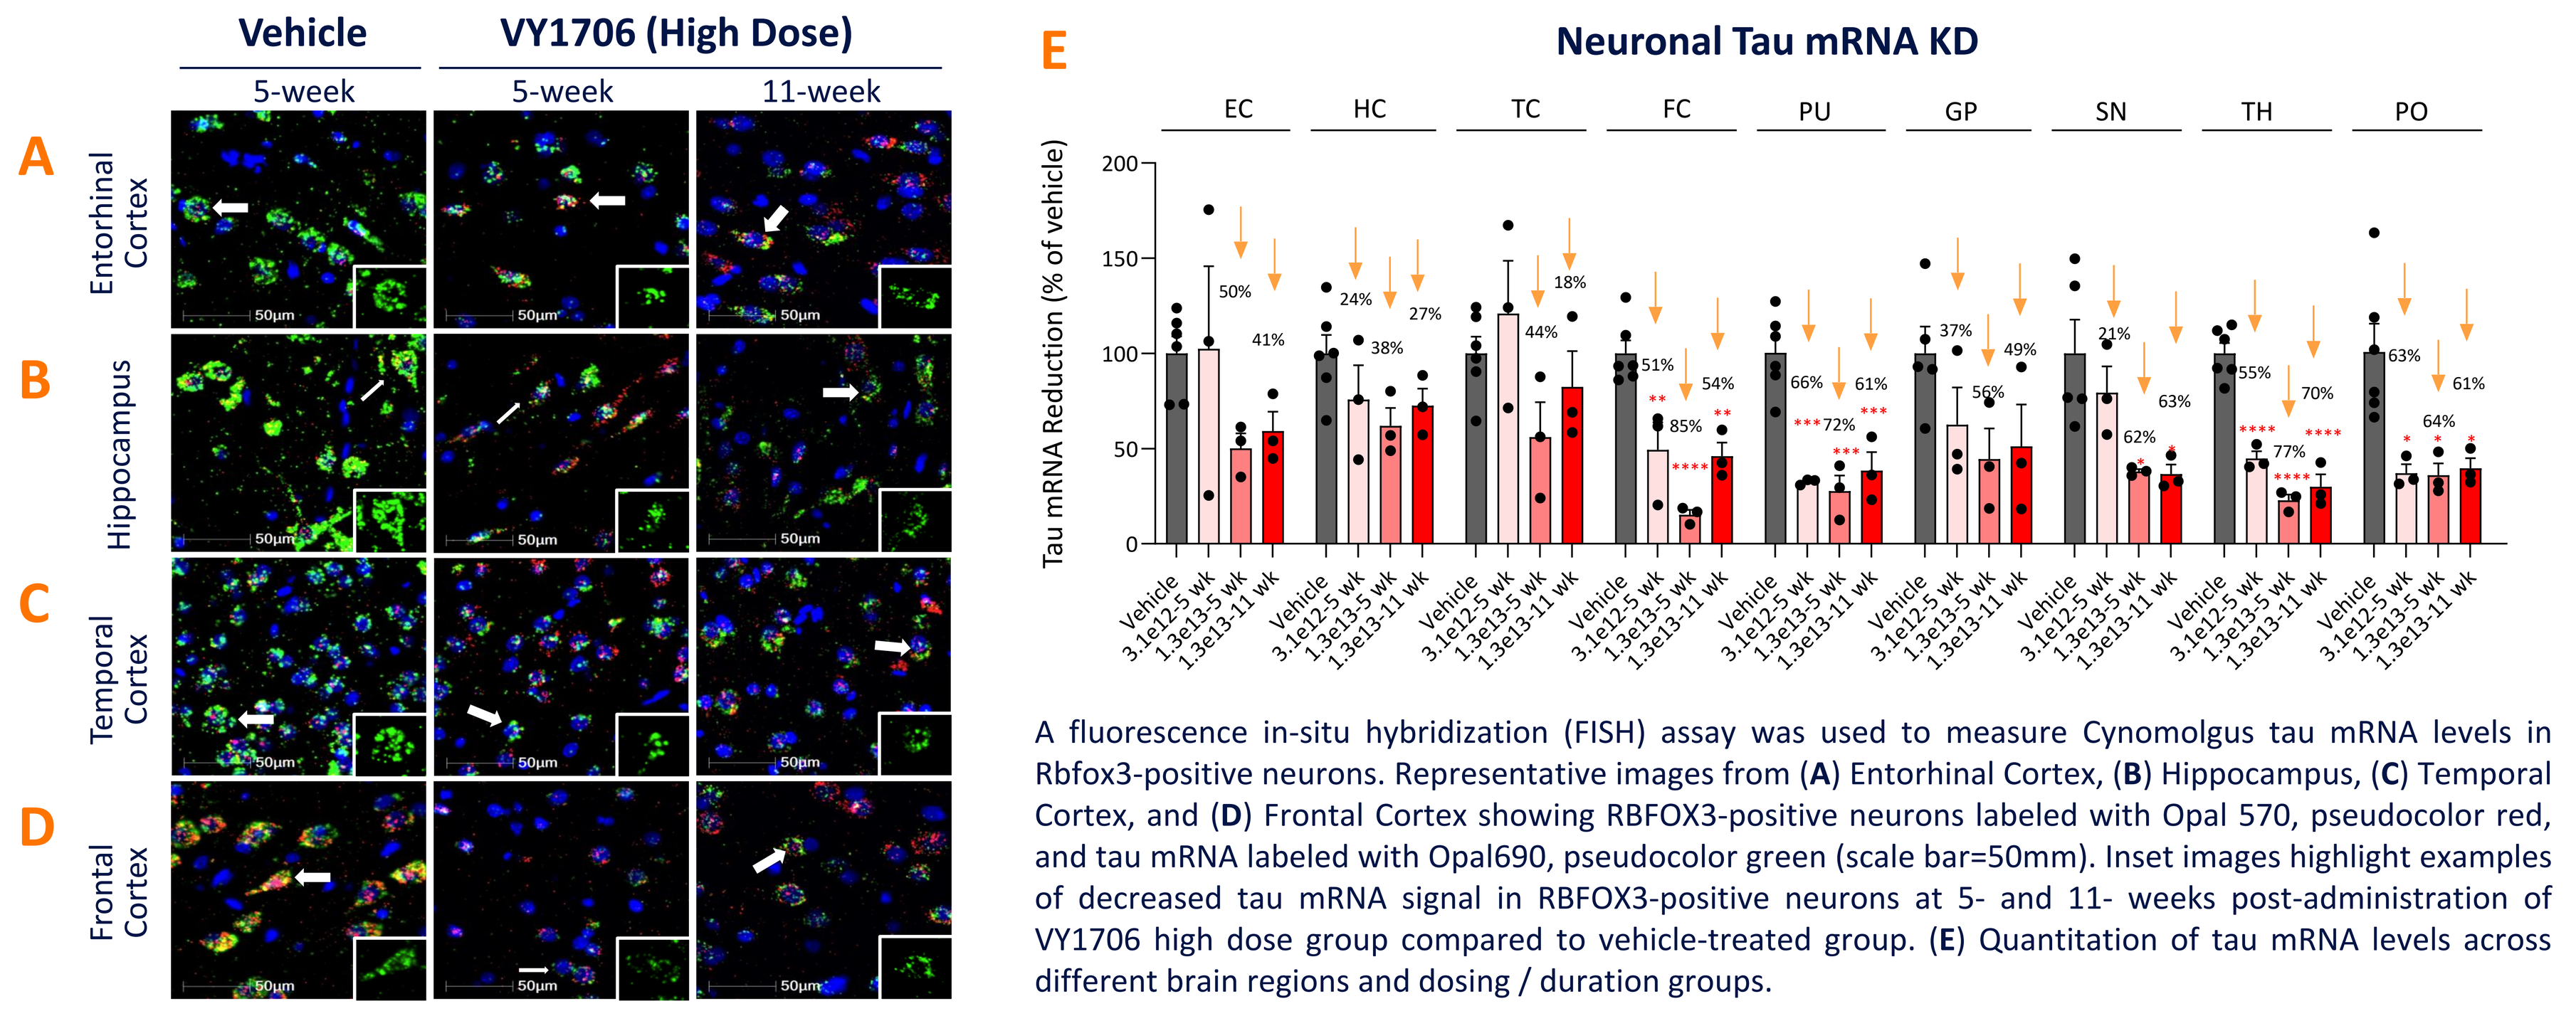
Vehicle
VY1706 (High Dose)
5-week
5-week
11-week
A
Entorhinal Cortex
B
Hippocampus
C
Temporal Cortex
D
Frontal Cortex
E
Neuronal Tau mRNA KD
Tau mRNA Reduction (% of vehicle)
EC HC TC FC PU GP SN TH PO

A fluorescence in-situ hybridization (FISH) assay was used to measure Cynomolgus tau mRNA levels in Rbfox3-positive neurons. Representative images from (**A**) Entorhinal Cortex, (**B**) Hippocampus, (**C**) Temporal Cortex, and (**D**) Frontal Cortex showing RBFOX3-positive neurons labeled with Opal 570, pseudocolor red, and tau mRNA labeled with Opal690, pseudocolor green (scale bar=50mm). Inset images highlight examples of decreased tau mRNA signal in RBFOX3-positive neurons at 5- and 11- weeks post-administration of VY1706 high dose group compared to vehicle-treated group. (**E**) Quantitation of tau mRNA levels across different brain regions and dosing / duration groups.

Figure 5. Robust Tau Protein Reduction in Neurons of AD and Primary Tauopathy Related Brain Regions of NHP Administered with High Dose of VY1706

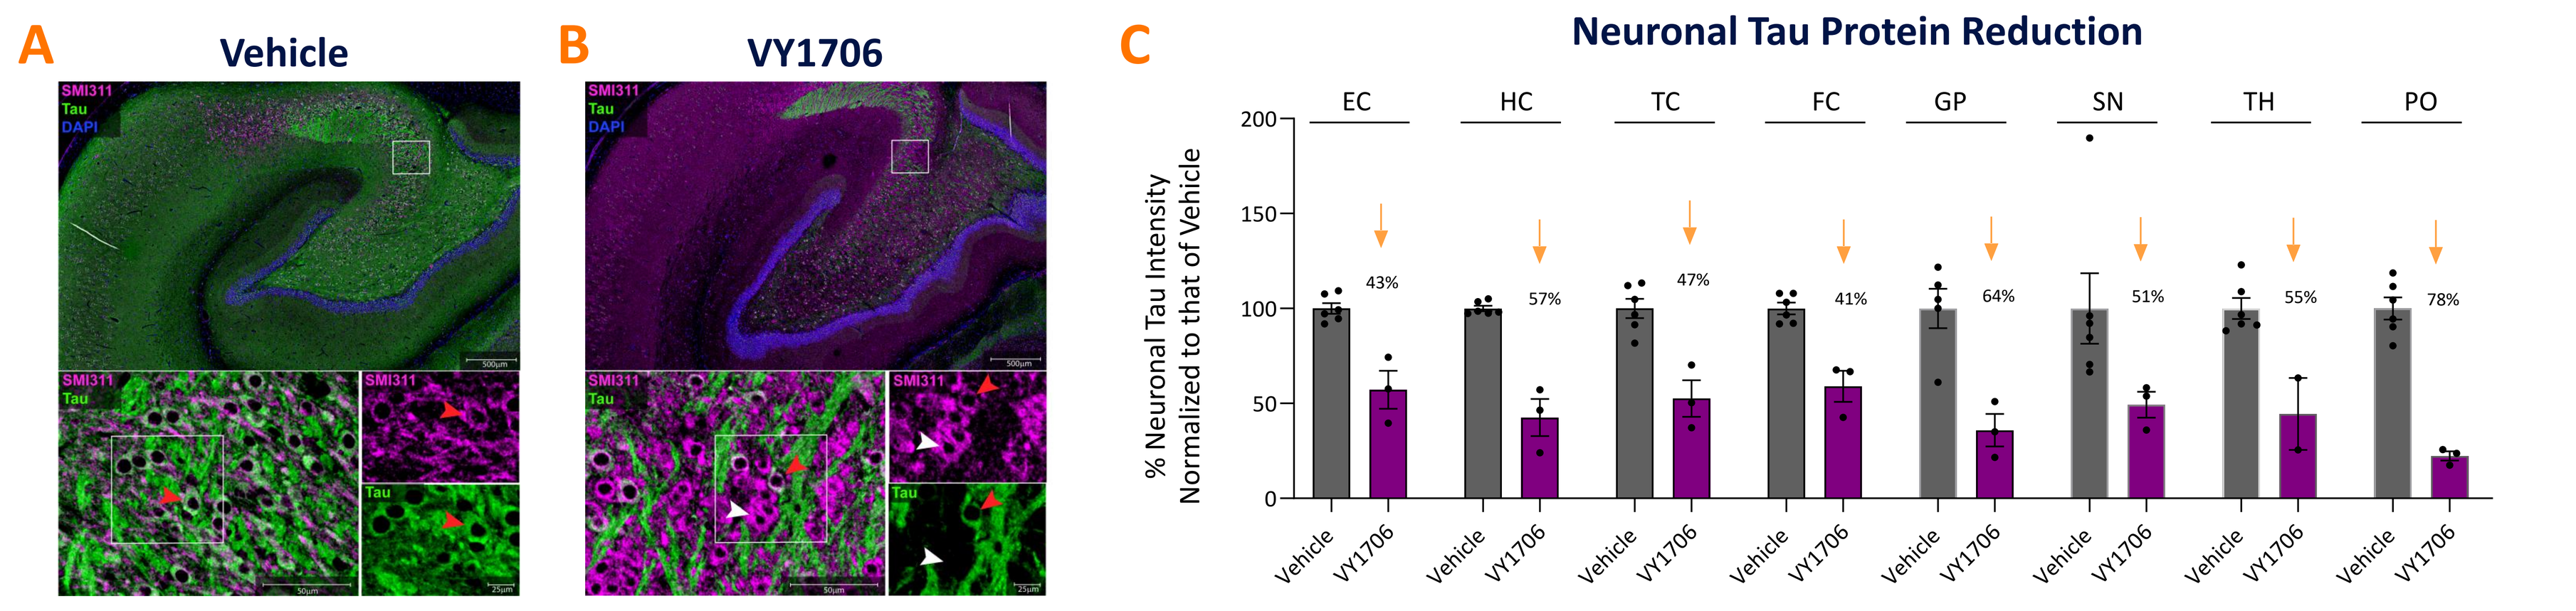
A
Vehicle
B
VY1706
C
Neuronal Tau Protein Reduction
% Neuronal Tau Intensity Normalized to that of Vehicle
EC HC TC FC GP SN TH PO

Reduction of Tau protein expression (Green) in SMI311+ve neurons (Magenta) was evident in Vehicle (**A**) versus VY1706 treated-NHP (high dose) (**B**) throughout the hippocampus in top panel and an example of zoomed in CA3 region of hippocampus (white box) was shown in the bottom panel. The treatment was indicated on the top. White arrows pointing towards an example of normal Tau protein (green) expression in SMI311 (magenta) positive neurons as shown in inset image. Red arrows pointing towards an example of loss of Tau (green) expression in the SMI311. The top insert of each image represents SMI311 stained neurons (magenta), while the bottom insert of each image represents Tau protein expression (green) (**C**) Relative tau intensity in the SMI311 positive neuronal soma shows a range of 41-78% reduction of tau immunoreactivity in evaluated brain regions : entorhinal cortex (EC), hippocampus (HC), temporal cortex (TC), frontal cortex (FC), globus pallidus (GP), substantia nigra (SN), thalamus (TH) and pons (PO).

Table 1. Small RNA-seq Processing Profiles

VY-1706						
	% relative to endogenous miRNA (safe profile: less than 5%)		Guide/Passenger ratio (safe profile: >10)		5' end processing precision (% N) (safe profile: >85%)	
Dose	3.1E+12	1.3E+13	3.1E+12	1.3E+13	3.1E+12	1.3E+13
Entorhinal CTX	0.2	1.1	50.5	33.1	96.5	94.3
Hippocampus	0.1	0.9	73.2	47.6	96.4	94.8
Frontal CTX	0.2	1.4	49.5	40.6	92.8	90
Temporal CTX	0.2	1.1	50.6	42.8	92.9	91
DRG	0	0.1	14	17.9	93.8	90
Liver	0.1	0.5	70.8	65	92.8	90

Favorable small RNA-seq profiles were observed in the entorhinal cortex, hippocampus, temporal cortex and frontal cortex. DRG, and liver of NHP dosed with VY1706.

Figure 6. RNA Processing Fidelity and miR Transcriptome Evaluation

No Differentially Expressed Endogenous miRNA were Observed (TX vs Vehicle)

TX	Dose (vg/kg)	Up	Down
VY1706	3.14E12	0	0
VY1706	1.3E13	0	0

No Differentially Expressed miRNA Frontal Cortex, 1.3E13 vg/kg vs. Veh

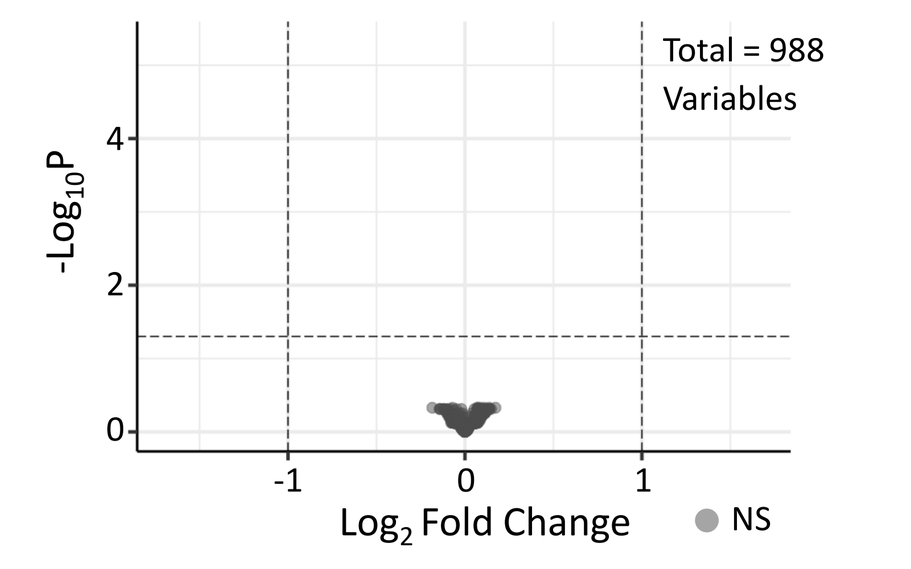
Total = 988 Variables
$-Log_{10}P$
Log_2 Fold Change
NS

There were no significant changes in the endogenous miRNA transcriptome. Representative data from the Frontal cortex is shown although identical trends were observed in the entorhinal cortex, hippocampus and temporal cortex, The volcano plot shows the –log10 BH-p adjusted value on the y axis and the log2 Fold change on the y axis. The y-value indicates statistical significance, or the False Discovery Rate. The x-value indicates the difference between the dosing groups. The dotted line indicates the p = 0.05, and +/- 2-fold changes on the y and x axis, respectively. No changes achieve significance using DESeq2 analysis.

Figure 7. NfL Levels in Serum

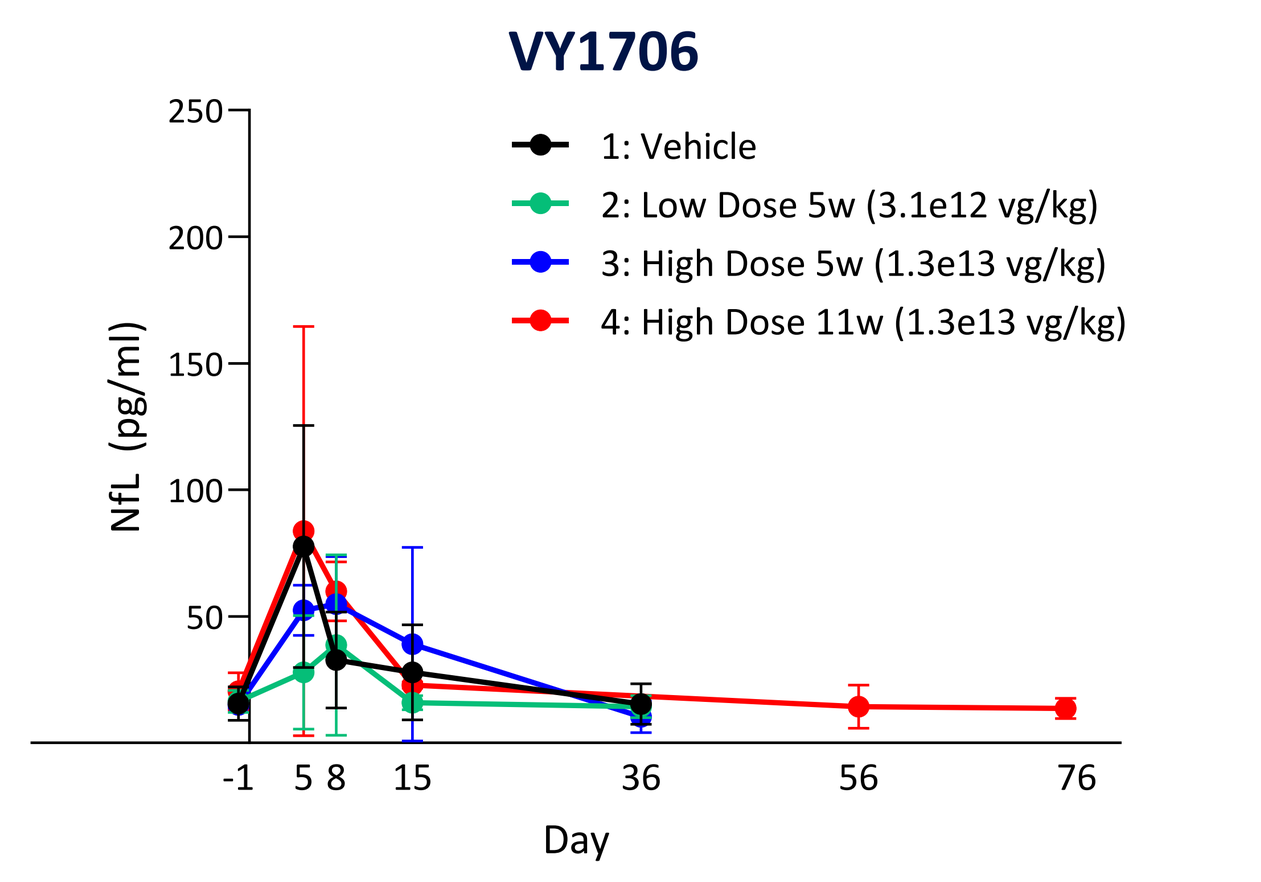
VY1706
1: Vehicle
2: Low Dose 5w (3.1e12 vg/kg)
3: High Dose 5w (1.3e13 vg/kg)
4: High Dose 11w (1.3e13 vg/kg)
NfL (pg/ml)
Day

NfL levels in serum from VY1706 treated- or vehicle groups show early, transient elevation that resolves to baseline prior to necropsy. The transient NfL increase observed at day 5 in the vehicle group is consistent with published observations in NHPs with other AAV constructs, suggesting that this early fluctuation is not treatment related.

CONCLUSIONS

- Potent vectorized siRNA achieves up to 73% MAPT mRNA reduction at 5 weeks and 55% Tau protein reduction at 11 weeks in AD- and Primary Tauopathy-relevant brain regions at a low 1.3e13 vg/kg dose.
- Up to 82% MAPT mRNA reduction in neurons in key, AD and Primary Tauopathy-relevant brain regions.
- Dose dependent reduction of Tau mRNA, Tau protein and Pathological Tau in mouse model(s) of Tauopathy.
- High fidelity miRNA processing with minimal impact on endogenous microRNAs as evidenced via small RNA seq.
- Well-tolerated - no TA-related clinical, macroscopic or microscopic adverse findings and no TA-related elevations in Serum NfL.
- GLP Tox study ongoing, IND-filing in 2026.

voyager therapeutics 75 Hayden Avenue, Lexington, MA 02421 | www.voyagertherapeutics.com

AD/PD | March 17-21, 2026 | Copenhagen, Denmark