Pharmacology and Safety Assessment of VY1706, a BBB-Crossing Tau-Lowering AAV Gene Therapy for Alzheimer's Disease, in a GLP Toxicology Study in NHPs

Vik Arora[1], Joydeep Ghosh[1], Yasuhiro Ito[1], Charlotte Chung[1], Wencheng Liu, Jeffrey Thompson[1], Hechen Bao[1], Cansu Colpan[1], Tyler Moyer[1], Brett Hoffman[1], Maneesha A. Paranjpe[1], Joydip Kundu[1], Su Jing Chan[1], Krishanu Mathur[1], Gonzalo Milet[1], Kevin Nguyen[1], Russ Udani[1], Patrick Carroll[1], Marina Feschenko[1], Sara Movassaghian[1], Robert Vass[1], Hima Ramachandrareddy[1], Camille Stengel[1], Kumar Dhanasekharan[1], Shamik Sharma[1], Tom Elich[1], Andrew Schrock[1], Bridget Shaughnessy[1], , Weitong Chen[1], Johnny Yao[1], **Mathieu Nonnenmacher[1]**, Toby Ferguson[1], Elena Ratti[1], Dinah W.Y. Sah[1], Todd Carter[1], Rajeev Sivasankaran[1], Raj Rajagovindan[1]

Voyager Therapeutics, Lexington, MA, USA

voyager therapeutics

INTRODUCTION

- Tau pathology is strongly linked to cognitive decline in Alzheimer's disease (AD), making tau reduction a compelling therapeutic strategy. Although antisense oligonucleotides have shown promise, the need for repeated intrathecal administration and heterogeneous CNS distribution represents potential challenges. A blood–brain barrier (BBB)-crossing adeno-associated virus (AAV) gene therapy could enable more uniform CNS distribution and durable tau lowering following a one-time intravenous (IV) administration.
- To address this challenge, we engineered VY1706, which combines a proprietary BBB-crossing AAV capsid with a potent vectorized microRNA to lower MAPT mRNA in the CNS. Importantly, the VY1706 capsid engages alkaline phosphatase (ALPL) to enable efficient BBB transport in rodents and primates.
- Conserved ALPL sequence and tissue expression across species, together with similar capsid engagement of mouse, cynomolgus macaque, and human ALPL, support the translatability of VY1706 to humans. In a mouse model of tauopathy, VY1706 produced robust, dose-dependent reductions in MAPT mRNA, tau protein, and pathological tau across key brain regions.
- In nonhuman primates (NHPs), a single IV administration of VY1706 resulted in significant reductions in MAPT mRNA and tau protein in AD-relevant brain regions at 11 weeks post-dose.
- In an IND-enabling GLP toxicology study in NHPs, VY1706 was evaluated across multiple dose levels, with comprehensive assessment of biodistribution (vector genome and amiRNA levels), pharmacology (MAPT mRNA and tau protein lowering), and safety, including safety pharmacology endpoints.
- These results demonstrate favorable tolerability together with durable, dose-dependent tau lowering in AD-relevant brain regions. Collectively, these findings position VY1706 as a one-time gene therapy capable of achieving widespread CNS delivery and durable tau reduction, supporting its potential as a treatment strategy for Alzheimer's disease.

Figure 1. VY1706, a Tau-lowering AAV Gene Therapy is Transported Across the BBB via the Highly Cross-species Conserved ALPL Receptor

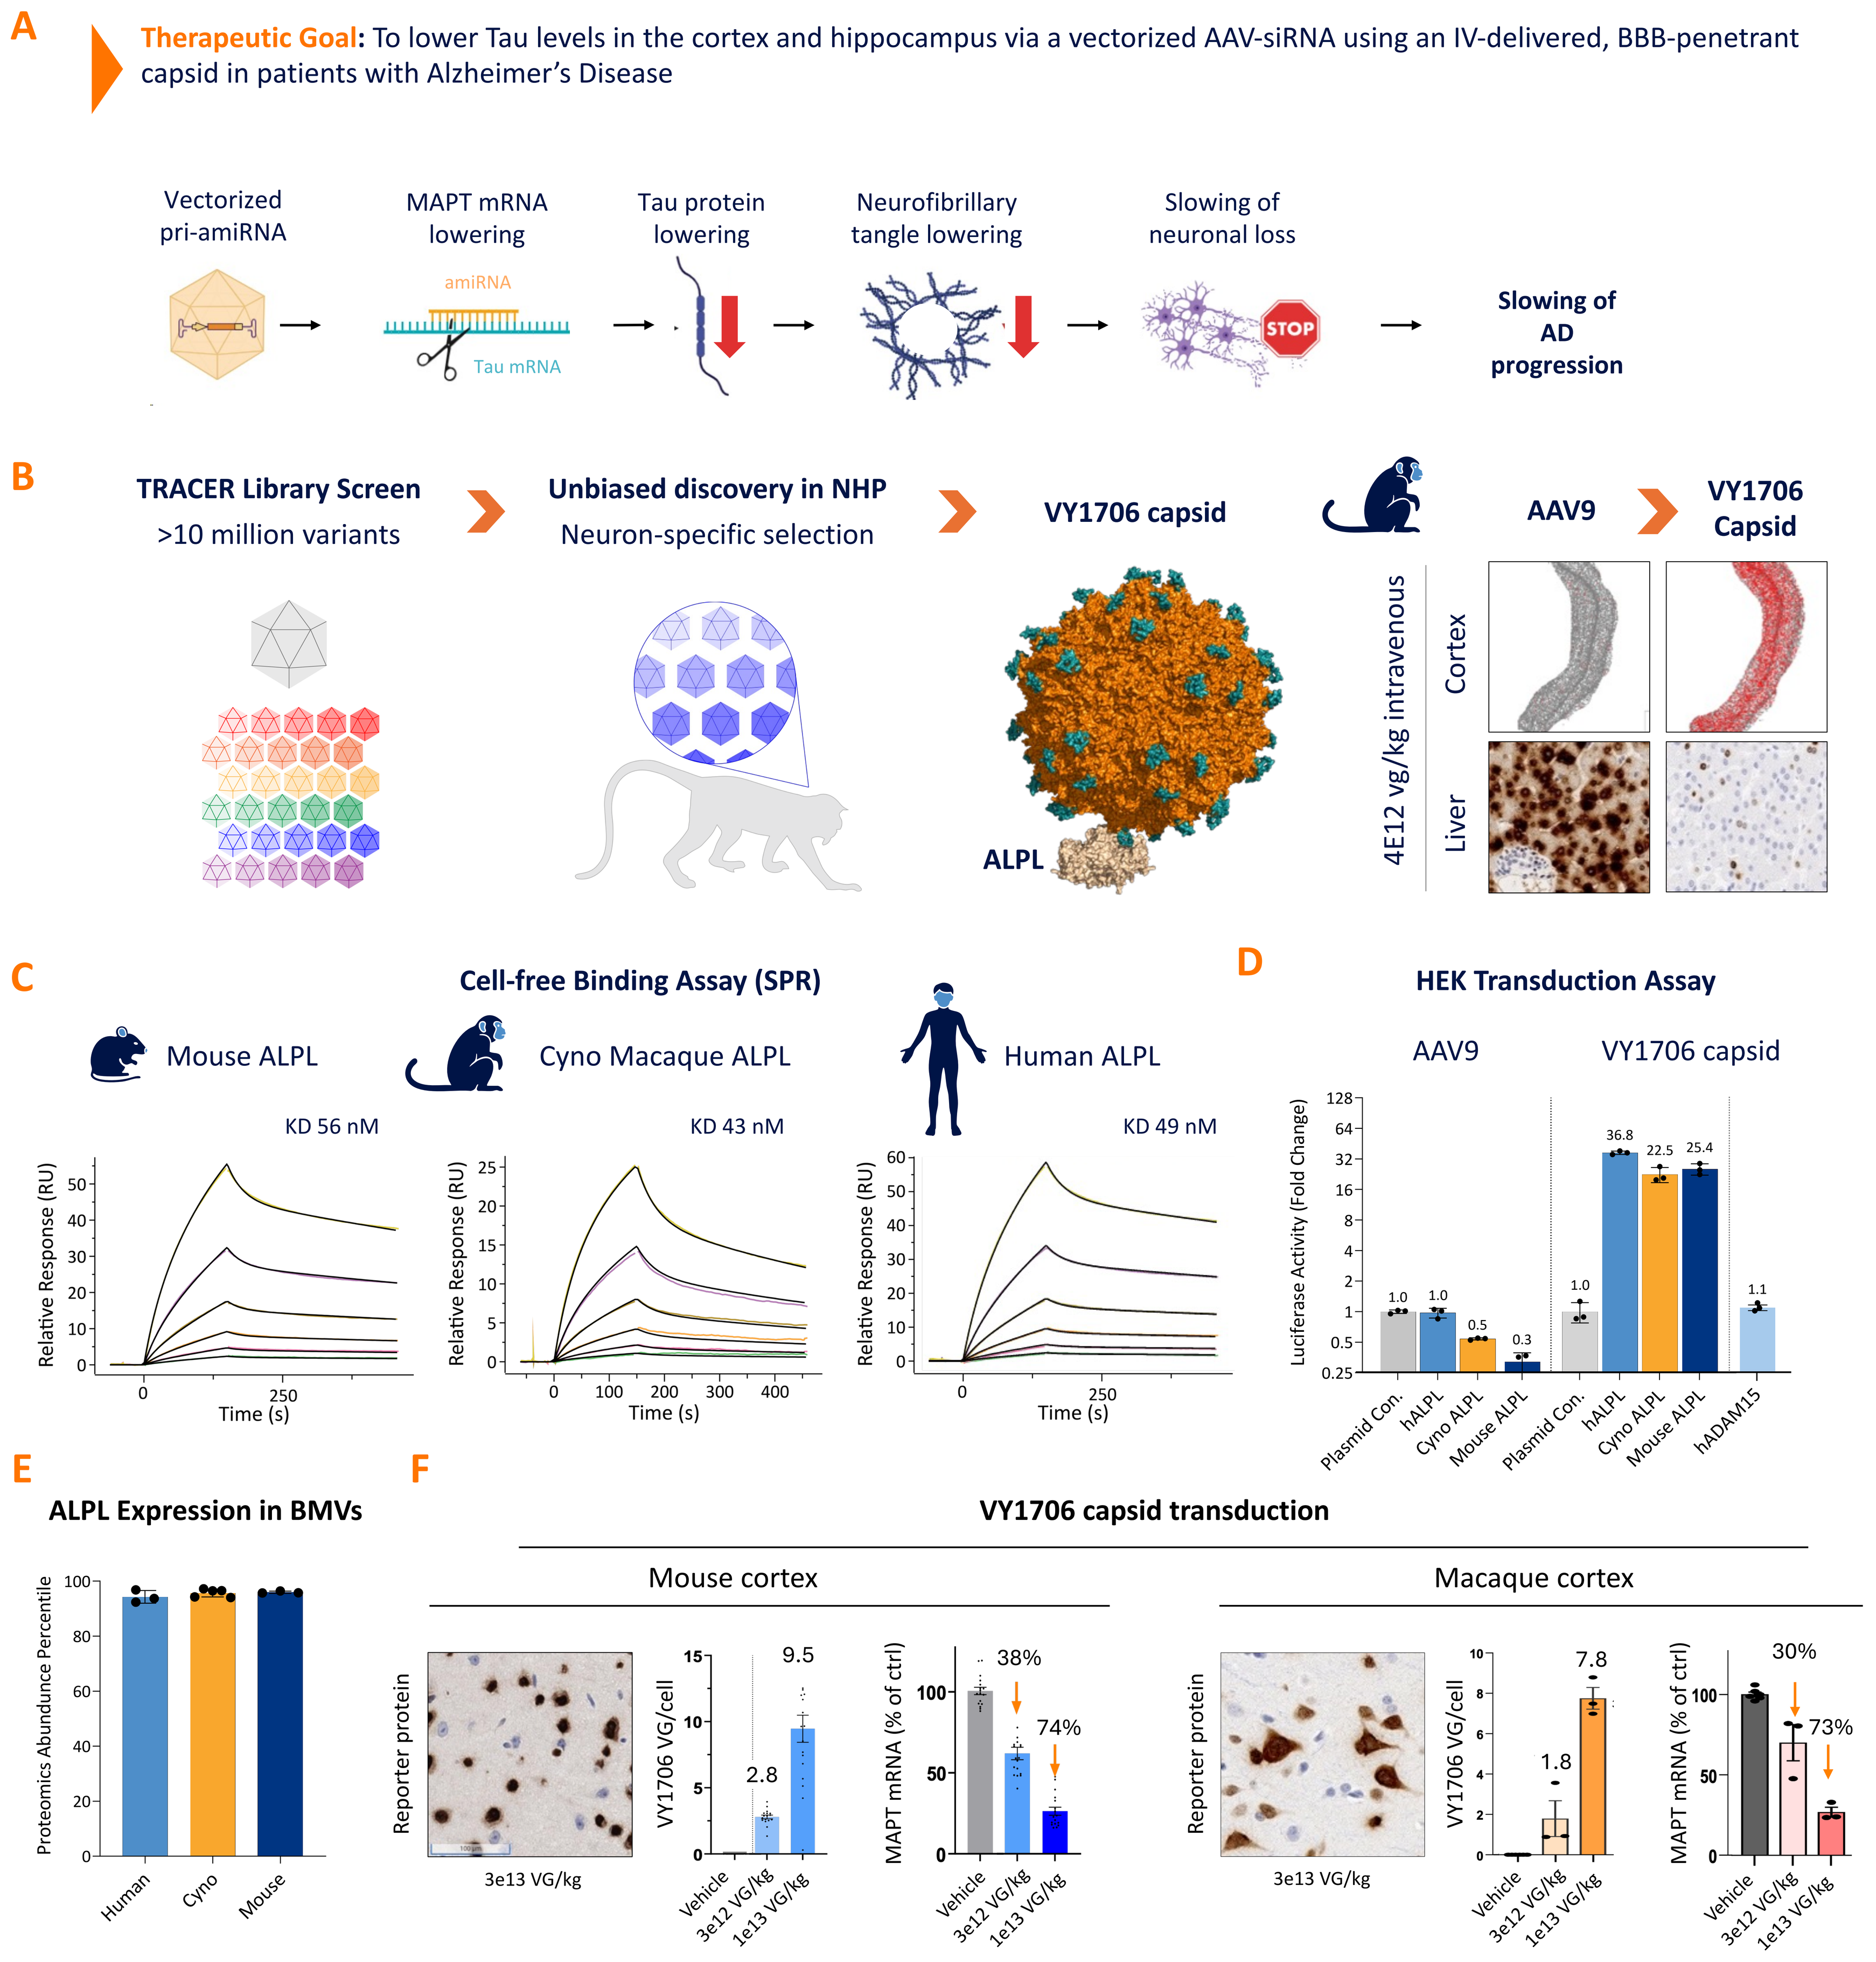
A
Therapeutic Goal: To lower Tau levels in the cortex and hippocampus via a vectorized AAV-siRNA using an IV-delivered, BBB-penetrant capsid in patients with Alzheimer's Disease
Vectorized pri-amiRNA
MAPT mRNA lowering
amiRNA
Tau mRNA
Tau protein lowering
Neurofibrillary tangle lowering
Slowing of neuronal loss
STOP
Slowing of AD progression
B
TRACER Library Screen
>10 million variants
Unbiased discovery in NHP
Neuron-specific selection
VY1706 capsid
ALPL
AAV9
VY1706 Capsid
4E12 vg/kg intravenous
Cortex
Liver
C
Cell-free Binding Assay (SPR)
Mouse ALPL
Cyno Macaque ALPL
Human ALPL
KD 56 nM
KD 43 nM
KD 49 nM
Relative Response (RU)
Time (s)
D
HEK Transduction Assay
AAV9
VY1706 capsid
Luciferase Activity (Fold Change)
Plasmid Con.
hALPL
Cyno ALPL
Mouse ALPL
hADAM15
E
ALPL Expression in BMVs
Proteomics Abundunce Percentile
Human
Cyno
Mouse
F
VY1706 capsid transduction
Mouse cortex
Macaque cortex
Reporter protein
3e13 VG/kg
VY1706 VG/cell
MAPT mRNA (% of ctrl)
Vehicle
3e12 VG/kg
1e13 VG/kg

VY1706-capsid mechanism and cross-species conservation. (**A**) Therapeutic goal and mechanism of action. (**B**) Evolution and characterization of VY1706 capsid, that interacts with the highly conserved brain vascular receptor ALPL to enable BBB-crossing. (**C, D**) Physical and functional interaction of VY1706 capsid with ALPL isoforms from various species measured by SPR (**C**) and transduction of HEK293T cells transfected with indicated control, ALPL or ADAM15 plasmids, measured by luciferase activity (**D**). (**E**) Comparative expression of ALPL protein in human, macaque and mouse brain microvessels (BMVs), from proteomics analysis. Values indicate the percentile distribution of ALPL reads among the entire proteome. ALPL scored ~95th percentile of protein reads in all three species, indicating high expression at the BBB. Percentiles are plotted as mean ± SEM (N≥3). (**F**) Comparative dose-response of VY1706 capsid in mouse and macaques cortex, measured with 1) reporter protein expression at 3e13VG/kg (IHC, left panel), 2) viral genome biodistribution of VY1706 in the cortex at 3e12 and 1e13 VG/kg (middle panels) and 3) MAPT mRNA relative to vehicle control at 3e12 and 1e13 VG/kg (righ panels).

Figure 2. VY1706 Shows Robust Tau Protein Knockdown in Neurons Across AD- and Primary Tauopathy-Relevant Brain Regions and is Liver Sparing in NHP Dose-range Finding Study

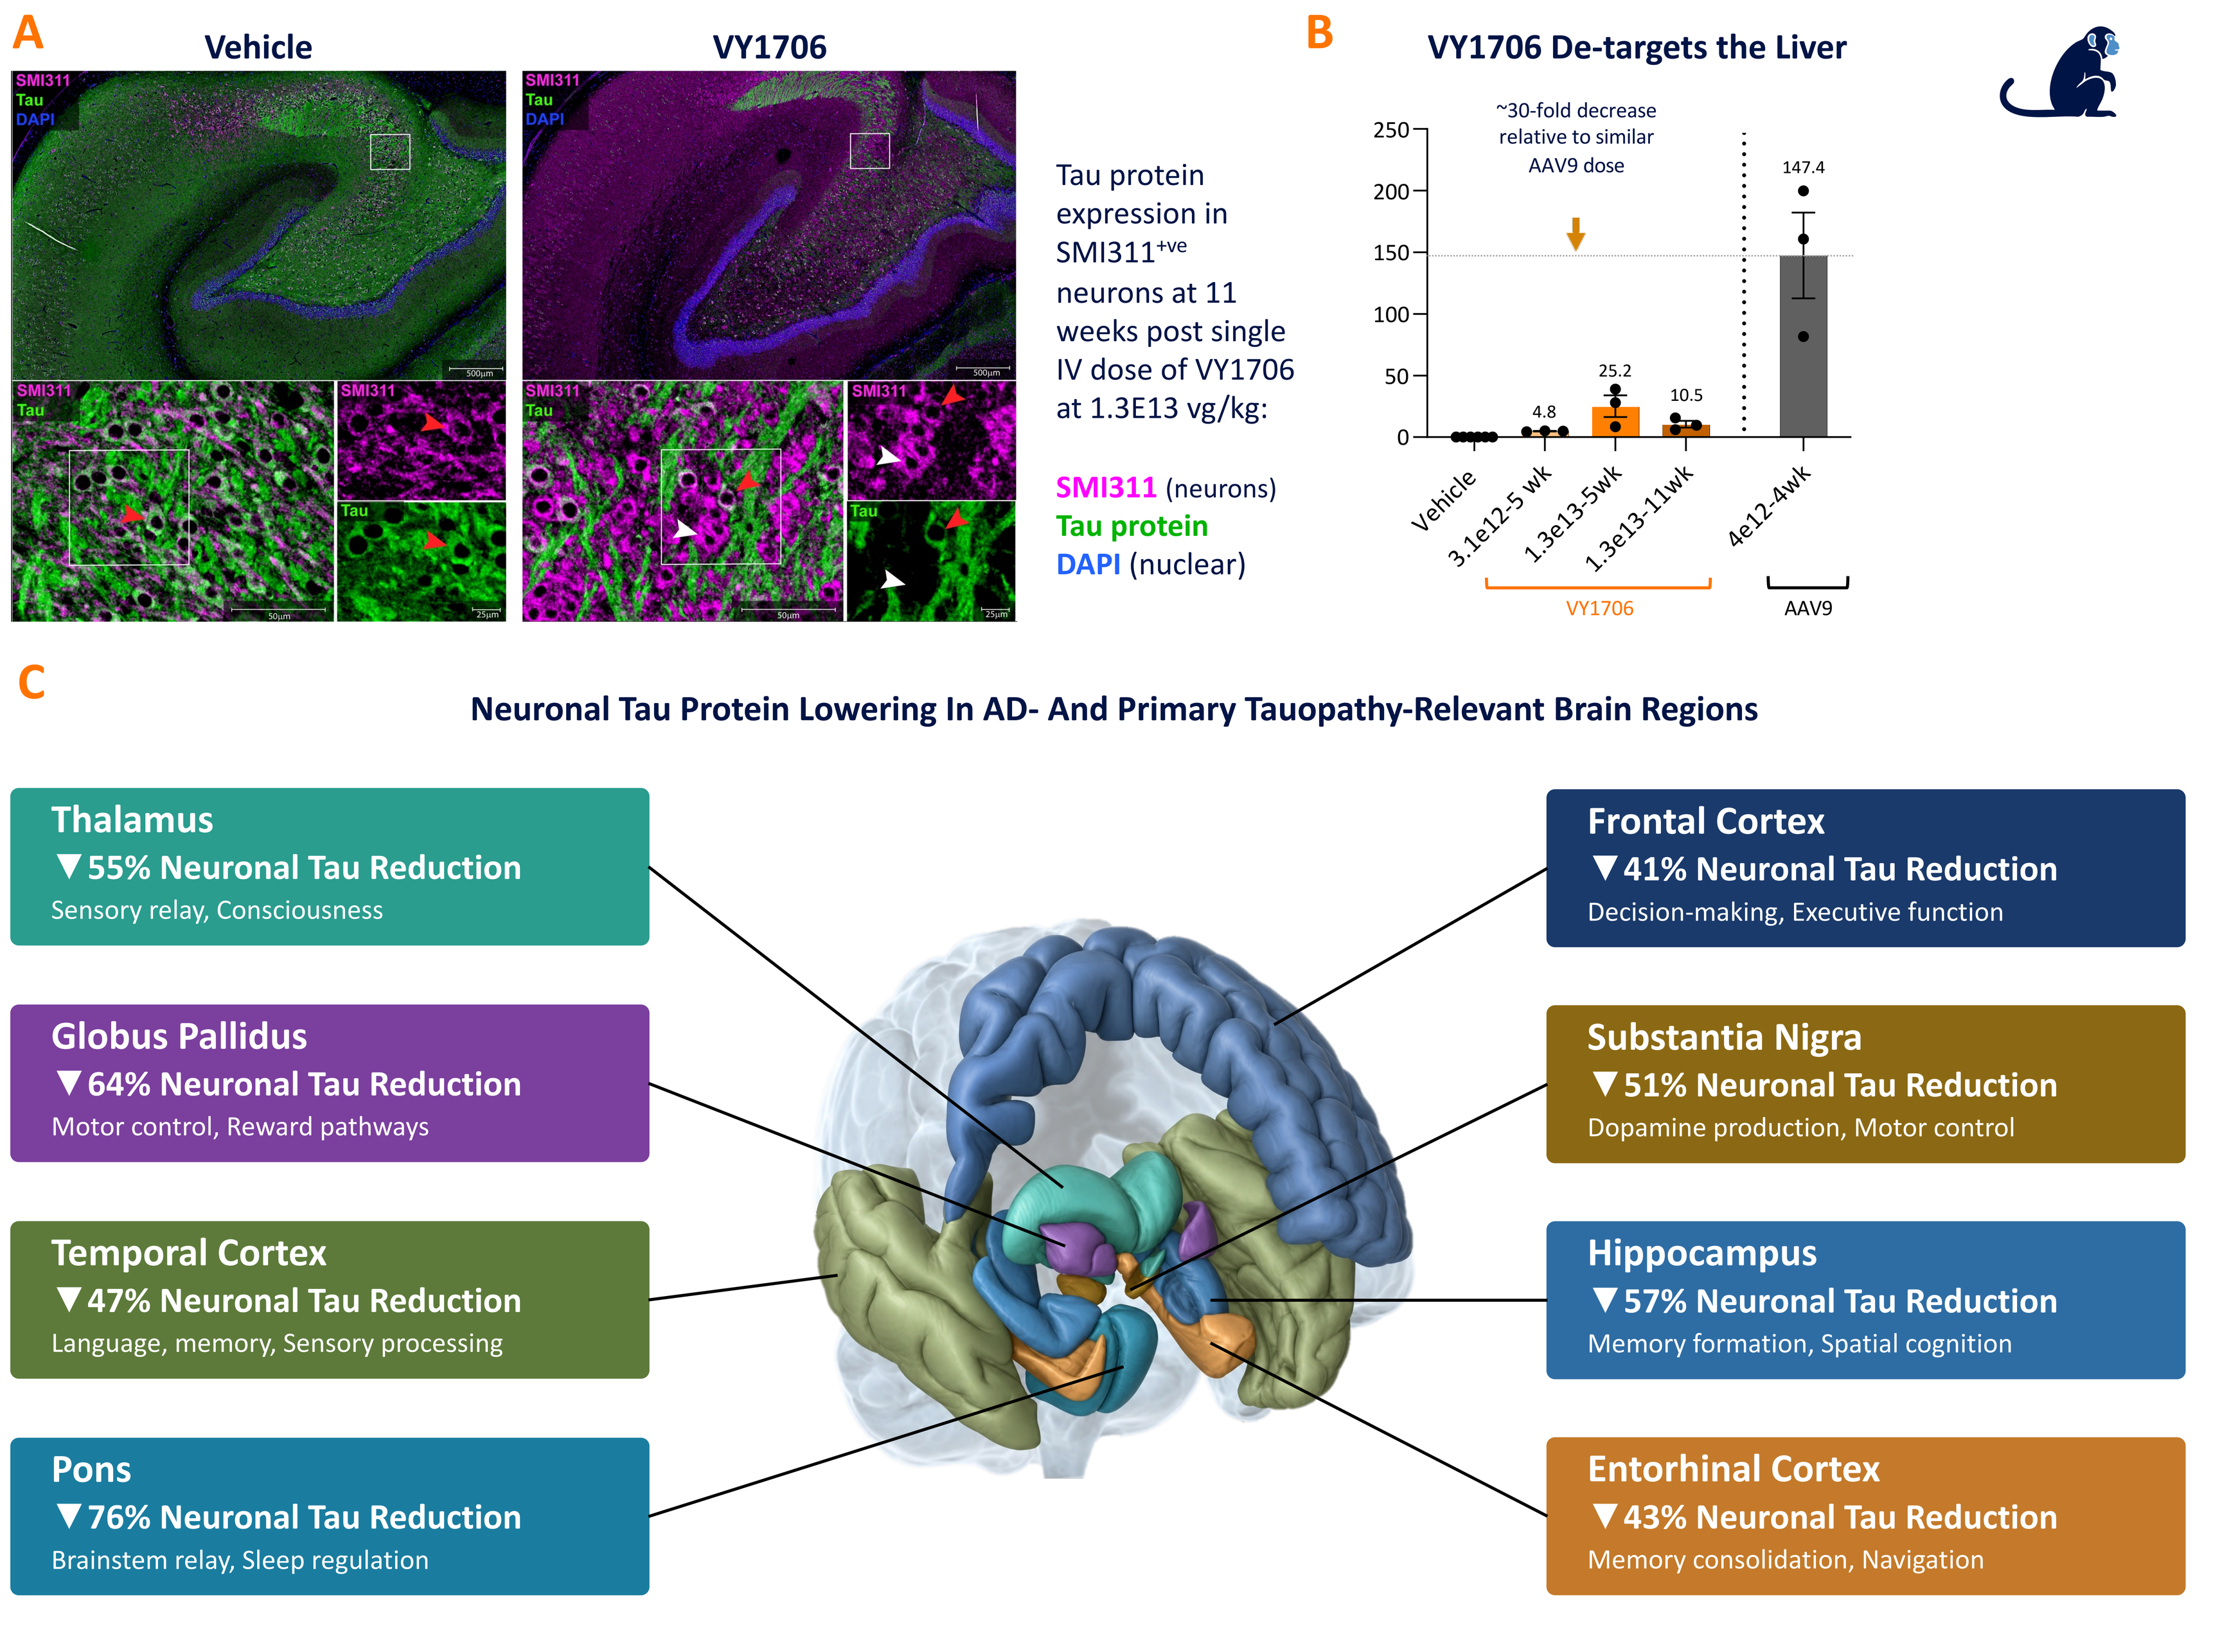
A
Vehicle
VY1706
SMI311
Tau
DAPI
Tau protein expression in SMI311+ve neurons at 11 weeks post single IV dose of VY1706 at 1.3E13 vg/kg:
SMI311 (neurons)
Tau protein
DAPI (nuclear)
B
VY1706 De-targets the Liver
~30-fold decrease relative to similar AAV9 dose
147.4
25.2
4.8
10.5
Vehicle
3.1e12-5 wk
1.3e13-5wk
1.3e13-11wk
4e12-4wk
VY1706
AAV9
C
Neuronal Tau Protein Lowering In AD- And Primary Tauopathy-Relevant Brain Regions
Thalamus
▼55% Neuronal Tau Reduction
Sensory relay, Consciousness
Globus Pallidus
▼64% Neuronal Tau Reduction
Motor control, Reward pathways
Temporal Cortex
▼47% Neuronal Tau Reduction
Language, memory, Sensory processing
Pons
▼76% Neuronal Tau Reduction
Brainstem relay, Sleep regulation
Frontal Cortex
▼41% Neuronal Tau Reduction
Decision-making, Executive function
Substantia Nigra
▼51% Neuronal Tau Reduction
Dopamine production, Motor control
Hippocampus
▼57% Neuronal Tau Reduction
Memory formation, Spatial cognition
Entorhinal Cortex
▼43% Neuronal Tau Reduction
Memory consolidation, Navigation

VY1706 treatment allows widespread Tau reduction in NHP neurons. (**A**) Representative immunofluorescence images of Tau protein expression (green) in SMI311-positive neurons (magenta) from vehicle and VY1706-treated NHPs in the hippocampus at 11 weeks after VY1706 administration (1.3e13 VG/KG). Scale bar = 500 µm. White box indicates inset of CA3 region shown below. Scale bar = 50 µm. Corresponding single channel representative images of inset show SMI311 protein (magenta, top) and Tau protein (green, bottom) immunofluorescence. Scale bar = 25 µm. (**B**) VY1706 exhibited liver de-targeting compared to historical data on WT-AAV9 expressing an alternative transgene and dosed at 4E12 vg/kg. (**C**) Summarized reduction of Tau protein immunofluorescence intensity in the SMI311-positive neuronal somata relative to the vehicle group (relative neuronal Tau intensity [% of vehicle]) in brain regions relevant to Alzheimer's disease: Data were collected from N=6 animals for vehicle group and N=3 for treatment group.

SUMMARY OF GLP TOX RESULTS

VY1706 was well-tolerated with no detectable adverse clinical pathology or histopathological findings in the CNS, DRGs, and peripheral organs up to the highest tested dose level of 5E13 vg/kg

- Well tolerated with no infusion reactions detected
- No detectable gross pathology or organ weight findings
- No effects on clinical pathology parameters were observed at the dose levels tested (including liver)
- No adverse neurological, ocular, or cardiac safety findings were observed at the dose levels tested
- No adverse DRG or neurological findings on histopathology or electrophysiology were detected; plasma NfL was generally stable with no dose-related increases
- No cellular immune activation; TAb/NAb responses were typical of AAV

Table 1. Study Design of the VY1706 GLP Toxicology Testing Conducted in Cynomolgus Macaques

	Group	Route of administration	Number			
			3 Month Cohort		6 Month Cohort	
			MALES (♂)	FEMALES (♀)	MALES (♂)	FEMALES (♀)
	Vehicle	Single Intravenous infusion	2	2	2	2
VY1706	LOW DOSE		2	2	2	2
	MID DOSE 1		2	2	2	2
	MID DOSE 2		2	2	2	2
	5E13 vg/kg		2	2	2	2

Immunosuppression: Daily oral methylprednisolone (Day -3, through 8 weeks, followed by tapering)

Study assessments: General toxicology, ophthalmic, cardiovascular and neurological safety pharmacology, histopathology, immunogenicity, biodistribution and pharmacology endpoints

Figure 3. VY1706 Shows No AAV-Typical Liver Transaminase Elevations at the Dose Level Evaluated Through 6 Months

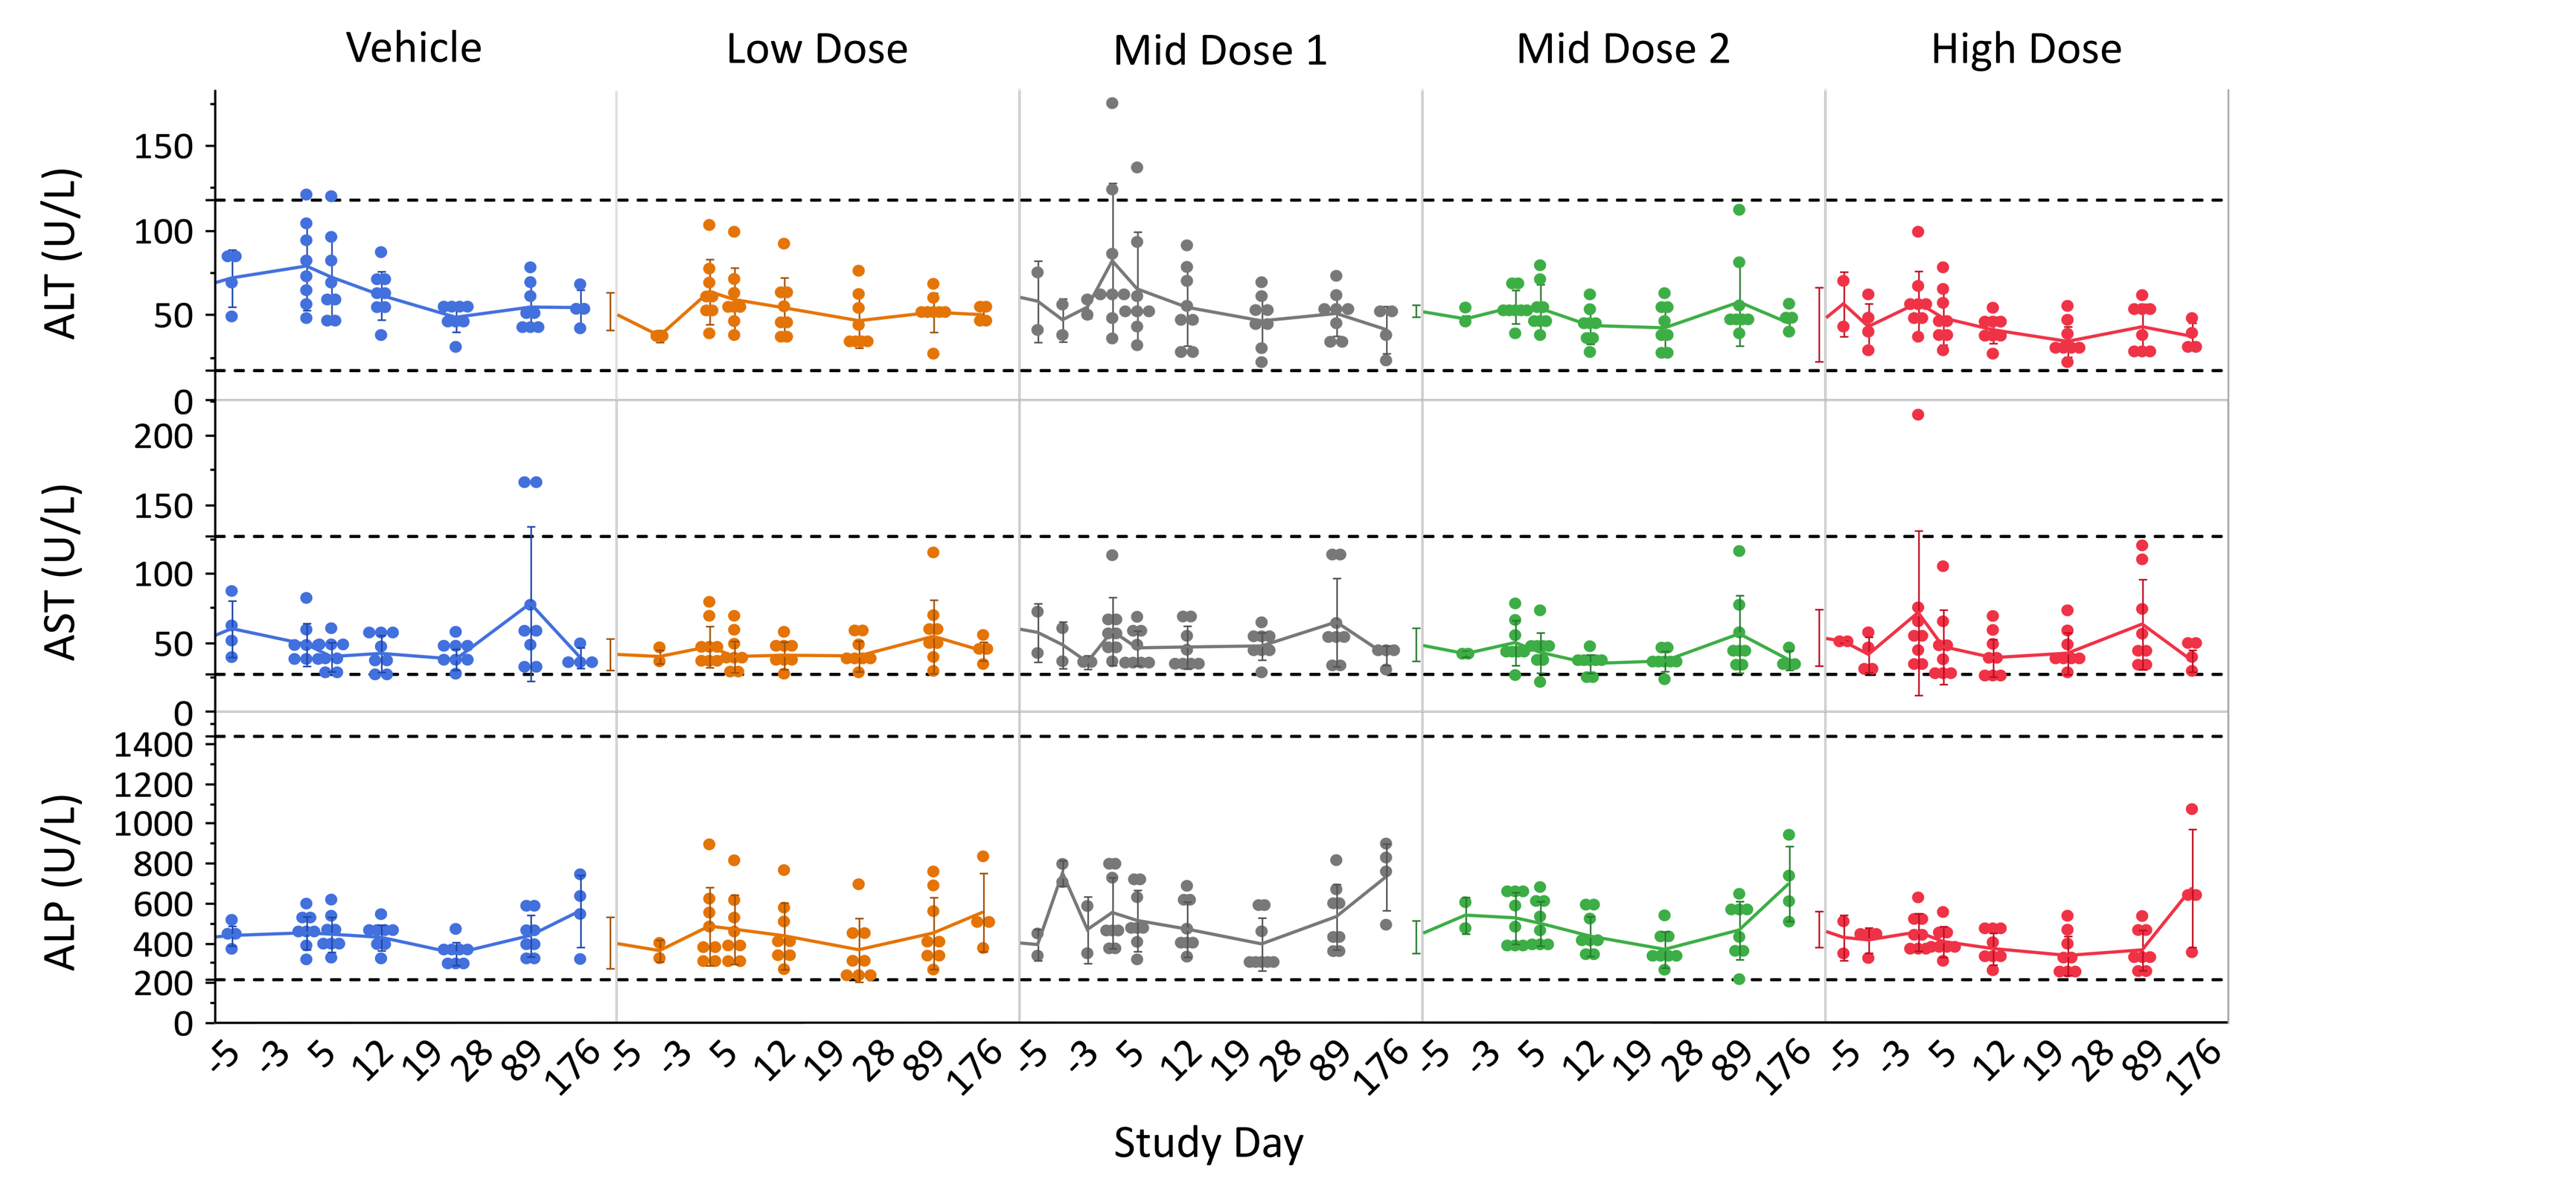
Vehicle
Low Dose
Mid Dose 1
Mid Dose 2
High Dose
ALT (U/L)
AST (U/L)
ALP (U/L)
-5 -3 5 12 19 28 89 176
Study Day

Representative liver function parameters (ALT: alanine aminotransferase, AST: aspartate aminotransferase & ALP: alkaline phosphatase) were evaluated as part of comprehensive serum clinical chemistry testing at following timepoints: Predose (multiple occasions), Post-dose Day 3, Day 5, Weeks 2, 4, 13 & 26. Broken horizontal lines in each panel bracket the typical normal range of these parameters for the test species. Data demonstrate absence of liver toxicity-associated serum level elevation of these parameters that are commonly reported with systemic AAV therapies, thereby corroborating the liver de-targeting property of capsid component of VY1706. Data are shown as mean ± SEM.

Figure 4. VY1706 Shows Dose-Dependent and Durable Increase in miRNA Level, Lowering of MAPT *mRNA* and Tau Protein in AD-relevant Brain Regions and is Liver Sparing in GLP NHP Toxicology Study

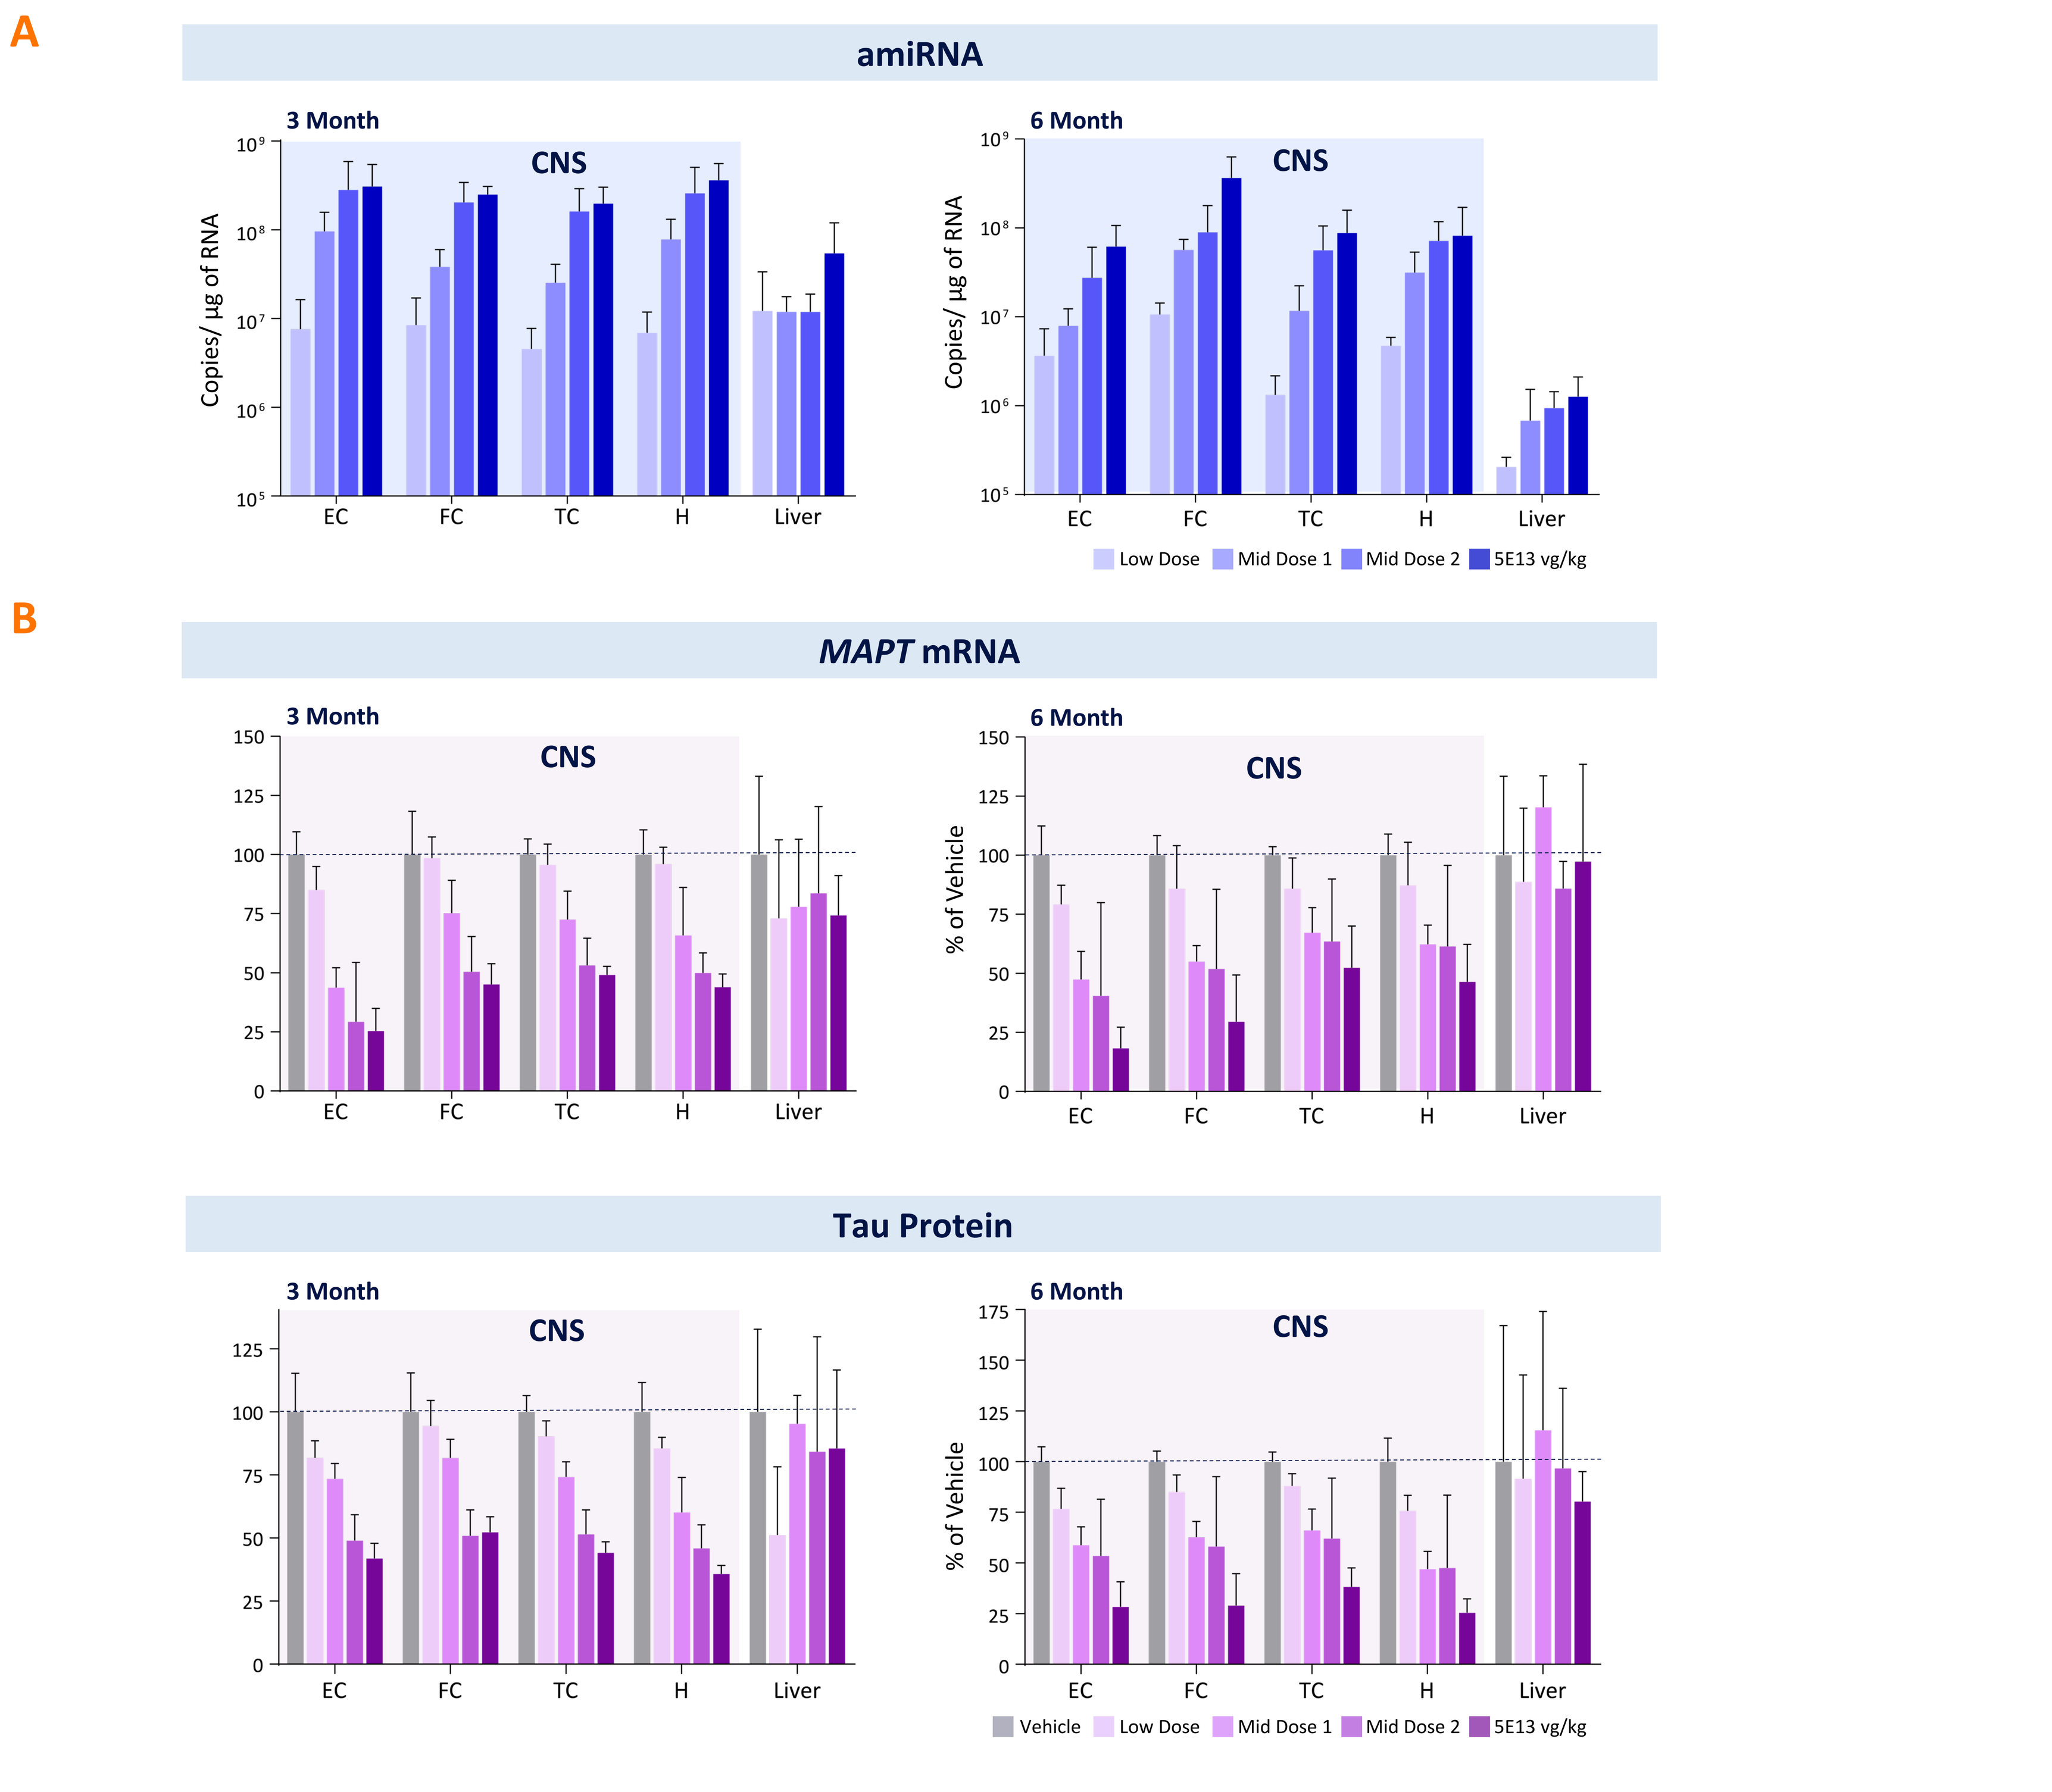
A
amiRNA
3 Month
6 Month
CNS
Copies/ µg of RNA
EC
FC
TC
H
Liver
Low Dose
Mid Dose 1
Mid Dose 2
5E13 vg/kg
B
MAPT mRNA
% of Vehicle
Tau Protein
Vehicle

(**A**) Robust expression of VY1706 amiRNA was observed in the AD-relevant brain regions. Substantially lower level of VY1706 amiRNA expression was observed in liver compared to the brain regions. (**B**) VY1706 reduced MAPT mRNA expression across AD-relevant brain regions in a dose-dependent manner. No substantial or dose-dependent MAPT mRNA expression change was observed in the liver. (**C**) VY1706 lowered Tau protein levels in AD-relevant brain regions in a dose-dependent manner. n = 3-4/group; Plots show mean + SD. EC, entorhinal cortex; FC, frontal cortex; TC, temporal cortex; H, hippocampus.

CONCLUSIONS

- A single IV dose of VY1706 in NHPs achieved broad, durable, and dose-dependent CNS delivery with up to 75% lowering of MAPT mRNA and Tau protein in key Alzheimer's disease relevant brain regions through 6-months
- ALPL, a well conserved brain vasculature endothelial receptor, mediates VY1706 blood-brain barrier transport, supporting cross-species translatability
- VY1706 was well-tolerated in the 3 and 6-month GLP NHP toxicology study, with no detectable adverse clinical pathology or histopathological findings in the CNS, DRGs, and peripheral organs (including liver) up to the highest dose tested (5E13 vg/kg)
- Nonclinical package supports advancement to first-in-human evaluation in patients with Alzheimer's disease
- VY1706 IND is active and dosing in participants with early Alzheimer's disease is expected to begin in the fourth quarter of 2026, pending patient enrollment.